Discovery and preclinical development of novel CD74-targeting antibody-drug conjugates (ADCs) with significant activity in multiple myeloma (MM) cell lines and xenograft models

Cristina Abrahams[1], Xiaofan Li[1], Abigail Yu[1], Stellanie Krimm[1], Jason Kahana[2], Rama Krishna Narla[2], Eric Schwartz[2], John Boylan[2], Heidi Hoffmann[1], Alexander Steiner[1], James Zawada[1], Heather Stephenson[1], Maureen Bruhns[1], Venita De Almeida[1], Shannon Matheny[1], Stuart Bussell[1], Adam Galan[1], Toni Kline[1], Nicki Vasquez[1], Alice Yam[1], Ryan Stafford[1], Henry Heinsohn[1], Aaron Sato[1], Arturo Molina[1], Trevor Hallam[1] and Mark Lupher[1]

[1]Sutro Biopharma, South San Francisco, CA [2] Celgene Corporation, San Diego, CA

BACKGROUND

Rationale for targeting CD74 (HLA-DR-associated invariant chain)

CD74 is a type II transmembrane glycoprotein that is rapidly internalized and recycled back to the membrane.

CD74 Functions

- Formation and transport of MHC class II
- B-cell maturation via a nuclear factor-kappa B (NF-KB) mediated pathway
- Involved in signal transduction of macrophage migration inhibitory factor (MIF)
- Mediation of disease progression in a variety of malignancies

CD74 Expression

- Normal tissues: HLA class II positive cells, including B-cells, monocytes, macrophages, Langerhans cells, dendritic cells, subsets of activated T cells and thymic epithelium.
- Overexpression in malignancy: Immunohistochemistry (IHC) using the anti-CD74 antibody LL1 demonstrated 19 / 22 multiple myeloma specimens stained positively for CD74, with 16 of these specimens demonstrating strong CD74 expression in > 95% of the myeloma cells[1]

Discovery of CD74-targeting ADCs

ADCs are emerging as a promising class of cancer biopharmaceuticals that combine the specificity of monoclonal antibodies with the anti-tumor activity of cytotoxic agents.

We have developed a novel anti-CD74 human IgG1 antibody, SP7219, and conjugated this at specific amino acids to non-cleavable maytansinoid linker-warheads (TCRS296 or SC236) with a drug-antibody ratio (DAR) of 2, to generate two potent ADCs, SP7676 and STRO-001.

Development of the CD74-targeting lead antibody and novel, specific and homogeneous ADCs.

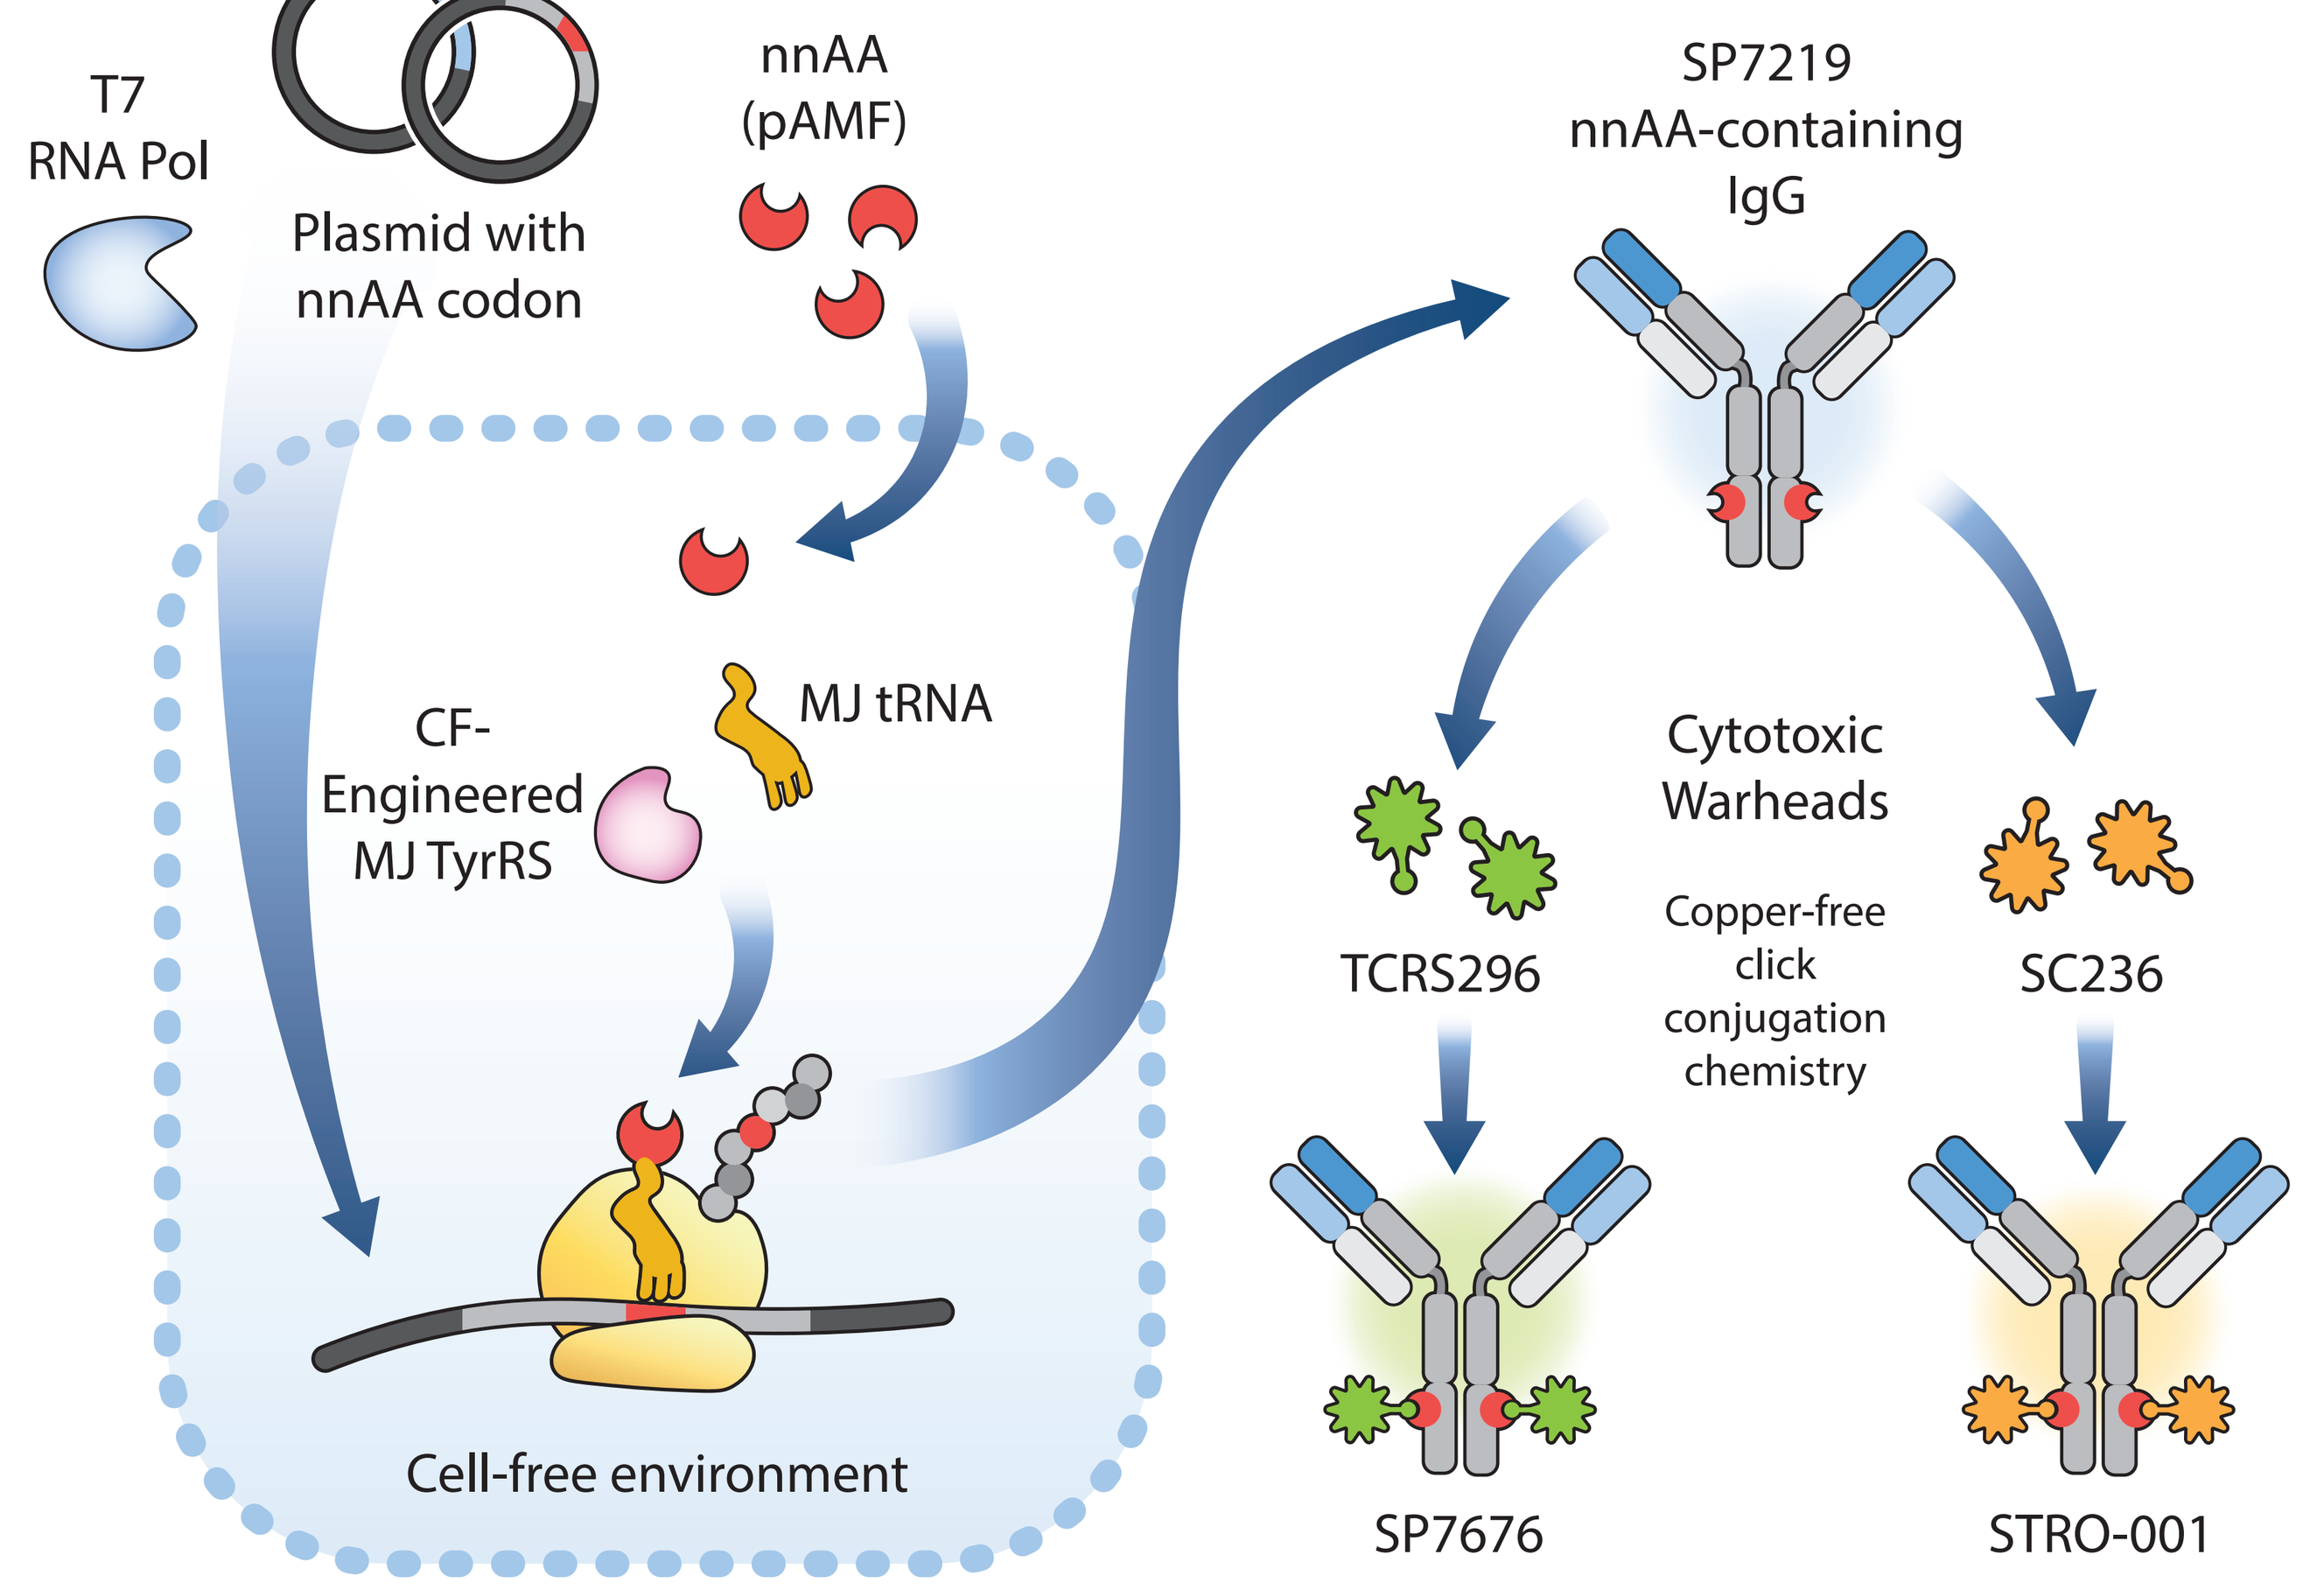
T7
RNA Pol
Plasmid with
nnAA codon
nnAA
(pAMF)
SP7219
nnAA-containing
IgG
CF-
Engineered
MJ TyrRS
MJ tRNA
Cytotoxic
Warheads
Copper-free
click
conjugation
chemistry
TCRS296
SC236
Cell-free environment
SP7676
STRO-001

Development of SP7219, SP7676 and STRO-001 using Sutro's proprietary Xpress Cell-Free (XpressCF+) system[2]. SP7219 was discovered from a Fab ribosome display library and screening platform based on XpressCF. SP7219 was selected based on optimal affinity, cell binding, internalization, biophysical properties, and immunogenicity potential. The non-natural amino acid (nnAA) pAMF was incorporated at different sites on SP7219, with the optimal sites selected based on conjugation efficiency, cell killing activity and PK in mice. SP7219 was conjugated at pAMF to the noncleavable maytansinoid linker-warheads TCRS296 or SC236 to generate SP7676 or STRO-001, respectively.

RESULTS

Figure 1. Multiple myeloma cell lines express CD74 on the cell surface

A)

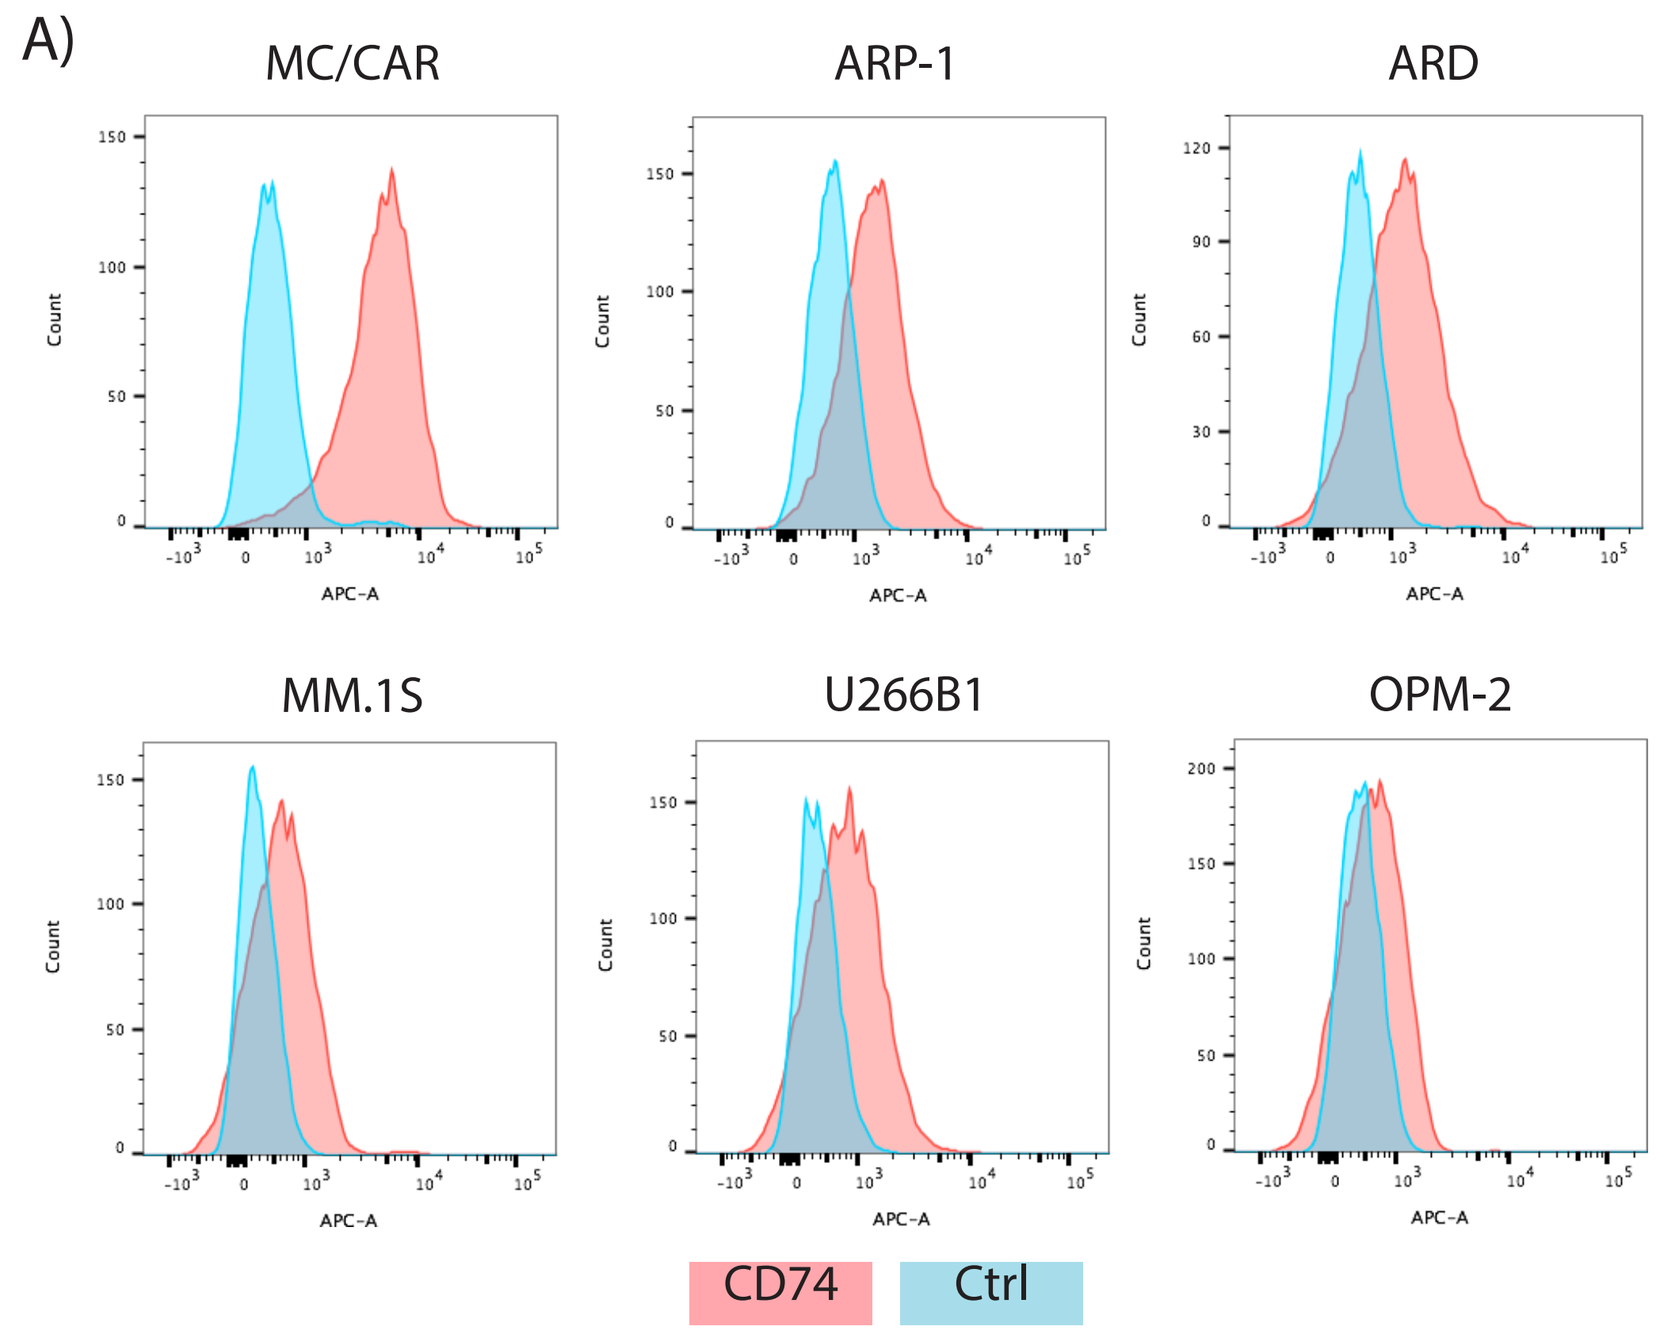
MC/CAR
ARP-1
ARD
MM.1S
U266B1
OPM-2
CD74
Ctrl

B)

MM Cell Line	SP7219 Binding	
	MFI	ABC
MC/CAR	3744	42981
ARP-1	924	88
ARD	841	BD
MM.1S	332	BD
U266B1	427	BD
OPM-2	218	BD

A) Histograms of the DBCO-Alexa647conjugated SP7219 binding to different MM cell lines by FACS analysis (red). Unstained cells were used as negative control (blue). B) Quantitation of cell binding activity of DBCO-Alexa647 SP7219 to MM cell lines. MFI=Median Fluorescent Intensity; ABC=Antibody Binding Capacity; BD=Below Detection.

Figure 2. STRO-001 ADC targeting of CD74 results in efficient cell killing *in vitro*

A)

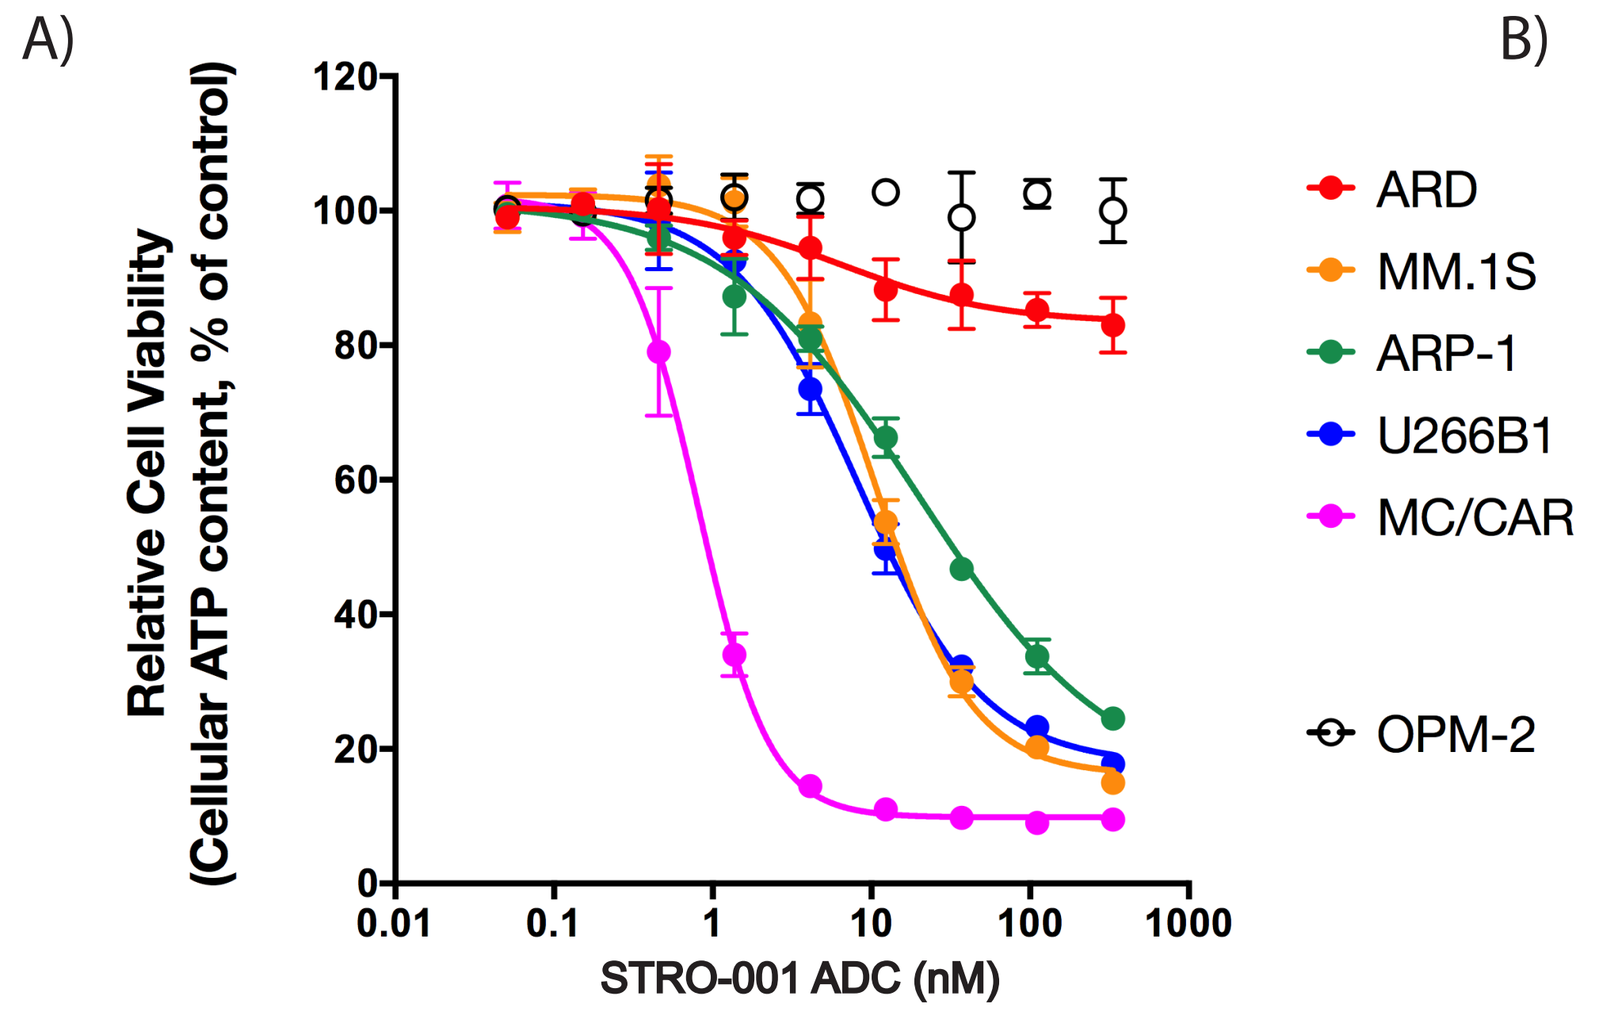
Relative Cell Viability
(Cellular ATP content, % of control)
STRO-001 ADC (nM)
ARD
MM.1S
ARP-1
U266B1
MC/CAR
OPM-2

B)

MM Cell Line	STRO-001 Cell Killing Activity	
	IC50 (nM)	Span (%)
MC/CAR	0.81	92
ARP-1	20	88
ARD	6.5	17
U266B1	8.2	84
MM.1S	11	86
OPM-2	NK	NK

STRO-001 ADC was used to determine the IC50 and percent span of killing in MM cell lines. A) Cell survival curve showing percentage of viable cells at different concentrations of STRO-001 for MM cell lines. B) Quantitation of IC50 and cell killing span (% of cells killed). NK=No Killing

Figure 3. SP7676 increases survival in the disseminated CAG MM model

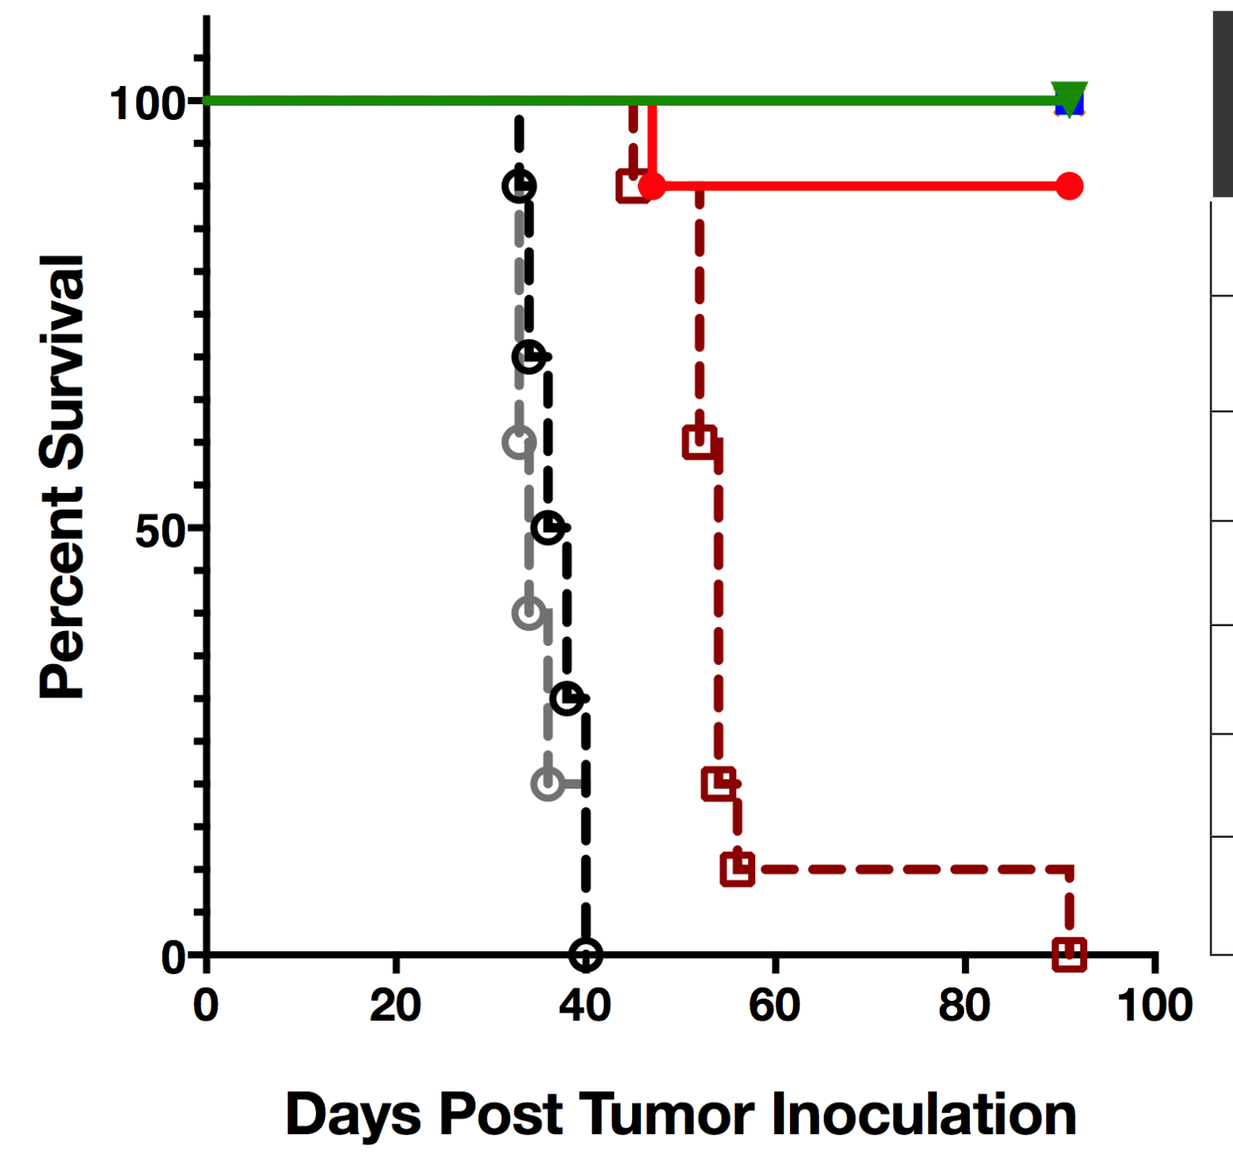
Percent Survival
Days Post Tumor Inoculation

Treatment	Median Survival (Days)	p-Value (Log-rank test)
SP7676 (1mg/kg Q3Dx5)	>92 (NR)	<0.0001
SP7676 (3mg/kg Q3Dx5)	>92 (NR)	<0.0001
SP7676 (10mg/kg Q7Dx3)	>92 (NR)	<0.0001
SP7676 (10mg/kg Q3Dx5)	>92 (NR)	<0.0001
Vehicle (Q3Dx5)	34	-
SP7219 (10mg/kg, Q3D)	37	NS
Melphalan (10mg/kg, Q7D)	54	<0.0001

Kaplan-Meier curve of SCID mice inoculated with 1×10^7 CAG MM cells. 14 days after tumor cell inoculation, 10 mice were randomized to each group and treated either with vehicle (PBS), SP7219, a range of doses of the ADC SP7676 or melphalan. The experiment was terminated at 92 days, and median survival was compared using a log-rank test. NS= not significant; NR= not reached.

Figure 4. STRO-001 and SP7676 significantly reduce tumor burden in disseminated ARP-1 MM model

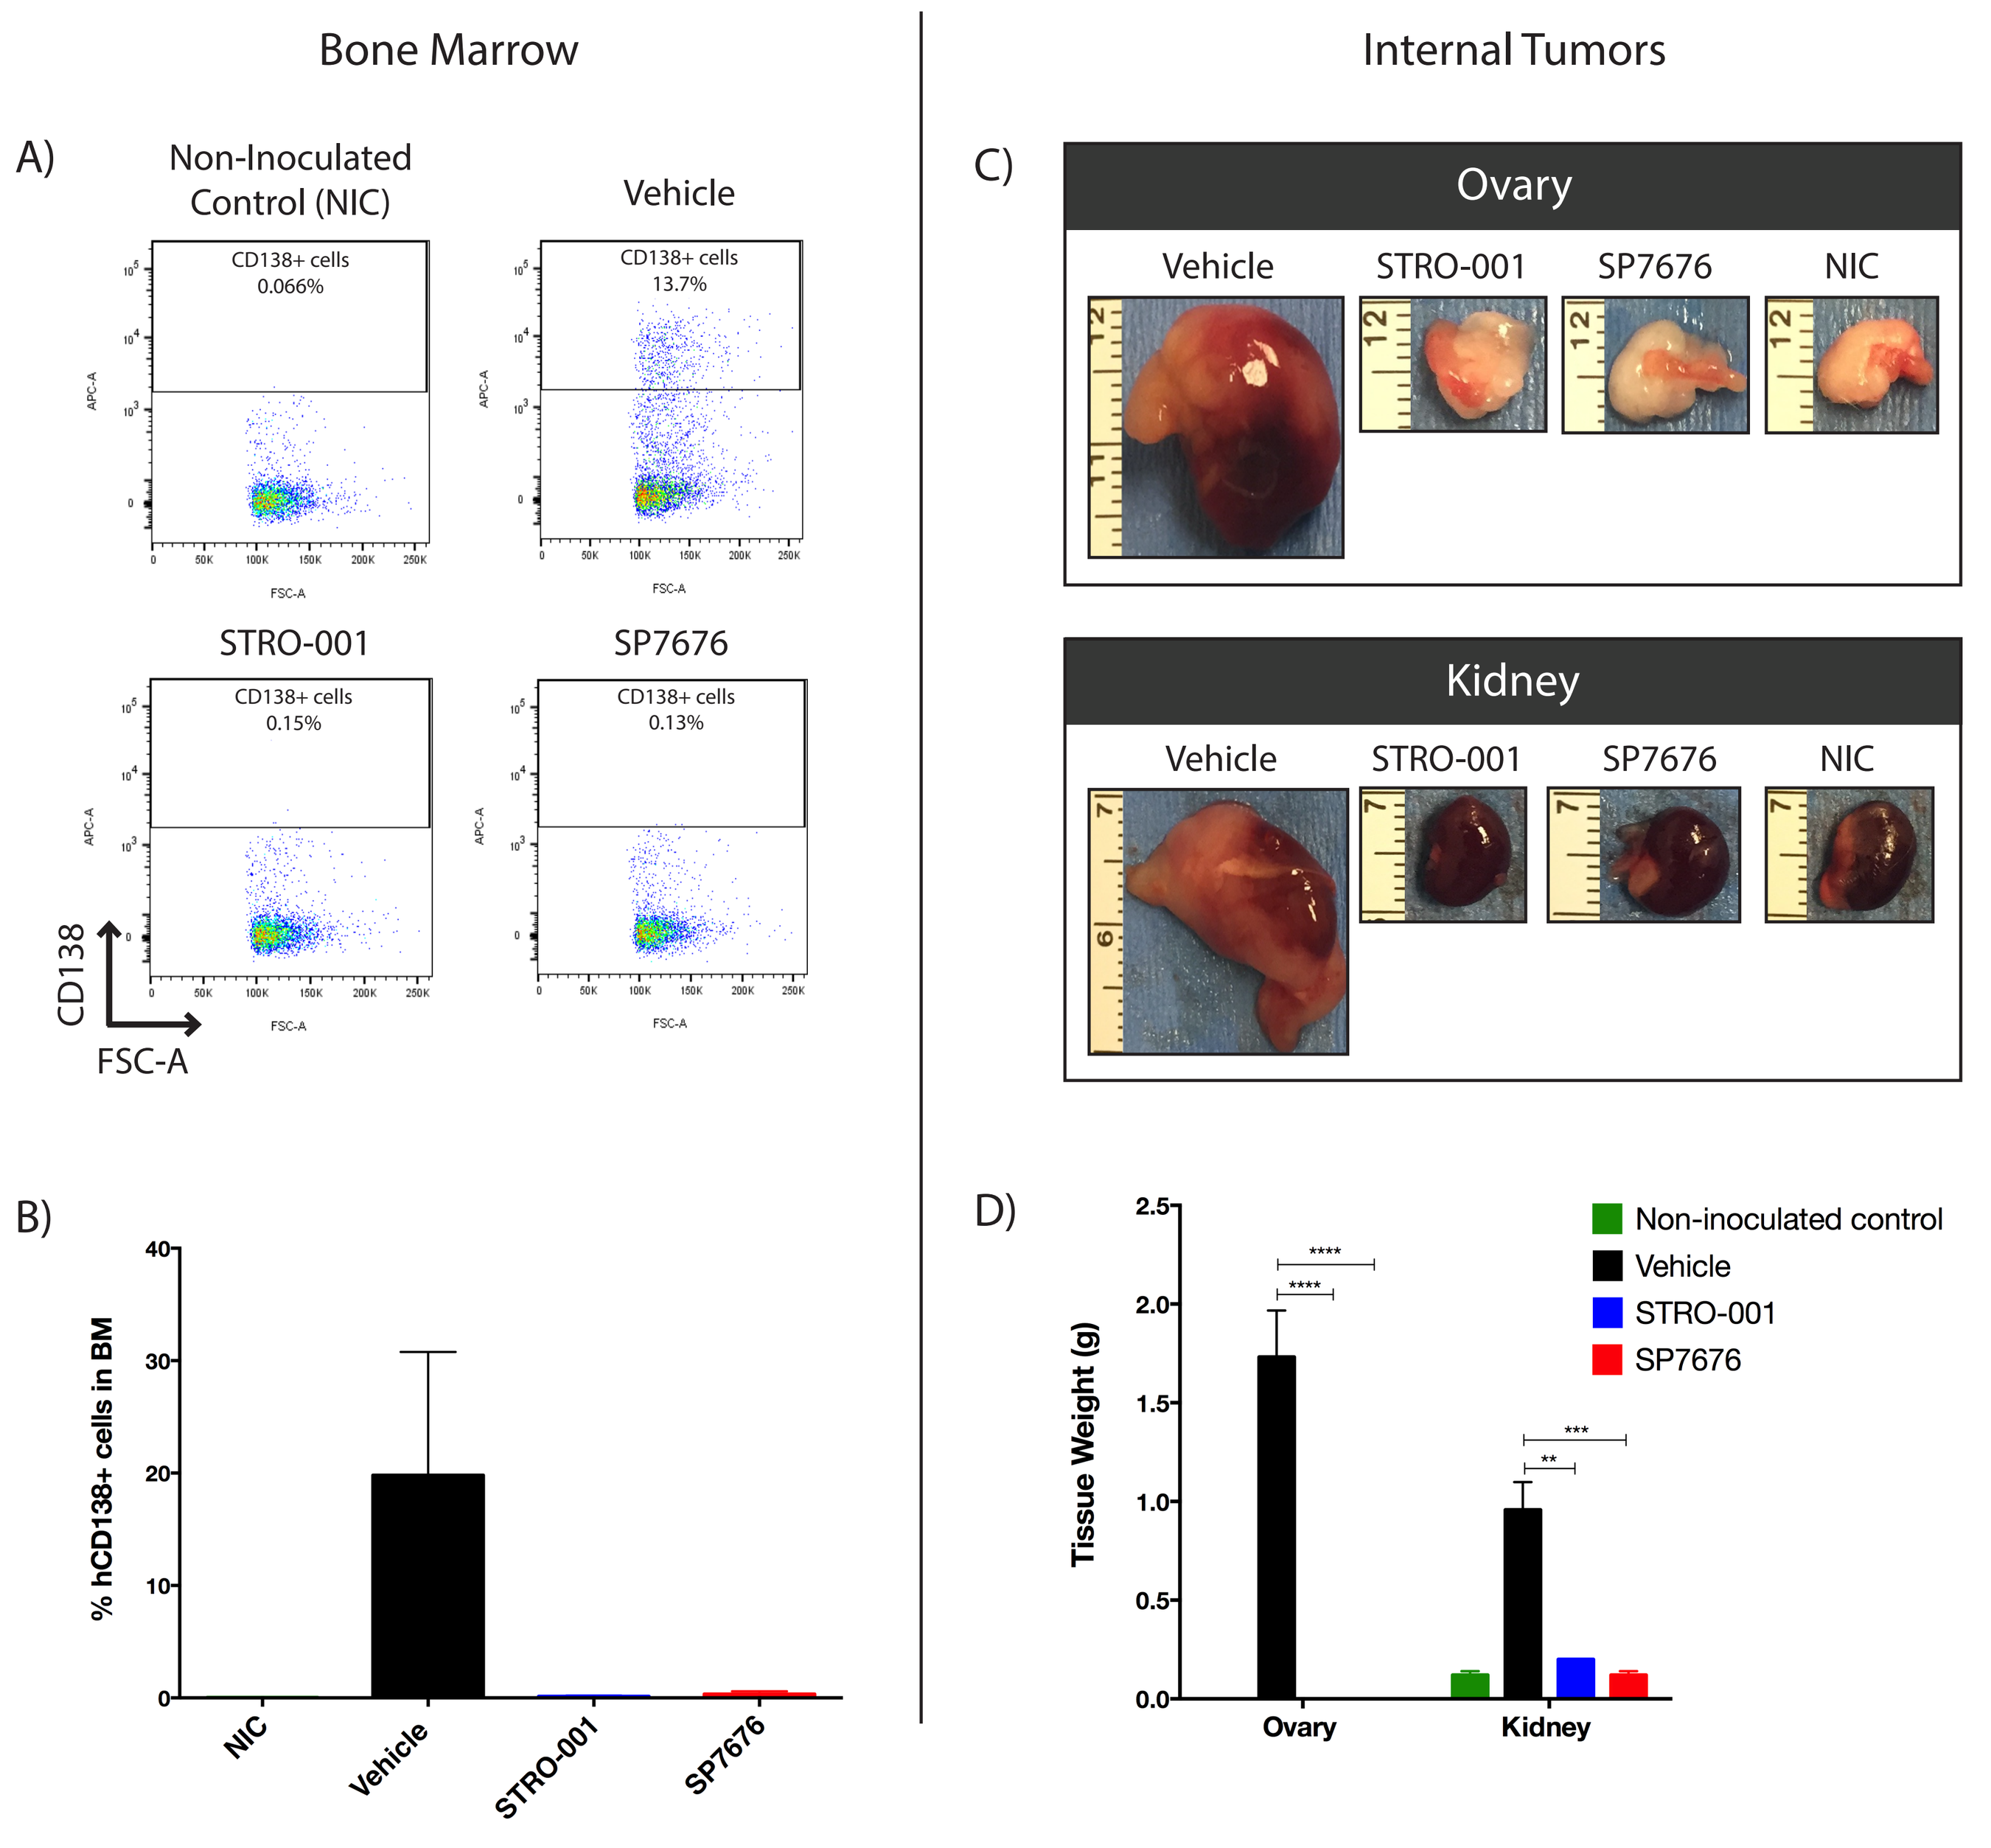
Bone Marrow
A)
Non-Inoculated Control (NIC)
Vehicle
STRO-001
SP7676
CD138+ cells 0.066%
CD138+ cells 13.7%
CD138+ cells 0.15%
CD138+ cells 0.13%
CD138
FSC-A
Internal Tumors
C)
Ovary
Kidney
Vehicle
STRO-001
SP7676
NIC
B)
% hCD138+ cells in BM
NIC
Vehicle
STRO-001
SP7676
D)
Tissue Weight (g)
Ovary
Kidney
Non-inoculated control
Vehicle
STRO-001
SP7676

SCID mice injected i.v. with ARP-1 cells were treated q7dx4 with vehicle, 3 mg/kg STRO-001 or 3 mg/kg SP7676 starting on d14 post-inoculation and harvested on d49. Non-inoculated controls did not receive cells or treatment. A) Representative dot plots showing STRO-001 and SP7676 inhibited ARP-1 growth in the bone marrow. B) Quantification of percent CD138+cells in BM. C) Formation of internal ARP-1 tumors was abrogated by STRO-001 and SP7676 treatment. D) Quantification of internal tumor tissue weight. Statistical analysis using one-way ANOVA with Dunnett's multiple comparison test. (****, p<0.0001; ***, p<0.001; **p<0.01). All graphs are depicted as average values +/- standard error of the mean (SEM).

Figure 5. SP7676 is a potent inhibitor of tumor growth in the ANBL-6 Melphalan refractory MM subcutaneous disease model

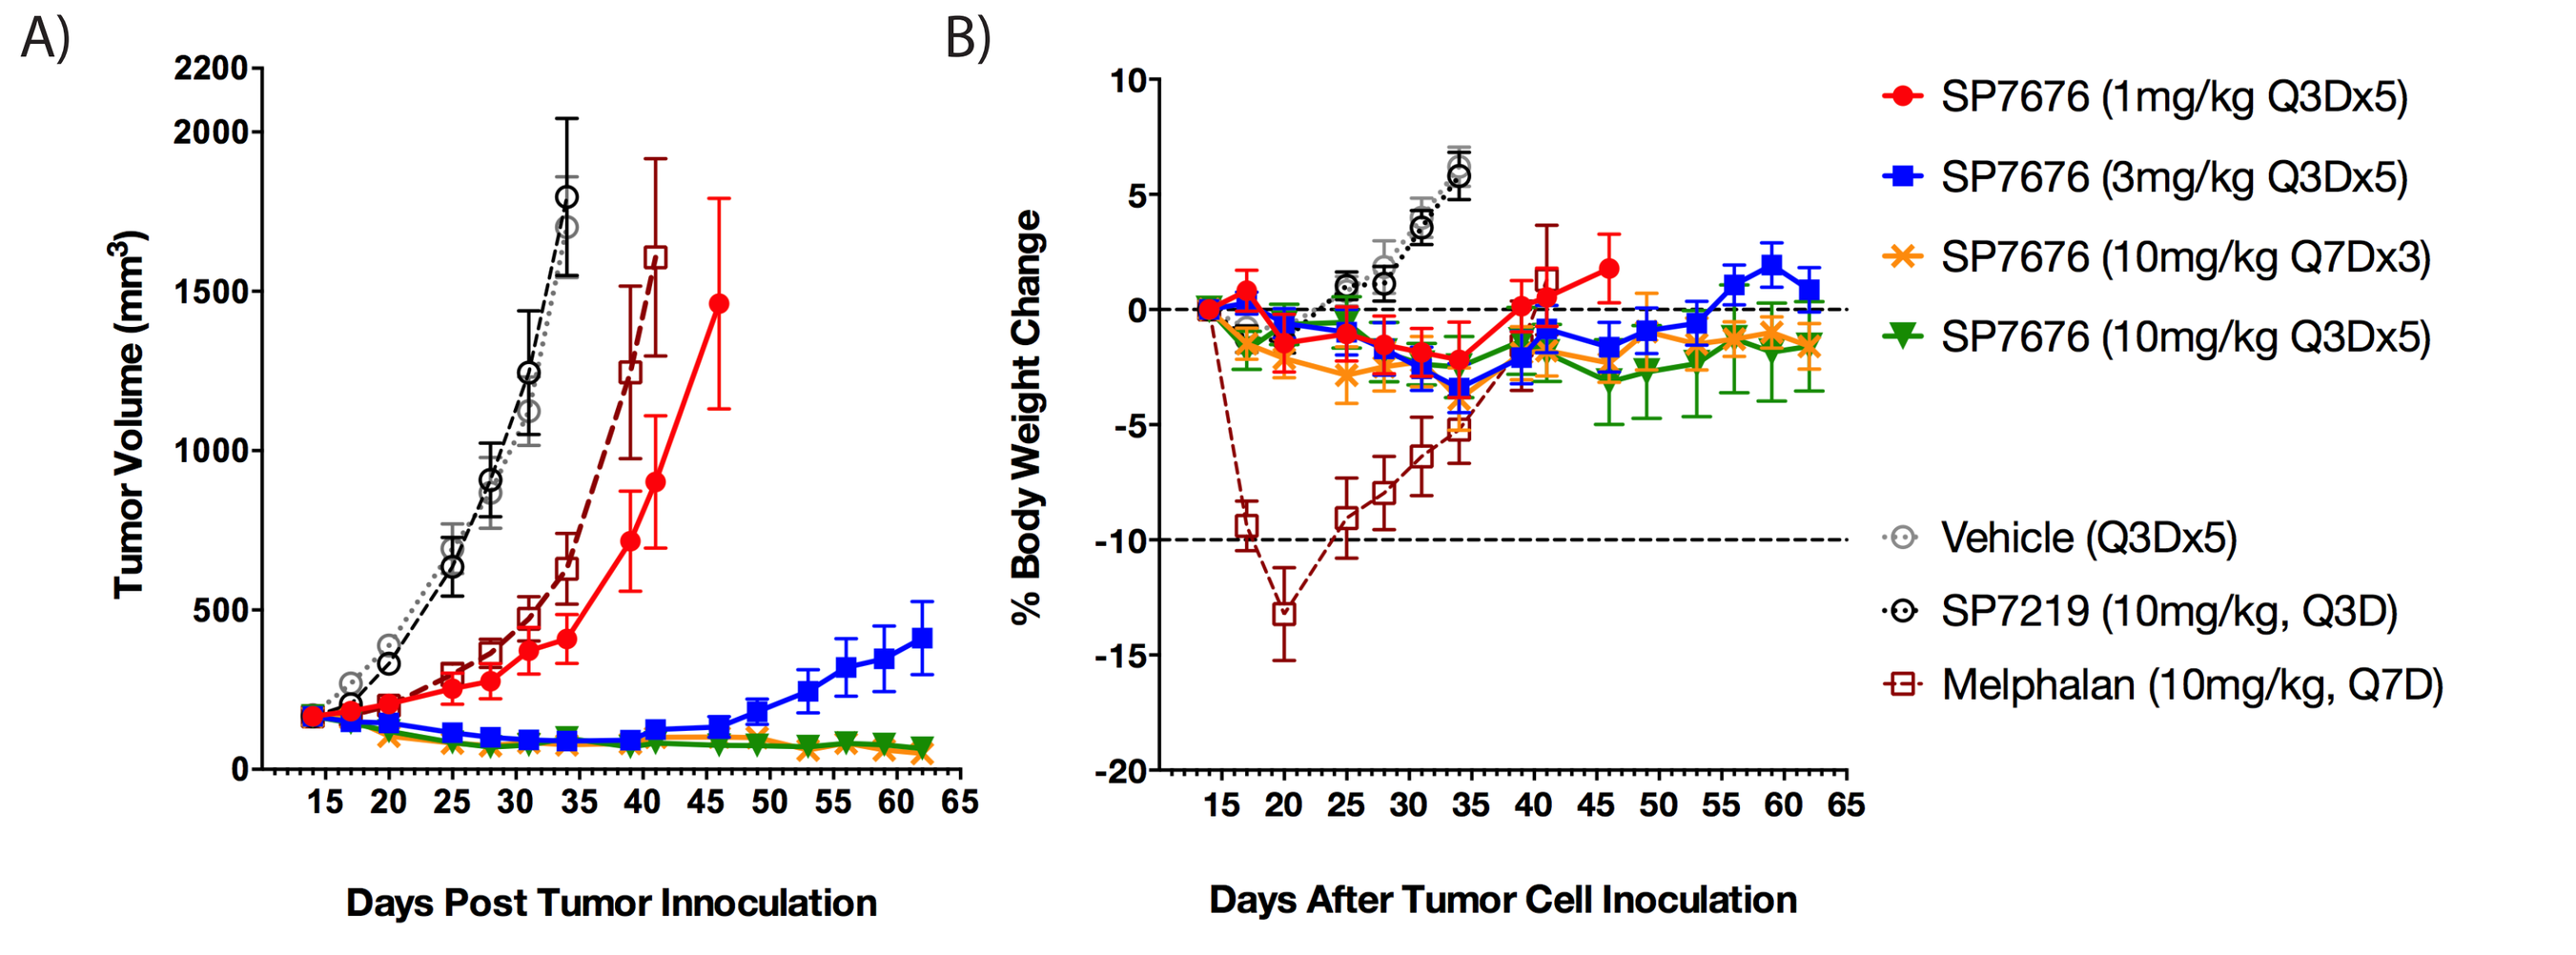
A)
Tumor Volume (mm³)
Days Post Tumor Innoculation
B)
% Body Weight Change
Days After Tumor Cell Inoculation
SP7676 (1mg/kg Q3Dx5)
SP7676 (3mg/kg Q3Dx5)
SP7676 (10mg/kg Q7Dx3)
SP7676 (10mg/kg Q3Dx5)
Vehicle (Q3Dx5)
SP7219 (10mg/kg, Q3D)
Melphalan (10mg/kg, Q7D)

A) Tumor growth of SCID mice following inoculation 2×10^7 ANBL-6 MM cells and treatment with vehicle, the SP7219 or SP7676.
B) Body weight change of all mice on study, calculated relative to weight at the start of treatment (day 14) as mean body weight± SEM.

Figure 6. STRO-001 induces dose-responsive ablation of B-cells in cynomolgus monkeys

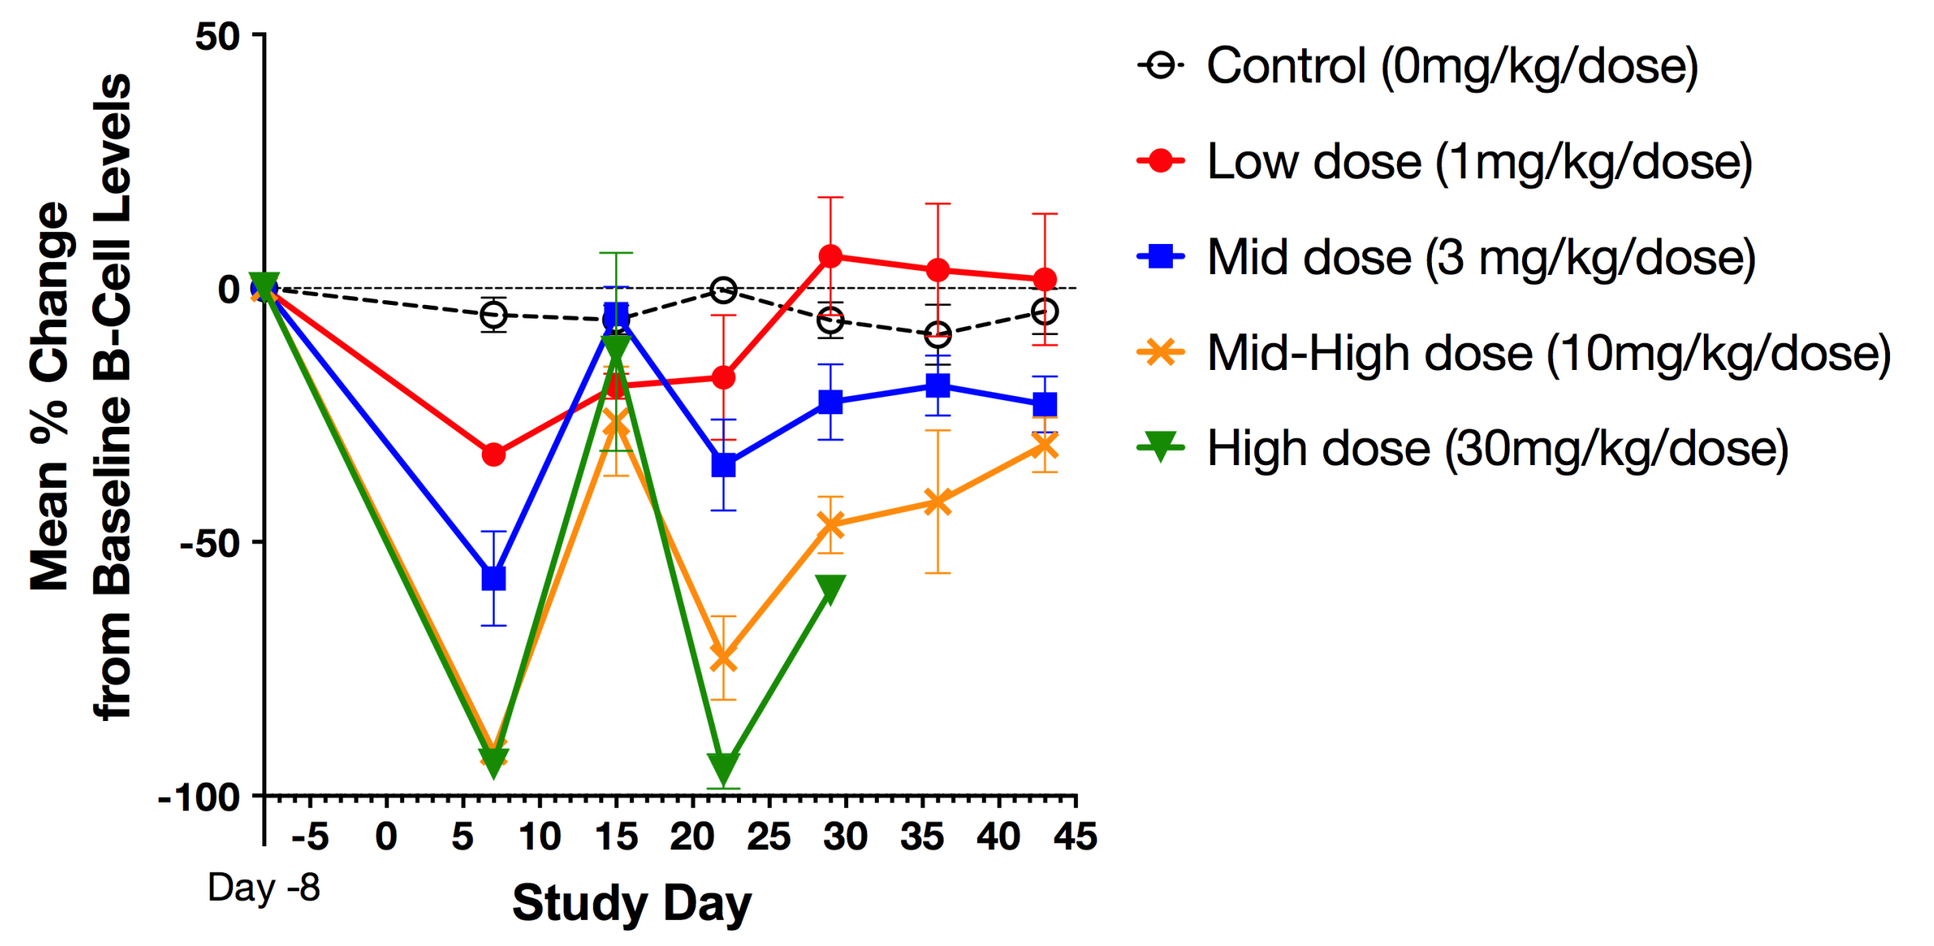
Mean % Change
from Baseline B-Cell Levels
Day -8
Study Day
Control (0mg/kg/dose)
Low dose (1mg/kg/dose)
Mid dose (3 mg/kg/dose)
Mid-High dose (10mg/kg/dose)
High dose (30mg/kg/dose)

B-cells were quantitated using flow cytometry. Total lymphocyte populations were identified using a gating strategy consisting of CD45 fluorescent staining and side-scatter characteristics (SSC) demarcation (CD45brightSSCdim) to delineate lymphocyte populations. The relative values for CD3-CD20+ cells obtained from the flow cytometer were multiplied by the absolute lymphocyte count from the hematology analysis to enumerate absolute cell counts. The graph above normalizes the relative B-cell numbers for each animal at each time point to the pre-dose B-cell numbers, depicted as average values +/- SEM.

CONCLUSIONS

- Sutro's technology allows for the generation of novel, specific, and homogenous ADCs targeting CD74.
- ADCs targeting CD74 produce efficient cell-killing in multiple MM cell lines.
- ADCs targeting CD74 led to the suppression of tumor growth in three MM models in vivo: ANBL-6, CAG, and ARP-1.
- Initial toxicology study in cynomolgus monkeys did not produce any unexpected findings; the main result of treatment with STRO-001 was B-cell depletion followed by recovery.
- IND-enabling studies are planned.

MATERIALS & METHODS

CELL BINDING AND DETERMINATION OF ANTIBODY BINDING CAPACITY (ABC) ON CELL SURFACE. Cells were blocked with Human Fc block from BD and then incubated with 100nM of DBCO-Alexa647 conjugated to SP7219 in FACS buffer (PBS+1% BSA) on ice for 60 minutes. Samples were then washed twice and analyzed on BD FACS Canto system. Median Fluorescent Intensity (MFI) was calculated by FlowJo. Antibody binding capacity (ABC) on cell surface was determined by Quantum Simply Cellular anti-human IgG beads from Bangs Laboratories, Inc.

ADC CELL KILLING ASSAYS. Cytotoxicity effects of the free drug linkers and ADCs were measured with a cell proliferation assay. Filter sterilized samples were serial diluted under sterile conditions and added onto cells on 384-well flat bottom white polystyrene plate. Plates were cultured at 37°C in a CO2 incubator for 72hrs. Cell viability was measured using Cell Titer-Glo® reagent. The relative luminescence measured by ENVISION® plate reader were converted to % viability using untreated cells as controls. Data was fitted using GraphPad Prism.

XENOGRAFT MODELS. CB17 SCID mice were intravenously injected with 1×10^7 CAG or ARP-1 cells via the tail vein or subcutaneously with 2×10^7 ANBL-6 cells + matrigel. Randomization and treatment was initiated 2 weeks post-tumor inoculation when tumors were established. Animals were administered with test articles, doses and dosing regimen as indicated in the figure legends. Body weights and tumor measurements (for ANBL-6 tumors only) were monitored twice weekly. For disseminated model, study endpoints included survival (significant change in body weight) and/or clinical signs of moribundity. In ARP-1 study, bone marrow was harvested from 14 days after last dose to assess human CD138+ cells.

EXPLORATORY TOXICOLOGY IN CYNOMOLGOUS MONKEY. An exploratory safety study was conducted in female cynomolgus monkeys, and animals were given i.v. doses of vehicle, 1, 3, 10 or 30 mg/kg on day 1 and day 15 followed by a 28-day observation period post-last dose. Animals were observed for clinical signs and evaluated for clinical pathology (hematology, coagulation and serum chemistry) and immunophenotyping, as well as analysis of pharmacokinetic properties of the molecule.

REFERENCES

1 Stein, M.R. Mattes, J., et al. Clin Cancer Res Sep 2007 (13) (18) 5556s-5563s.

2 Zimmerman, E.S. Heibeck, T.H. et al. Bioconjug Chem Feb 2014 25(2): 351-61.

Disclosures - All co-authors were employees of either Sutro or Celgene when this research was conducted.