Characterization and pre-clinical development of STRO-001, a novel CD74-targeting antibody-drug conjugate (ADC) for the treatment of B-cell malignancies

Cristina Abrahams, Xiaofan Li, Venita De Almeida, Millicent Embry, Abigail Yu, Stellanie Krimm, Heidi Hoffmann, James Zawada, Maureen Bruhns, Shannon Matheny, Stuart Bussell, Toni Kline, Alice Yam, Ryan Stafford, Trevor Hallam, Mark Lupher, and Arturo Molina.

Sutro Biopharma, South San Francisco, CA

BACKGROUND

- CD74 is a transmembrane glycoprotein involved in MHC protein formation & transport
- CD74 is expressed in ~90% of B-cell cancers including myeloma and lymphoma
- Normal tissues have minimal CD74 expression
- CD74 is rapidly internalized

Immunohistology of patient biopsy specimens[1]

Diagnosis	No. positive / no. tested	% Target cells stained
Follicular lymphoma	8/9	> 95%
Diffuse large B-cell lymphoma	4/4	~ 80%
Other NHL	31/35	Not determined
Small lymphocytic lymphoma / CLL	14/14	> 90%
Multiple myeloma	19/22	16/22, >95% 3/22, ~ 50%

Discovery of CD74-targeting ADCs

ADCs are emerging as a promising class of cancer biopharmaceuticals that combine the specificity of monoclonal antibodies with the anti-tumor activity of cytotoxic agents.

We have developed a novel anti-CD74 human IgG1 antibody, SP7219, and conjugated this at specific amino acids to non-cleavable maytansinoid linker-warhead (SC236) with a drug-antibody ratio (DAR) of 2, to generate a potent ADC, STRO-001.

Figure 1. Generation of the CD74-targeting lead antibody and a novel, specific and homogeneous ADC, STRO-001

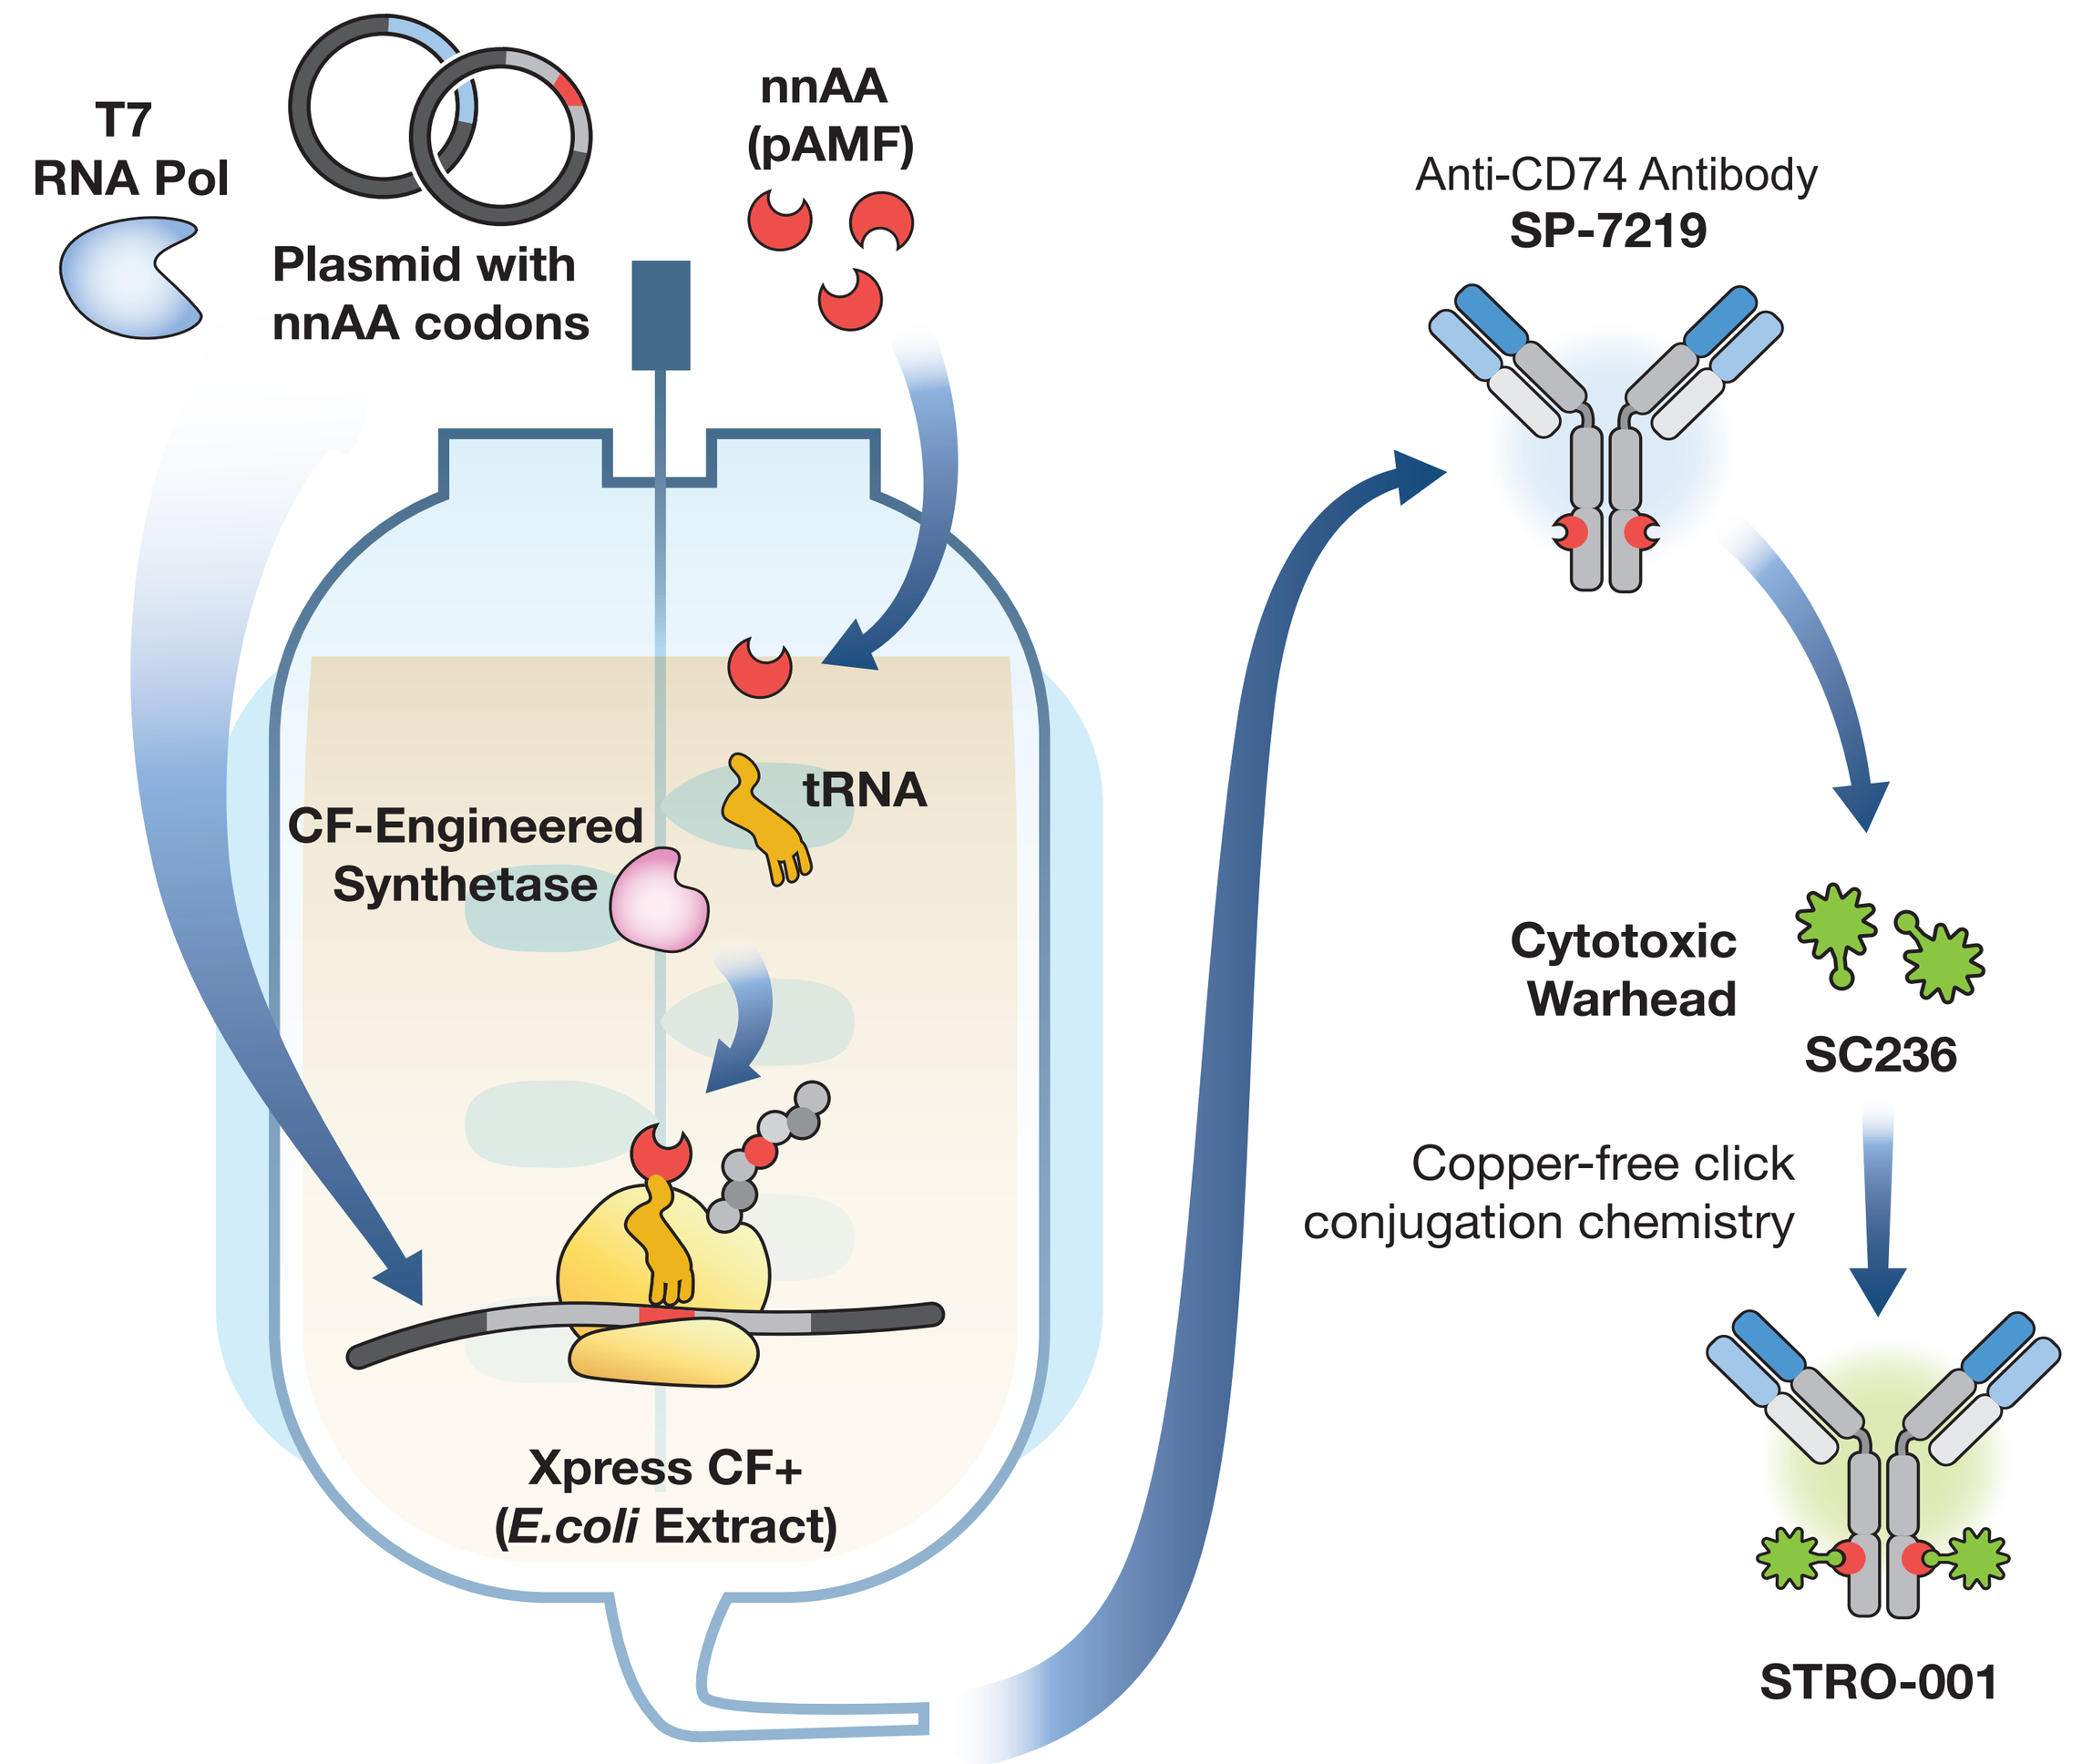
T7
RNA Pol
Plasmid with
nnAA codons
nnAA
(pAMF)
CF-Engineered
Synthetase
tRNA
Xpress CF+
(E.coli Extract)
Anti-CD74 Antibody
SP-7219
Cytotoxic
Warhead
SC236
Copper-free click
conjugation chemistry
STRO-001

Development of SP7219 and STRO-001 using Sutro's proprietary Xpress Cell-Free (XpressCF+™) system[2]. SP7219 was discovered from a Fab ribosome display library and screening platform based on XpressCF™. SP7219 was selected based on optimal affinity, cell binding, internalization, biophysical properties, and immunogenicity potential. The non-natural amino acid (nnAA) pAMF was incorporated at different sites on SP7219, with the optimal sites selected based on conjugation efficiency, cell killing activity and PK in mice. SP7219 was conjugated at pAMF to the noncleavable maytansinoid linker-warhead SC236 to generate STRO-001.

RESULTS

Figure 2. STRO-001 is cytotoxic to CD74+ lymphoma cells while catabolites have limited toxicity irrespective of CD74 expression

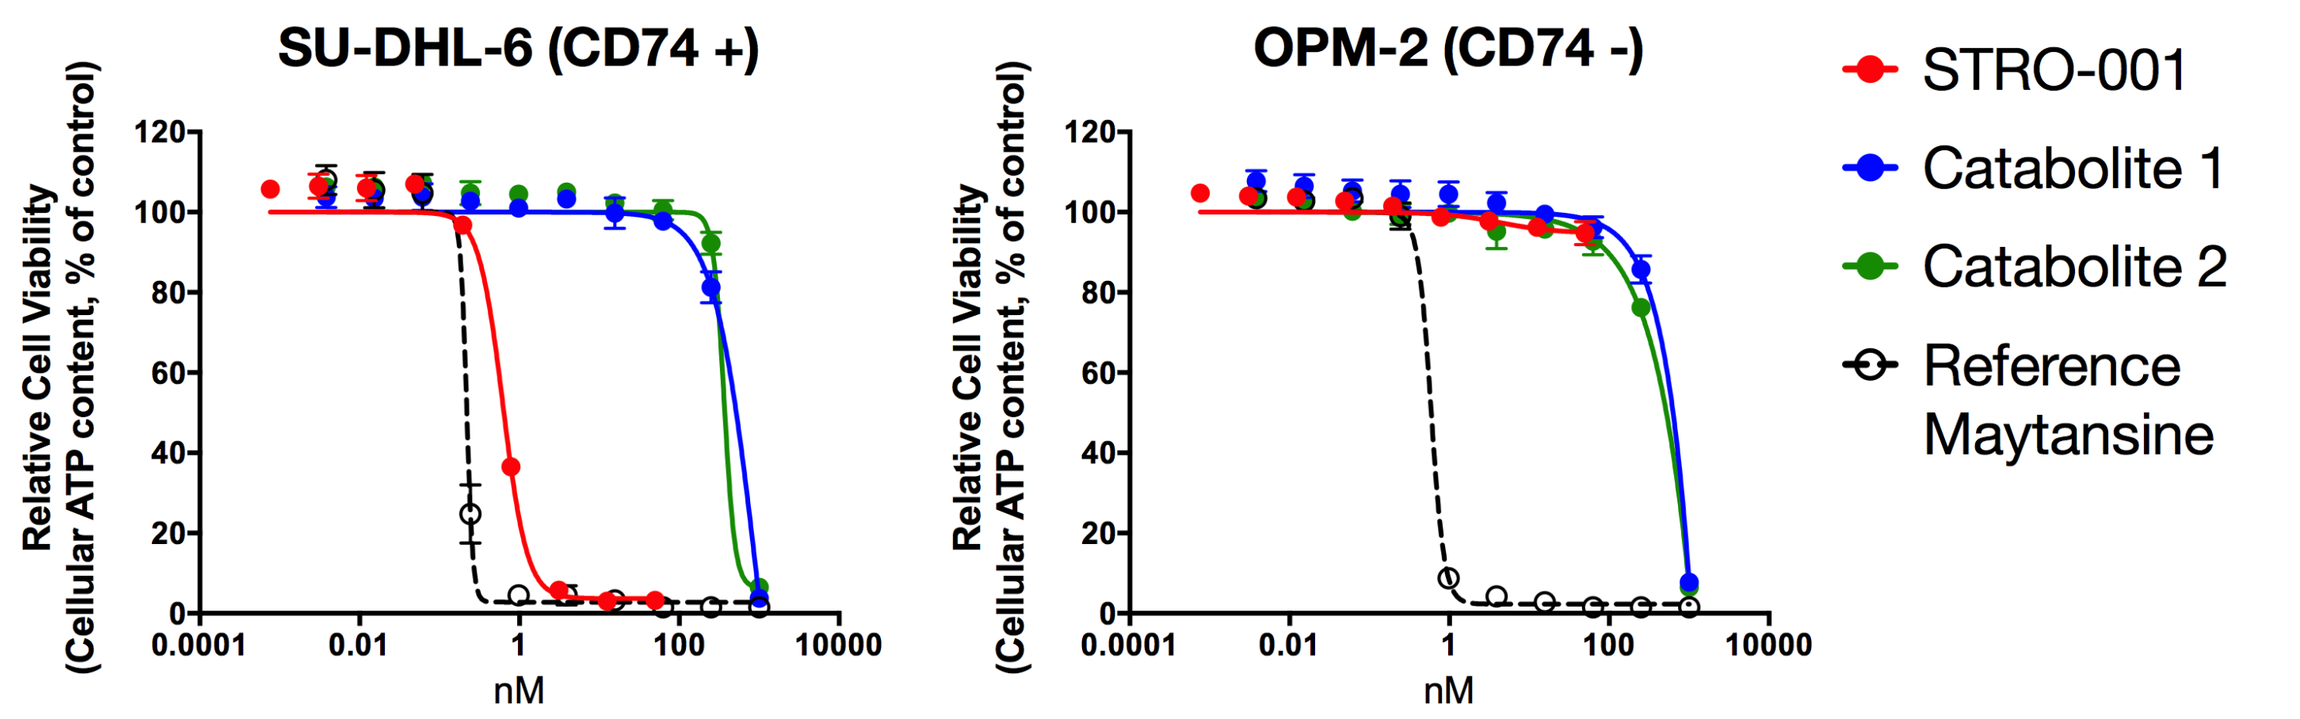
SU-DHL-6 (CD74 +)
OPM-2 (CD74 -)
Relative Cell Viability (Cellular ATP content, % of control)
nM
STRO-001
Catabolite 1
Catabolite 2
Reference Maytansine

Cell Line	STRO-001 EC50 (nM)	STRO-001 Span (%)	Reference Maytansine EC50 (nM)	Reference Maytansine Span (%)	Catabolite 1 EC50 (nM)	Catabolite 1 Span (%)	Catabolite 2 EC50 (nM)	Catabolite 2 Span (%)
SU-DHL-6	0.62	96	0.22	97	NC	NC	NC	NC
OPM-2	NK	NK	0.59	98	NC	NC	NC	NC

Comparison of cytotoxicity of reference maytansine, the active STRO-001 catabolites 1 and 2 (regioisomers), and STRO-001 on SU-DHL-6 (CD74 +) and OPM-2 (CD74 -) cells. NC- not calculable, NK-no killing.

Figure 3. STRO-001 expression and activity in a diverse panel of B-cell tumor lines

A)

Disease	Cell Line	SP-7219 Receptor Copy Number	STRO-001 Cell Killing EC50 (nM)	STRO-001 Cell Killing Span (%)
Multiple Myeloma	MC/CAR	42,981	0.81	92
	ARP-1	9,523	20	88
	ARD	8,605	>5	>15
	MM.1S	< 8,445	11	86
	U266B1	< 8,445	8.2	84
GCB-like DLBCL	WSU-DLCL-2	51,090	0.17	97
	WSU-NHL	49,925	0.69	96
	SU-DHL-4	49,603	0.24	97
	Pfei er	47,123	0.54	88
	SU-DHL-6	23,235	0.56	96
	OCI-Ly1	21,529	0.69	96
	HT	20,788	0.34	68
	NUDUL-1	13,040	0.3	98
	OCI-Ly19	< 4,321	1.3	38
ABC-like DLBCL	OCI-Ly3	77,435	0.46	96
	U2932	10,649	1.3	80
	RIVA (RI-1)	< 4,321	3.3	77
	SU-DHL-2	< 4,321	>100	>20
Mantle Cell Lymphoma	Mino	28,117	0.5	97
	JVM-2	23,672	1.2	60
	JeKo-1	14,754	0.4	97
	Rec-1	8,247	>15	>50

B)

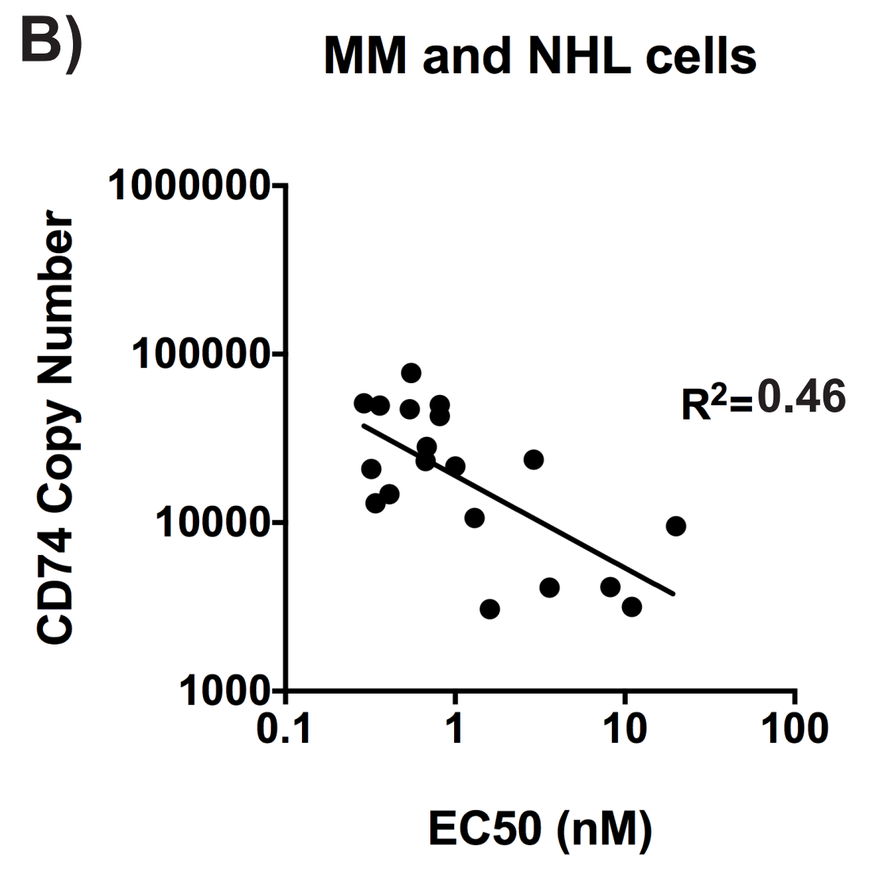
MM and NHL cells
CD74 Copy Number
EC50 (nM)
R^2=0.46

A) Expression of CD74 (receptor copy number) and the cell killing activity of STRO-001 in multiple myeloma (MM) and Non-Hodgkin's Lymphoma (NHL) (including germinal center B-cell (GCB)-like diffuse large B-cell lymphoma (DLBCL), activated B-cell (ABC) DLBCL and mantle cell lymphoma) cell lines. **B)** CD74 receptor number correlation with EC50 is not significant.

Figure 4. STRO-001 significantly reduces tumor burden in ARP-1 myeloma model

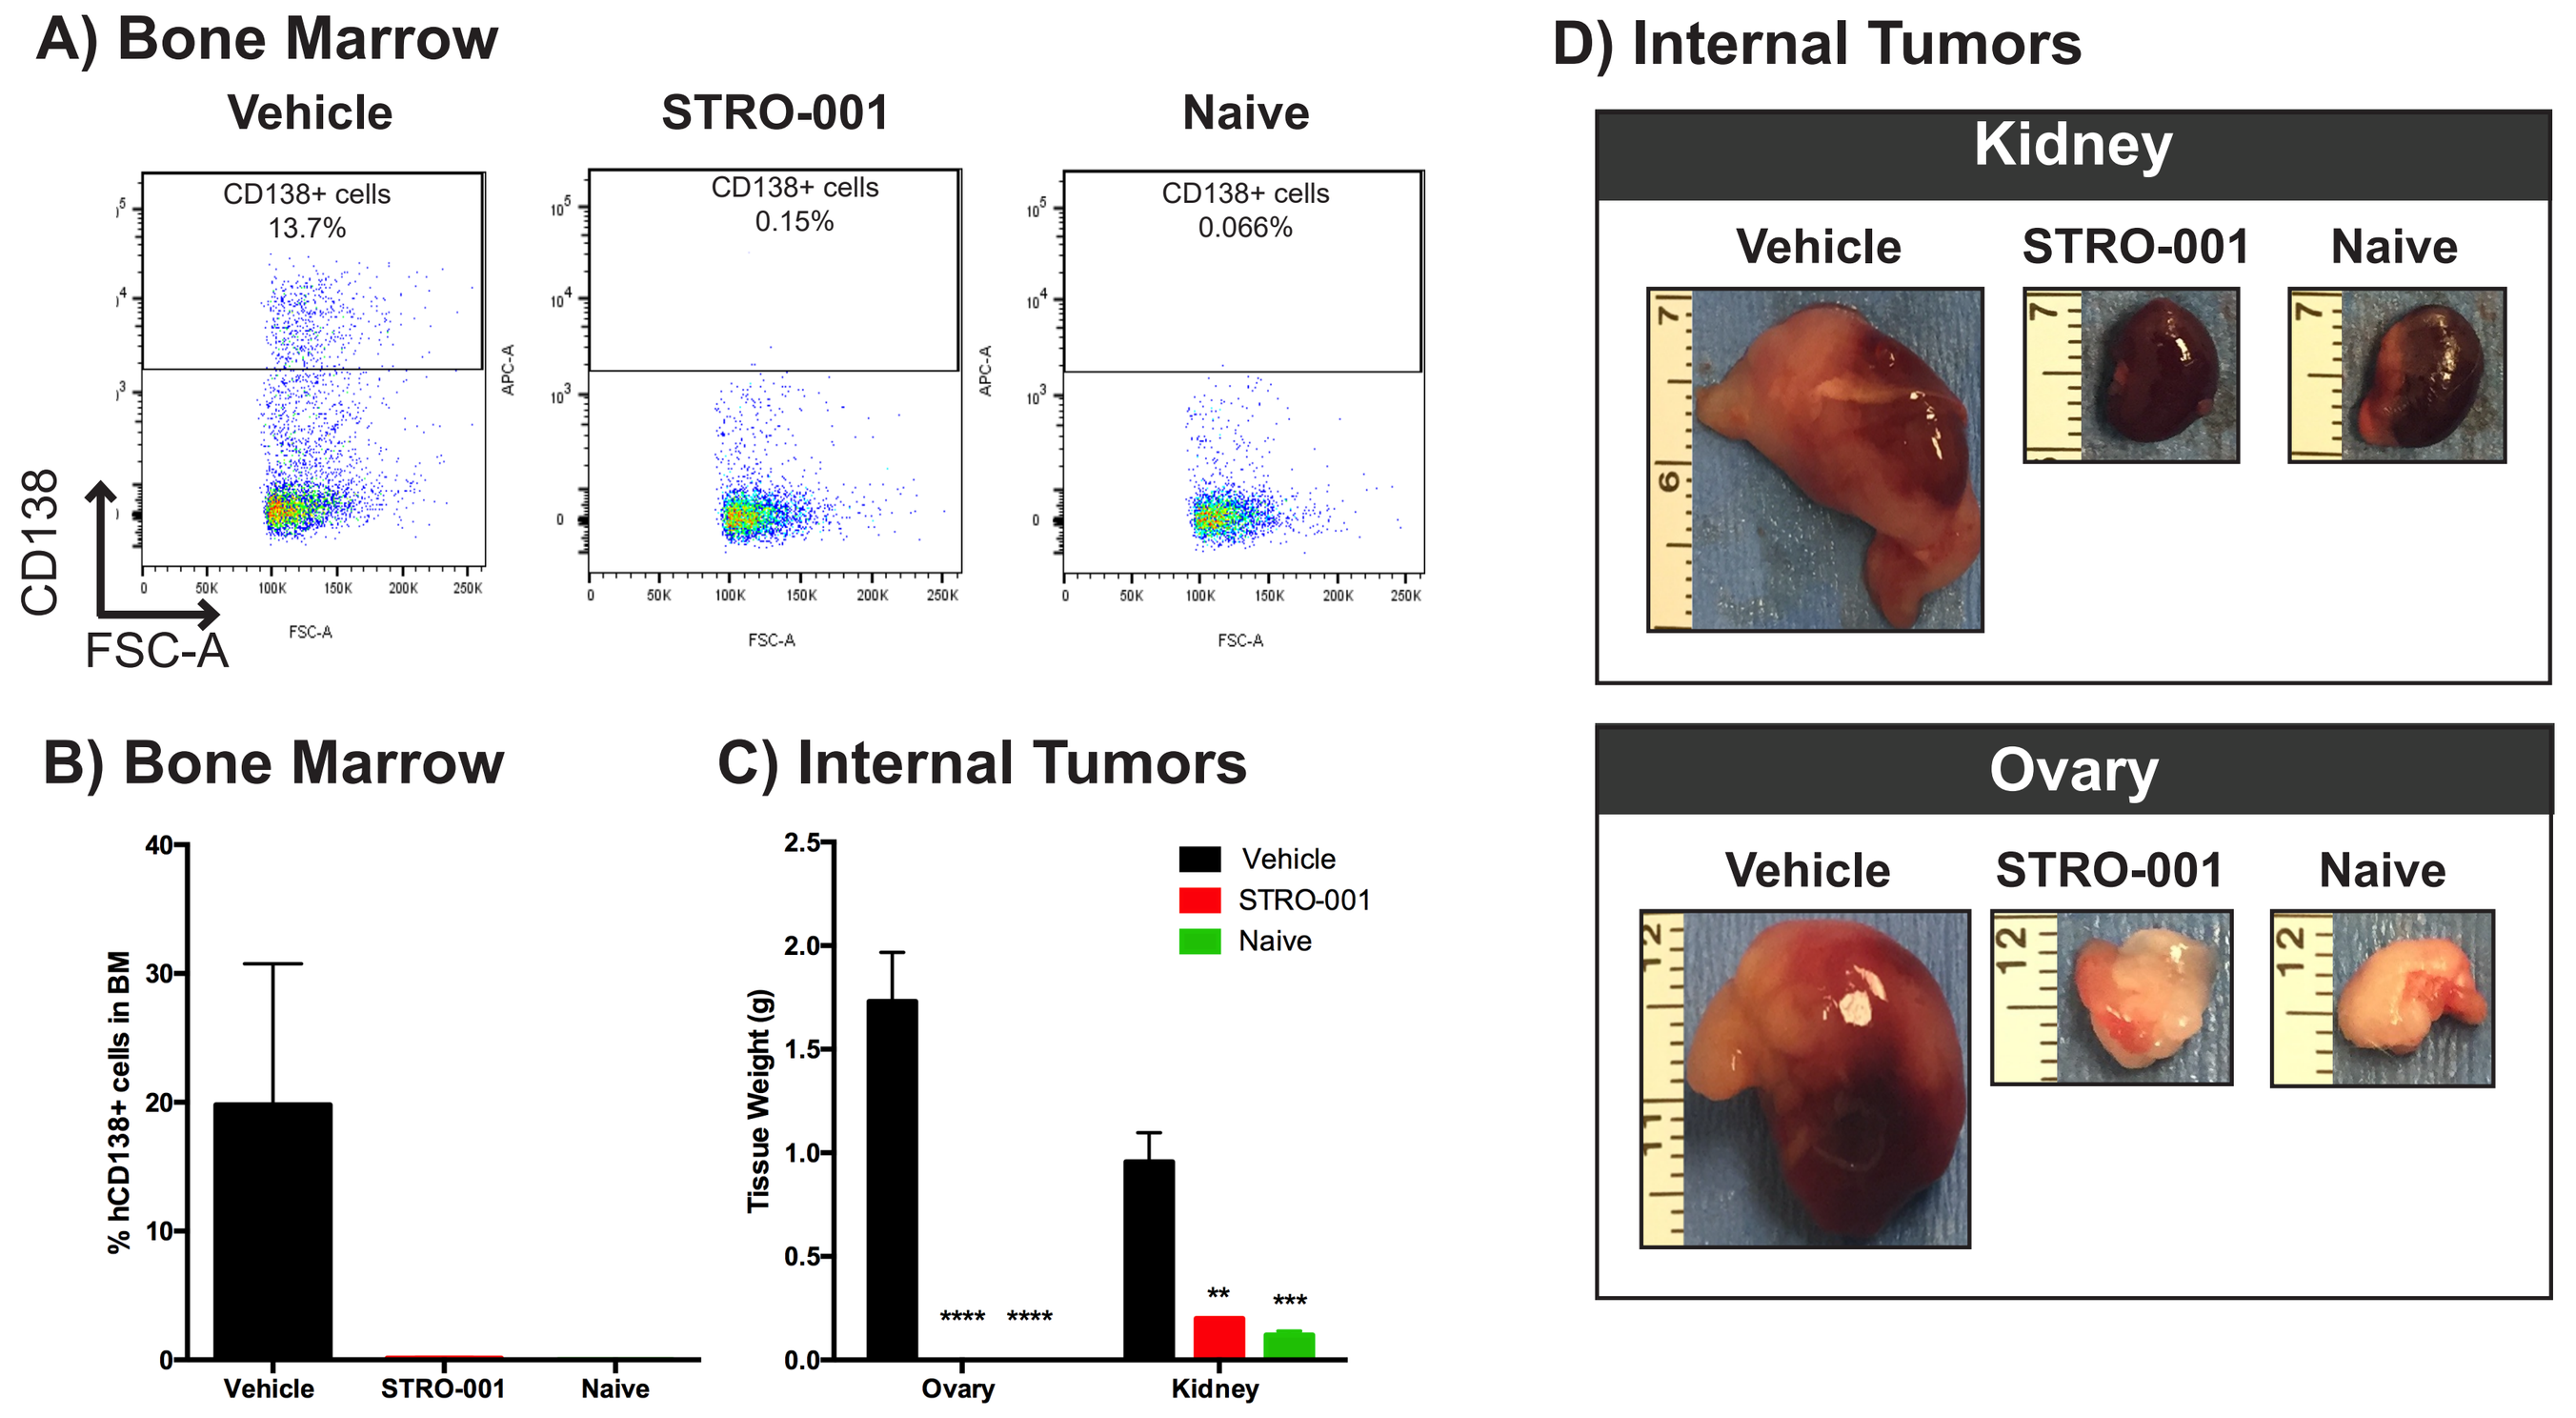
A) Bone Marrow
Vehicle
STRO-001
Naive
CD138+ cells 13.7%
CD138+ cells 0.15%
CD138+ cells 0.066%
CD138
FSC-A
B) Bone Marrow
% hCD138+ cells in BM
C) Internal Tumors
Tissue Weight (g)
Ovary
Kidney
D) Internal Tumors
Kidney
Ovary

SCID mice injected IV with ARP-1 cells treated q7dx4 with vehicle or 3 mg/kg STRO-001 starting on d14 post-inoculation and harvested on d49. Naive controls did not receive cells or treatment. **A)** Representative dot plots showing CD138+ cells in the bone marrow. to evaluate tumor burden. **B)** Quantification of % CD138+cells in bone marrow. **C-D)** ARP-1 model results in formation of internal tumors. Quantification of weights **(C)** and representative images of ovary and kidneys **(D)** in response to treatment. Statistical analysis was performed using one-way ANOVA with Dunnett's multiple comparison test.

Figure 5. STRO-001 eradicates malignant bone marrow plasma cells and prolongs survival in MM1.S myeloma model

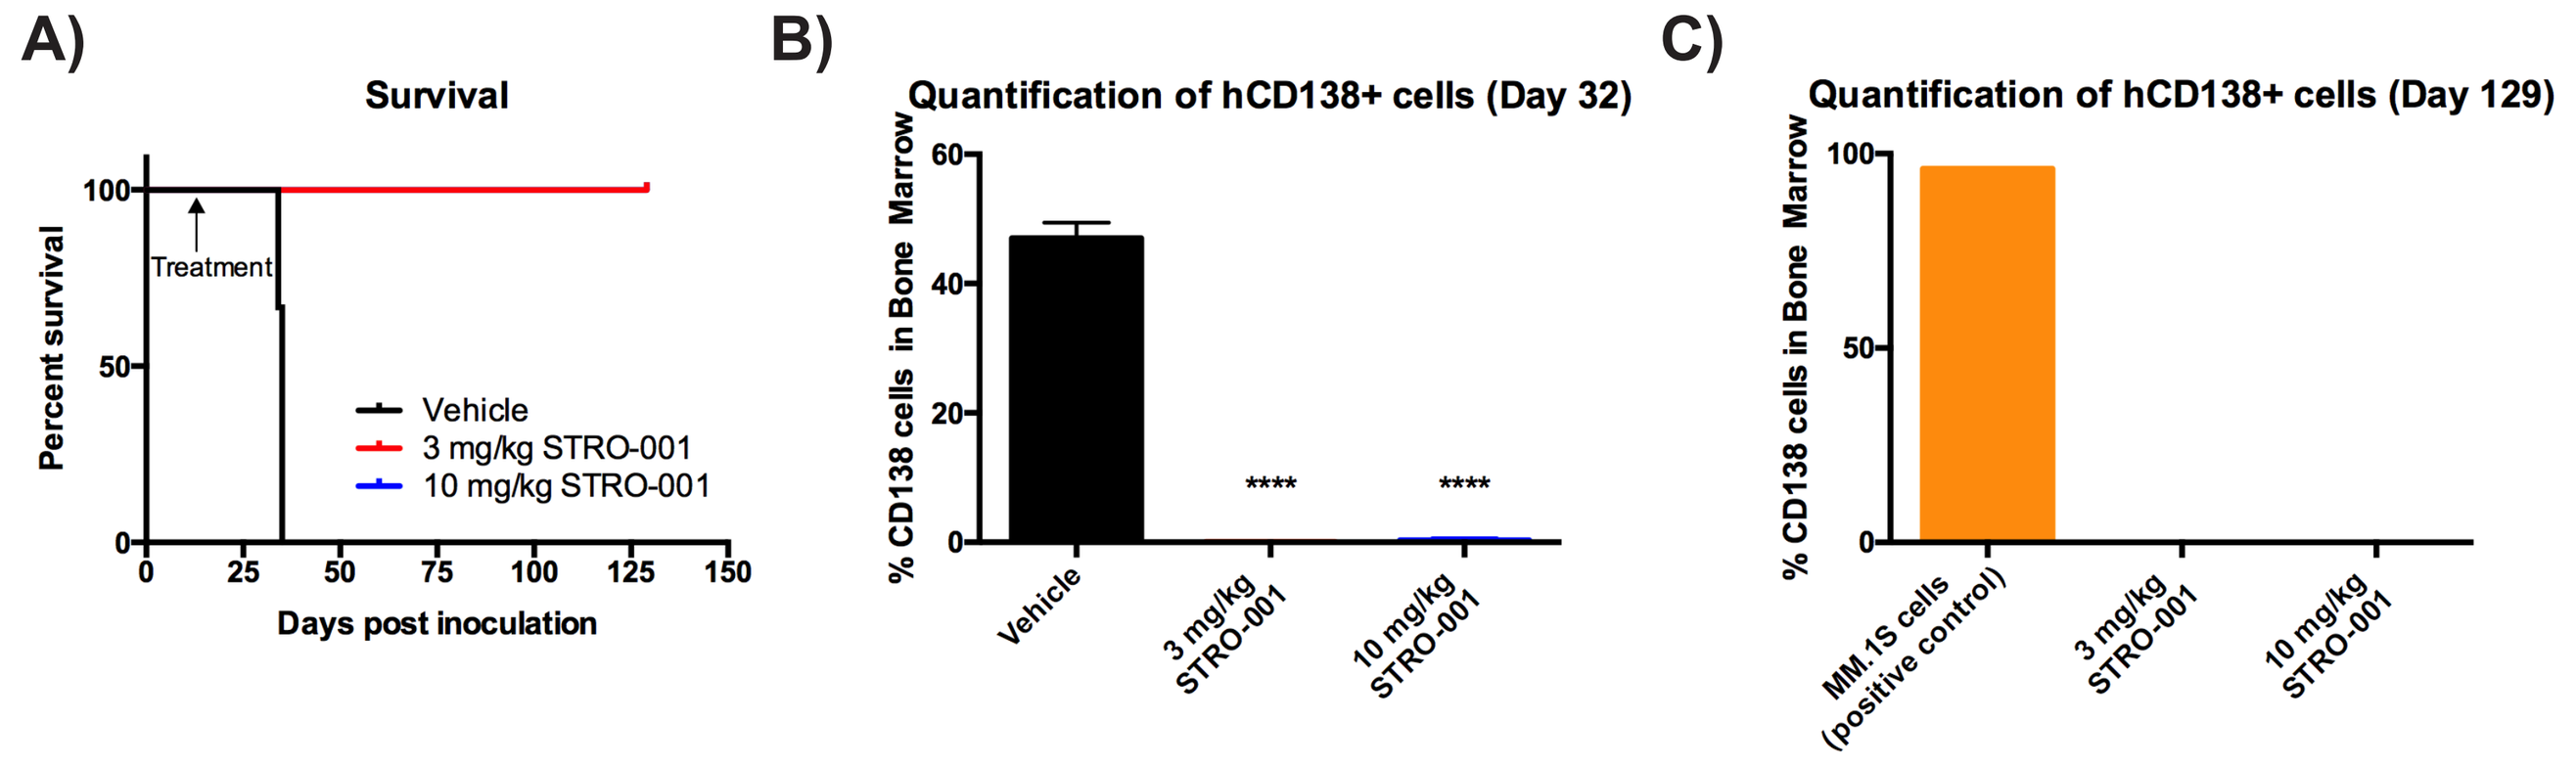
A)
Survival
Percent survival
Days post inoculation
Treatment
Vehicle
3 mg/kg STRO-001
10 mg/kg STRO-001
B)
Quantification of hCD138+ cells (Day 32)
C)
Quantification of hCD138+ cells (Day 129)
% CD138 cells in Bone Marrow
MM.1S cells (positive control)

NSG mice were inoculated with MM.1S cells. Treatment was started at day 11 with vehicle control, 3mg/kg STRO-001 or 10mg/kg STRO-001 (q7dx3). **A)** Kaplan-Meier curves in response to STRO-001 treatment or vehicle. **B-C)** Quantification of CD138+ in the bone marrow on day 32 **(B)** or day 129 **(C)**. Statistical analysis was performed using one-way ANOVA with Dunnett's multiple comparison test.

Figure 6. STRO-001 exhibits dose-dependent tumor growth inhibition in SU-DHL-6 lymphoma xenografts

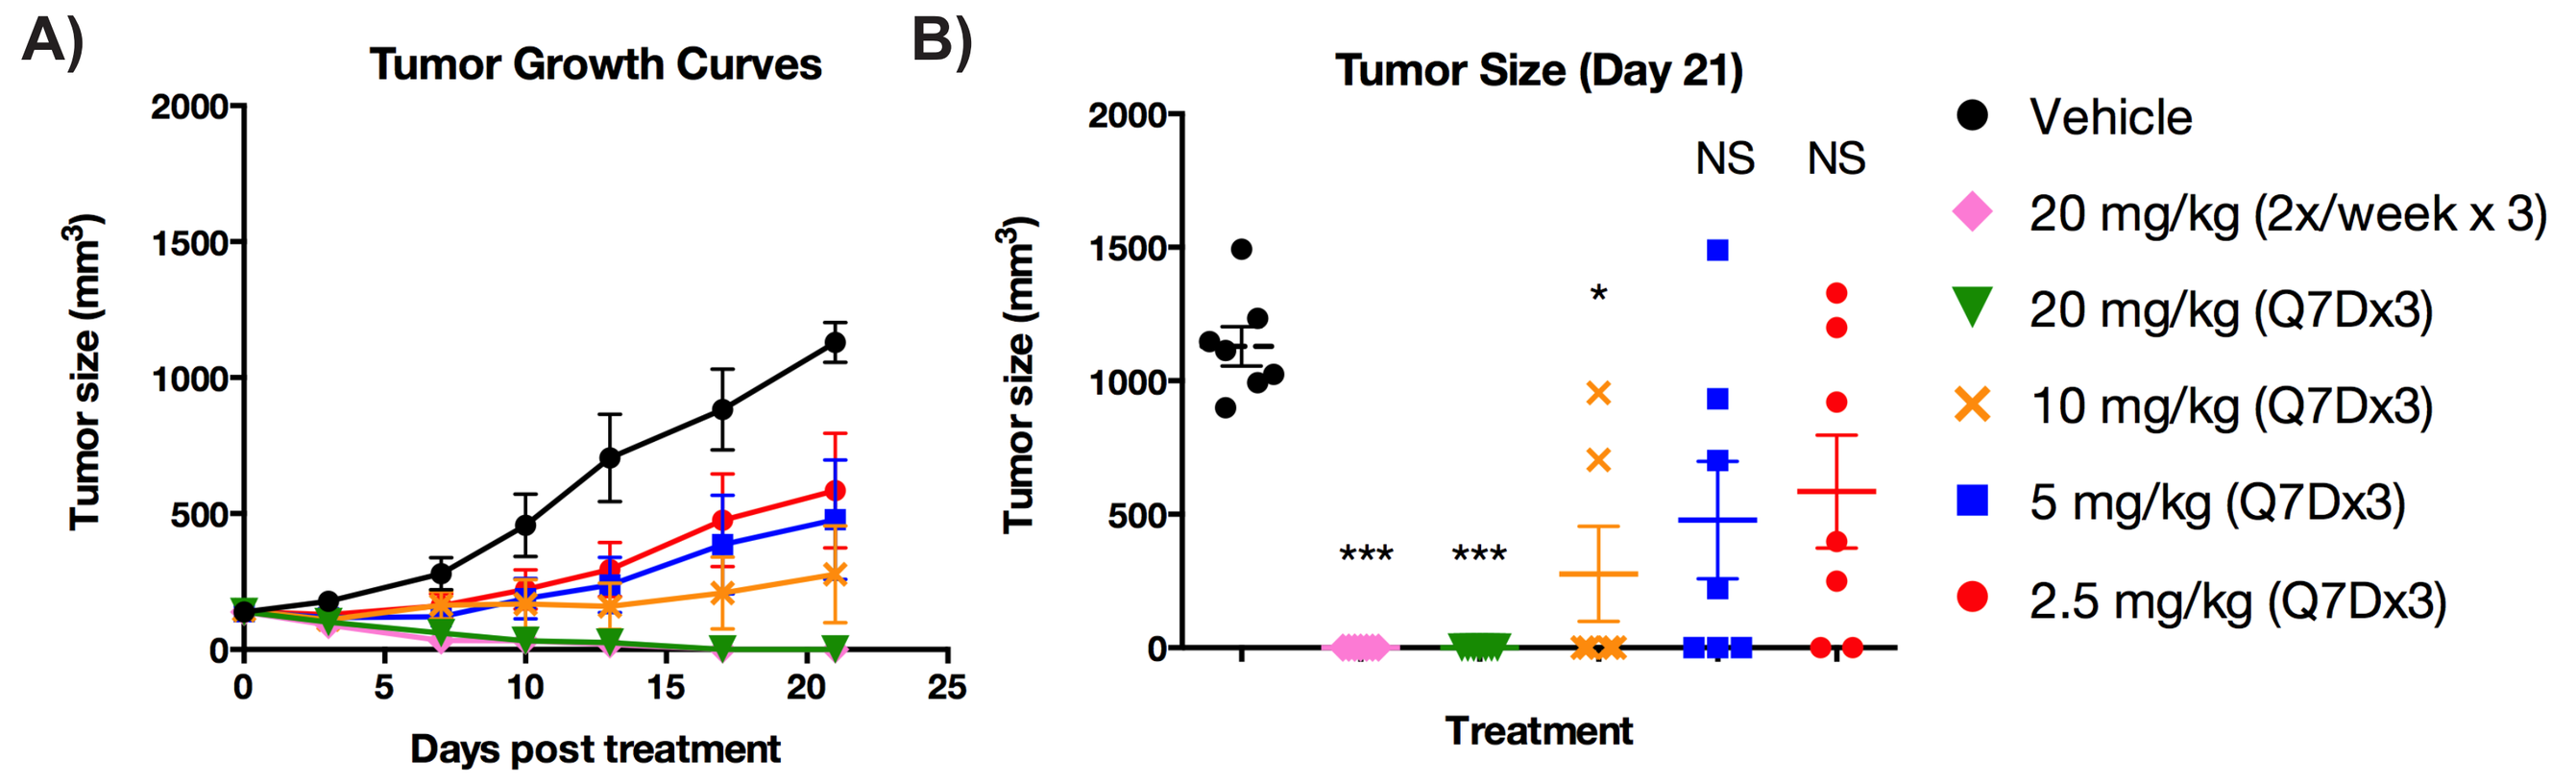
A)
Tumor Growth Curves
Tumor size (mm³)
Days post treatment
B)
Tumor Size (Day 21)
Treatment
Vehicle
20 mg/kg (2x/week x 3)
20 mg/kg (Q7Dx3)
10 mg/kg (Q7Dx3)
5 mg/kg (Q7Dx3)
2.5 mg/kg (Q7Dx3)

A) SU-DHL-6 tumor growth curves in response to STRO-001 treatment. **B)** Scatter plot of individual tumor size on day 21 when mean of control tumors >1000 mm³. Statistical analysis was performed using one-way ANOVA with Dunn's multiple comparison test.

Figure 7. Combination of STRO-001 + Rituximab/Bendamustine improves control of tumor growth in SU-DHL-6 lymphoma xenografts

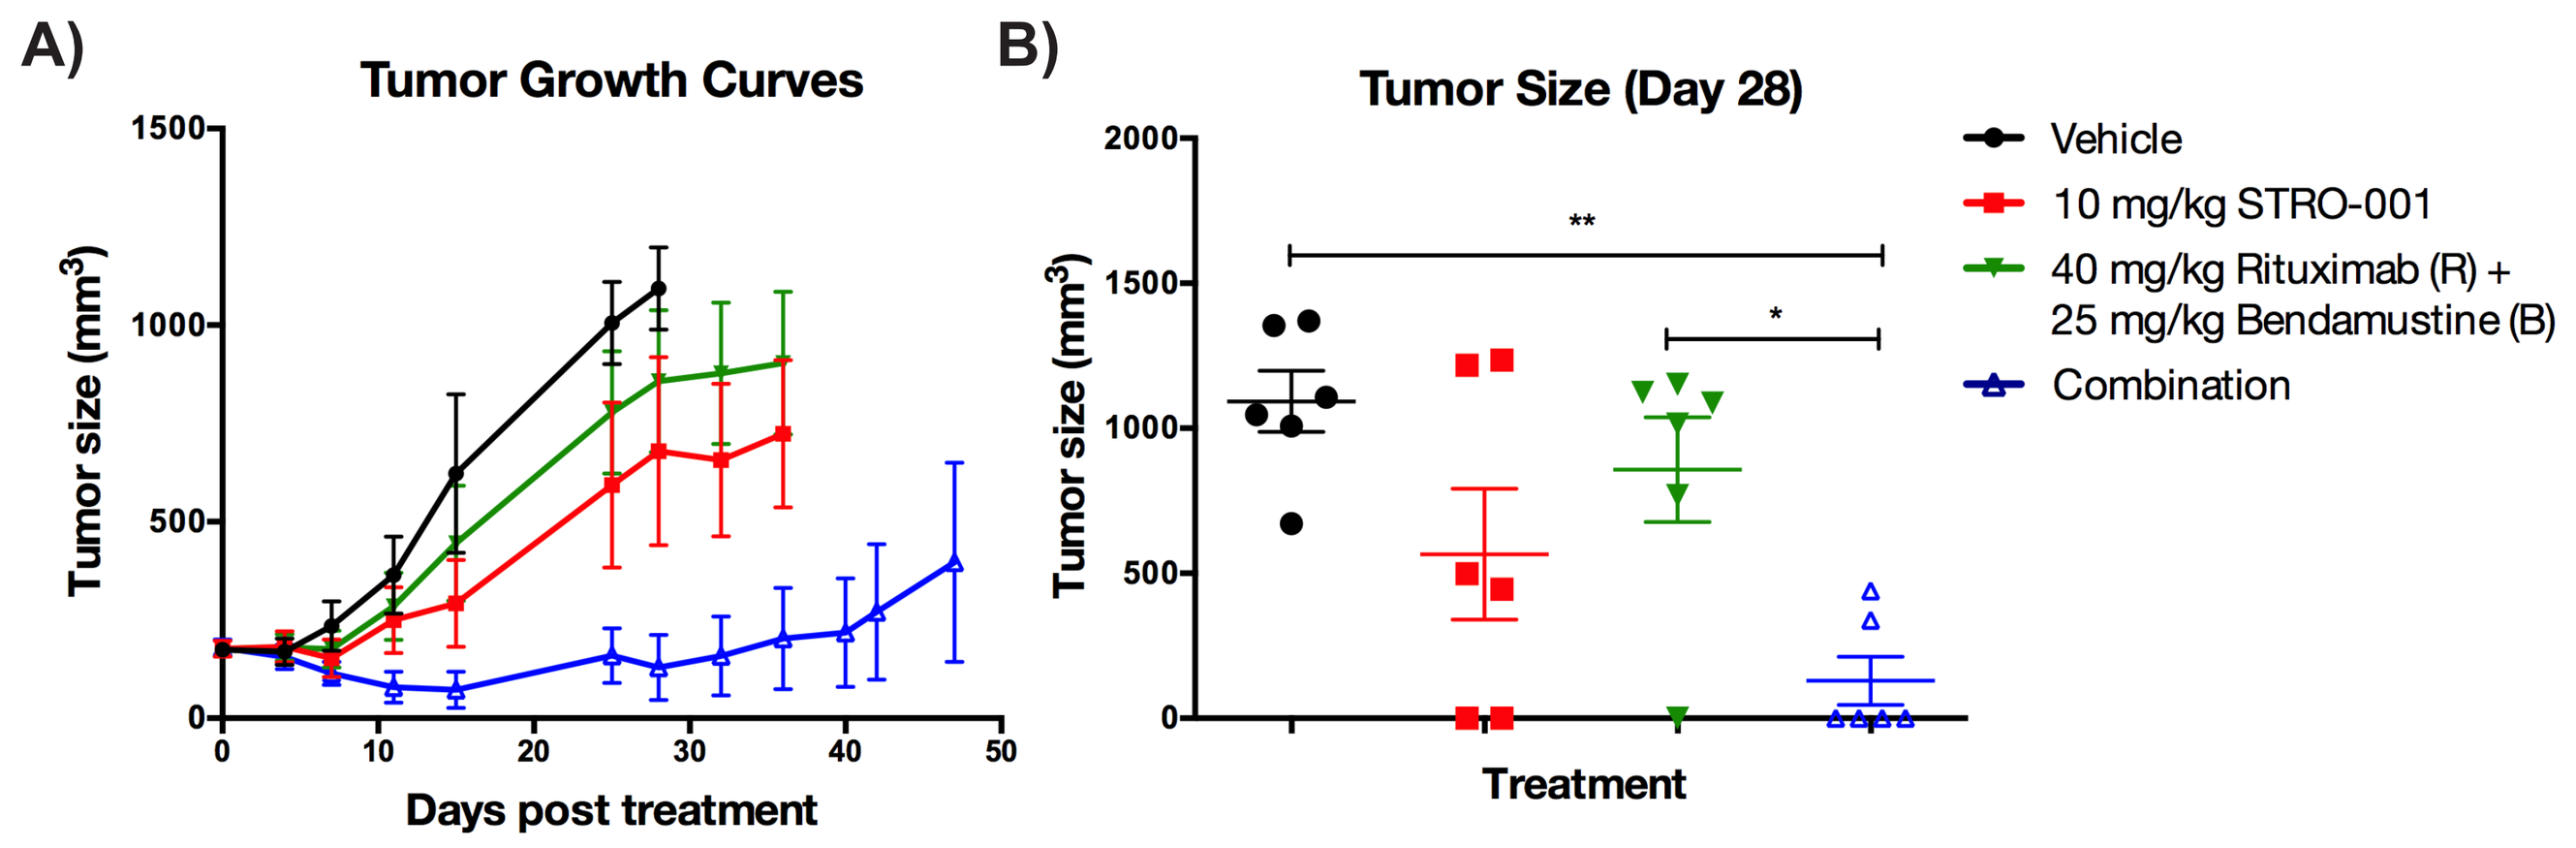
A)
Tumor Growth Curves
Tumor size (mm³)
Days post treatment
B)
Tumor Size (Day 28)
Treatment
Vehicle
10 mg/kg STRO-001
40 mg/kg Rituximab (R) +
25 mg/kg Bendamustine (B)
Combination

A) SU-DHL-6 tumor growth curves in response to indicated treatment. **B)** Scatter plot of individual tumor size on day 28 when mean of control tumors >1000 mm³. Statistical analysis was performed using one-way ANOVA with Tukey's multiple comparison test.

Figure 8. STRO-001 significantly inhibits tumor growth in Jeko-1 mantle cell lymphoma xenograft model

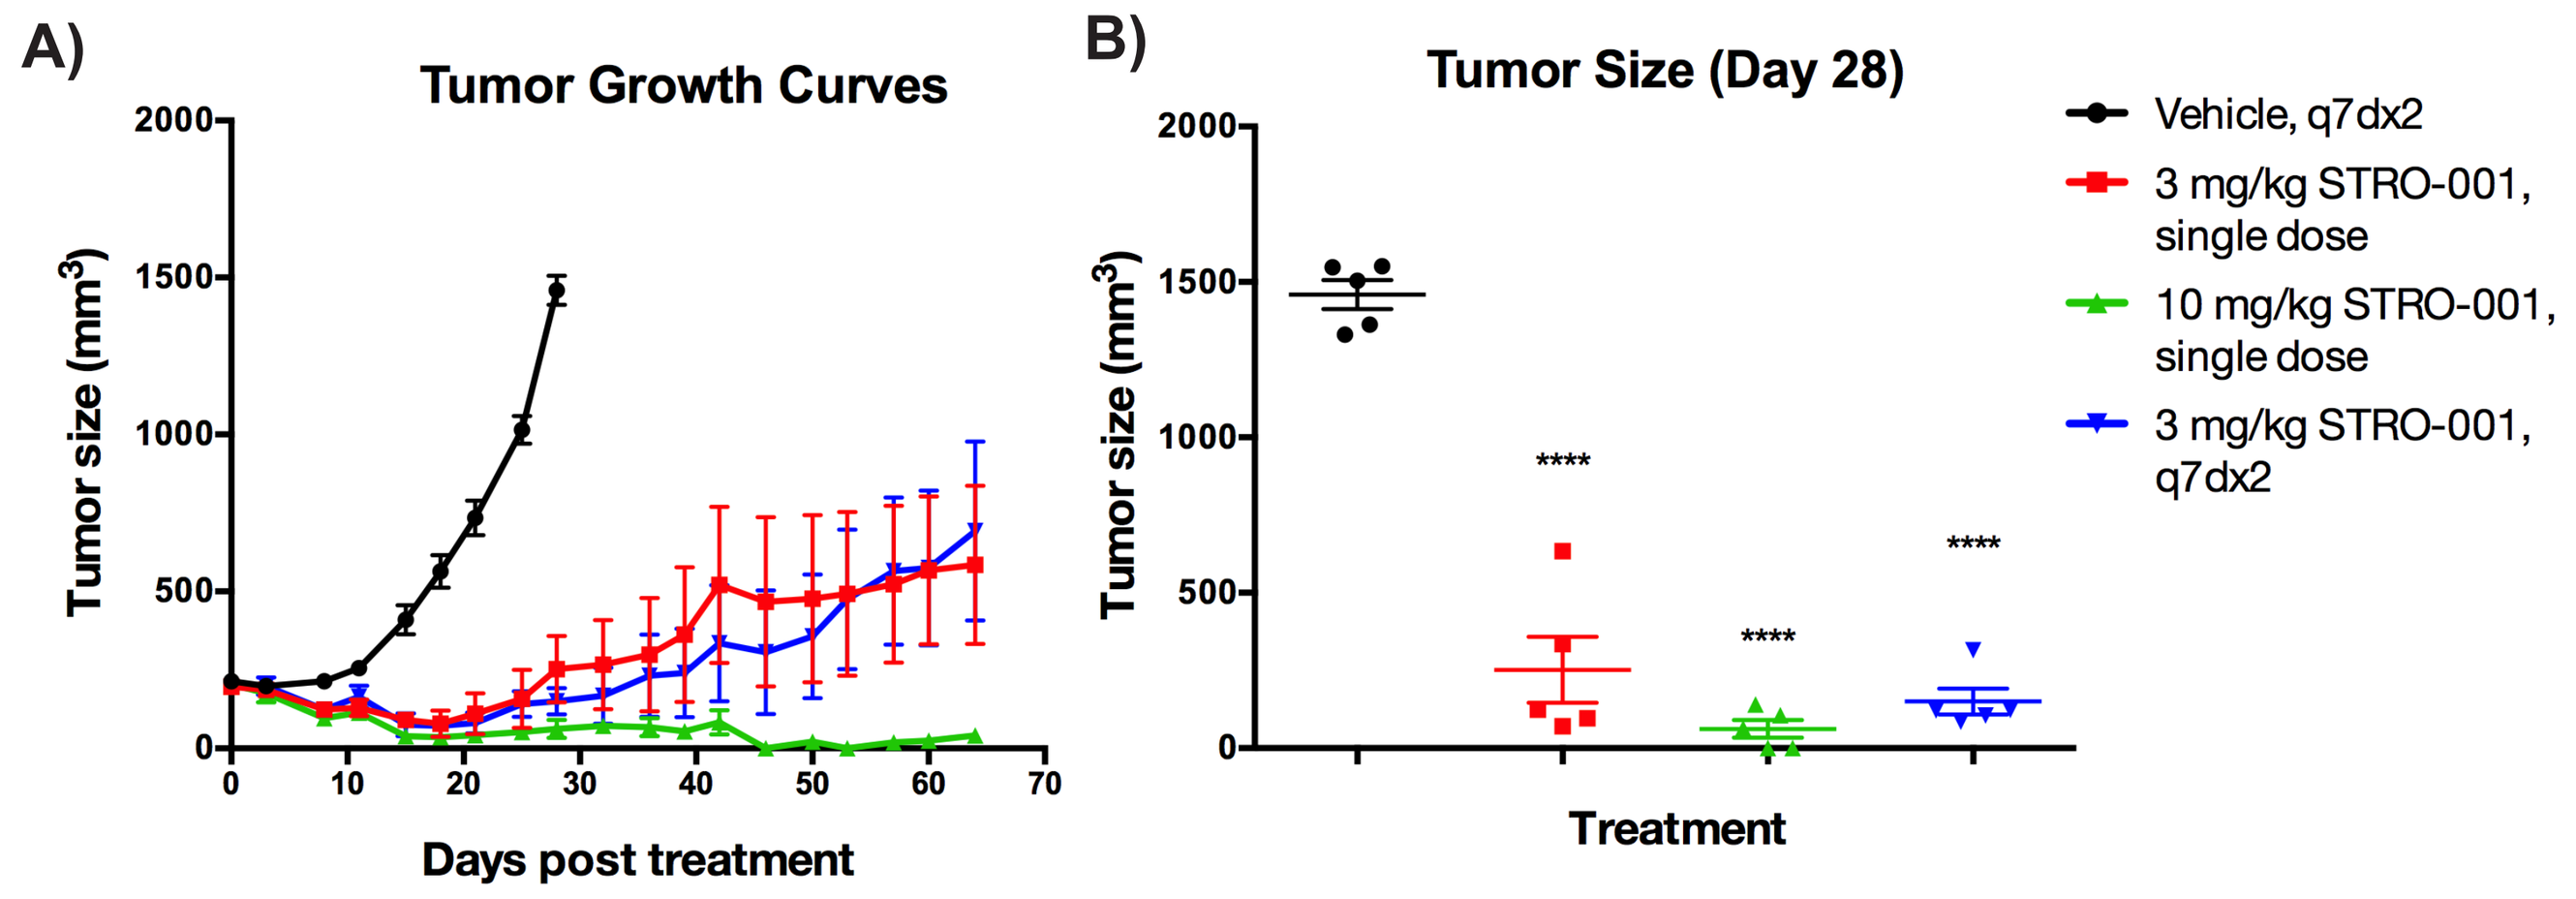
A)
Tumor Growth Curves
Tumor size (mm³)
Days post treatment
B)
Tumor Size (Day 28)
Treatment
Vehicle, q7dx2
3 mg/kg STRO-001, single dose
10 mg/kg STRO-001, single dose
3 mg/kg STRO-001, q7dx2

A) Jeko-1 tumor growth curves in response to STRO-001 treatment. **B)** Scatter plot of individual tumor size on day 28 when mean of control tumors ~1500 mm³. Statistical analysis was performed using one-way ANOVA with Dunnett's multiple comparison test.

Figure 9. STRO-001 induces dose-responsive ablation of B-cells in cynomolgus monkeys

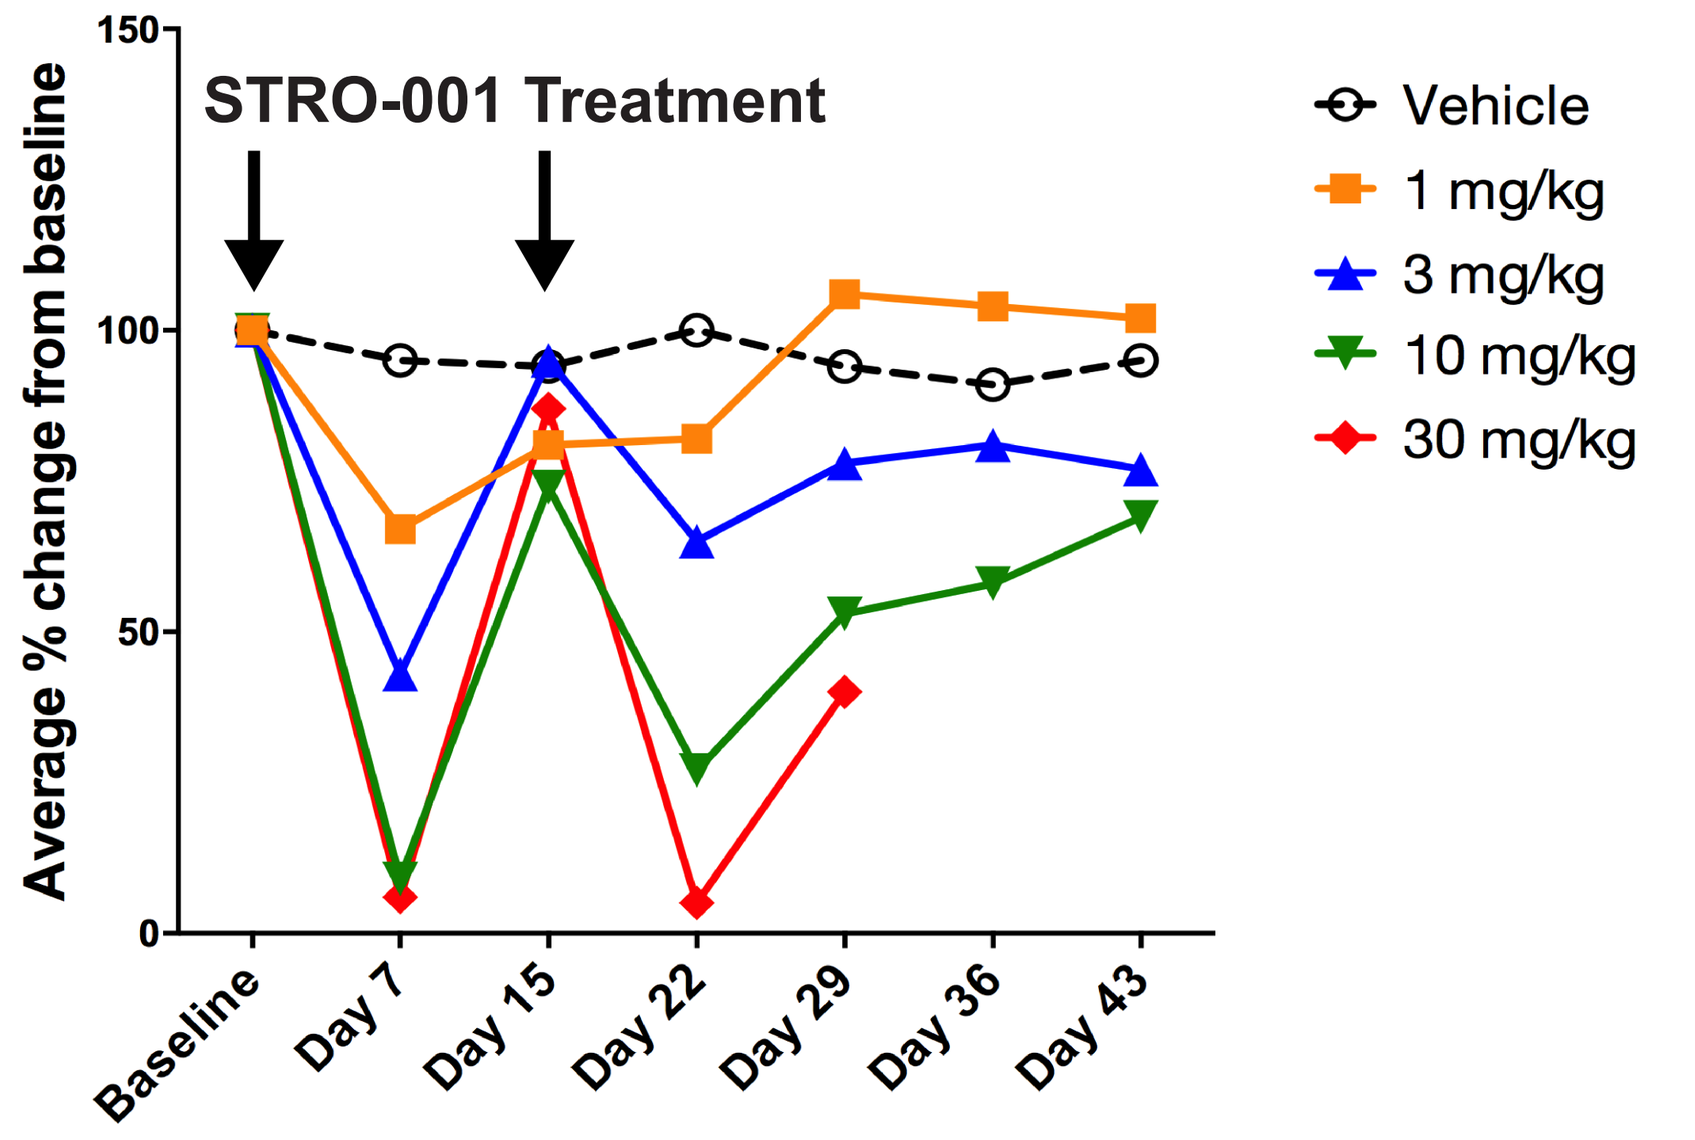
STRO-001 Treatment
Average % change from baseline
Baseline
Day 7
Day 15
Day 22
Day 29
Day 36
Day 43
Vehicle
1 mg/kg
3 mg/kg
10 mg/kg
30 mg/kg

B-cells were quantitated using flow cytometry. The graph above normalizes the relative B-cell numbers for each animal at each time point to the pre-dose B-cell numbers for the dose groups.

CONCLUSIONS

- Sutro's technology allows for the generation of novel, specific, and homogenous ADCs.
- STRO-001 targets CD74 and demonstrates potent *in vitro* cytotoxicity in MM and NHL cell lines.
- STRO-001 catabolites exhibit low cytotoxicity irrespective of CD74 status, suggesting a low likelihood of off-target toxicity and potential for improved therapeutic index.
- STRO-001 exhibits significant anti-tumor activity in MM (ARP-1 and MM.1S), NHL-DLBCL (SU-DHL-6) and NHL-MCL (Jeko-1) xenograft models *in vivo*.
- Exploratory toxicology study in cynomolgus monkeys did not produce any unexpected findings; treatment with STRO-001 resulted in B-cell depletion, the intended pharmacodynamic effect.
- IND enabling studies are underway and clinical trials of STRO-001 are planned.

MATERIALS & METHODS

Cell binding and determination of antibody binding capacity (ABC) on cell surface. Cells were blocked with Human Fc block and stained with 100nM of DBCO-Alexa647 conjugated to SP7219. Samples were analyzed on BD FACS CantoII system. Median Fluorescent Intensity (MFI) was calculated by FlowJo. Antibody binding capacity (ABC) on cell surface was determined by Quantum Simply Cellular anti-human IgG beads from Bangs Laboratories, Inc.

ADC cell killing assays. Cells were incubated with serial diluted free drugs or ADC samples for 72hrs. Cell viability was measured using Cell Titer-Glo® reagent. The relative luminescence measured by ENVISION® plate reader was converted to % viability using untreated cells as controls. Data was fitted using GraphPad Prism.

Mouse models. CB17 SCID or NSG mice were intravenously (IV) injected with ARP-1 or MM1.S cells via the tail vein or subcutaneously with SU-DHL-6 cells or Jeko-1 cells + matrigel. Randomization and treatment as indicated on figure legends was initiated when tumors were established. For disseminated models, the study endpoints was survival and tumor burden in the bone marrow (BM) was assessed by detection of human CD138+ cells by flow cytometry. Statistical tests are listed in figure legends. A probability of less than 5% ($p<0.05$) was considered as significant. All graphs are presented as mean or individual values ± SEM. Legend: ****, p<.0.0001; ***, p<0.001; **p<0.01, *p<0.05

Exploratory toxicology in cynomolgous monkey. An exploratory safety study was conducted in female cynomolgus monkeys, and animals were given iv doses of vehicle, 1, 3, 10 or 30 mg/kg on day 1 and day 15 followed by a 28-day observation period post-last dose. Animals were observed for clinical signs and evaluated for clinical pathology (hematology, coagulation and serum chemistry) and immunophenotyping, as well as analysis of pharmacokinetic properties of the molecule. Total lymphocyte populations were identified using a gating strategy consisting of CD45 fluorescent staining and side-scatter characteristics (SSC) demarcation (CD45brightSSCdim) to delineate lymphocyte populations. The relative values for CD3-CD20+ cells obtained from the flow cytometer were multiplied by the absolute lymphocyte count from the hematology analysis to enumerate absolute cell counts.

REFERENCES & DISCLAIMERS

1. Stein R, Goldenberg DM, et al.. CD74: A New Candidate Target For the Immunotherapy of B-Cell Neoplasms. Clin. Cancer Res. 2007.
2. Zimmerman, E.S. Heibeck, T.H. et al. Bioconjug Chem Feb 2014 25(2): 351-61.

Disclosures - All co-authors were employees of Sutro when this research was conducted.