STRO-002, an anti-FolRα ADC, Demonstrates Immune-Modulating Properties and Potentiates PD-L1 Blockade

Poster # 2250

Millicent Embry*, Sihong Zhou*, Christine Cheng, Janice Yu, Cristina Abrahams, Xiaofan Li, Jeff Hanson, Cuong Tran, Gang Yin, Shamim Ahmad, Krishna Bajjuri, Venita De Almeida, Mark Lupher, and Trevor Hallam. Sutro Biopharma, South San Francisco, CA, USA

* Contributed equally

SUTRO BIOPHARMA

Introduction

- Folate receptor alpha (FolRα) is a cell-surface protein overexpressed in ovarian and endometrial cancer, and an attractive target for therapeutic development.
- We have used a platform based on Sutro's proprietary XpressCF+™ cell-free expression system and site specific conjugation to generate STRO-002, a novel FolRα-targeting antibody drug conjugate (ADC).
- STRO-002 is composed of the anti-FolRα human IgG1 antibody, SP8166, conjugated to a novel proprietary cleavable drug-linker (SC239) at specific sites Y180 and F404 on the antibody heavy chain.
- The active metabolite of STRO-002, 3-aminophenyl hemiasterlin (SC209), is tubulin-targeting cytotoxic agent with bystander activity.
- Certain classes of cytotoxins have been shown to elicit immunogenic cell death (ICD) leading to T-cell recruitment into the tumor microenvironment and increased sensitivity to immunomodulatory agents.
- Here we investigate the ability of STRO-002 to induce ICD and evaluate efficacy of STRO-002 in combination with PD1/PD-L1 blockade.

Generation of STRO-002, A Homogenous FolRα-Targeting ADC Through Cell-Free Antibody Synthesis And Site-Specific Conjugation

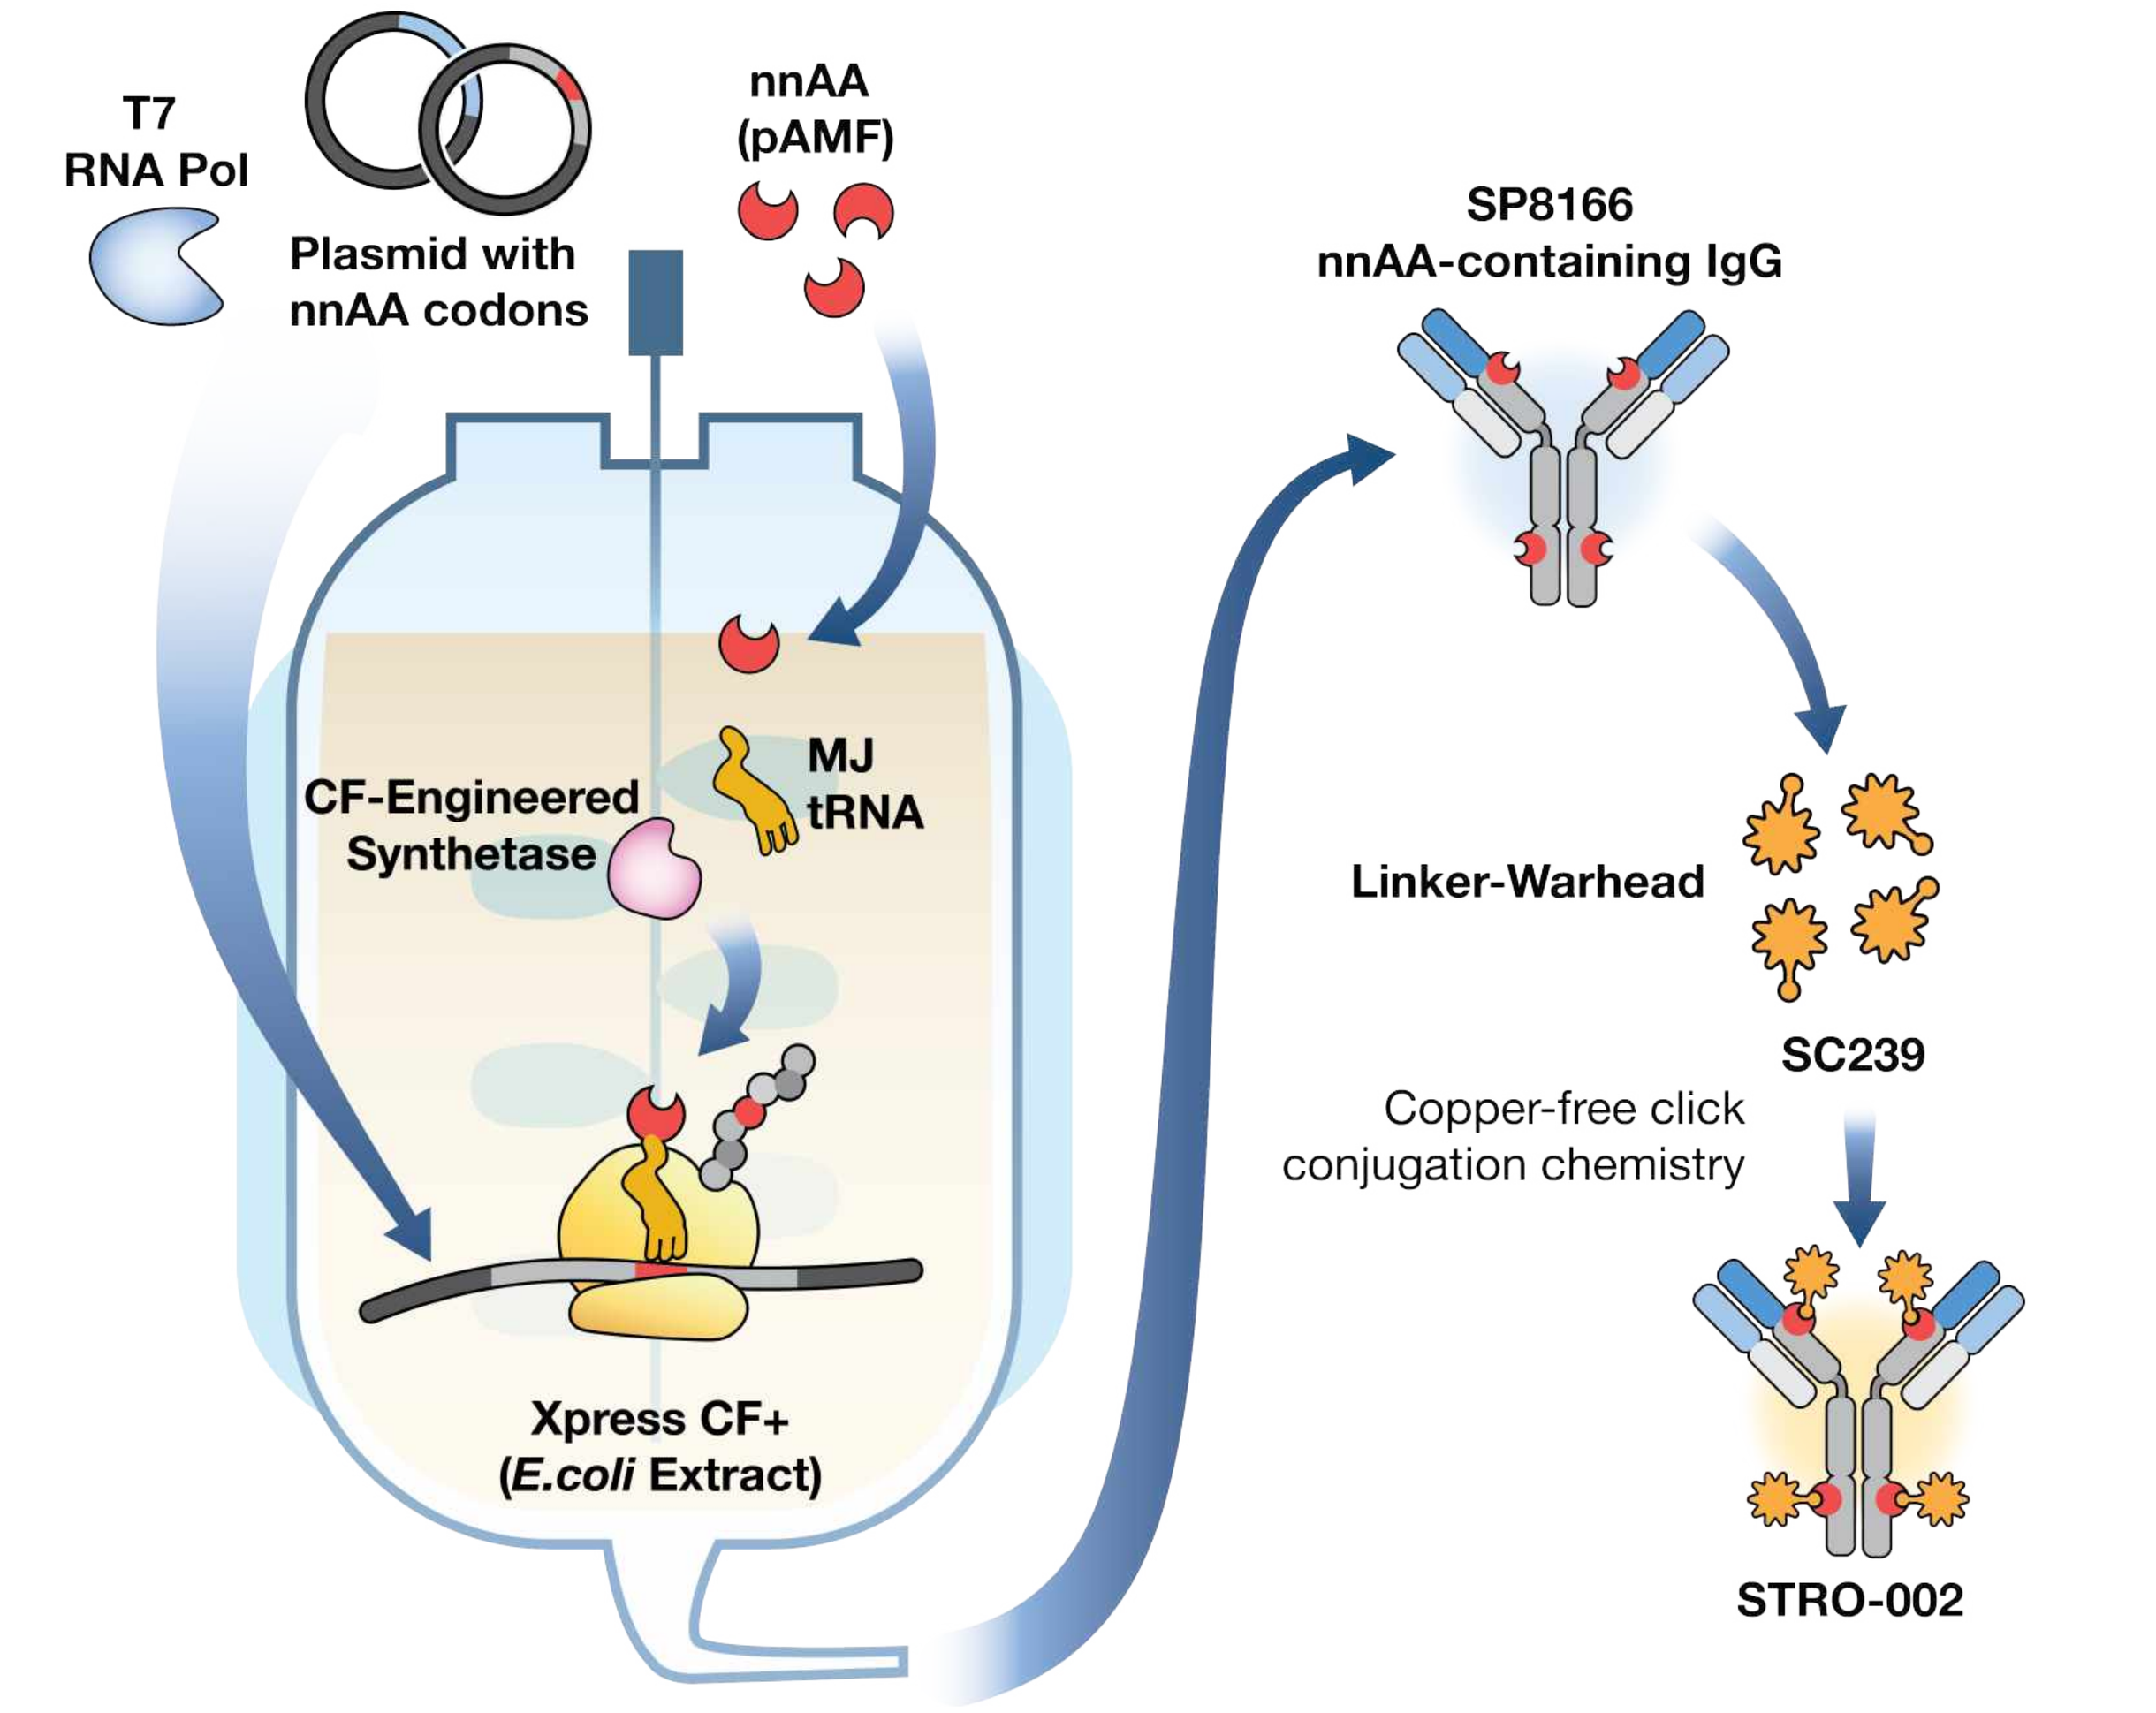
T7
RNA Pol
Plasmid with
nnAA codons
nnAA
(pAMF)
MJ
tRNA
CF-Engineered
Synthetase
Xpress CF+
(E.coli Extract)
SP8166
nnAA-containing IgG
Linker-Warhead
SC239
Copper-free click
conjugation chemistry
STRO-002

Generation of SP8166 and STRO-002 using Sutro's proprietary Xpress Cell-Free (XpressCF+TM) system. The non-natural amino acid (pAMF) was incorporated at two sites on the heavy chain of SP8166, with the sites selected based on conjugation efficiency, cell killing activity and in vivo activity in mice. SP8166 was conjugated at pAMF to the cleavable 3-aminophenyl hemiasterlin drug-linker SC239 to generate STRO-002.

In vivo methods

Human KB, human Igrov-1, or murine MC38-hFolRα cells were implanted subcutaneously into the right hind flank of athymic nude, SCID Beige, or C57BL/6 mice, respectively. Treatment with indicated test articles was initiated when tumors were established. Clinical grade Avelumab was used for in vivo studies. For quantitation of CD8⁺ T cells and evaluation of PD-1 expression, tumors were harvested at day 7 or day 5 post-treatment respectively. Statistical analysis was performed for TGI and quantitation of CD8+ cells using Prism using one-way ANOVA with Dunnett's or Tukey's multiple comparisons test, respectively. A probability of less than 5% ($p<0.05$) was considered as significant. Legend: ****, p<.0.0001; ***, p<0.001; **, p<0.01.

STRO-002 Exhibits Significant Cytotoxic Activity in FolRα Expressing Cells in vitro

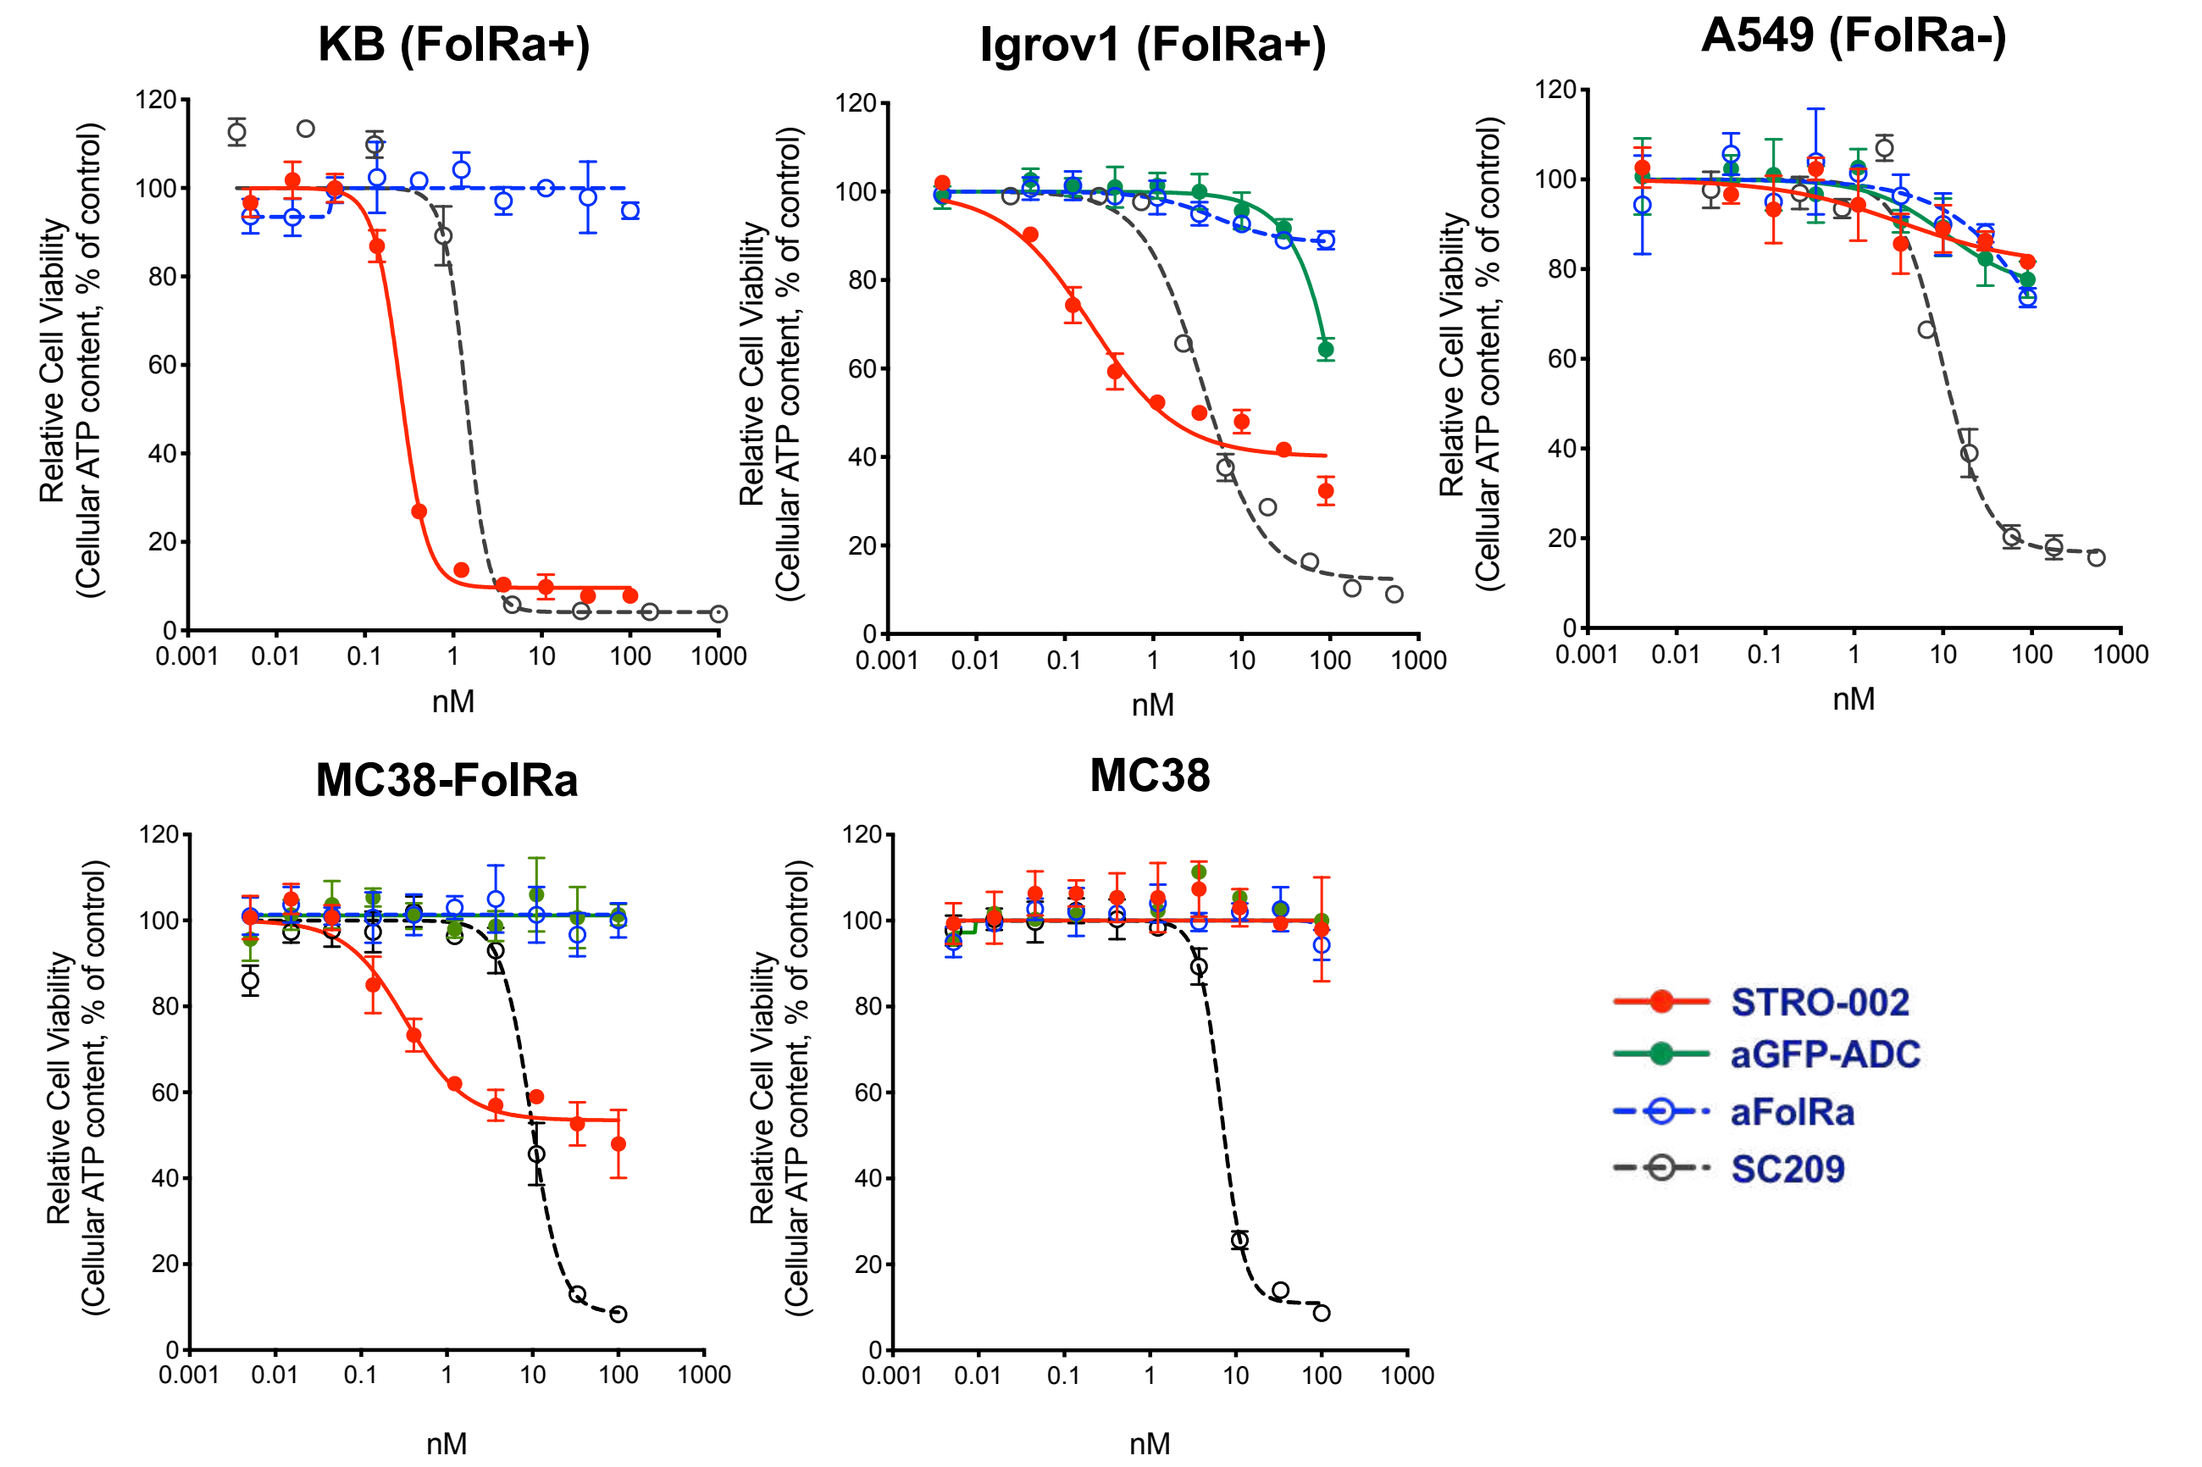
KB (FolRa+)
Igrov1 (FolRa+)
A549 (FolRa-)
MC38-FolRa
MC38
Relative Cell Viability
(Cellular ATP content, % of control)
nM
STRO-002
aGFP-ADC
aFolRa
SC209

- Cytotoxicity was assessed in vitro in a 5-day cell proliferation assay. Cell viability was measured using Cell Titer-Glo® reagent and normalized using untreated cells as control. Data was fitted using GraphPad Prism.
- STRO-002 showed target dependent cell killing activity on human FolRα positive KB and Igrov1 cells and murine MC38 cells engineered to express hFolRα (MC38-hFolRα).
- SC209, the active metabolite of STRO-002, showed cytotoxic effects on all cell lines irrespective of FolRα expression.

STRO-002 Exhibits Significant Anti-Tumor Activity in FolRα Expressing Models

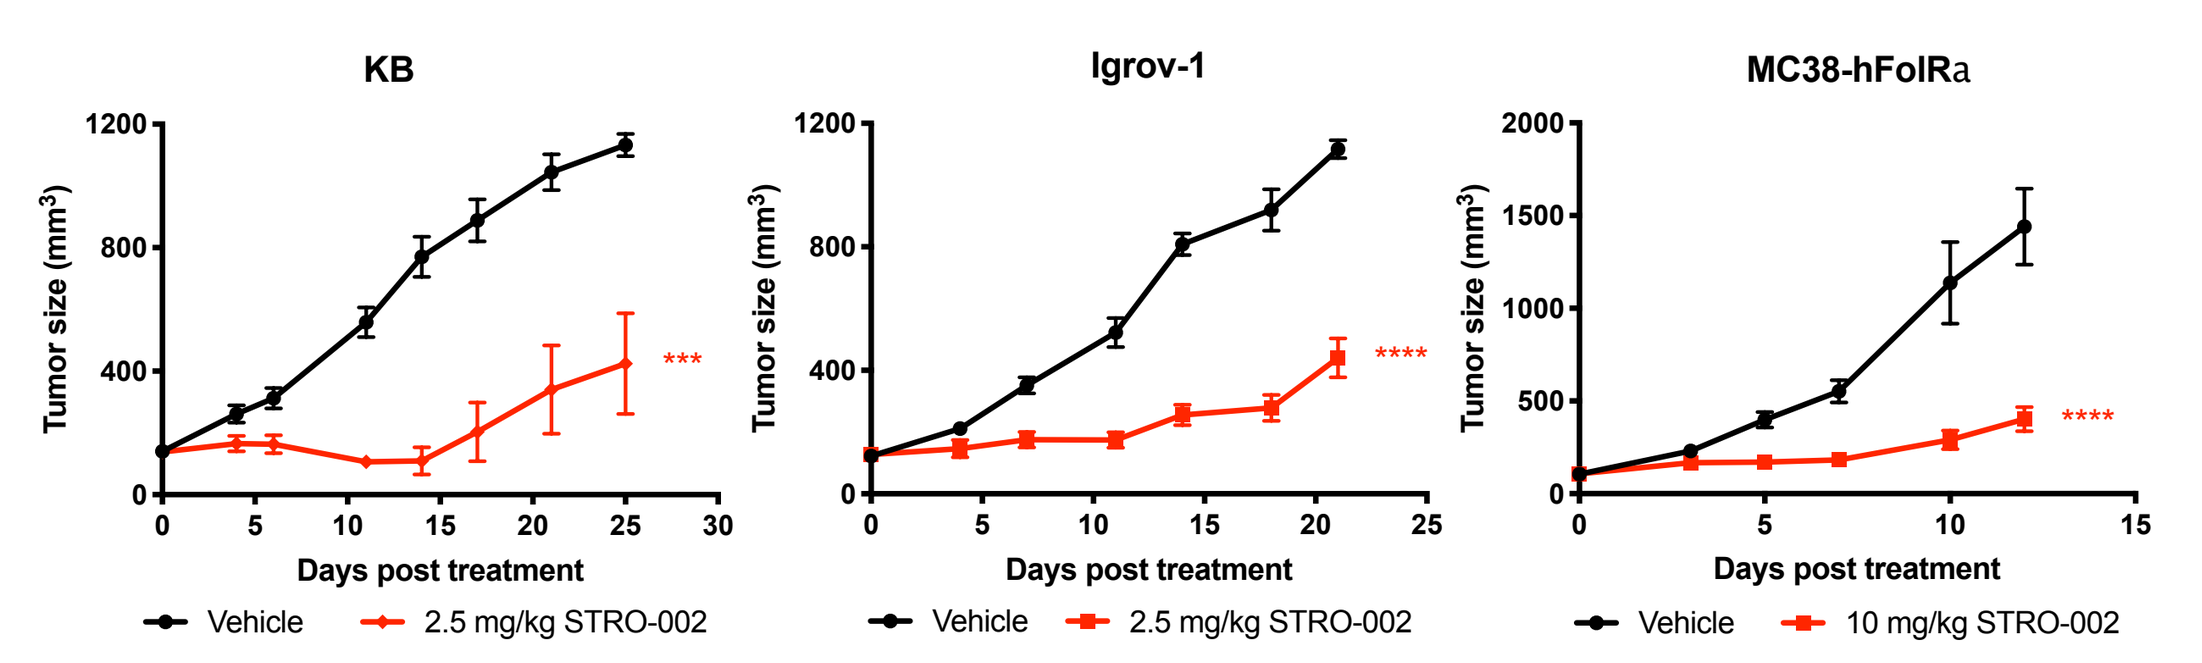
KB
Igrov-1
MC38-hFolRa
Tumor size (mm³)
Days post treatment
Vehicle
2.5 mg/kg STRO-002
10 mg/kg STRO-002

- Response to treatment with a single dose of STRO-002 or vehicle. Data is depicted as mean +/- SEM.
- STRO-002 significantly delayed tumor growth in human tumor models, KB and Igrov-1, with endogenous hFolRα expression as well as in the engineered murine MC38-hFolRα tumors.

STRO-002 Induces Hallmarks of Immunogenic Cell Death in FolRα Expressing Tumor Cells

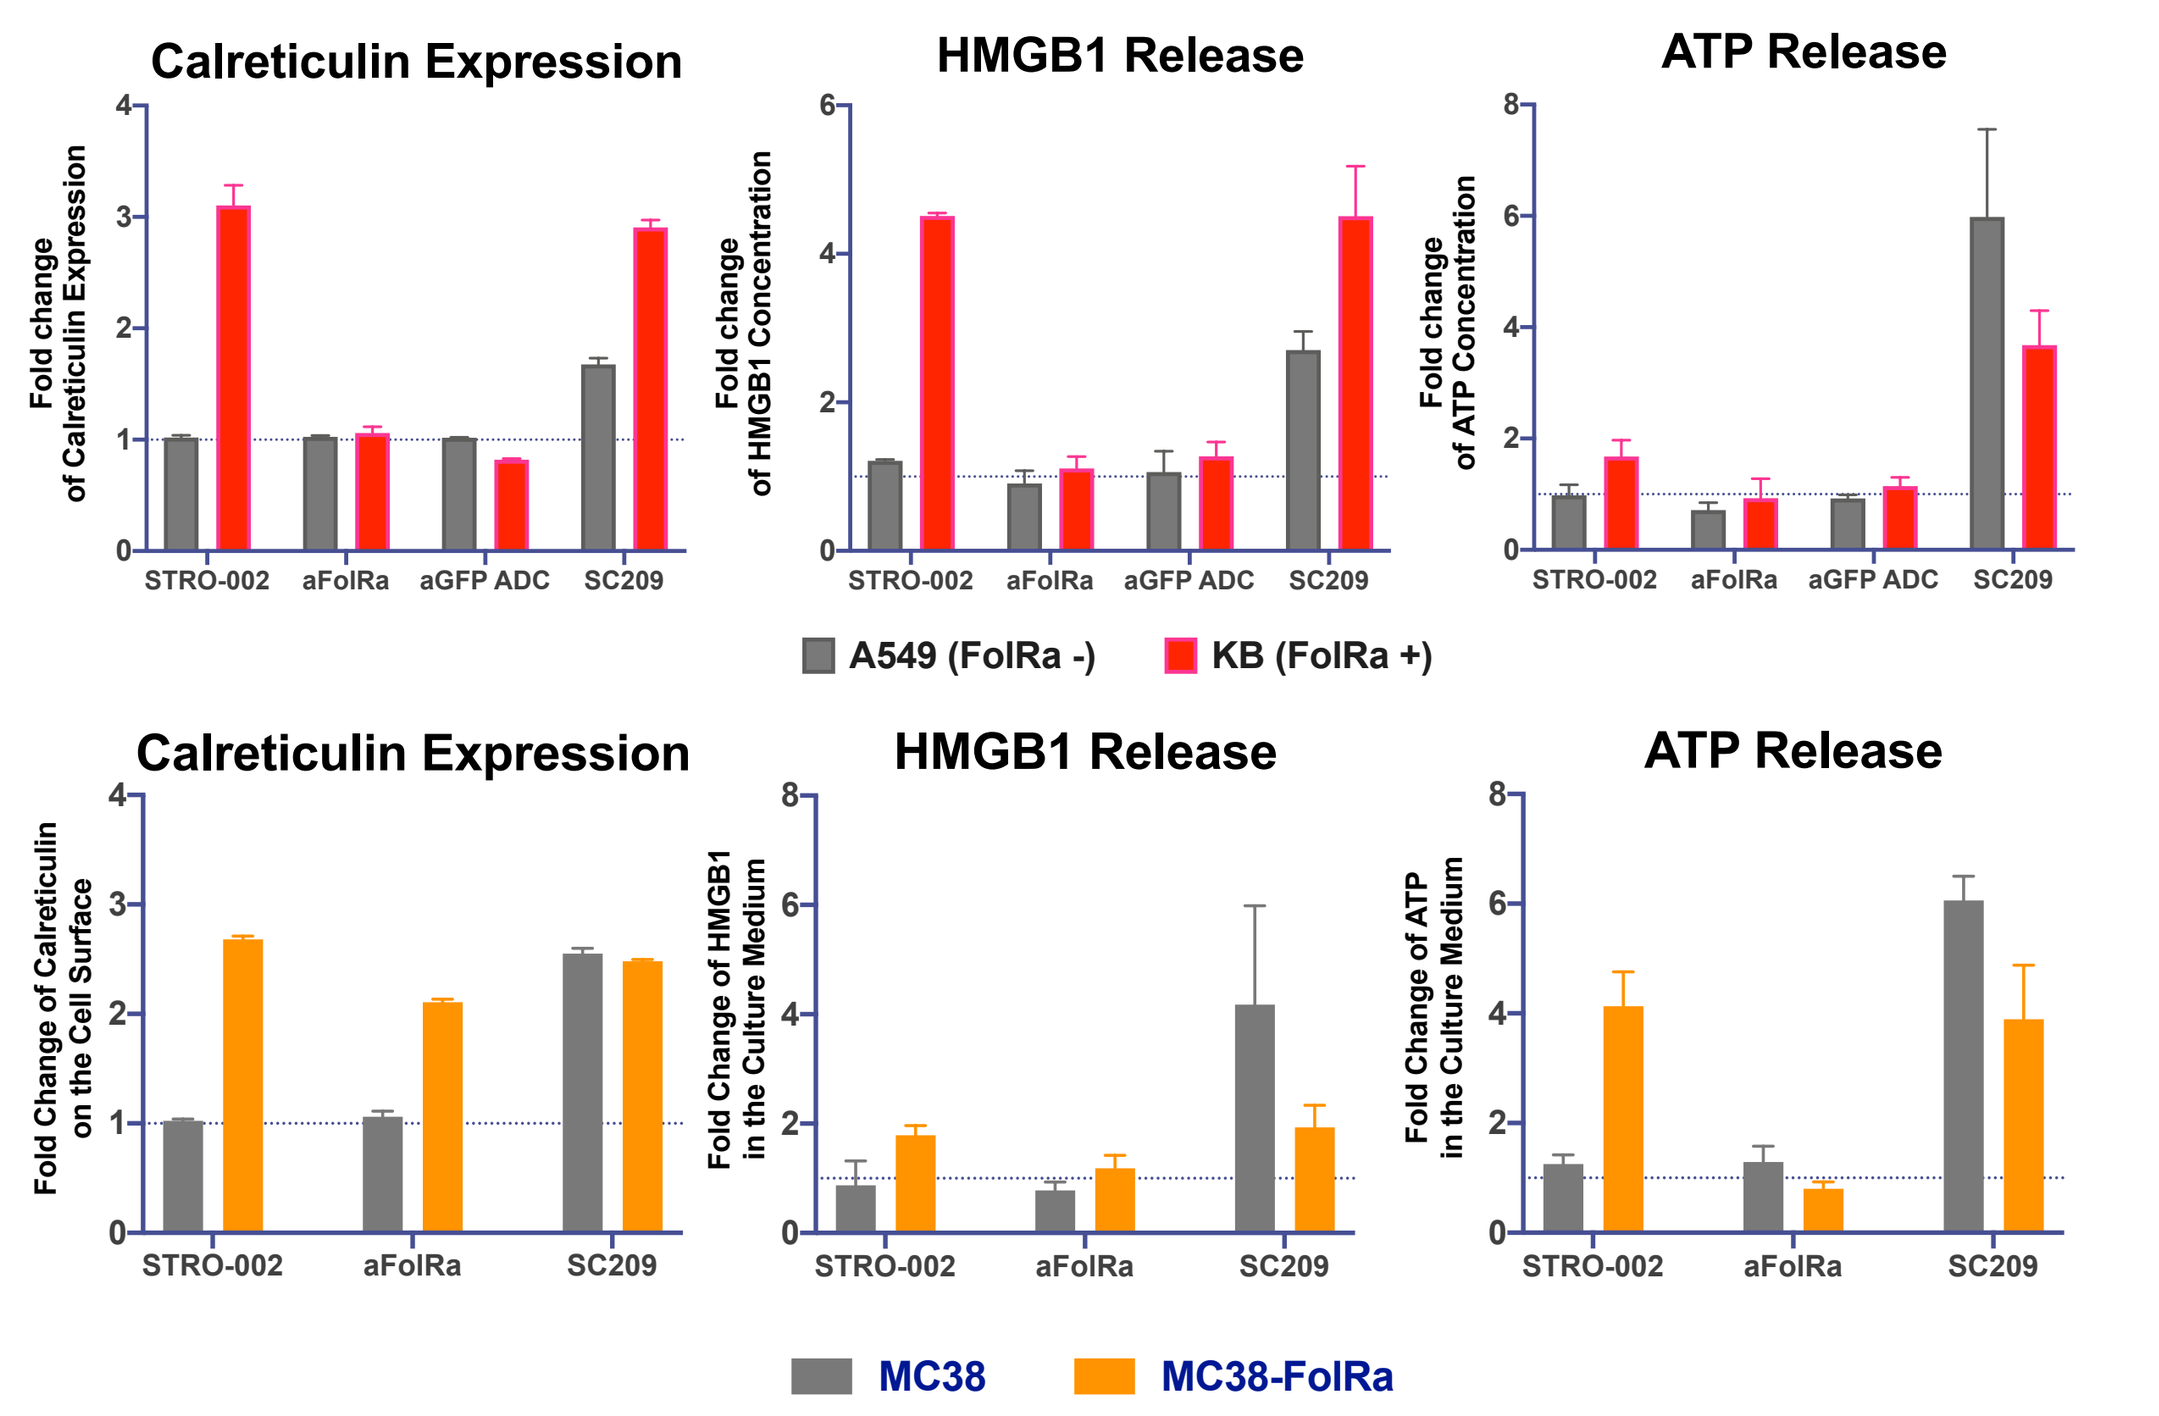
Calreticulin Expression
HMGB1 Release
ATP Release
Fold change of Calreticulin Expression
Fold change of HMGB1 Concentration
Fold change of ATP Concentration
STRO-002
aFolRa
aGFP ADC
SC209
A549 (FolRa -)
KB (FolRa +)
Fold Change of Calreticulin on the Cell Surface
Fold Change of HMGB1 in the Culture Medium
Fold Change of ATP in the Culture Medium
MC38
MC38-FolRa

- FolRα positive and negative cells were treated for 2 days to evaluate ICD. Calreticulin was measured by FACS using a fluorescent labeled anti-calreticulin antibody, HMGB1 measured by ELISA and ATP release measured by a chemiluminescence based assay.
- STRO-002 induced ICD on FolRα positive cells, while the active metabolite of STRO-002, SC209, induced ICD markers on both FolRα positive and negative cells. Unconjugated aFolRα antibody and an aGFP-SC209 conjugate were used as controls. ICD was indicated by translocation of calreticulin on the cell surface as well as release of HMGB1 and ATP into the cell culture medium.

STRO-002 Activates Monocytes When Co-Cultured with FolRα Expressing Tumor Cells

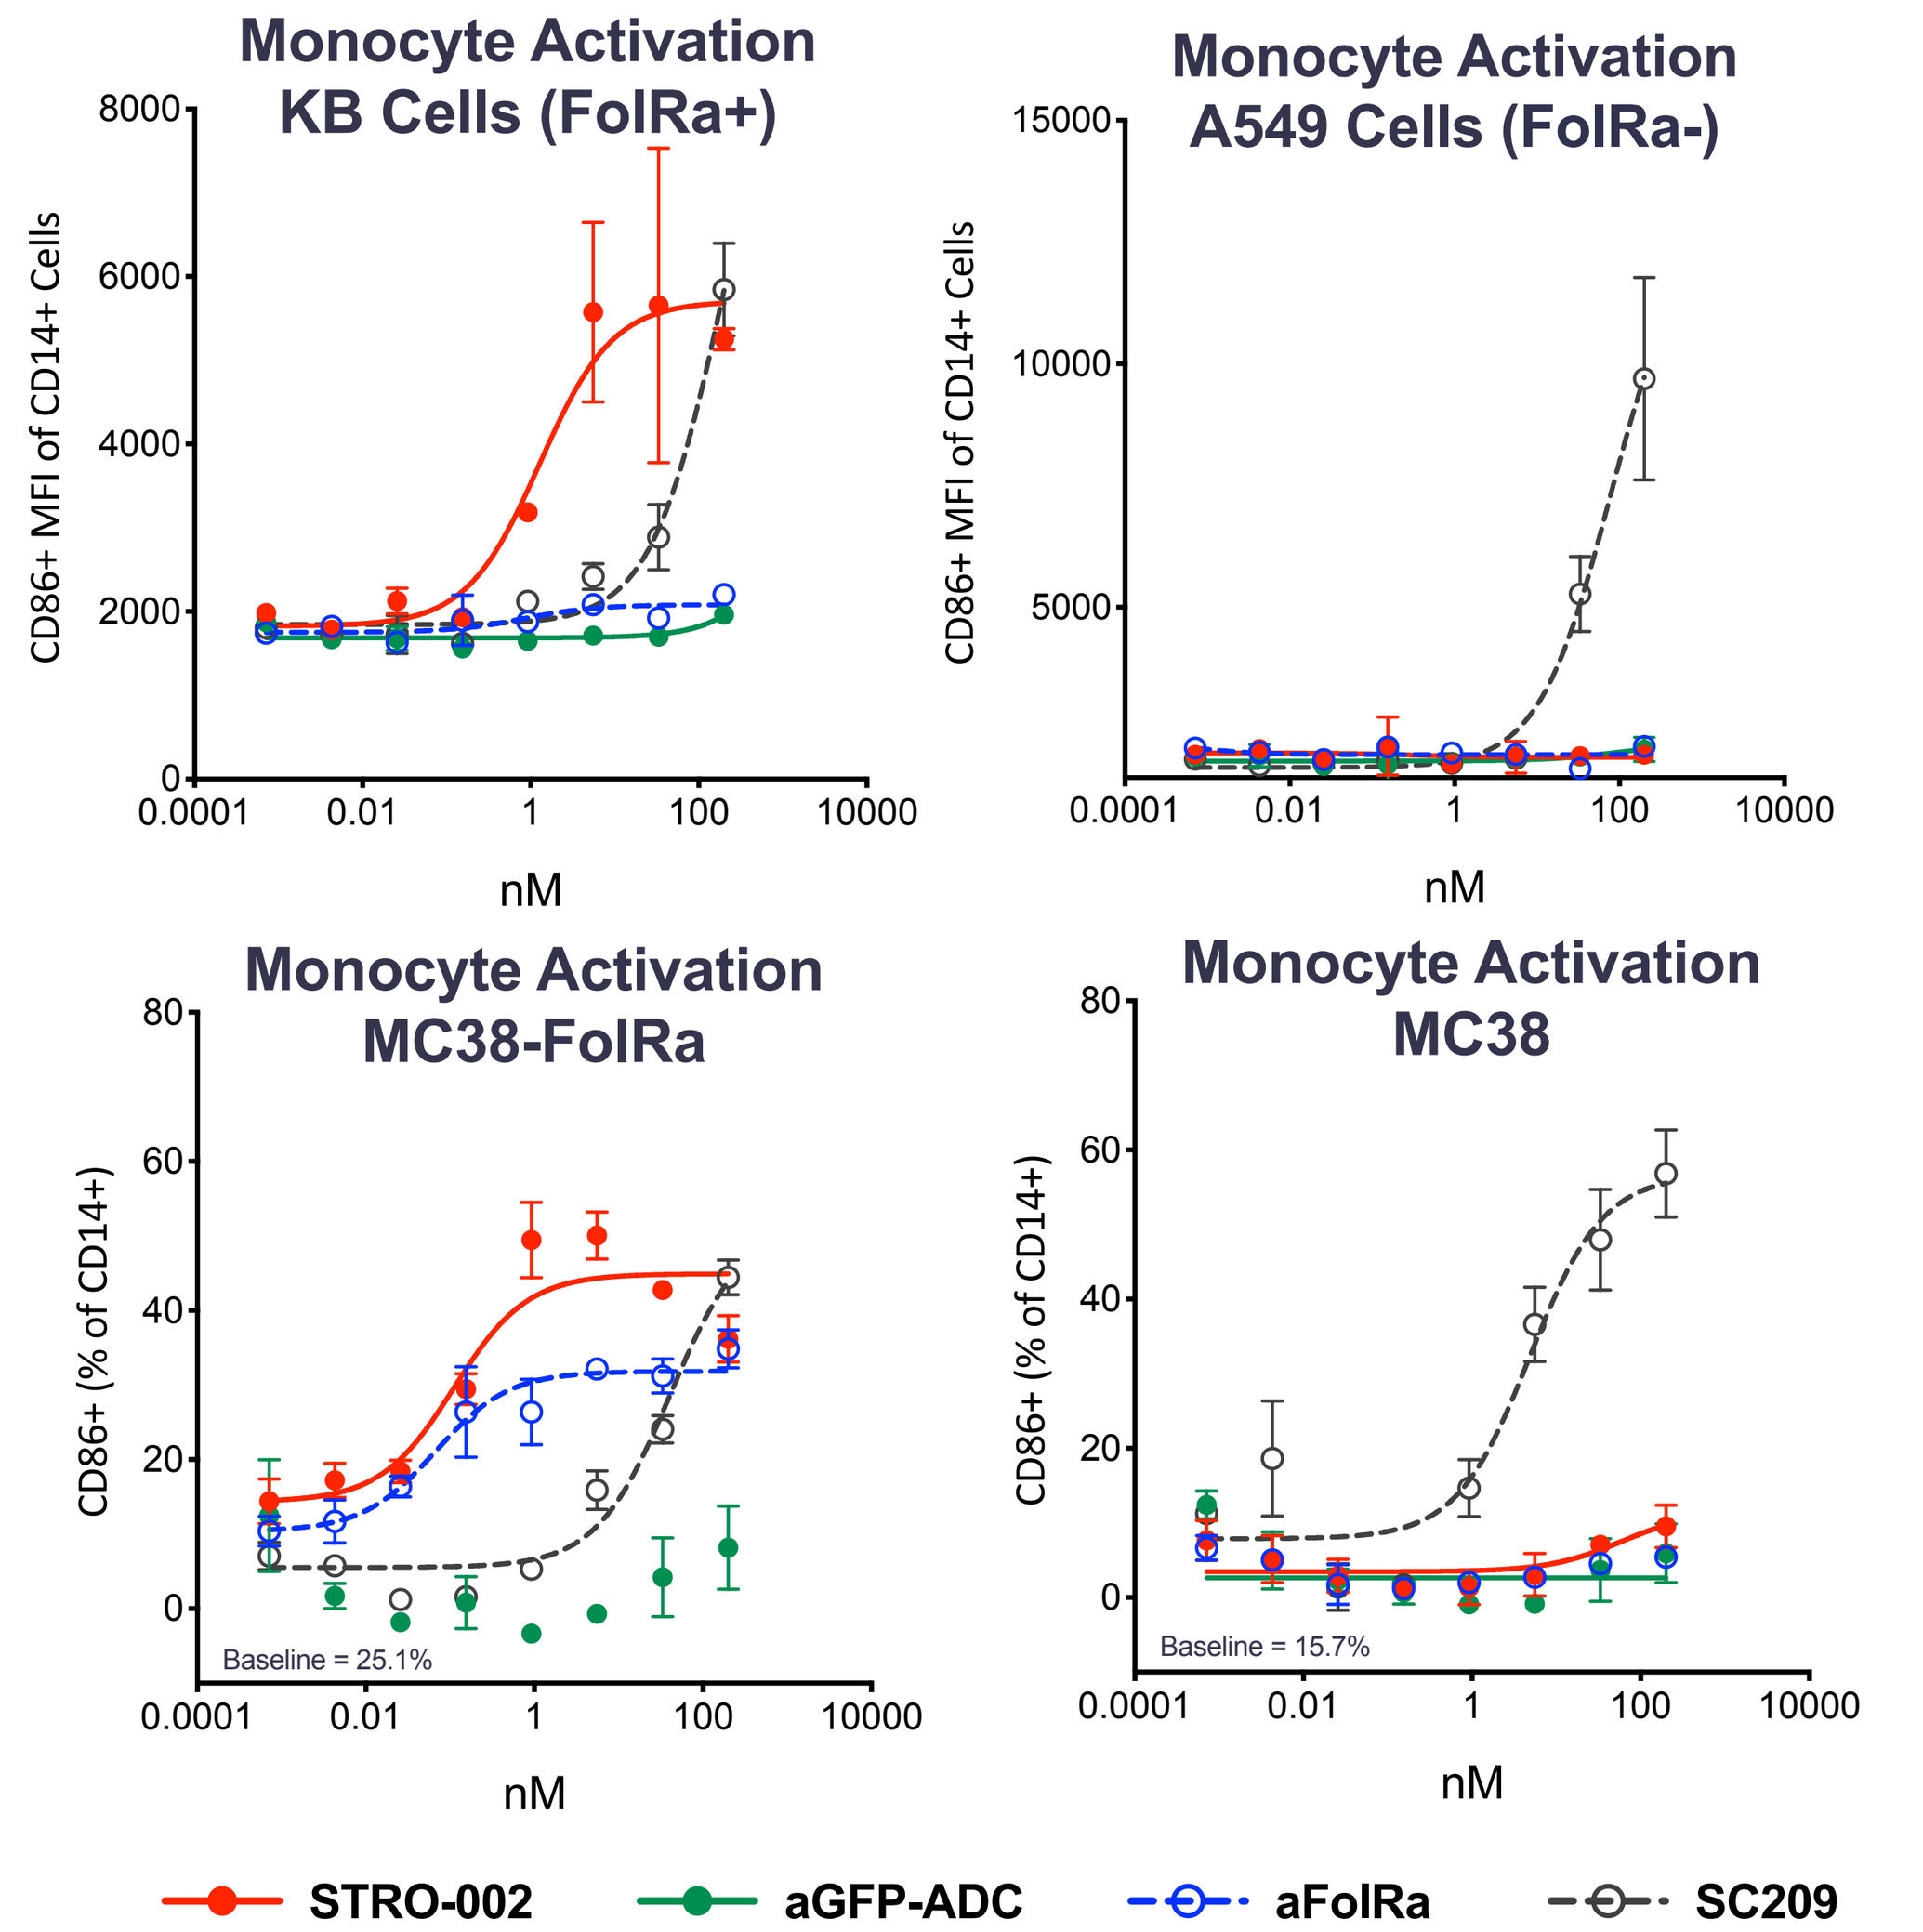
Monocyte Activation
KB Cells (FolRa+)
Monocyte Activation
A549 Cells (FolRa-)
Monocyte Activation
MC38-FolRa
Monocyte Activation
MC38
CD86+ MFI of CD14+ Cells
CD86+ (% of CD14+)
nM
Baseline = 25.1%
Baseline = 15.7%
STRO-002
aGFP-ADC
aFolRa
SC209

- FolRα positive and negative cells were co-cultured with human peripheral blood mononuclear cells (PBMCs) and treated for 2 days. Monocyte activation was indicated by increase of CD86 expression on CD14⁺ cells. The unconjugated aFolRα antibody and an aGFP-SC209 conjugate were used as controls.
- STRO-002 induced ICD on the FolRα positive cells, KB and MC38-hFolRα, which resulted in monocyte activation when the FolRα positive cells were co-cultured with human PBMCs.
- Consistent with its ability to induce ICD, SC209 activated monocytes in all cell lines irrespective of FolRα expression.

Therapeutic Synergy and Durable Anti-tumor Immunity Observed With a Single Dose of STRO-002 in Combination with Avelumab

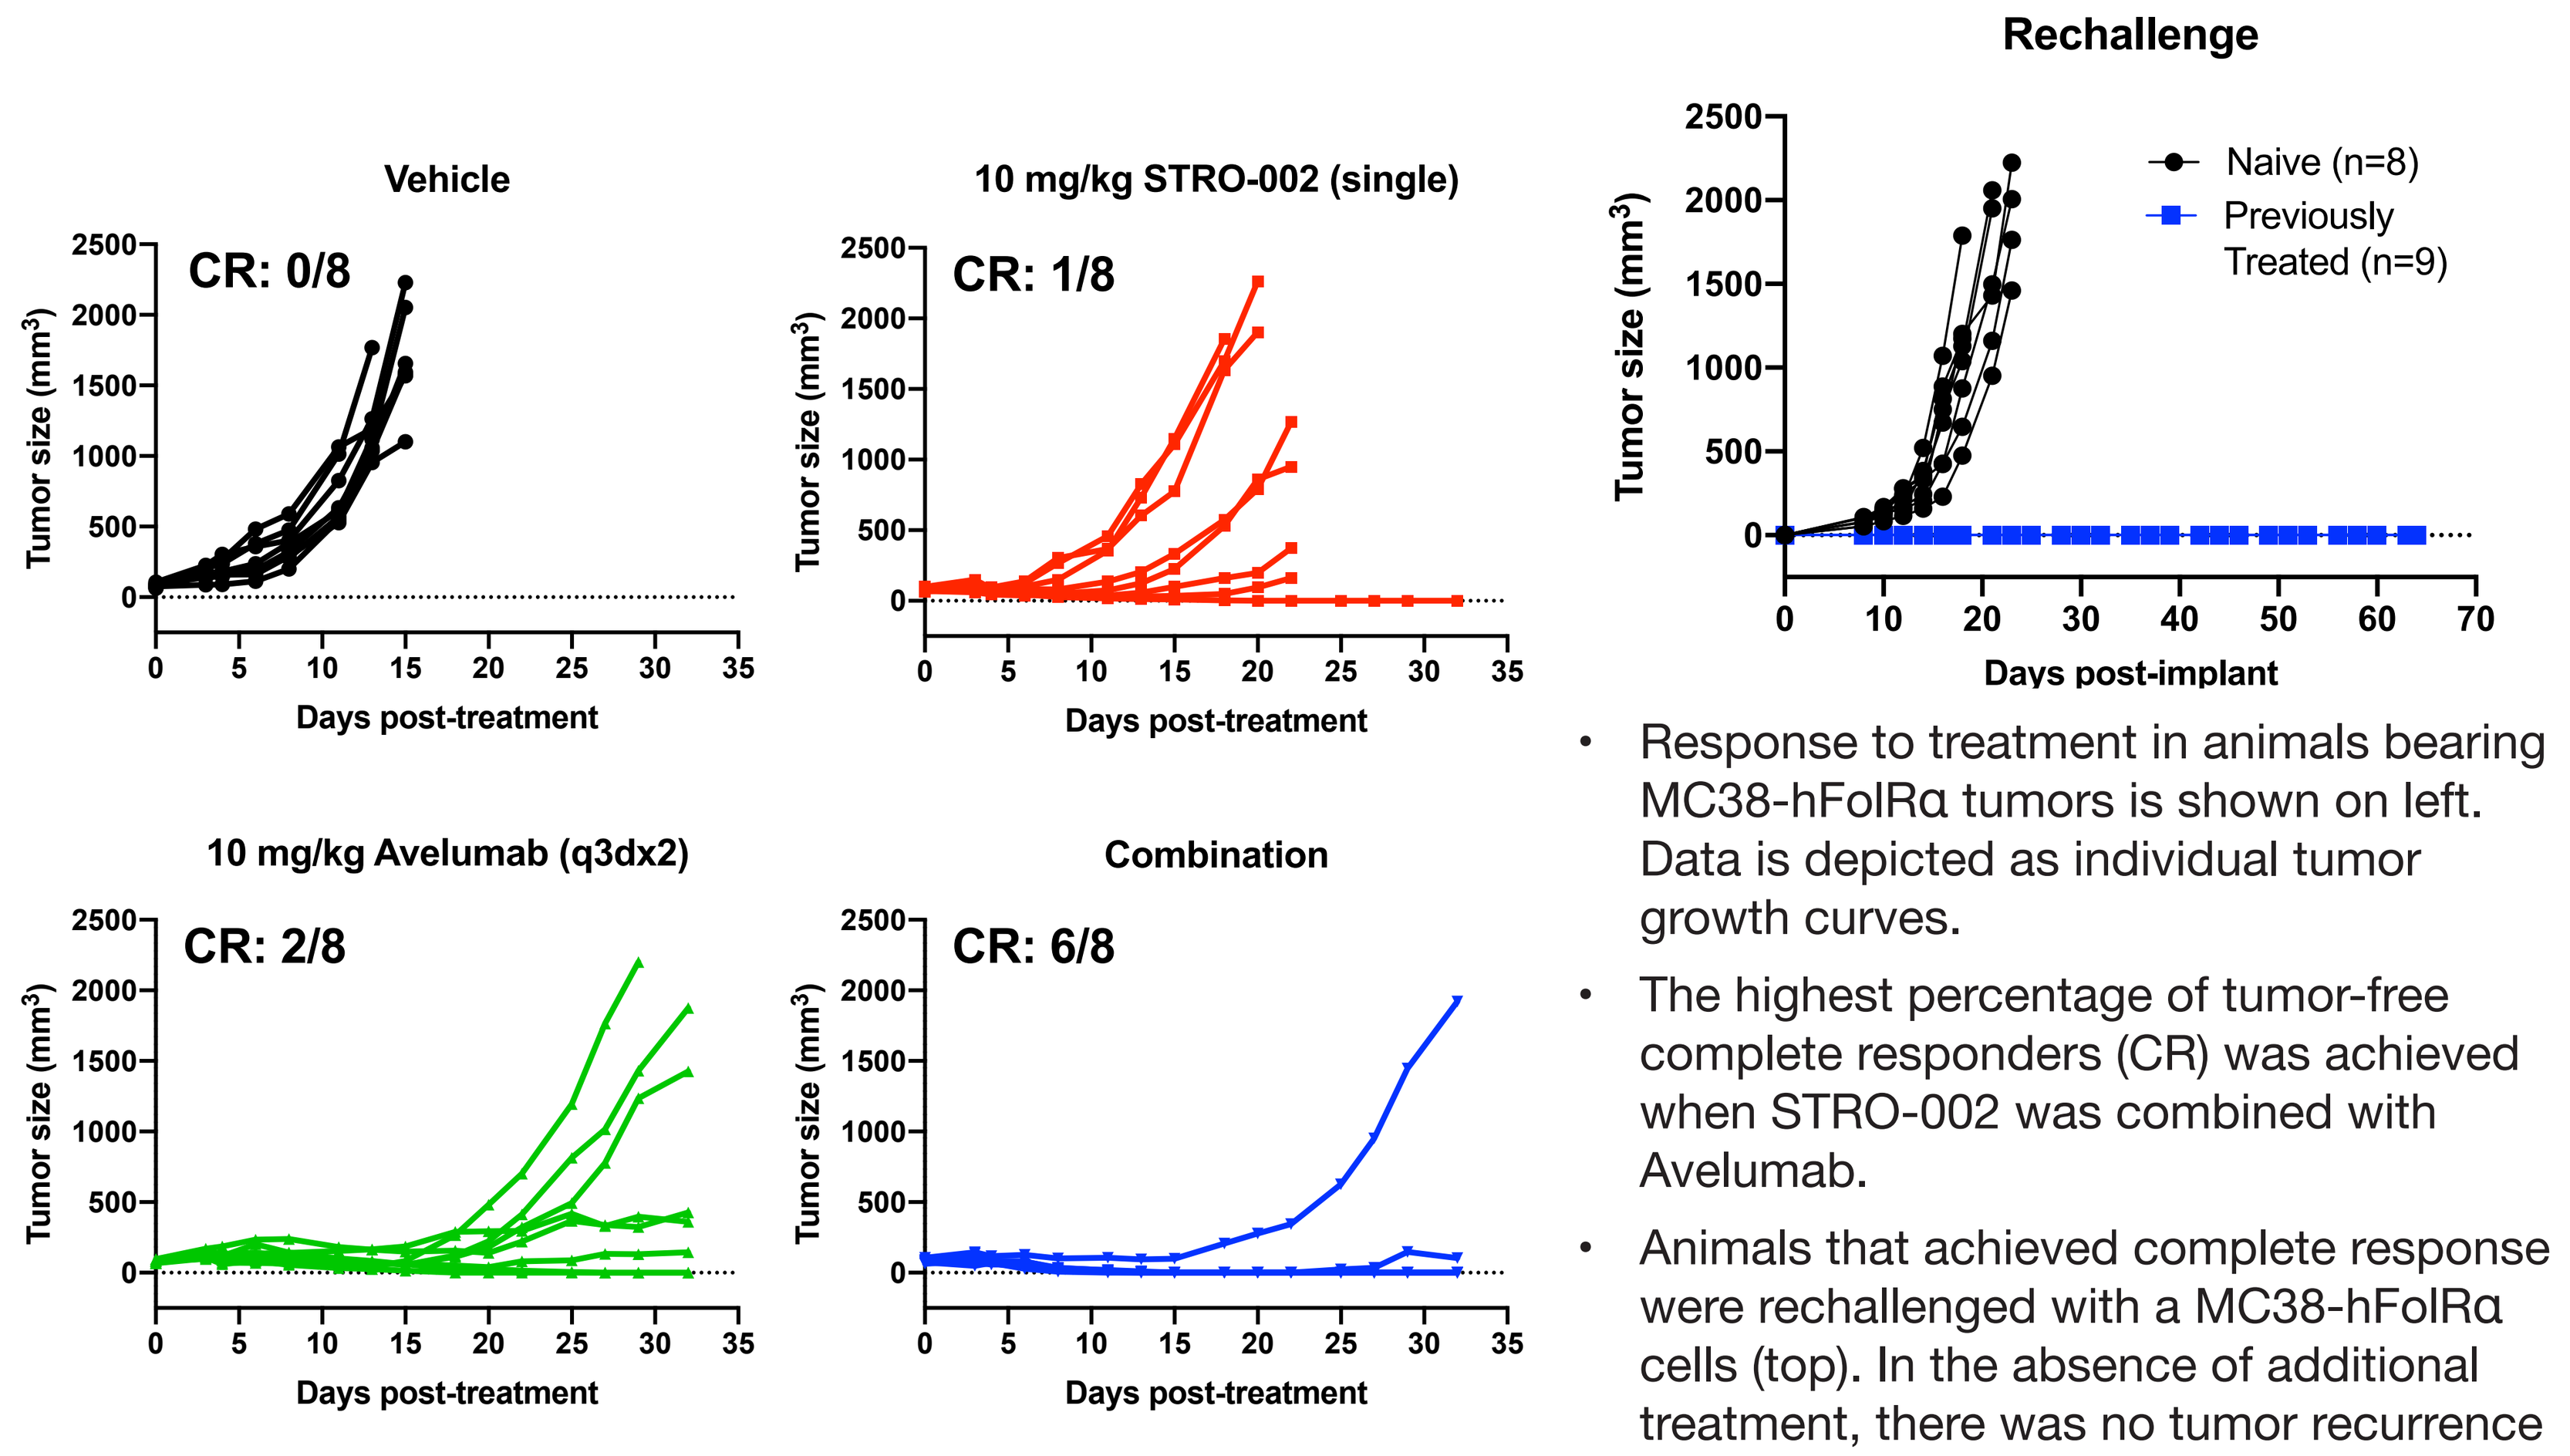
Vehicle
CR: 0/8
10 mg/kg STRO-002 (single)
CR: 1/8
10 mg/kg Avelumab (q3dx2)
CR: 2/8
Combination
CR: 6/8
Tumor size (mm³)
Days post-treatment
Rechallenge
Naive (n=8)
Previously
Treated (n=9)
Days post-implant

- Response to treatment in animals bearing MC38-hFolRα tumors is shown on left. Data is depicted as individual tumor growth curves.
- The highest percentage of tumor-free complete responders (CR) was achieved when STRO-002 was combined with Avelumab.
- Animals that achieved complete response were rechallenged with a MC38-hFolRα cells (top). In the absence of additional treatment, there was no tumor recurrence in previously treated animals while control naïve animals rapidly developed tumors.

Combination Treatment of STRO-002 and Avelumab Significantly Increased Infiltration of CD8⁺ T Cells in MC38-hFolRα Tumors

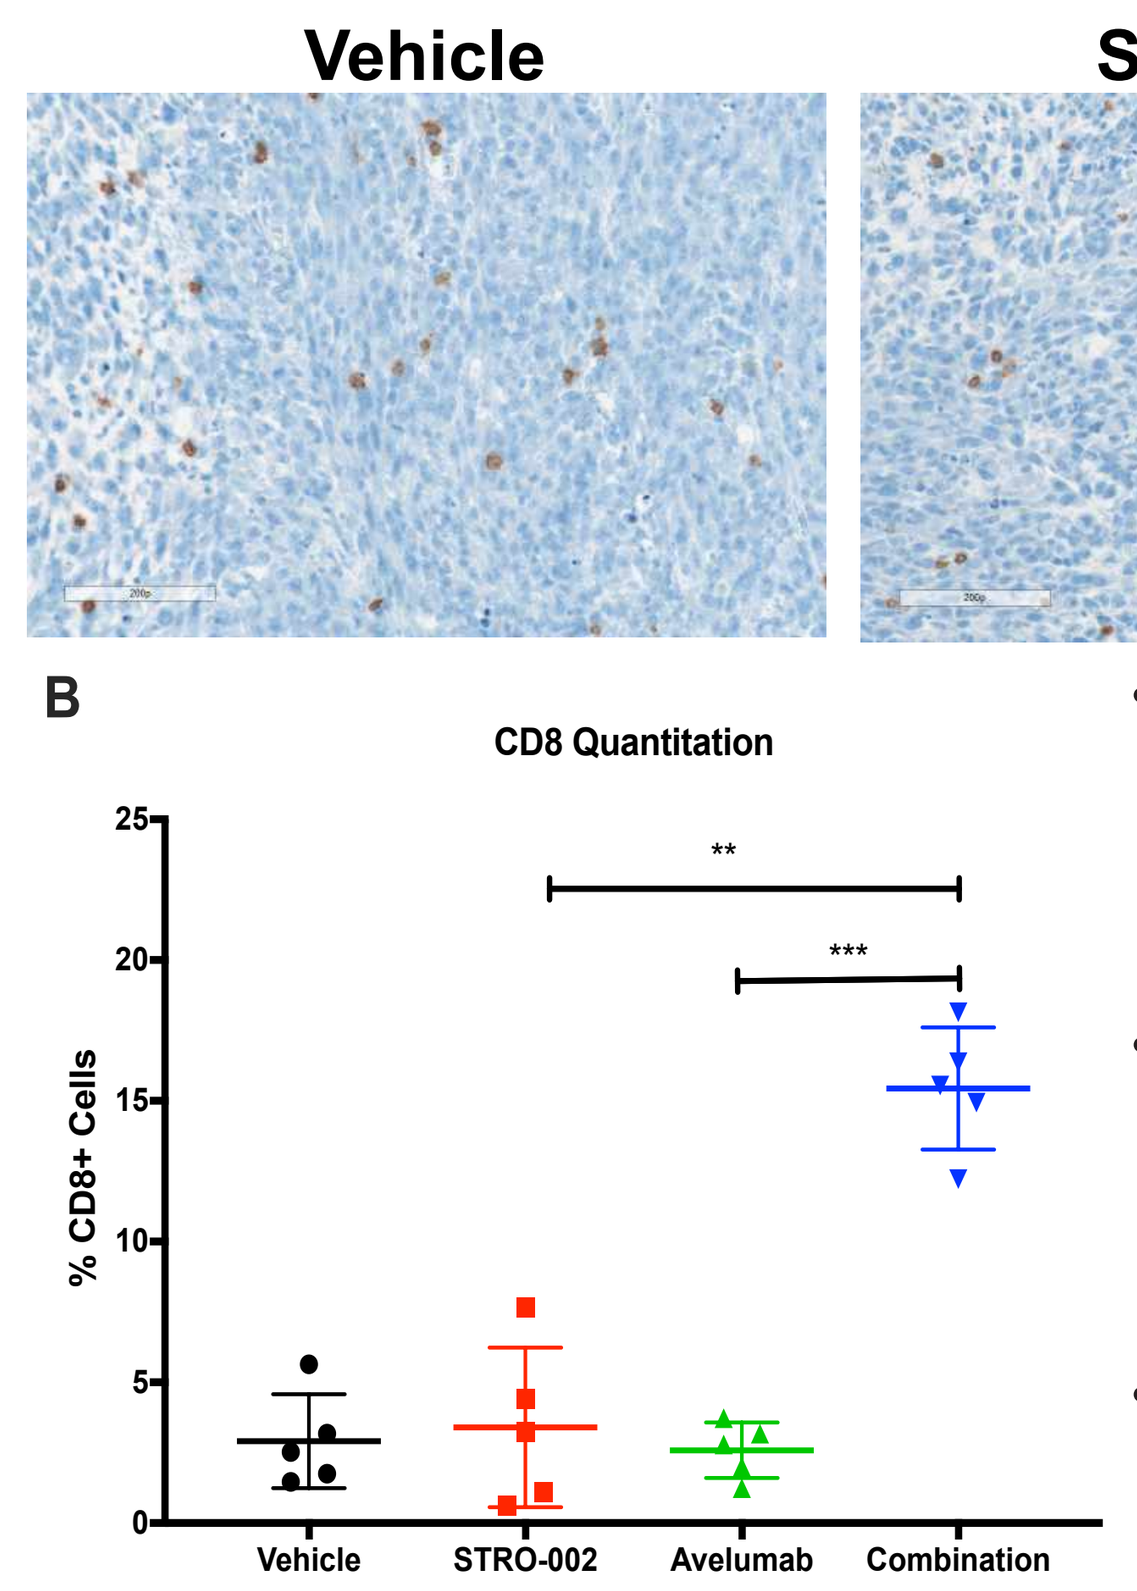
Vehicle
STRO-002
Avelumab
Combination
B
CD8 Quantitation
% CD8+ Cells
**

- Animals bearing MC38-hFolRα tumors were treated with a single dose of vehicle, 10 mg/kg STRO-002, 20 mg/kg Avelumab or the combination of both. Tumors were resected seven days after treatment and FFPE sections stained for CD8⁺ T cells by immunohistochemistry.
- Representative images of CD8 staining (brown) with nuclei counterstain (blue) is shown on top. The percentage of CD8⁺ cells (left) was quantitated using the HALO Multiplex IHC detection software (Indica Labs).
- Combination treatment of STRO-002 and Avelumab resulted in a striking increase in CD8⁺ T cell infiltration into the tumor microenvironment.

Increased PD-1 Expression in Response to STRO-002 Treatment Supports Enhanced Efficacy of STRO-002 in Combination With Avelumab

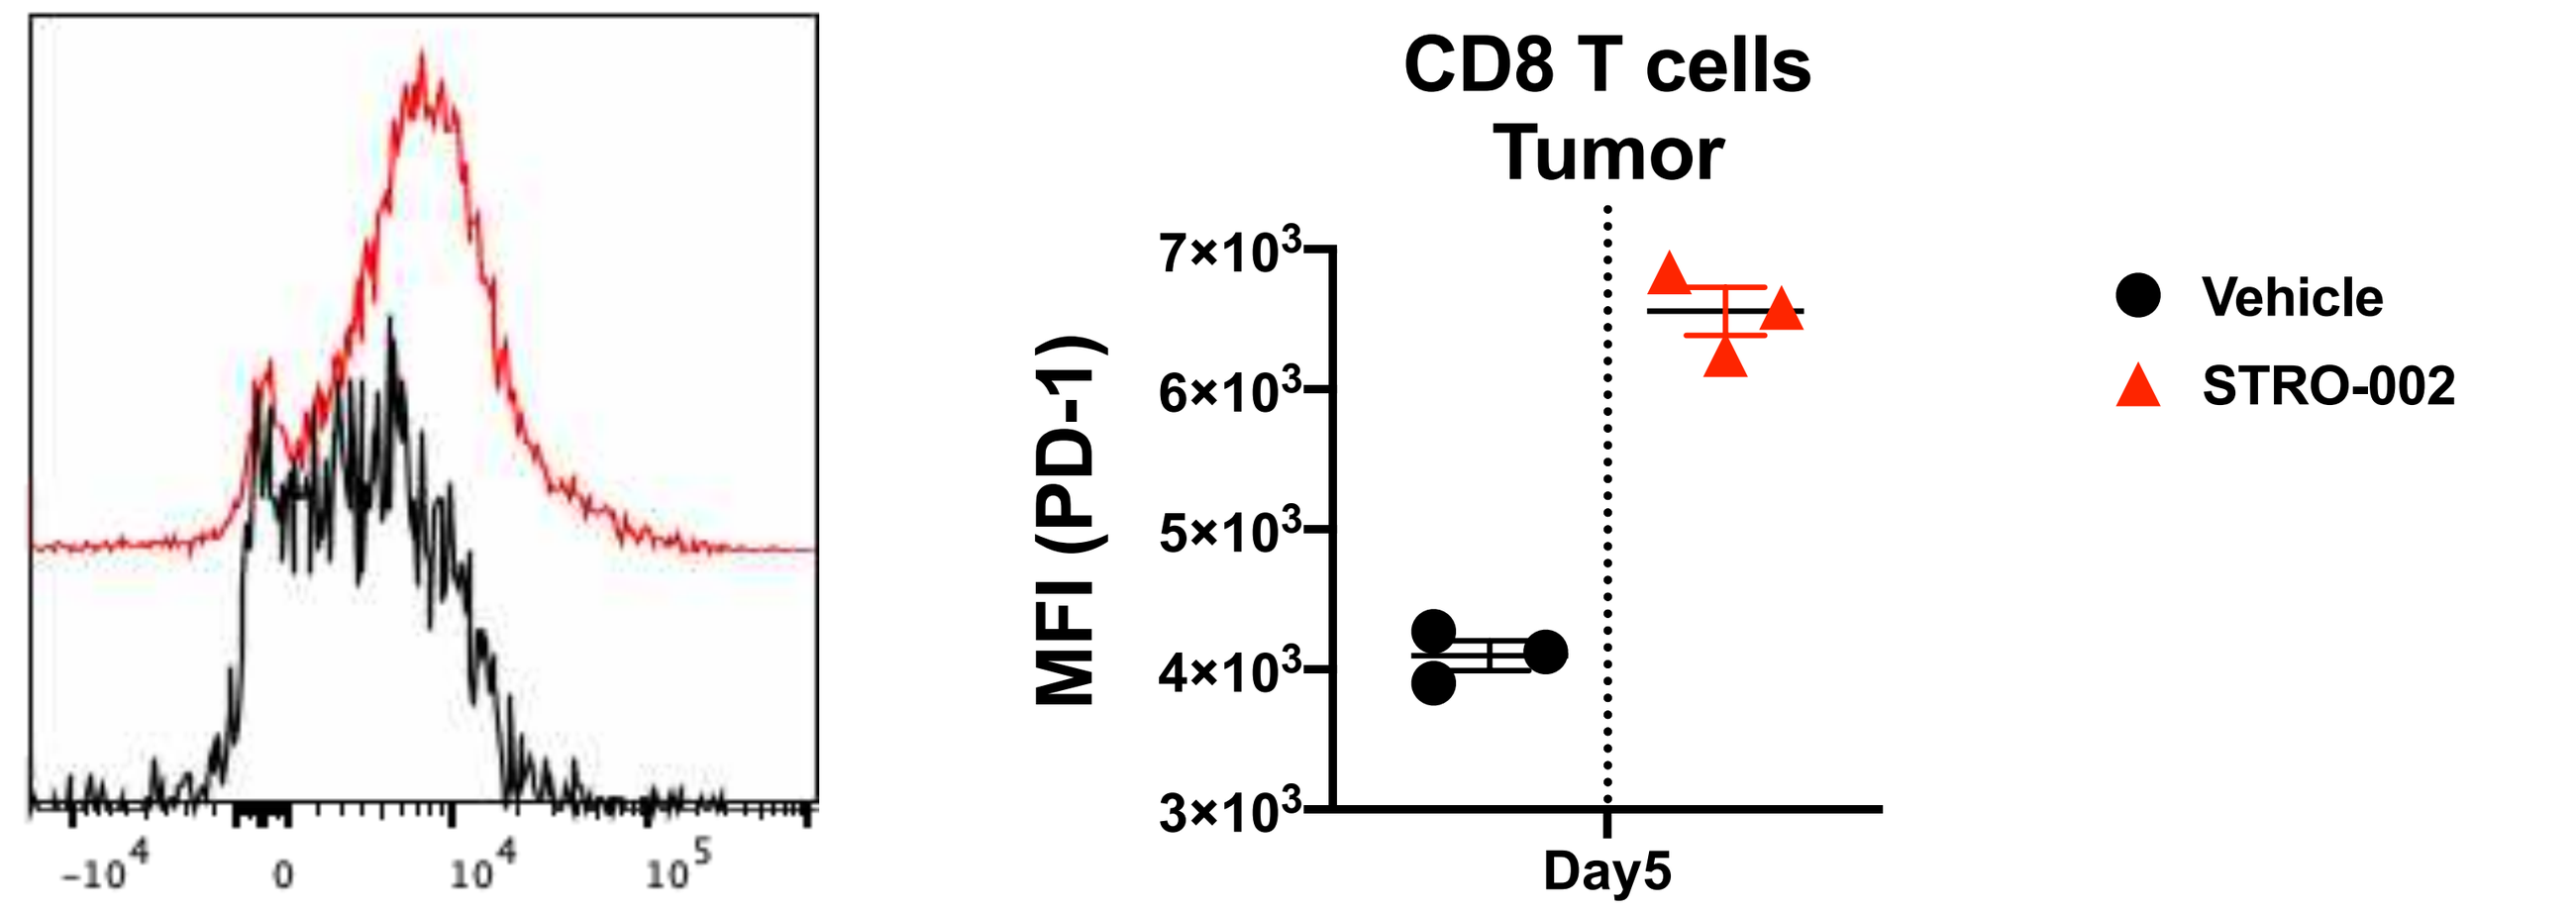
CD8 T cells
Tumor
MFI (PD-1)
Day5
Vehicle
STRO-002

- Animals bearing MC38-hFolRα tumors were treated with a single dose of 10 mg/kg STRO-002 or vehicle. Tumors were resected five days after treatment and analyzed for PD-1 expression by flow cytometry.
- Histograms (left) and measurement of mean fluorescence intensity (MFI, right) in CD8⁺ T cells reveal an upregulation of PD-1 expression in STRO-002 treated tumors, which may have further sensitized the MC38-hFolRα tumors to the combination treatment.

Summary

- Our studies demonstrate that in addition to its potent cytotoxic activity, STRO-002 demonstrates a complementary mechanism of action involving engagement of the host immune system to potentiate antitumor activity in a target dependent manner.
- Combination of a single dose of STRO-002 with Avelumab resulted in complete remission and durable protective anti-tumor immunity after tumor re-challenge.
- IHC analysis revealed a significant increase in CD8⁺ T cell infiltration in the tumors of animals treated with a combination of STRO-002 and Avelumab.
- Our data suggests that STRO-002 mediated induction of ICD and upregulation of PD-1 expression contribute to the added benefit observed with combination of STRO-002 and Avelumab.
- STRO-002 is currently being evaluated in a Phase 1 clinical trial (NCT03748186) and this preclinical data provides rationale to support clinical evaluation of STRO-002 in combination with anti-PD1/PD-L1 agents.