5550

Phase 1 Dose-Escalation Study of STRO-002, an Anti–Folate-Receptor Alpha Antibody Drug Conjugate, in Patients With Advanced, Progressive, Platinum-Resistant/-Refractory Epithelial Ovarian Cancer

R. Wendel Naumann,[1] Fadi S. Braiteh,[2] Lainie P. Martin,[3] Erika Hamilton,[4] John P. Diaz,[5] Sami Diab,[6] Russell J. Schilder,[7] John W. Moroney,[8] Denise Uyar,[9] David M. O'Malley,[10] Richard T. Penson,[11] Clifford DiLea,[12] Michael Palumbo,[13] Venita DeAlmeida,[13] Shannon Matheny,[13] Lin Lu,[13] Craig J. Berman,[13] Arturo Molina[13]

[1]Levine Cancer Institute, Atrium Health, Charlotte, NC; [2]Comprehensive Cancer Centers of Nevada, Las Vegas, NV; [3]University of Pennsylvania, Abramson Cancer Center, Philadelphia, PA; [4]Sarah Cannon Research Institute, Tennessee Oncology PLLC, Nashville, TN; [5]Miami Cancer Institute at Baptist Health, Miami, FL; [6]Rocky Mountain Cancer Center, Aurora, CO; [7]Sidney Kimmel Cancer Center at Thomas Jefferson University, Philadelphia, PA and Sarah Cannon Research Institute, Nashville, TN; [8]University of Chicago, Chicago, IL; [9]Medical College of Wisconsin, Milwaukee, WI; [10]Ohio State University, Wexner Medical Center, Columbus, OH; [11]Massachusetts General Hospital, Boston, MA; [12]Aclairo Pharmaceutical Development Group, Vienna, VA; [13]Sutro Biopharma, Inc., South San Francisco, CA

INTRODUCTION

- Folate-receptor alpha (FRα) is a cell surface glycoprotein overexpressed in ovarian cancer and endometrial adenocarcinoma[1,2]
- STRO-002 is a novel FRα-targeting antibody drug conjugate (ADC) with a precise drug-antibody ratio of 4 using site-specific conjugation technology circumventing limitations of current ADCs[3,4]
 - The potential dual mechanism includes
 - A potent tubulin-targeting cytotoxin containing 3-aminophenyl hemiasterlin warhead SC209 that is a weak substrate for P-glycoprotein
 - Immunogenic response upon cell death
- STRO-002-GM1 (NCT03748186) is an ongoing phase 1, open-label study in patients with advanced platinum-resistant or -refractory epithelial ovarian cancer (EOC)

METHODS

- Patients received intravenous STRO-002 on day 1 of each 21-day cycle until disease progression
 - Ocular exams were performed at baseline and every other cycle
 - Prophylactic corticosteroid eye drops were not administered
 - FRα expression was not required for eligibility
 - Retrospective analysis of FRα expression in archival tumor tissue is ongoing
- Results from the dose-escalation cohort are reported here; evaluation of the dose-expansion cohort is ongoing (**Figure 1**)

Figure 1. STRO-002-GM1 study design

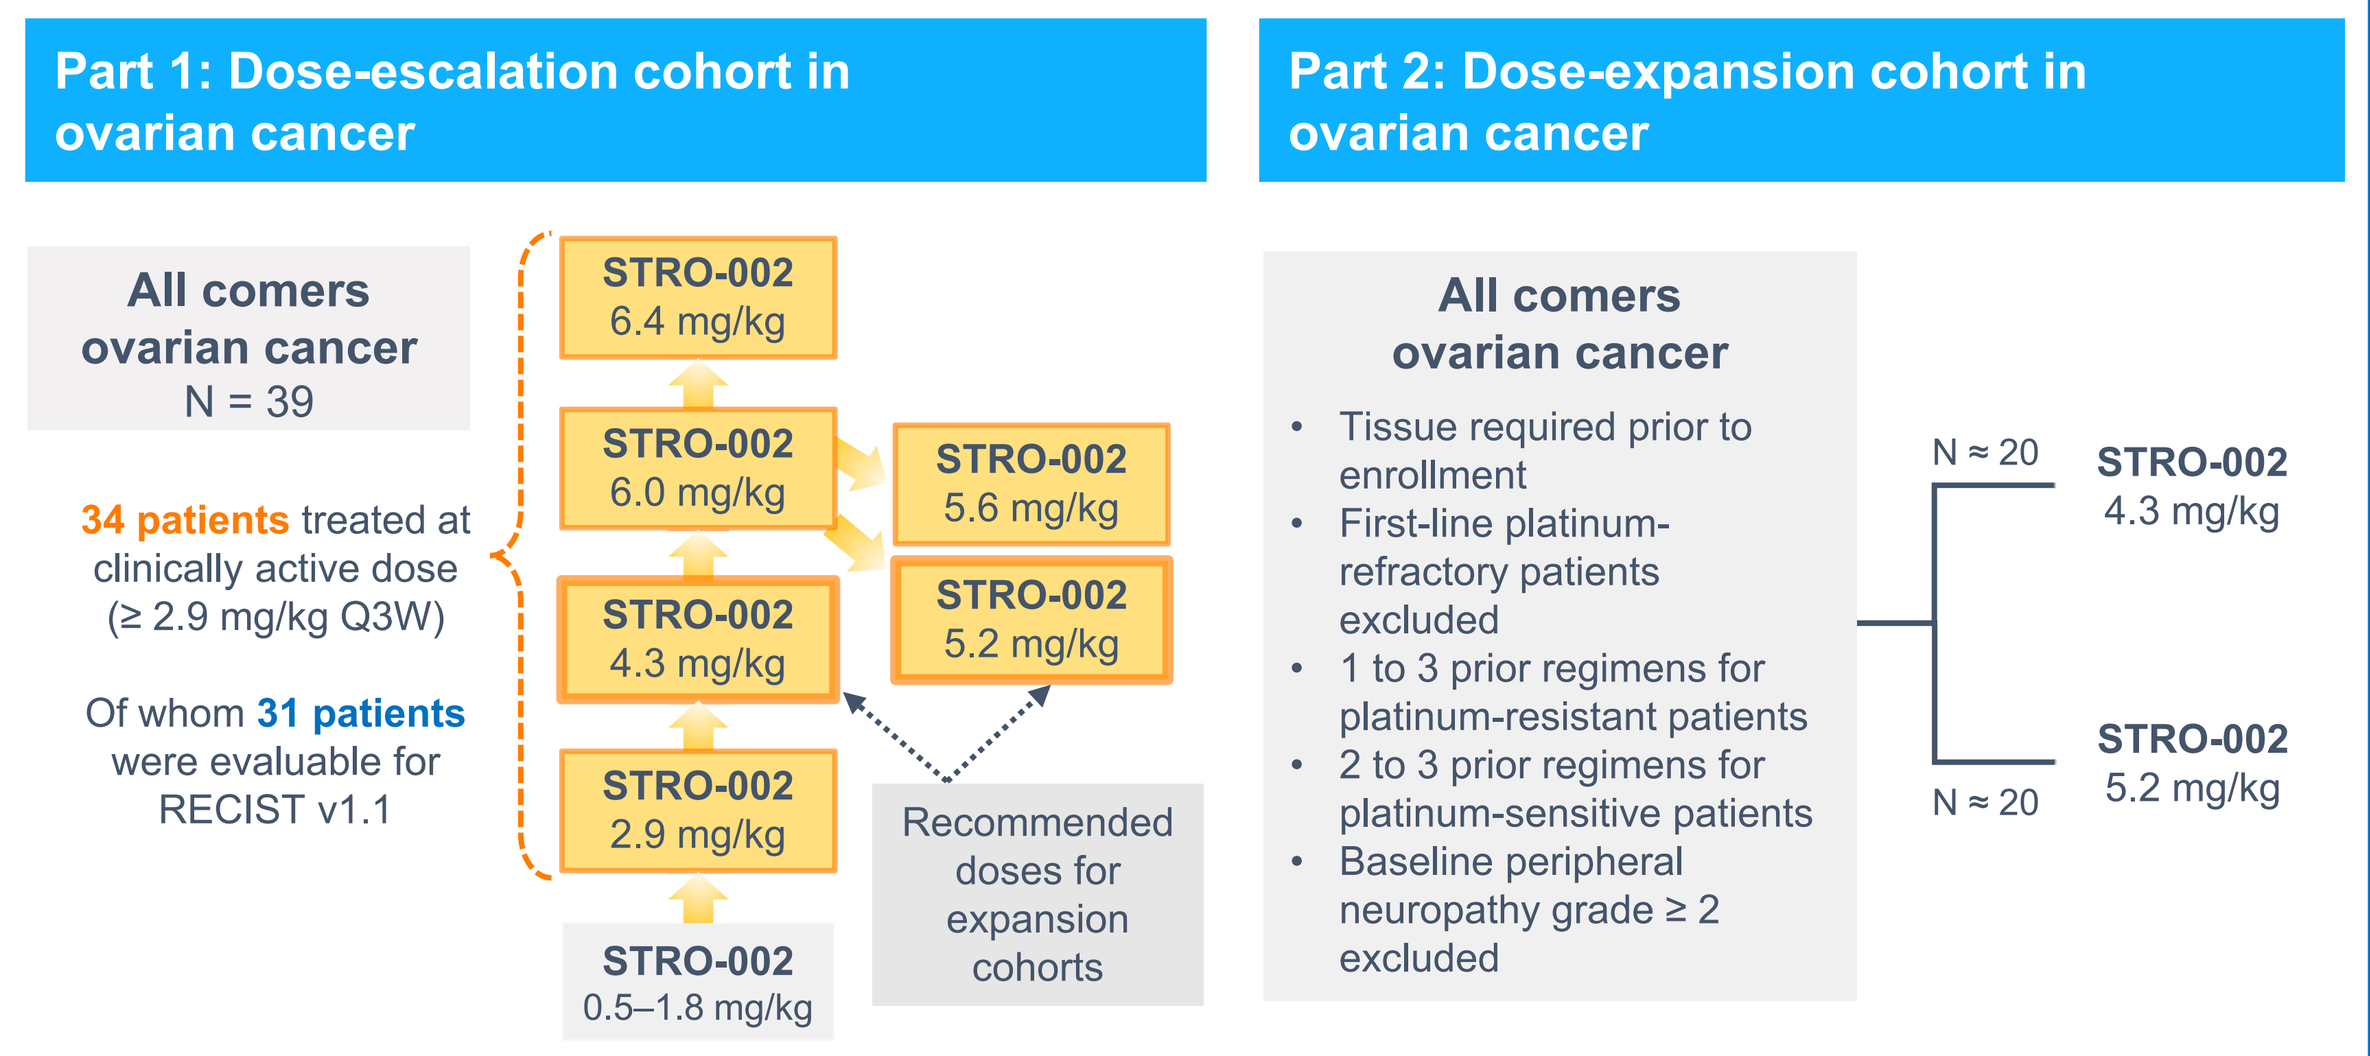

Part 1: Dose-escalation cohort in ovarian cancer
Part 2: Dose-expansion cohort in ovarian cancer
All comers ovarian cancer N = 39
34 patients treated at clinically active dose (≥ 2.9 mg/kg Q3W)
Of whom 31 patients were evaluable for RECIST v1.1
STRO-002 6.4 mg/kg
STRO-002 6.0 mg/kg
STRO-002 4.3 mg/kg
STRO-002 2.9 mg/kg
STRO-002 0.5–1.8 mg/kg
STRO-002 5.6 mg/kg
STRO-002 5.2 mg/kg
Recommended doses for expansion cohorts
All comers ovarian cancer
Tissue required prior to enrollment
First-line platinum-refractory patients excluded
1 to 3 prior regimens for platinum-resistant patients
2 to 3 prior regimens for platinum-sensitive patients
Baseline peripheral neuropathy grade ≥ 2 excluded
N ≈ 20
STRO-002 4.3 mg/kg
N ≈ 20
STRO-002 5.2 mg/kg

Key endpoints: Safety (DLTs and TEAEs), ORR, PK profile, DOR, PFS, OS, biomarkers

DLT, dose-limiting toxicity; DOR, duration of response; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PK, pharmacokinetic; RECIST, Response Evaluation Criteria in Solid Tumors; Q3W, every 3 weeks; TEAE, treatment-emergent adverse event.

RESULTS

- A total of 39 patients were enrolled, and data cutoff was April 23, 2021
- Patients had a median age of 61 years, with median time since diagnosis of 3.9 years and a median of 6 prior lines of therapy (**Table 1**)
- Patients were treated with 9 dose levels; 34 patients were treated at clinically active doses of ≥ 2.9 mg/kg

Table 1. Baseline characteristics

Baseline characteristic	All patients (N = 39)
Age, median (range), years	61 (48–79)
Time since diagnosis, median (range), years	3.9 (0.7–17.0)
No. of prior lines of therapy, median (range)	6 (1–11)
Previous therapies, n (%)	
Platinum	39 (100)
≥ 3 prior platinum regimens	18 (46)
Taxanes	38 (97)
Bevacizumab	32 (82)
Poly (ADP-ribose) polymerase inhibitors	23 (59)
Checkpoint inhibitors	8 (21)
Experimental therapy	14 (36)

ADP, adenosine diphosphate.

- Most TEAEs (86%) were grade 1–2 (**Table 2**)
 - No ocular toxicity signal was observed
 - The most frequent grade 3–4 adverse event was neutropenia (64%, which included neutropenia, febrile neutropenia, and neutrophil count decreased)
- DLTs were observed in 1 patient each at 6 mg/kg (grade 2 neuropathy/ grade 3 arthralgia) and at 6.4 mg/kg (grade 3 bone pain)
- Two grade 5 events were reported, both unrelated to study drug by investigator assessment (1 death not otherwise specified, 1 acute gastrointestinal bleed)

Table 2. Most frequent TEAEs > 25% by grade

All safety-evaluable patients, n (%)	Grade 1	Grade 2	Grade 3	Grade 4	Overall (N = 39)
Fatigue	7 (18)	19 (49)	4 (10)	0	30 (77)
Nausea	15 (39)	11 (28)	0	0	26 (67)
Constipation	12 (31)	13 (33)	0	0	25 (64)
Neutropenia[a]	0	0	8 (21)	17 (44)	25 (64)
Decreased appetite	10 (26)	10 (26)	0	0	20 (51)
Arthralgia	7 (18)	7 (18)	5 (13)	0	19 (49)
Neuropathy[b]	3 (8)	13 (33)	3 (8)	0	19 (49)
Abdominal pain	7 (18)	6 (15)	3 (8)	0	16 (41)
Vomiting	8 (21)	7 (18)	0	0	15 (39)
AST increased	10 (26)	3 (8)	2 (5)	0	15 (39)
Diarrhea	8 (21)	3 (8)	1 (3)	0	12 (31)
Dizziness	9 (23)	3 (8)	0	0	12 (31)
Dry eye	4 (10)	8 (21)	0	0	12 (31)
Anemia	3 (8)	6 (15)	2 (5)	0	11 (28)
Pyrexia	8 (21)	3 (8)	0	0	11 (28)
Headache	7 (18)	3 (8)	0	0	10 (26)
Insomnia	6 (15)	4 (10)	0	0	10 (26)

[a]Neutropenia included the following preferred terms: neutropenia, febrile neutropenia, and neutrophil count decreased. [b]Neuropathy included the following preferred terms: neuropathy peripheral and peripheral sensory neuropathy. AST, aspartate aminotransferase.

KEY FINDINGS

In this phase 1 dose-escalation study, the novel FRα-targeting ADC STRO-002 exhibited a generally well-tolerated safety profile and clinical efficacy as demonstrated by objective responses (1 CR, 4 PRs, and 5 PRus), disease control rate of 61% and 55% at ≥ 16 and 24 weeks, respectively, and median PFS of 7.2 months in heavily pretreated patients with progressive EOC

CONCLUSIONS

- STRO-002 is a novel FRα-targeting ADC that showed a preliminary clinical efficacy signal at multiple doses (≥ 2.9 mg/kg Q3W) in patients with heavily pretreated relapsed/refractory EOC
- Dose reductions were not associated with loss of antitumor activity
- STRO-002 treatment was well tolerated, including in patients with long-term responses (≥ 74 weeks); the majority of TEAEs (86%) were grade 1 or 2
- Antitumor activity was observed across a broad range of FRα expression levels based on PS2+ scoring and H-score
- During the dose-expansion part of the STRO-002-GM-1 study, patients with less heavily pretreated ovarian cancer will be enrolled
- Preliminary data from the dose-expansion cohort is expected later in 2021

REFERENCES

1. Kalli KR et al. *Gynecol Oncol*. 2008;108:619-626.
2. O'Shannessy DJ et al. *Int J Gynecol Pathol*. 2013;32:258-268.
3. Li X et al. *Cancer Res.* 2018;78(13 suppl):abstract 1782.
4. Zimmerman ES et al. *Bioconjug Chem.* 2014;255:351-361.

ACKNOWLEDGMENTS

This study was funded by Sutro Biopharma, Inc. Writing assistance was provided by Anita Misra-Press, PhD, and Benjamin Ricca, PhD, of Parexel, and was funded by Sutro Biopharma.

RESULTS (continued)

- 31 of 34 patients were evaluable for RECIST v1.1 response
 - 10 of 31 patients had objective responses: 1 complete response (CR), 4 confirmed partial response (PR), and 5 unconfirmed PR (PRu; **Figure 2**)

Figure 2. Maximum change[a] in tumor target lesions in RECIST-evaluable patients treated with STRO-002 ≥ 2.9 mg/kg Q3W (N = 31)

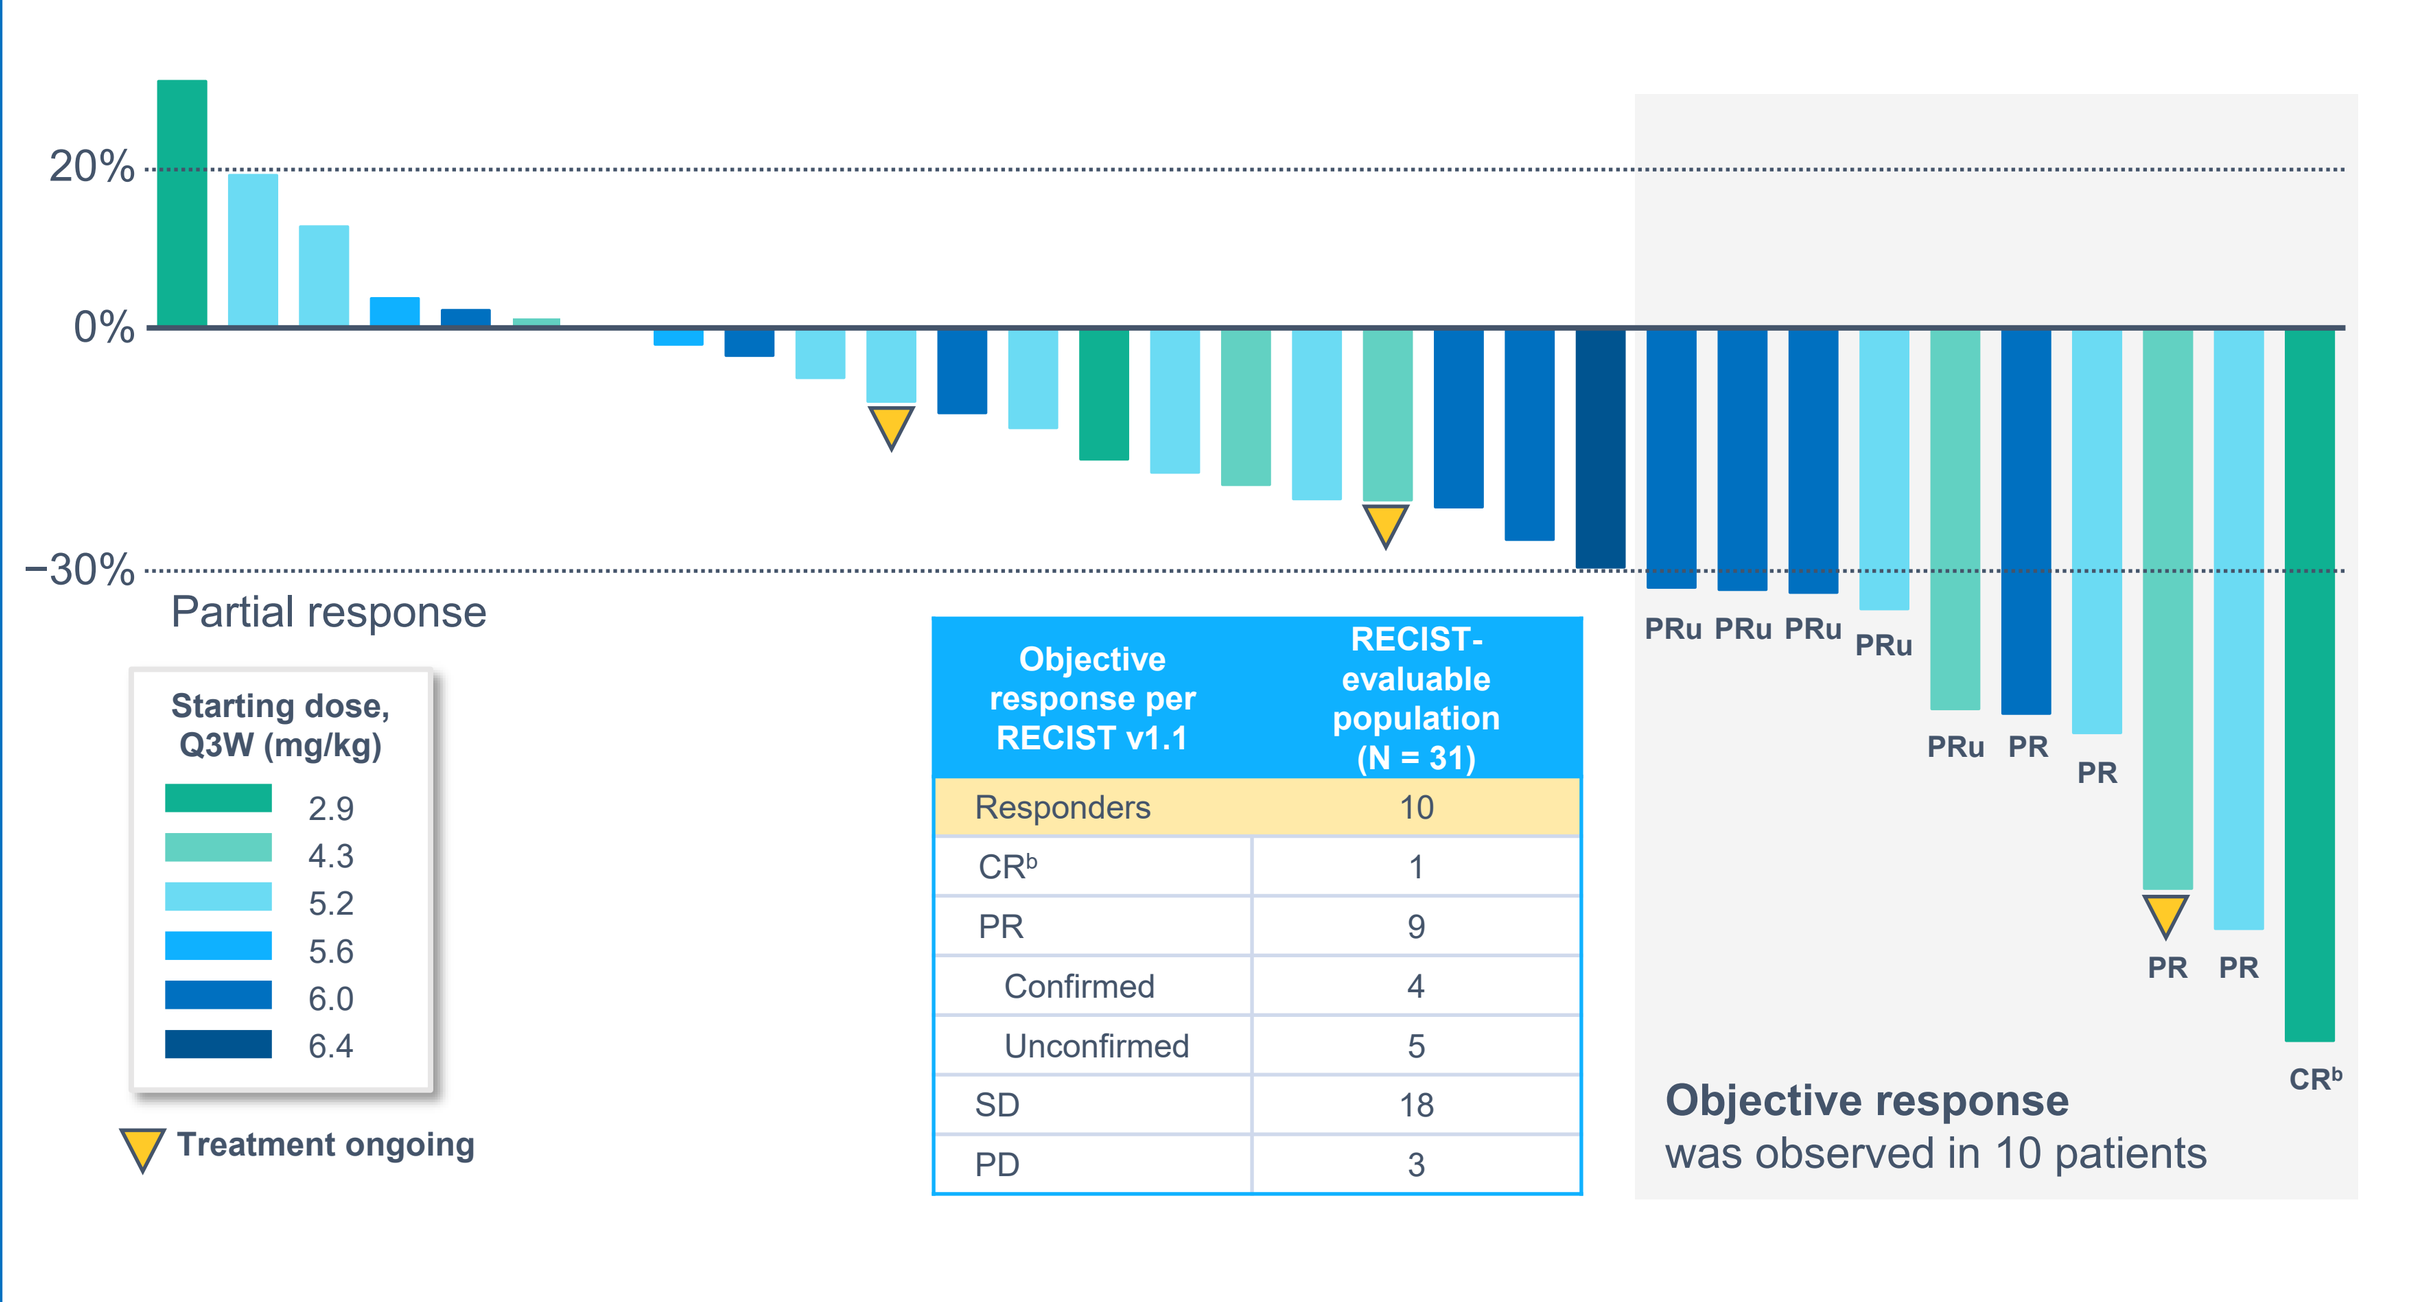

20%
0%
−30%
Partial response
Starting dose, Q3W (mg/kg)
2.9
4.3
5.2
5.6
6.0
6.4
Treatment ongoing
PRu PRu PRu PRu
PRu PR PR
PR PR
CR[b]
Objective response was observed in 10 patients

Objective response per RECIST v1.1	RECIST-evaluable population (N = 31)
Responders	10
CR[b]	1
PR	9
Confirmed	4
Unconfirmed	5
SD	18
PD	3

[a]Maximum percentage change from baseline in sum of longest diameter in evaluable patients treated with STRO-002 at ≥ 2.9 mg/kg Q3W (N = 31). [b]CR in patient treated at 2.9 mg/kg with resolution of peritoneal disease. PD, progressive disease; SD, stable disease.

- Disease control rate (CR+PR+SD) was 61% at ≥ 16 weeks and 55% at ≥ 24 weeks, with 3 patients on treatment at ≥ 74 weeks (**Figure 3**)

Figure 3. Treatment duration[a] and response in RECIST-evaluable patients treated with STRO-002 ≥ 2.9 mg/kg Q3W (N = 31)

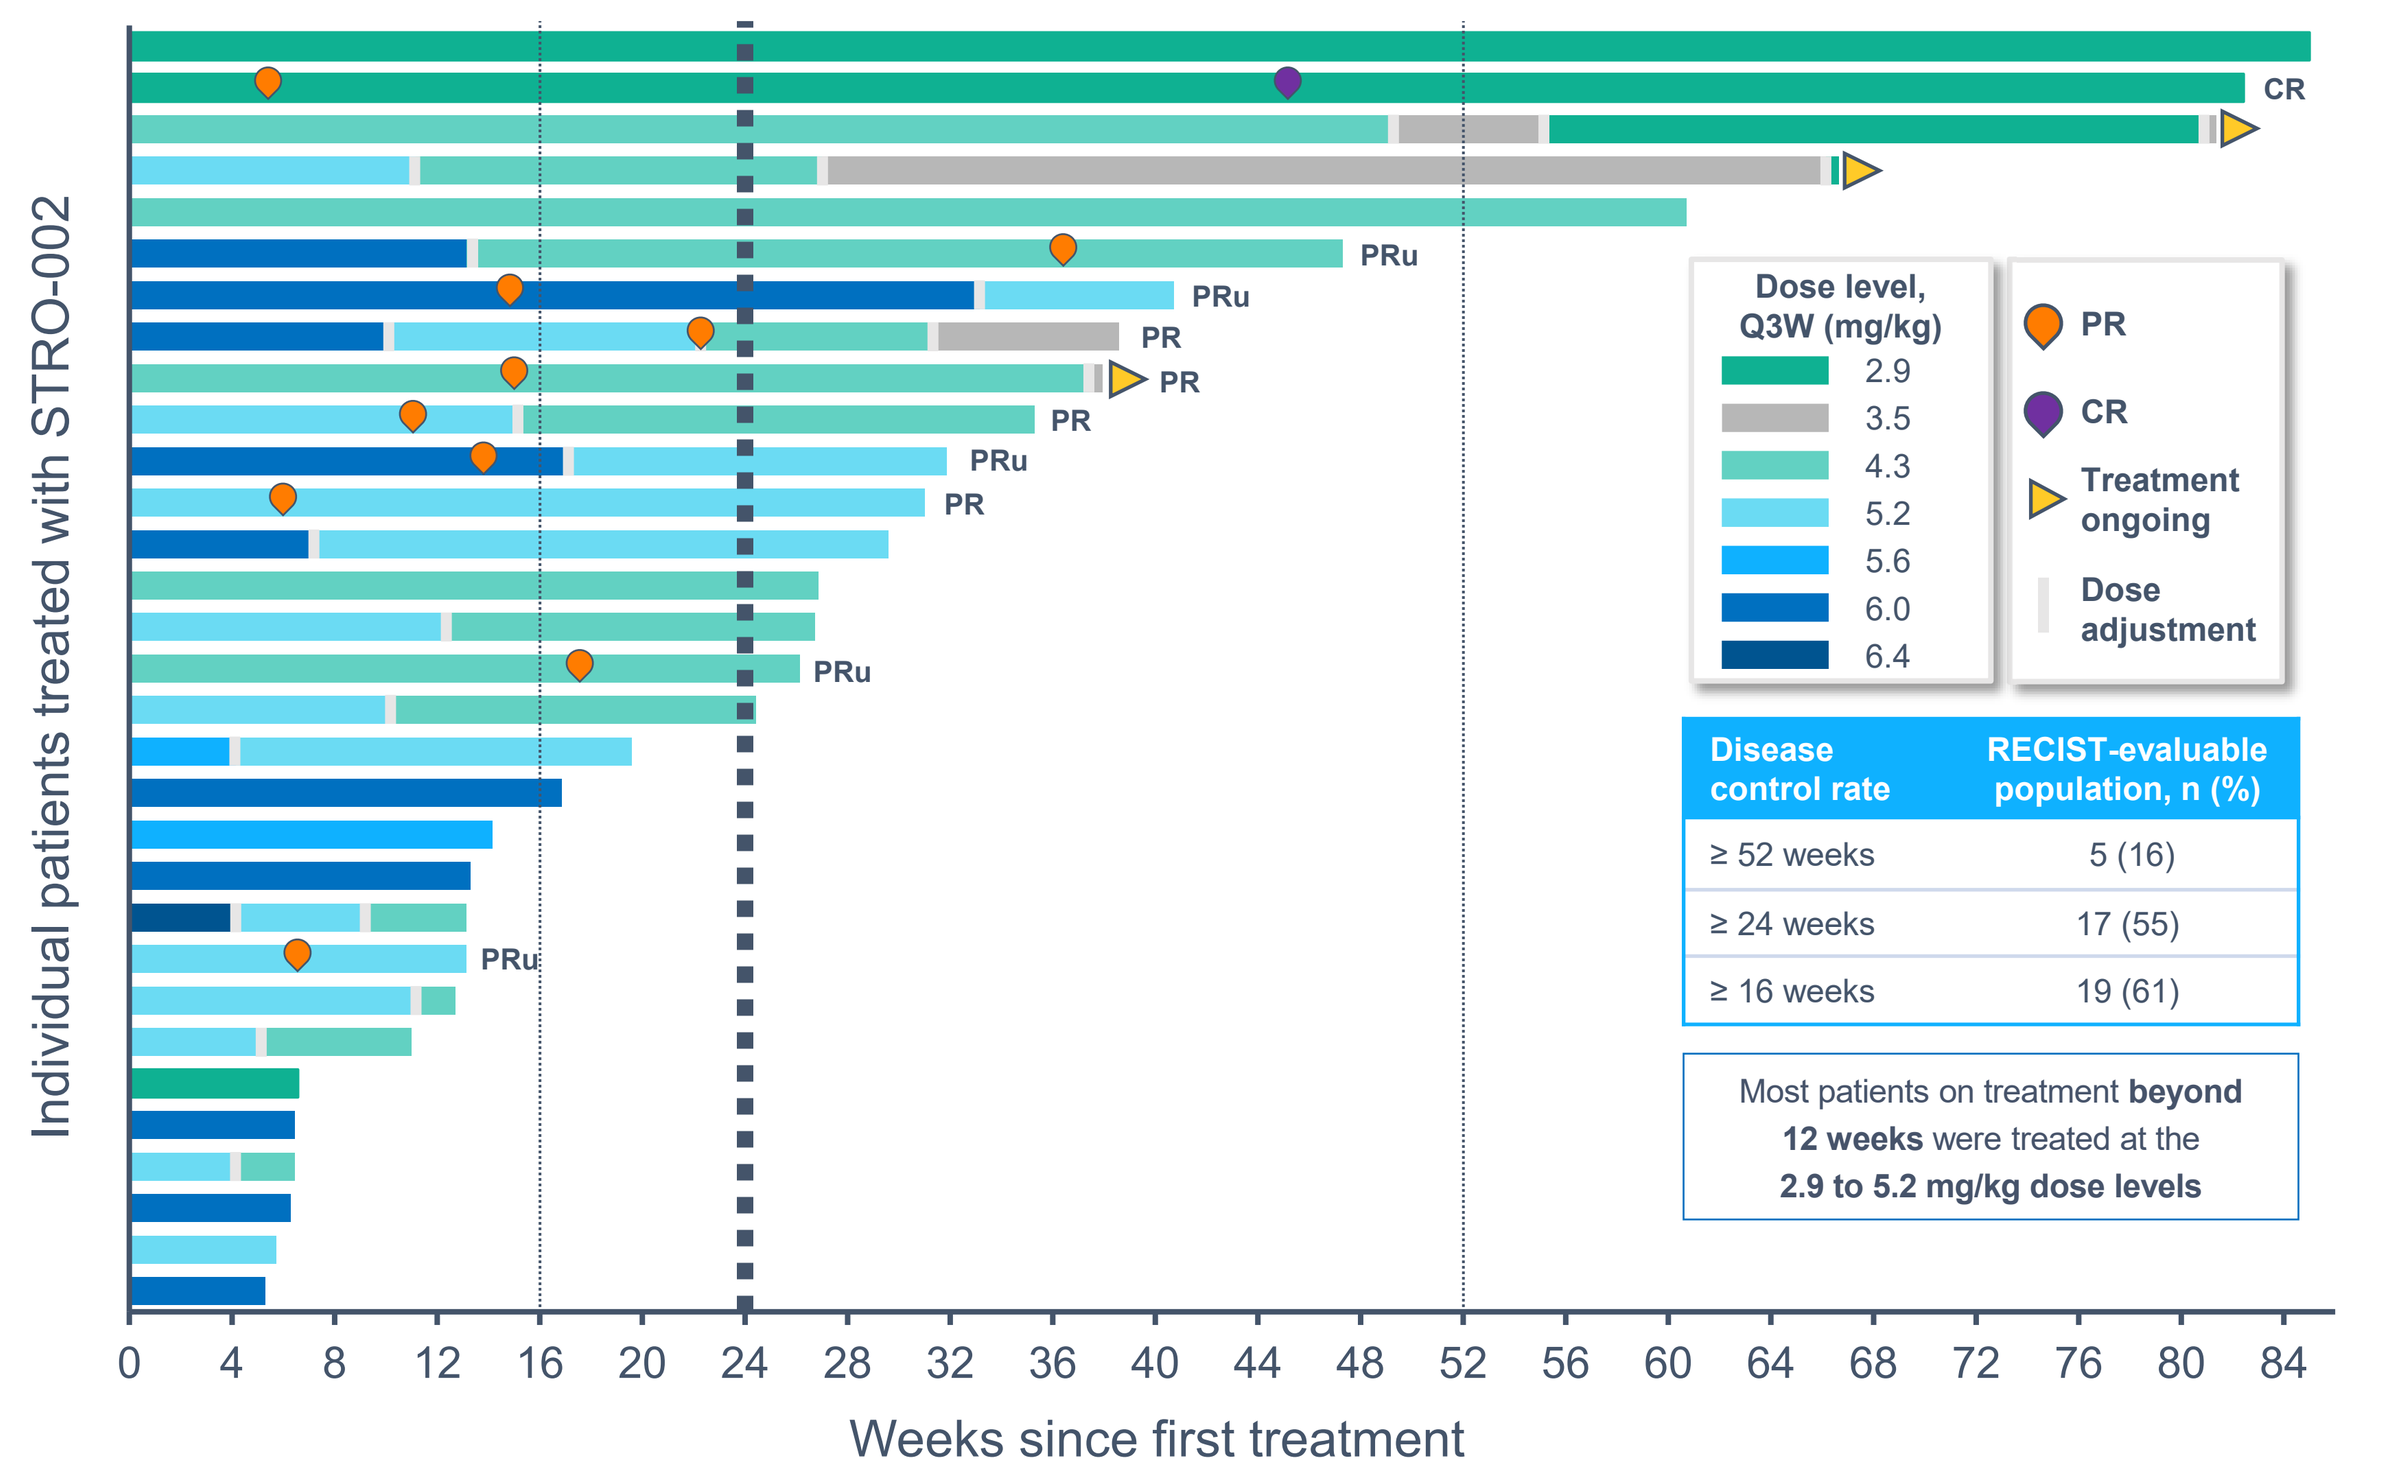

Individual patients treated with STRO-002
CR
PRu
PRu
PR
PR
PR
PRu
PR
PRu
PRu
Dose level, Q3W (mg/kg)
2.9
3.5
4.3
5.2
5.6
6.0
6.4
PR
CR
Treatment ongoing
Dose adjustment
Most patients on treatment beyond 12 weeks were treated at the 2.9 to 5.2 mg/kg dose levels
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84
Weeks since first treatment

Disease control rate	RECIST-evaluable population, n (%)
≥ 52 weeks	5 (16)
≥ 24 weeks	17 (55)
≥ 16 weeks	19 (61)

[a]Duration calculated as date of PD or time from first dose to last dose given (including 4 patients deriving clinical benefit post progression per investigator assessment).

- Based on Kaplan-Meier estimates, median PFS and median DOR were 31.1 and 25 weeks, respectively (**Table 3**)

Table 3. Kaplan-Meier estimates of PFS and DOR[a]

Endpoint		Median (95% CI)
PFS, weeks	(N = 39)	31.1 (19.6, 47.0)
DOR,[a] weeks	(N = 5)	25 (8.8, NE)

[a]DOR for confirmed responses. NE, not estimable.

- Deepening responses in patients with tumor regression or a trend toward disease control over time were observed in most patients (**Figure 4**)

Figure 4. Change in sum of diameters for target lesions over time (N = 31)

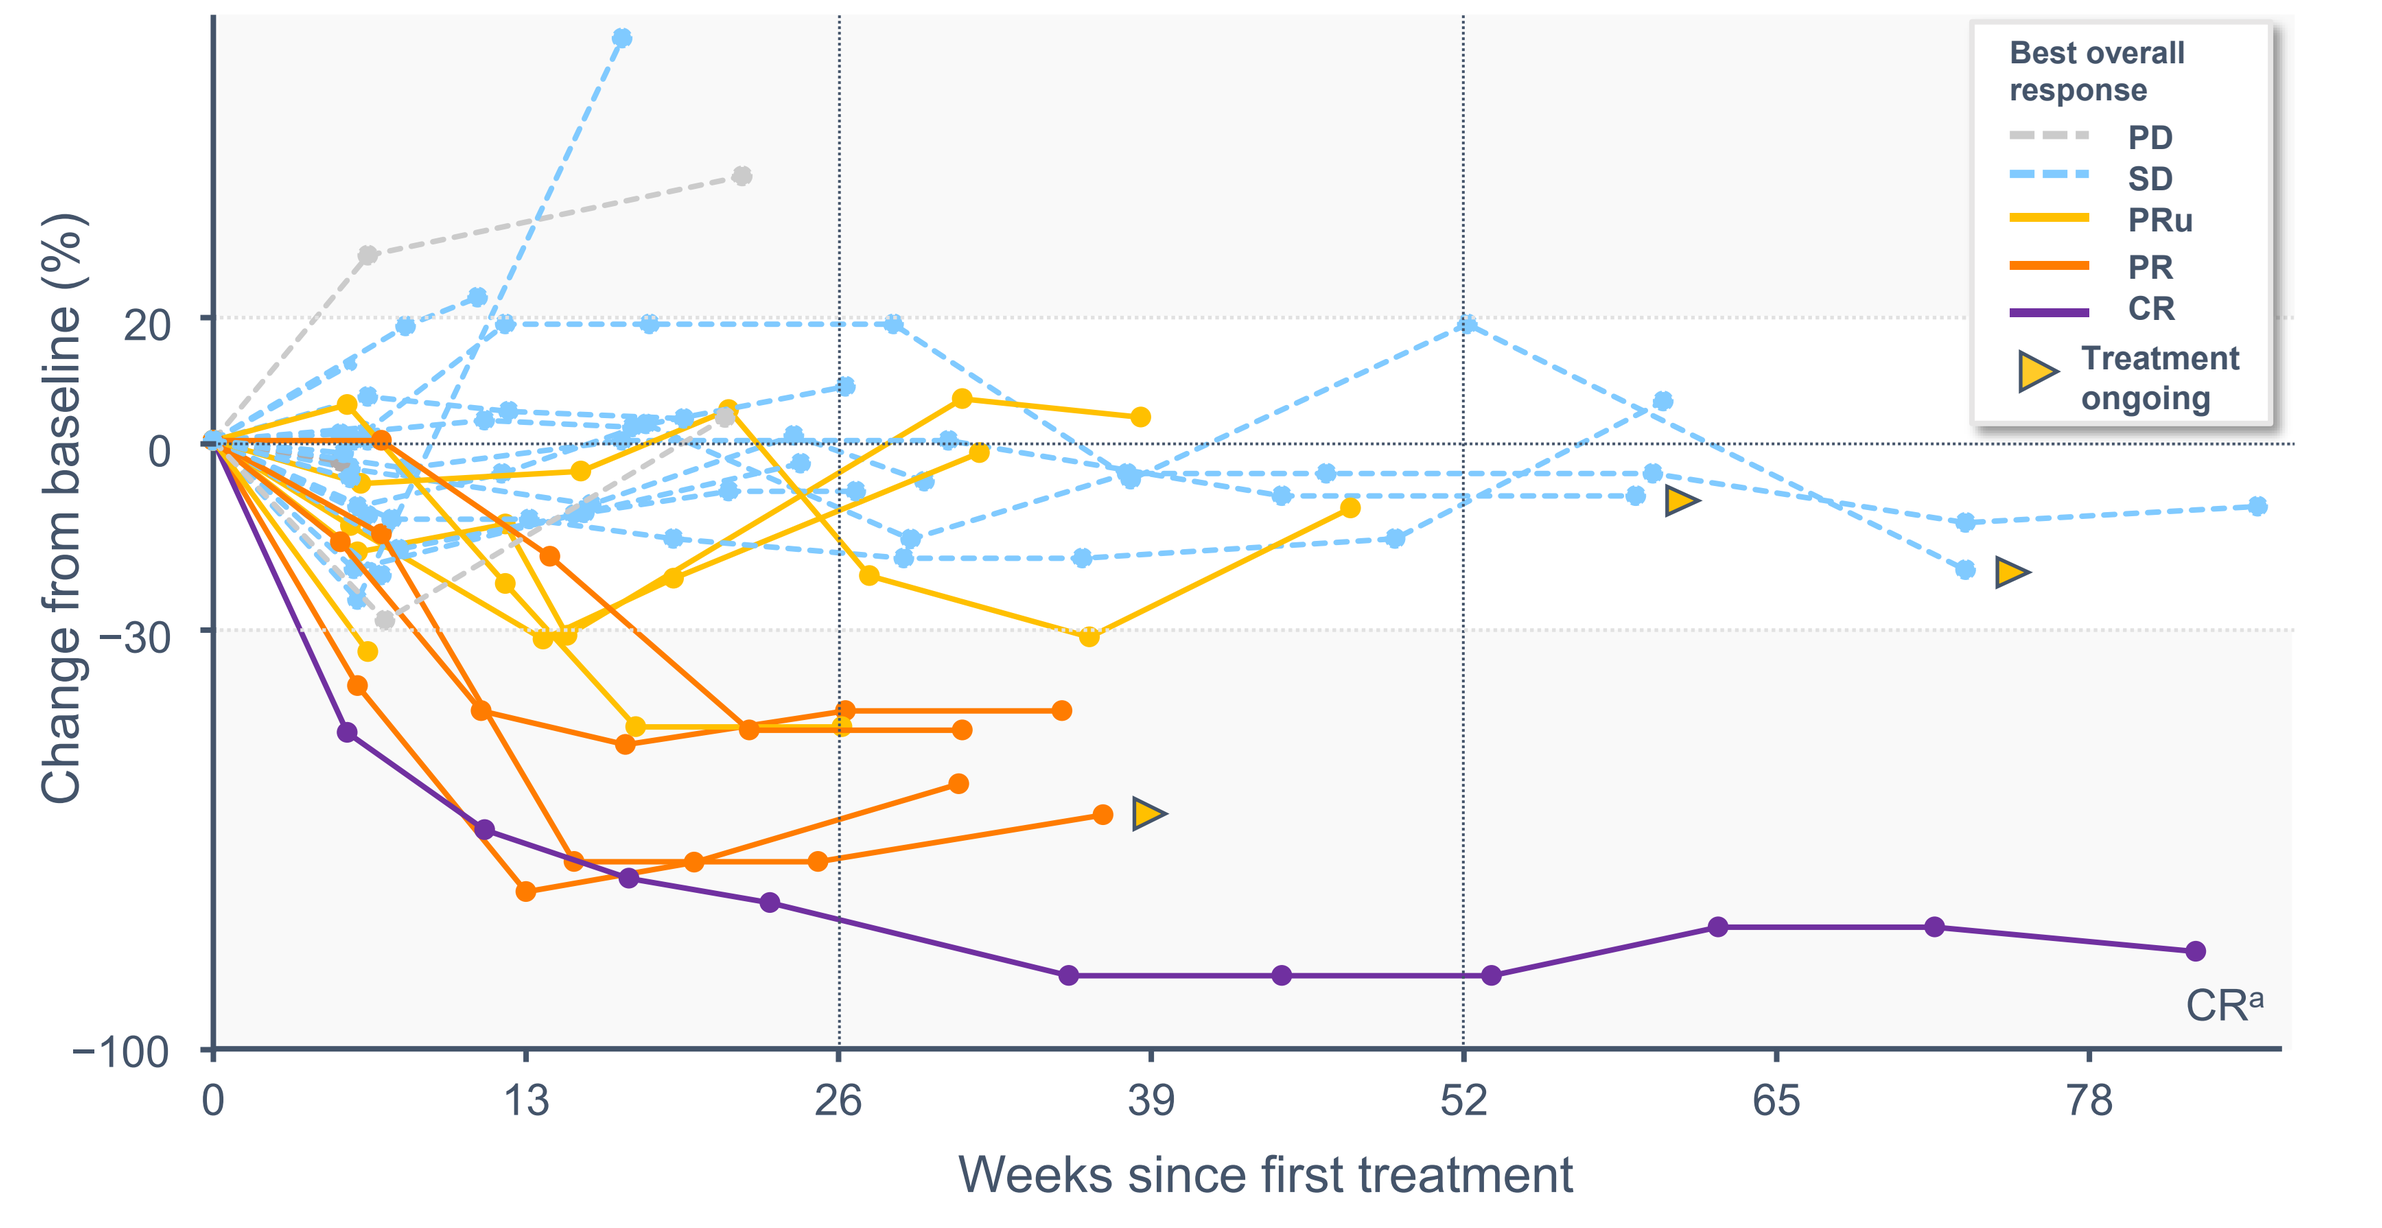

Best overall response
PD
SD
PRu
PR
CR
Treatment ongoing
Change from baseline (%)
20
0
−30
−100
CR[a]
0 13 26 39 52 65 78
Weeks since first treatment

[a]CR in patient treated at 2.9 mg/kg with resolution of peritoneal disease.

- Responses and antitumor activity were observed across various FRα expression levels assessed by immunohistochemistry (**Figure 5**)

Figure 5. FRα expression by immunohistochemistry[a] in individual patients treated with STRO-002 ≥ 2.9 mg/kg Q3W (N = 18)

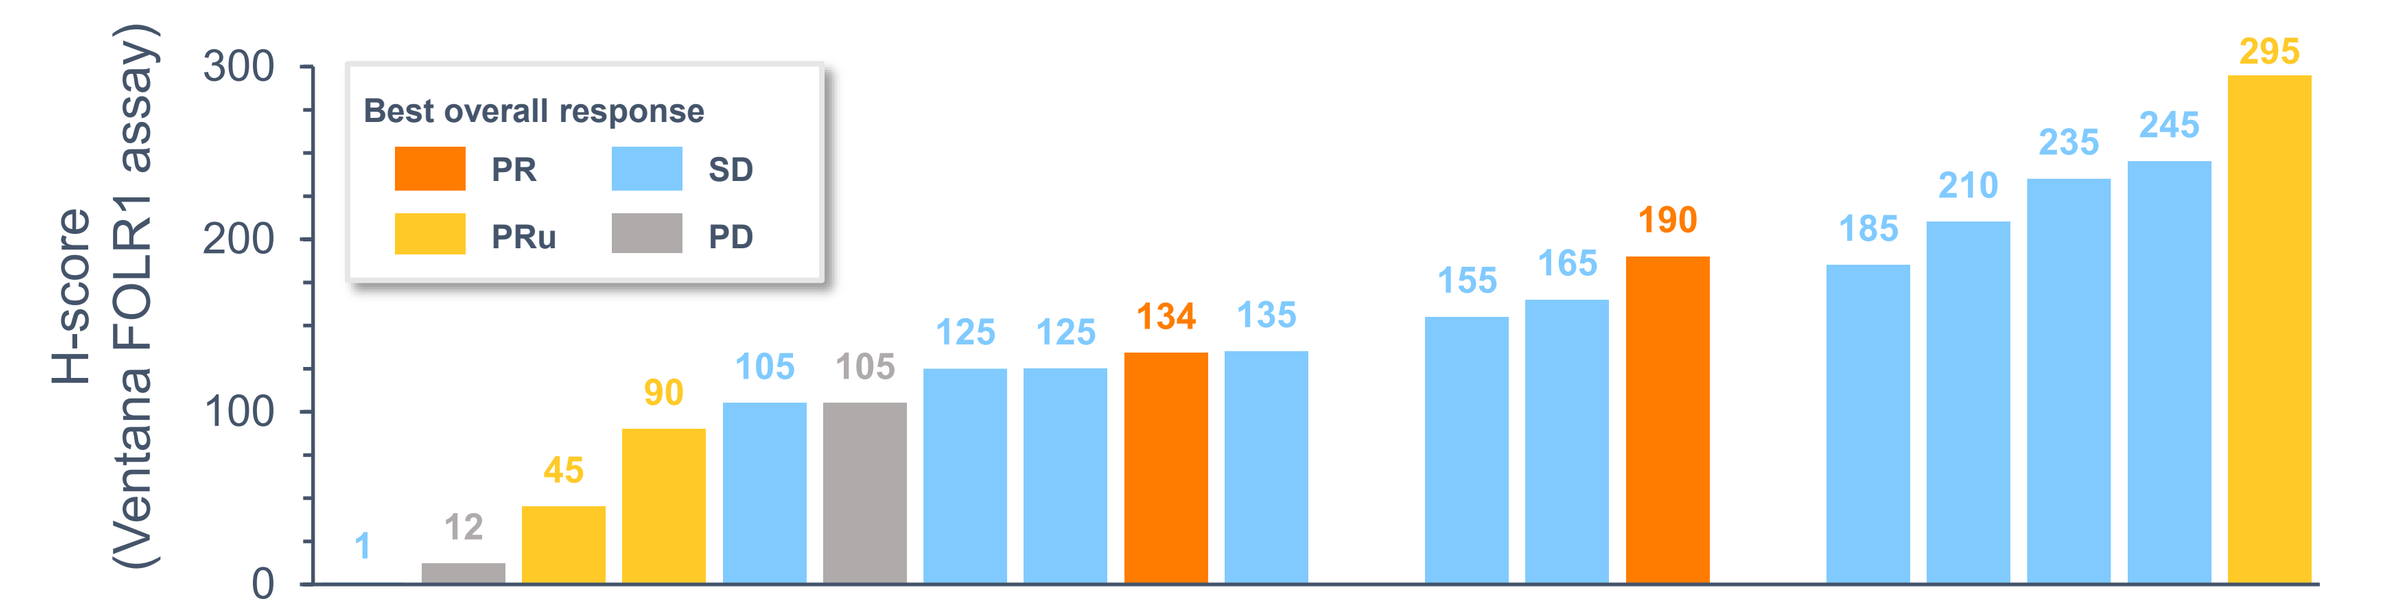

H-score (Ventana FOLR1 assay)
300
200
100
0
Best overall response
PR
SD
PRu
PD
1 12 45 90 105 105 125 125 134 135 155 165 190 185 210 235 245 295

FOLR1 PS2+ category	Weak/absent expression	Moderate expression	High expression
PR	1	1	0
PRu	2	0	1
SD	5	2	4
PD	2	0	0

[a]Assessed by Ventana FOLR1 expression assay based on available archival tissue from dose-escalation patients and scored using H-score and PS2 methods. FOLR1, folate receptor alpha.

- Pharmacokinetics: maximum plasma concentrations of STRO-002 were achieved by the end of the 1-hour infusion; exposure increased in a dose-proportionate manner

Correspondence: R. Wendel Naumann (wendel.naumann@mac.com) and Arturo Molina (amolina@sutrobio.com)

Presented at the 2021 ASCO Annual Meeting; June 4–8, 2021; Virtual.