Next-generation Immunostimulatory Antibody-Drug Conjugate (iADC) Combines Direct Tumor Killing and Innate Immune Stimulation to Provide Protective Anti-Tumor Immunity

Robert Yuan, Dayson Moreira, Jennifer Smith, Xiaofan Li, Christine Cheng, Abigail Yu, Trevor Hallam, and Kristin Bedard (presenting author)

Sutro Biopharma, Inc. South San Francisco, CA

SUTRO BIOPHARMA

Introduction

In recent years, combination therapies of antibody-drug conjugates (ADCs) and immune system activators have garnered much interest due to their potential to unlock greater durable clinical benefit. Recently, we demonstrated that STRO-002, our novel anti-folate receptor alpha (FolRα) ADC currently in Phase I clinical trials, can engage the immune system through induction of immunogenic cell death (ICD). We further showed that by complementing the ICD-inducing property of STRO-002 with combination treatment with Avelumab, we could enhance infiltration of CD8+ T cells into the tumor and generate potent anti-tumor immunity.

To develop the combination concept further, we leveraged Sutro's breakthrough XpressCF+® cell-free technology, which utilizes precise site-specific conjugation to generate complex molecules, to engineer the immunostimulatory ADC (iADC), a next-generation ADC molecule that combines tumor-targeted cell killing and immune activation in a single modality. Using murine MC38 tumors engineered to express human FolRa, we demonstrated that delivery of a hemiasterlin/TLR agonist dual-conjugate anti-FolRα iADC resulted in a more robust anti-tumor response compared to either modality administered alone. This improved response was associated with innate immune cell activation and increased $CD8^+$ T cell infiltration in the tumor. Moreover, a greater number of complete responses was also observed following treatment with the anti-FolRα iADC, and complete responders developed broad and robust immune memory that was able to reject MC38-hFolRα rechallenge in a $CD8^+$ T cell-dependent manner and parental MC38 rechallenge. Thus, the iADC concept unites two complementary mechanisms of tumor control in a single molecule to attain greater therapeutic benefit.

Sutro's drug discovery platform enables potential for best-in-class molecules

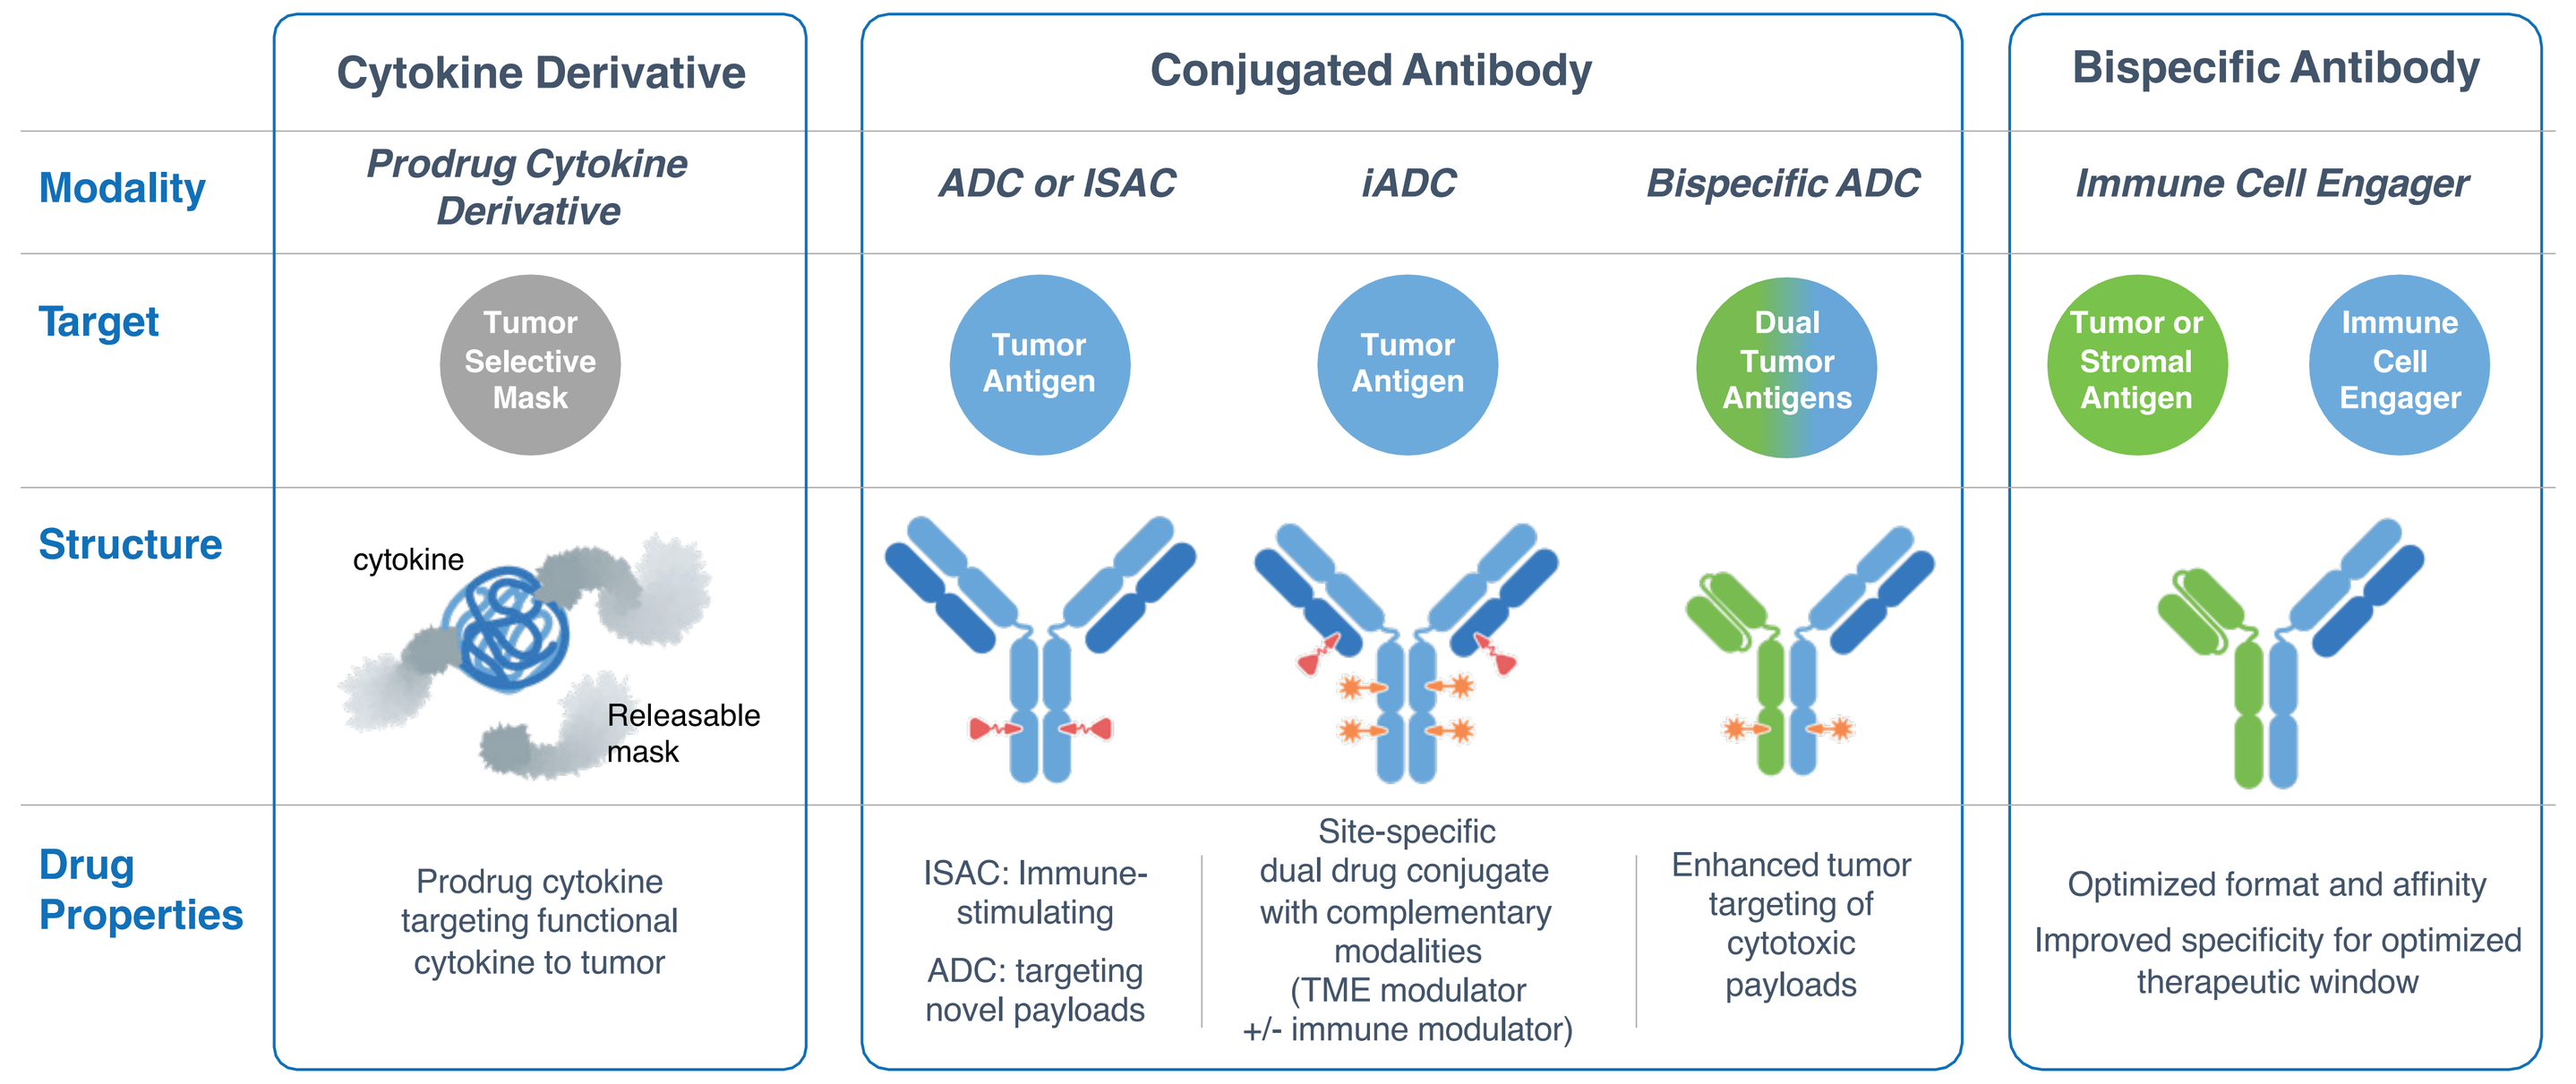
Cytokine Derivative
Conjugated Antibody
Bispecific Antibody
Modality
Prodrug Cytokine Derivative
ADC or ISAC
iADC
Bispecific ADC
Immune Cell Engager
Target
Tumor Selective Mask
Tumor Antigen
Tumor Antigen
Dual Tumor Antigens
Tumor or Stromal Antigen
Immune Cell Engager
Structure
cytokine
Releasable mask
Drug Properties
Prodrug cytokine targeting functional cytokine to tumor
ISAC: Immune-stimulating
ADC: targeting novel payloads
Site-specific dual drug conjugate with complementary modalities (TME modulator +/- immune modulator)
Enhanced tumor targeting of cytotoxic payloads
Optimized format and affinity
Improved specificity for optimized therapeutic window

Figure 1. Sutro's technology allows precise and novel designs to enhance efficacy and safety across multiple modalities and targets.

Sutro's robust pipeline through wholly-owned and partnered programs

Modality	Program	Target	Indication	Discovery	Preclinical	Phase 1/1b	Phase 2/3	Partner
Antibody-Drug Conjugate	STRO-002	FolRα ADC	Ovarian Cancer	Fast Track Designation				天士力生物 (Greater China)
			Ovarian Cancer (bevacizumab combo)					
			Endometrial Cancer					
			NSCLC/Non-Gyn Cancers					
	STRO-001	CD74 ADC	Lymphomas					BioNova Pharma (Greater China)
			Multiple Myeloma	Orphan Drug Designation				
	CC-99712	BCMA ADC	Multiple Myeloma	Orphan Drug Designation				Bristol Myers Squibb
			Multiple Myeloma (GSI combo)					
	Discovery	ROR1, Tissue Factor	Solid Tumors					
Bispecific ADC	M1231	MUC1-EGFR ADC	NSCLC & Esophageal Cancer					EMD Serono (1)
T-Cell Engager	Preclinical	5T4-CD3 TCE	Solid Tumors					
Cytokine Derivative	Not Disclosed	Cytokine target	Cancer	*2 Molecules*				MERCK (2)
	Discovery	IFNα, IL-12, IL-18	Solid Tumors					
Vaccine	VAX-24	24-valent pneumococcal conjugate vaccine	Invasive Pneumococcal Disease					Vaxcyte

(1) EMD Serono is the biopharmaceutical business of Merck KGaA, Darmstadt Germany in the US
(2) Cytokine Derivative program with Merck includes two molecules derived from one undisclosed target.

Figure 2. Sutro's pipeline has five product candidates advancing in the clinic and numerous late-stage discovery programs.

Sutro's next-generation tumor-targeting immunostimulatory ADC (iADC)

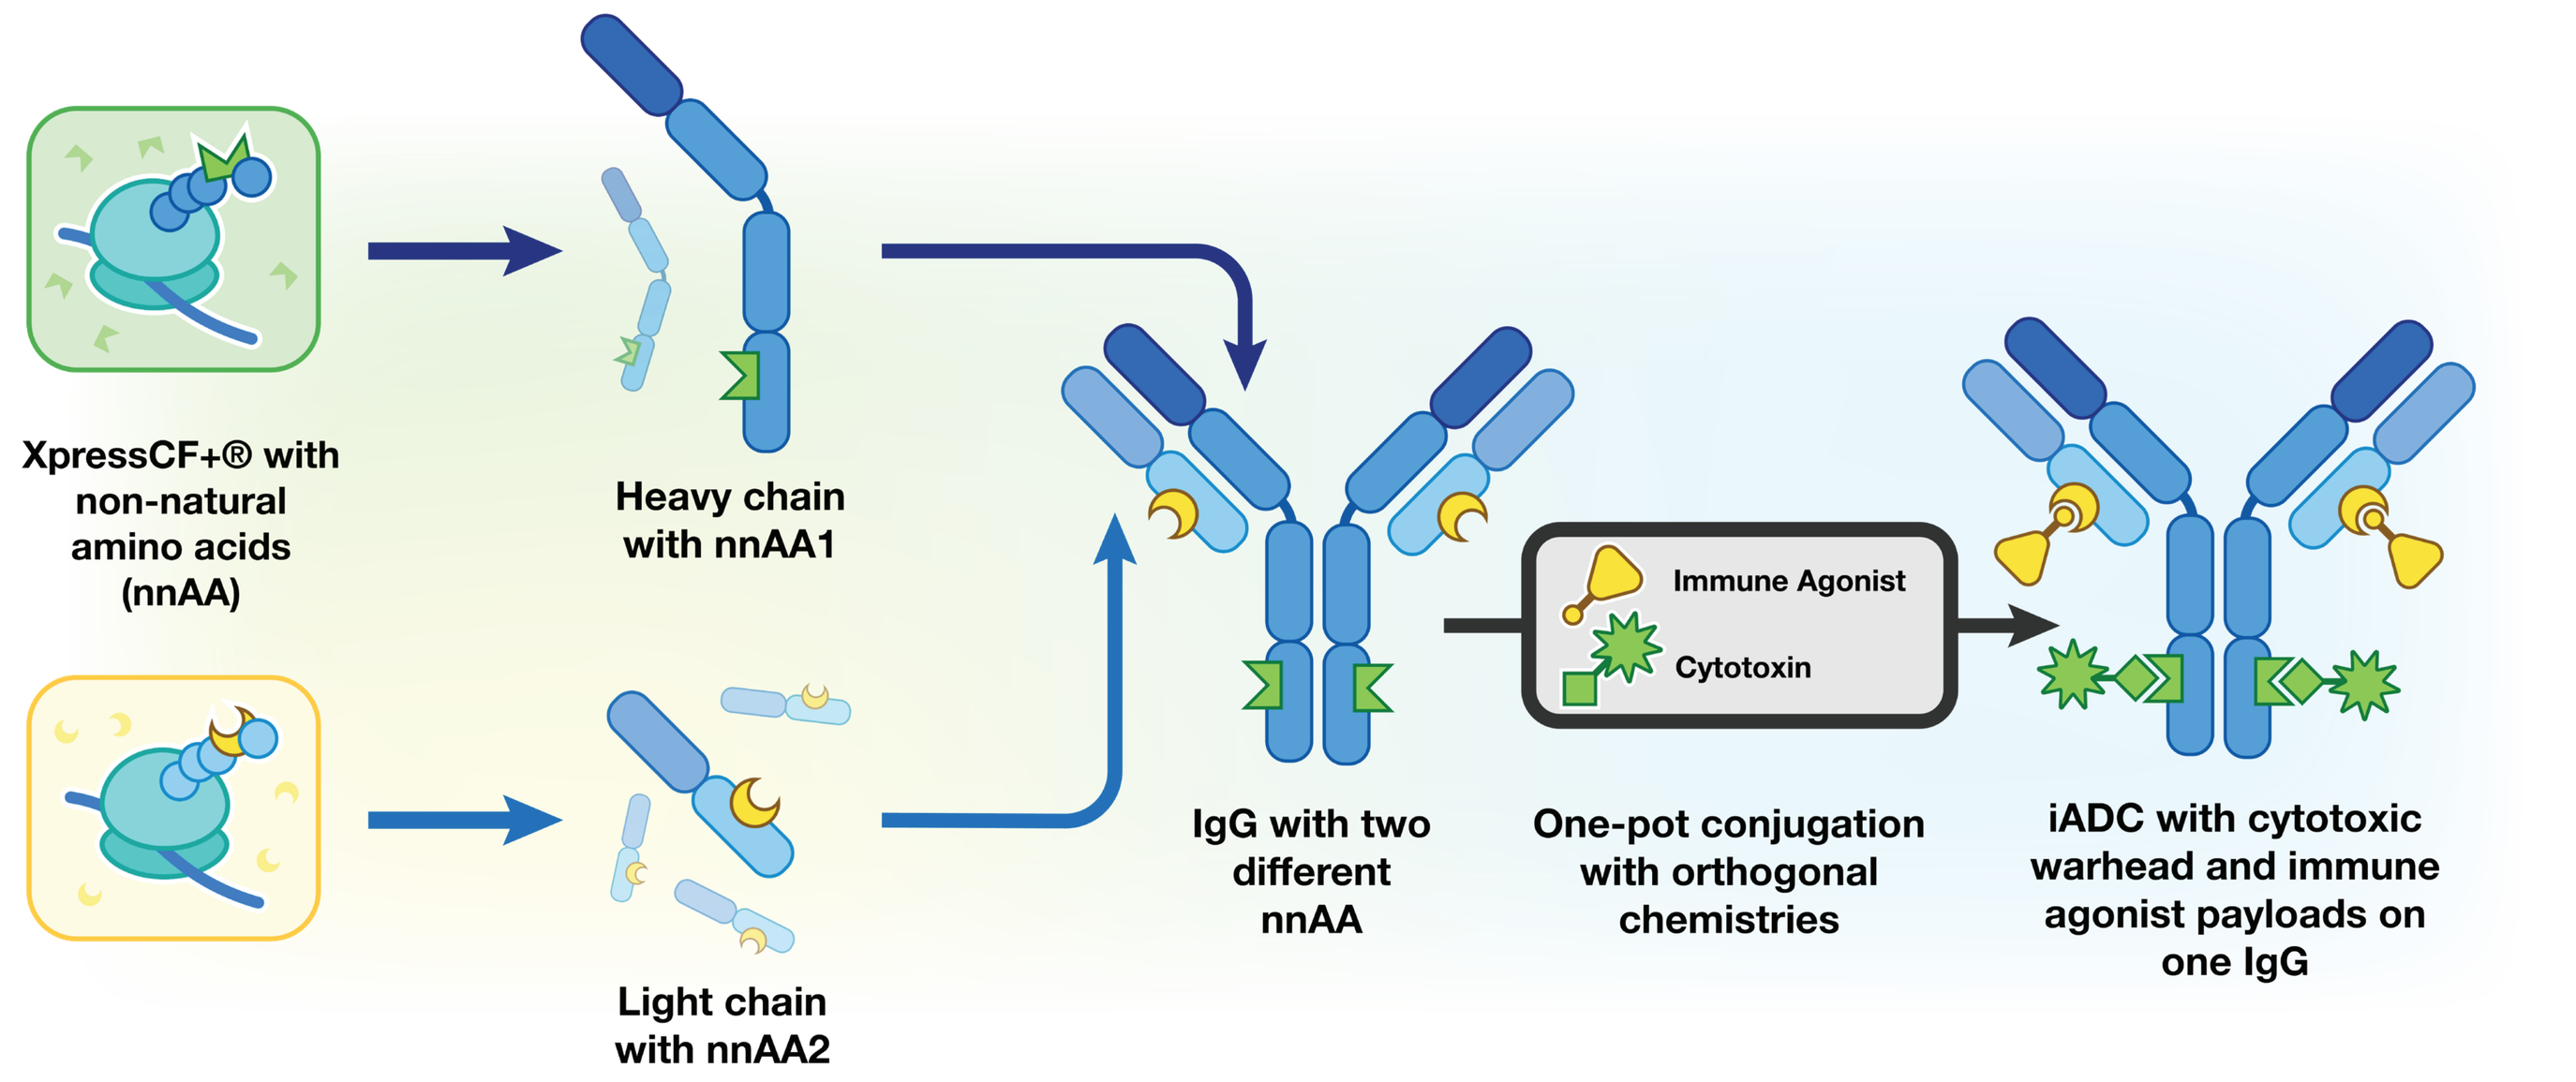
XpressCF+® with non-natural amino acids (nnAA)
Heavy chain with nnAA1
Light chain with nnAA2
IgG with two different nnAA
Immune Agonist
Cytotoxin
One-pot conjugation with orthogonal chemistries
iADC with cytotoxic warhead and immune agonist payloads on one IgG

Figure 3. Sutro's XpressCF+® system enables homogenous dual conjugation of a cytotoxic and immune agonist payload on a single antibody. The novel immunostimulatory ADC (iADC) design employs precise tumor-targeting of both the cytotoxin and the immune activator to allow systemic delivery and is intended to merge direct tumor cell killing and priming of an adaptive anti-tumor response in a monotherapy. iADC molecules made in cell free are not glycosylated, do not bind to Fc gamma receptors, and do not result in ADCC.

iADCs enable off-the-shelf, systemically administered *in situ* immunization

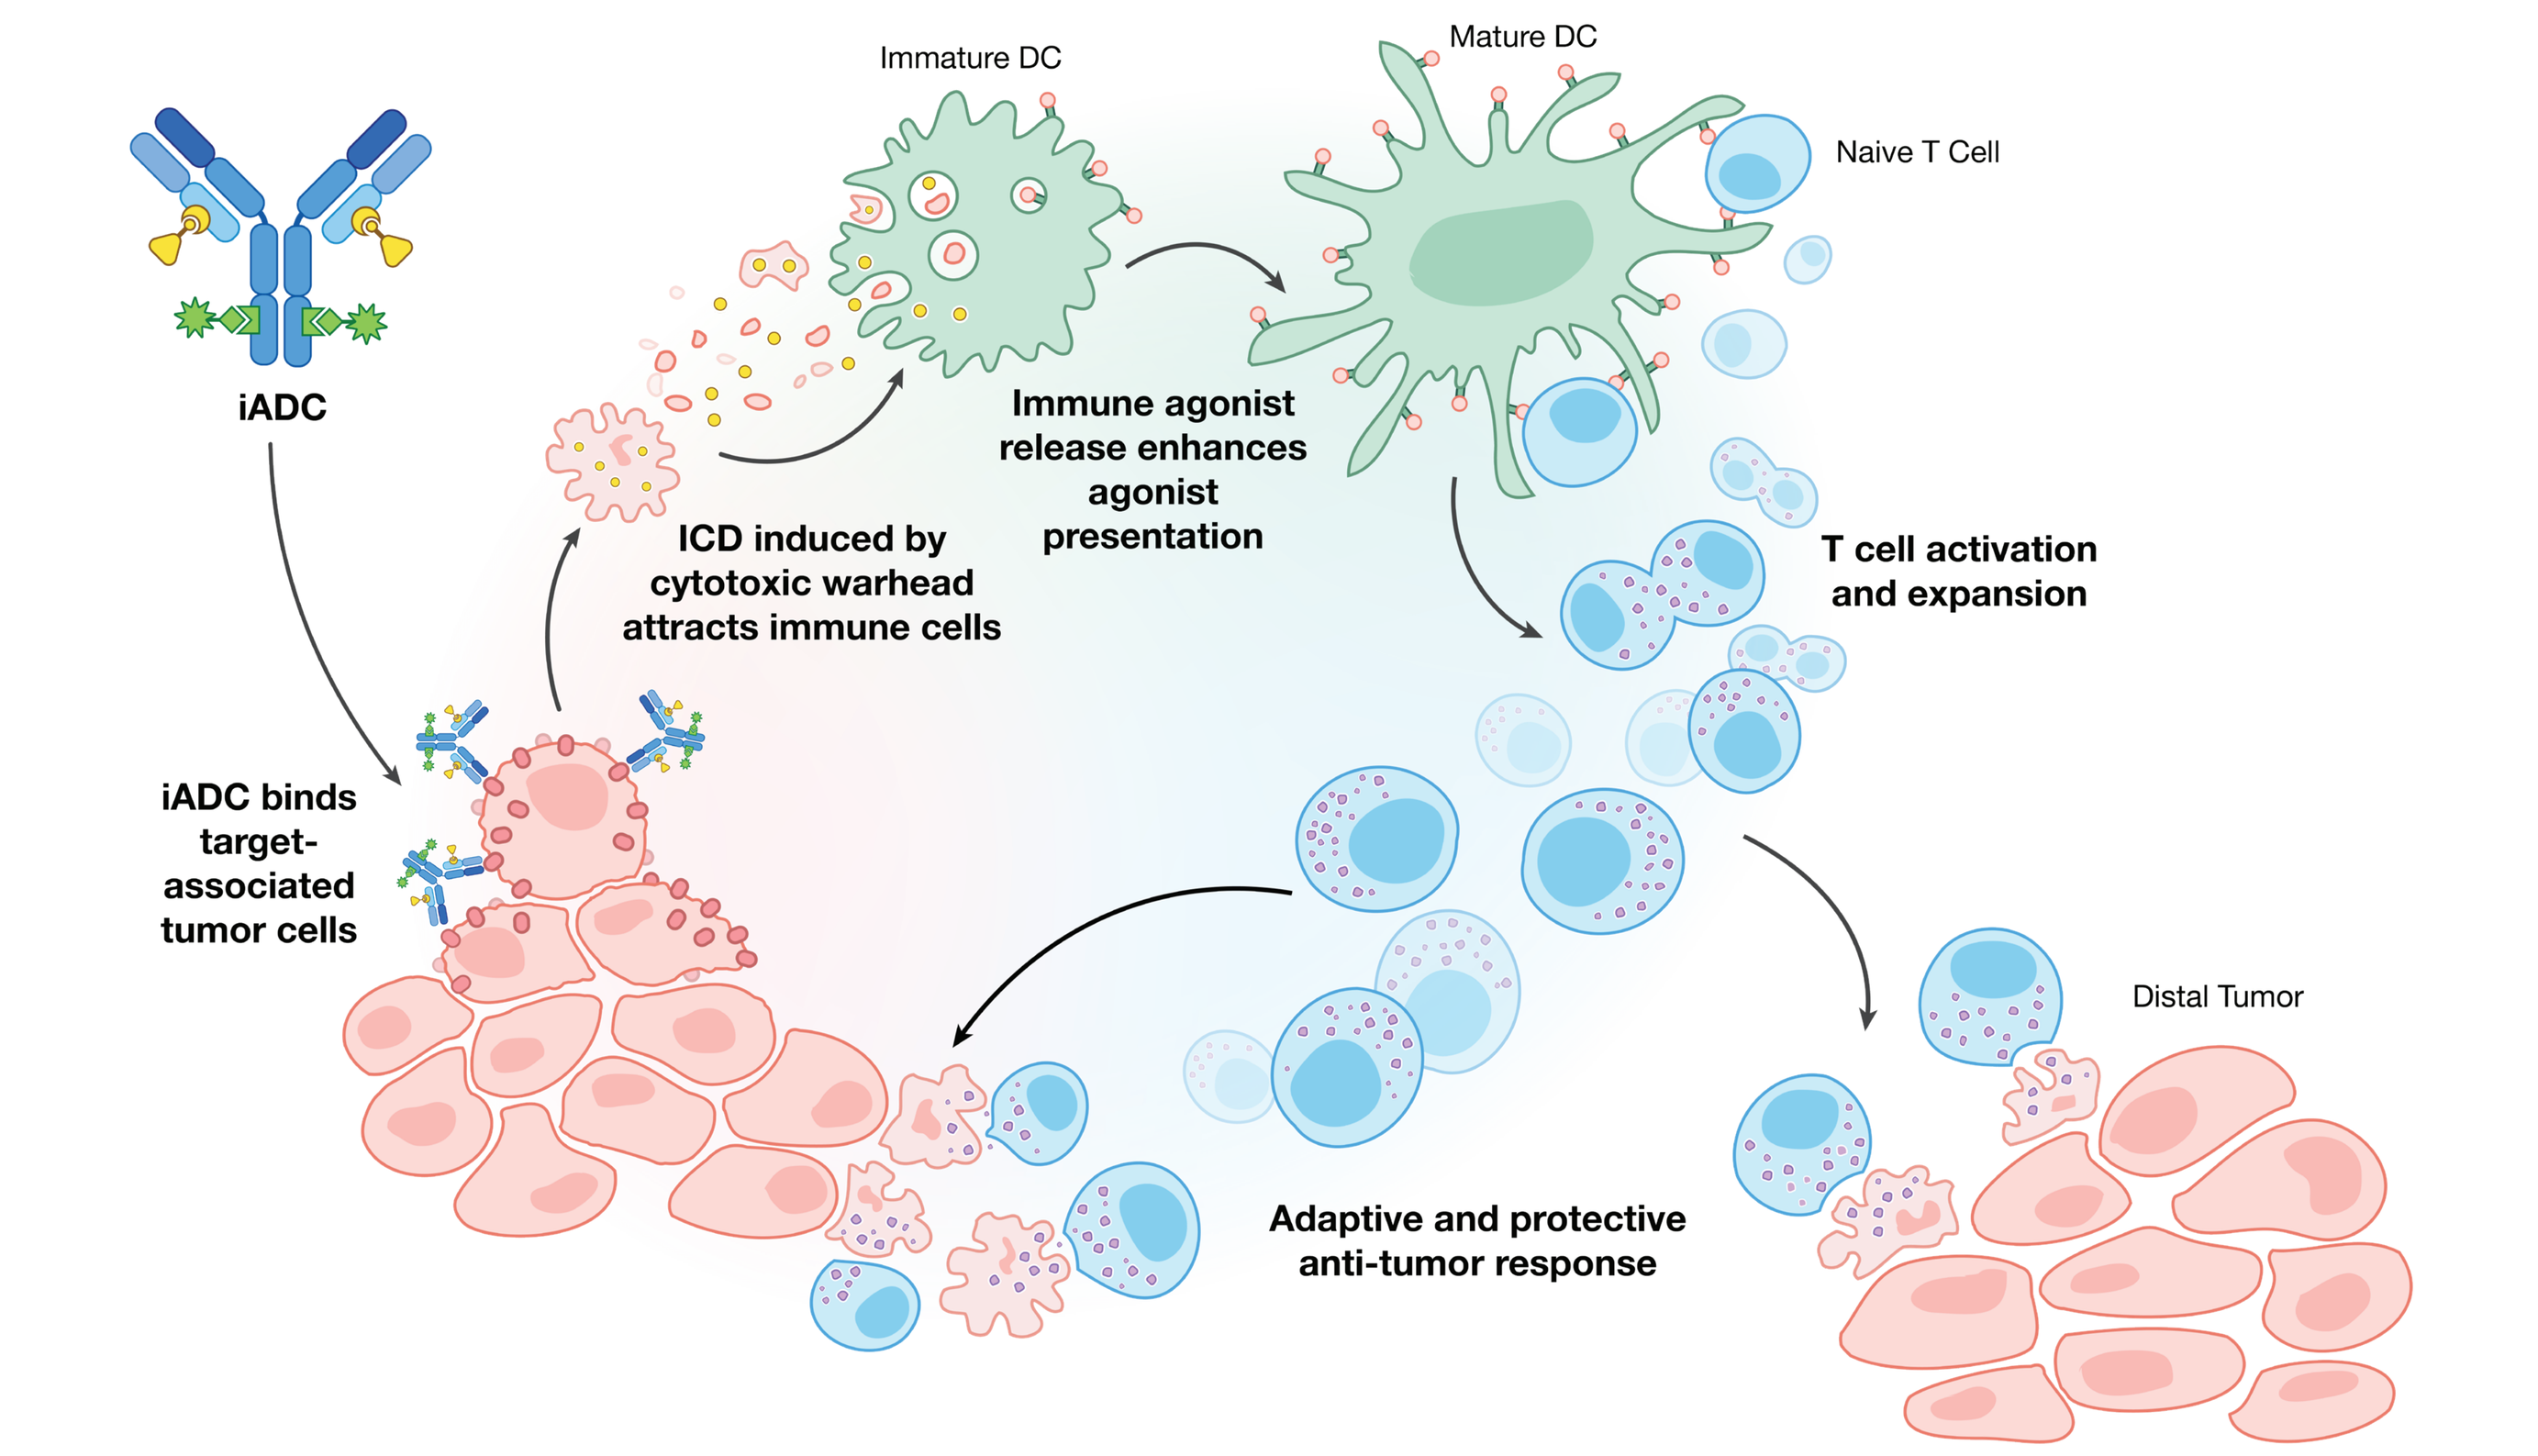
iADC
Immature DC
Mature DC
Naive T Cell
ICD induced by cytotoxic warhead attracts immune cells
Immune agonist release enhances agonist presentation
T cell activation and expansion
iADC binds target-associated tumor cells
Distal Tumor
Adaptive and protective anti-tumor response

Figure 4. iADCs promote anti-tumor immunity by specifically delivering cytotoxic and immune-activating payloads to target-expressing tumor cells. The cytotoxic payload induces tumor cell death and release of the immune agonist, in conjunction with tumor antigens, resulting in development of enhanced protective anti-tumor immunity.

Proof-of-concept hemiasterlin and TLR7 agonist iADC maintains FolRα-dependent cell killing

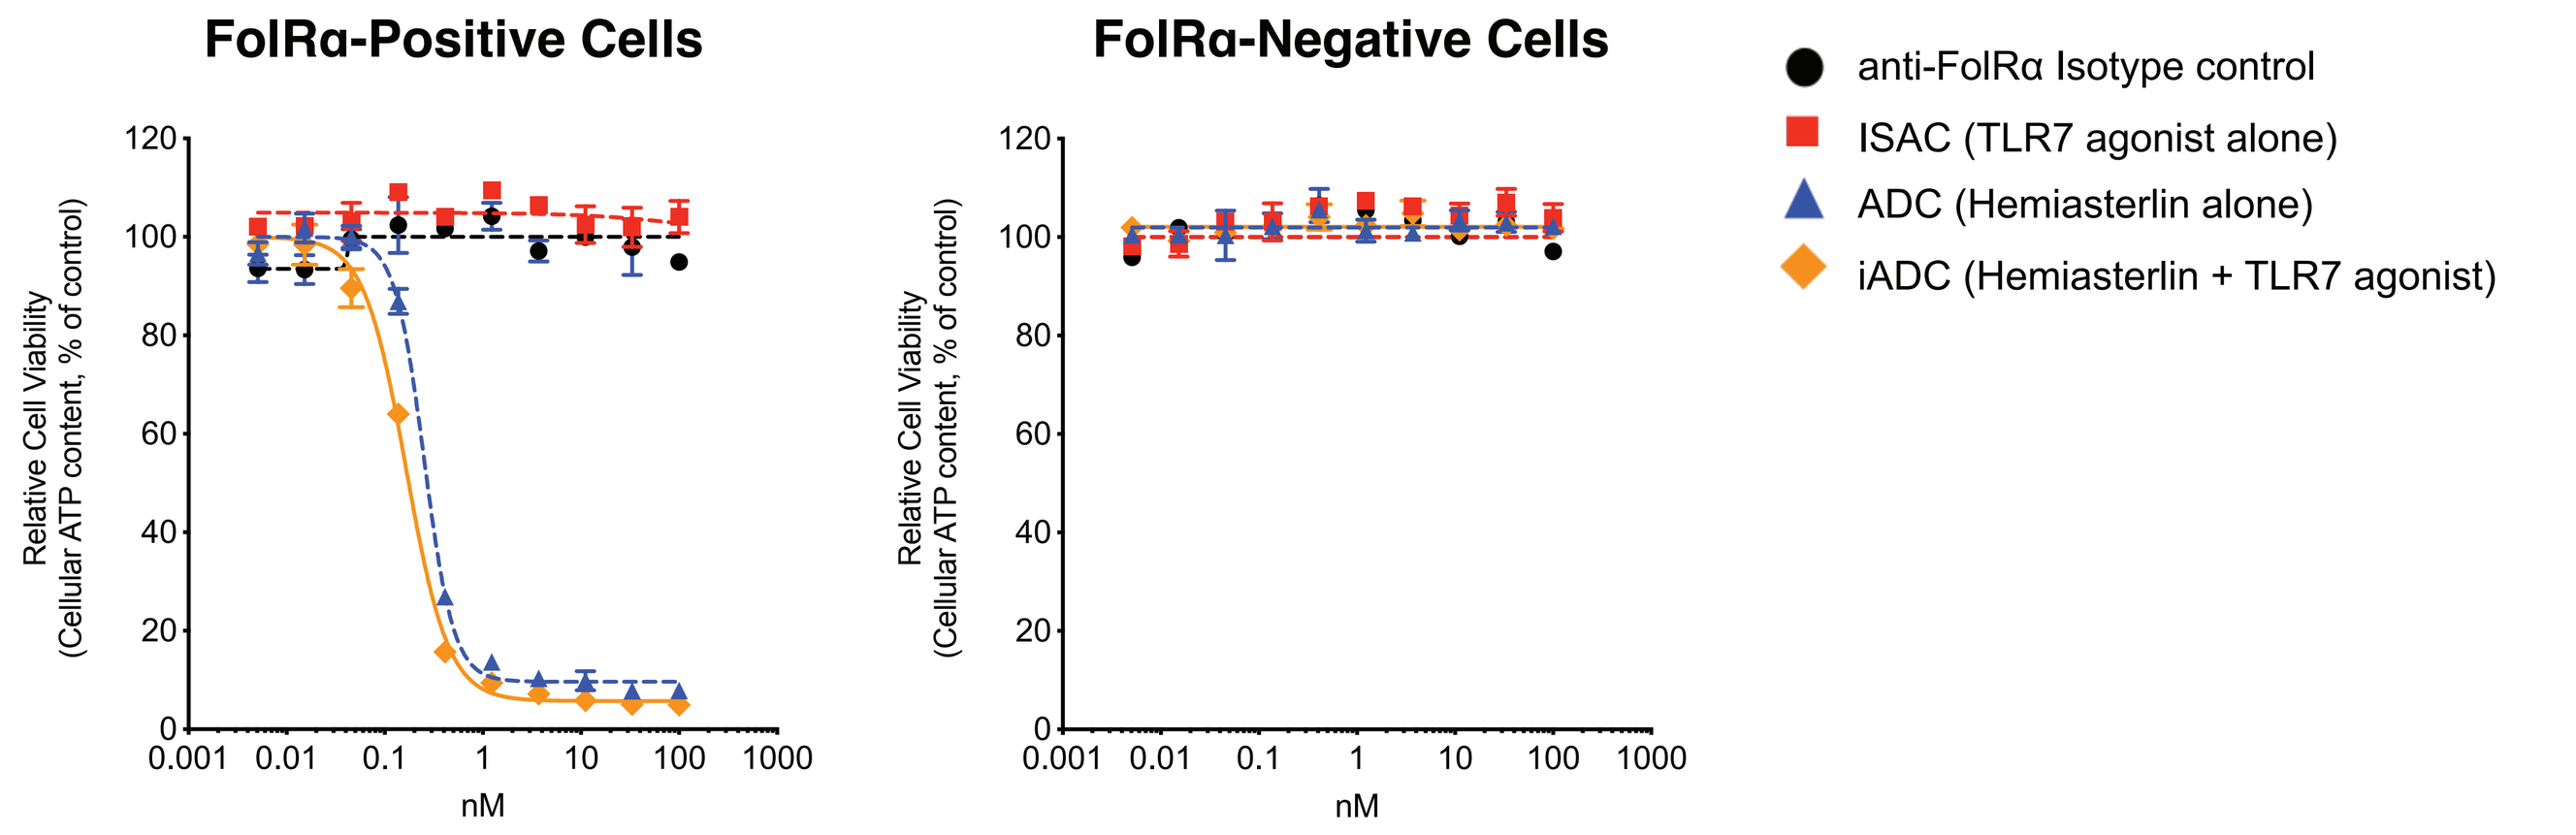
FolRα-Positive Cells
FolRα-Negative Cells
Relative Cell Viability (Cellular ATP content, % of control)
nM
anti-FolRα Isotype control
ISAC (TLR7 agonist alone)
ADC (Hemiasterlin alone)
iADC (Hemiasterlin + TLR7 agonist)

Figure 5. hFolRα⁺ tumor cells (target positive) or hFolRα⁻ cells (target negative) were incubated *in vitro* with an anti-FolRα ISAC (TLR7 agonist only), ADC (hemiasterlin only), or iADC (hemiasterlin + TLR7 agonist) for 5 days before measuring cell viability. Incubation with the ADC and iADC led to potent cell killing of target-positive cells but not target-negative cells, indicating retention of target-dependent cell killing with the iADC molecule. The control and ISAC molecules did not result in any measurable cell killing of either target-positive or target-negative cells.

iADC induces potent activation of immune cells in human PBMC and tumor cell co-cultures

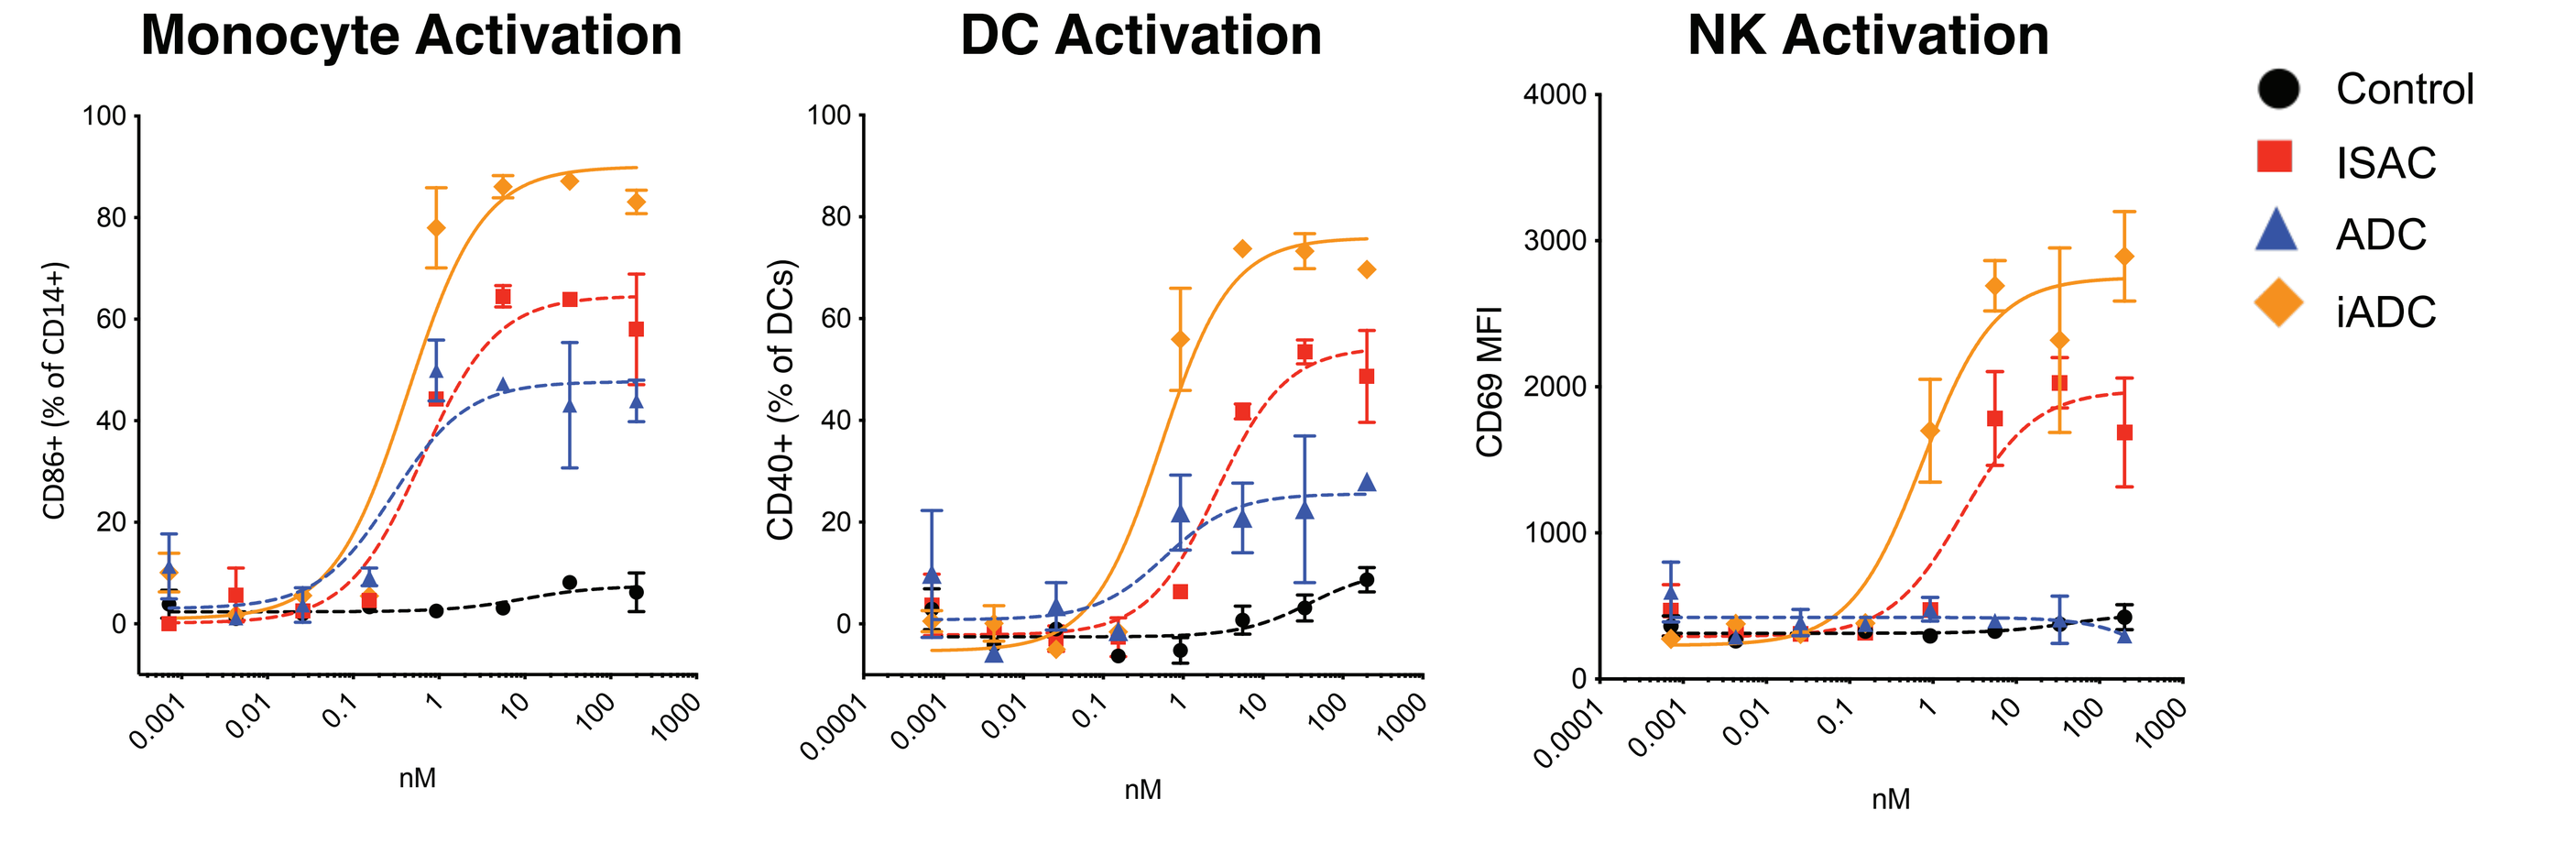
Monocyte Activation
DC Activation
NK Activation
CD86+ (% of CD14+)
CD40+ (% of DCs)
CD69 MFI
nM
Control
ISAC
ADC
iADC

Figure 6. Human PBMCs and hFolRα⁺ tumor cells were co-cultured *in vitro* and treated with the ISAC, ADC, or iADC for 48 hours. All molecules led to activation of the human monocytes and dendritic cells (DCs), but notably, the iADC showed enhanced activation compared to the ADC or ISAC alone. For natural killer (NK) cells, the ADC alone did not induce significant activation, but the iADC, again, showed more potent activation of the immune cells compared to the ISAC.

iADC treatment results in a greater number of complete regressions compared to treatment with ADC alone

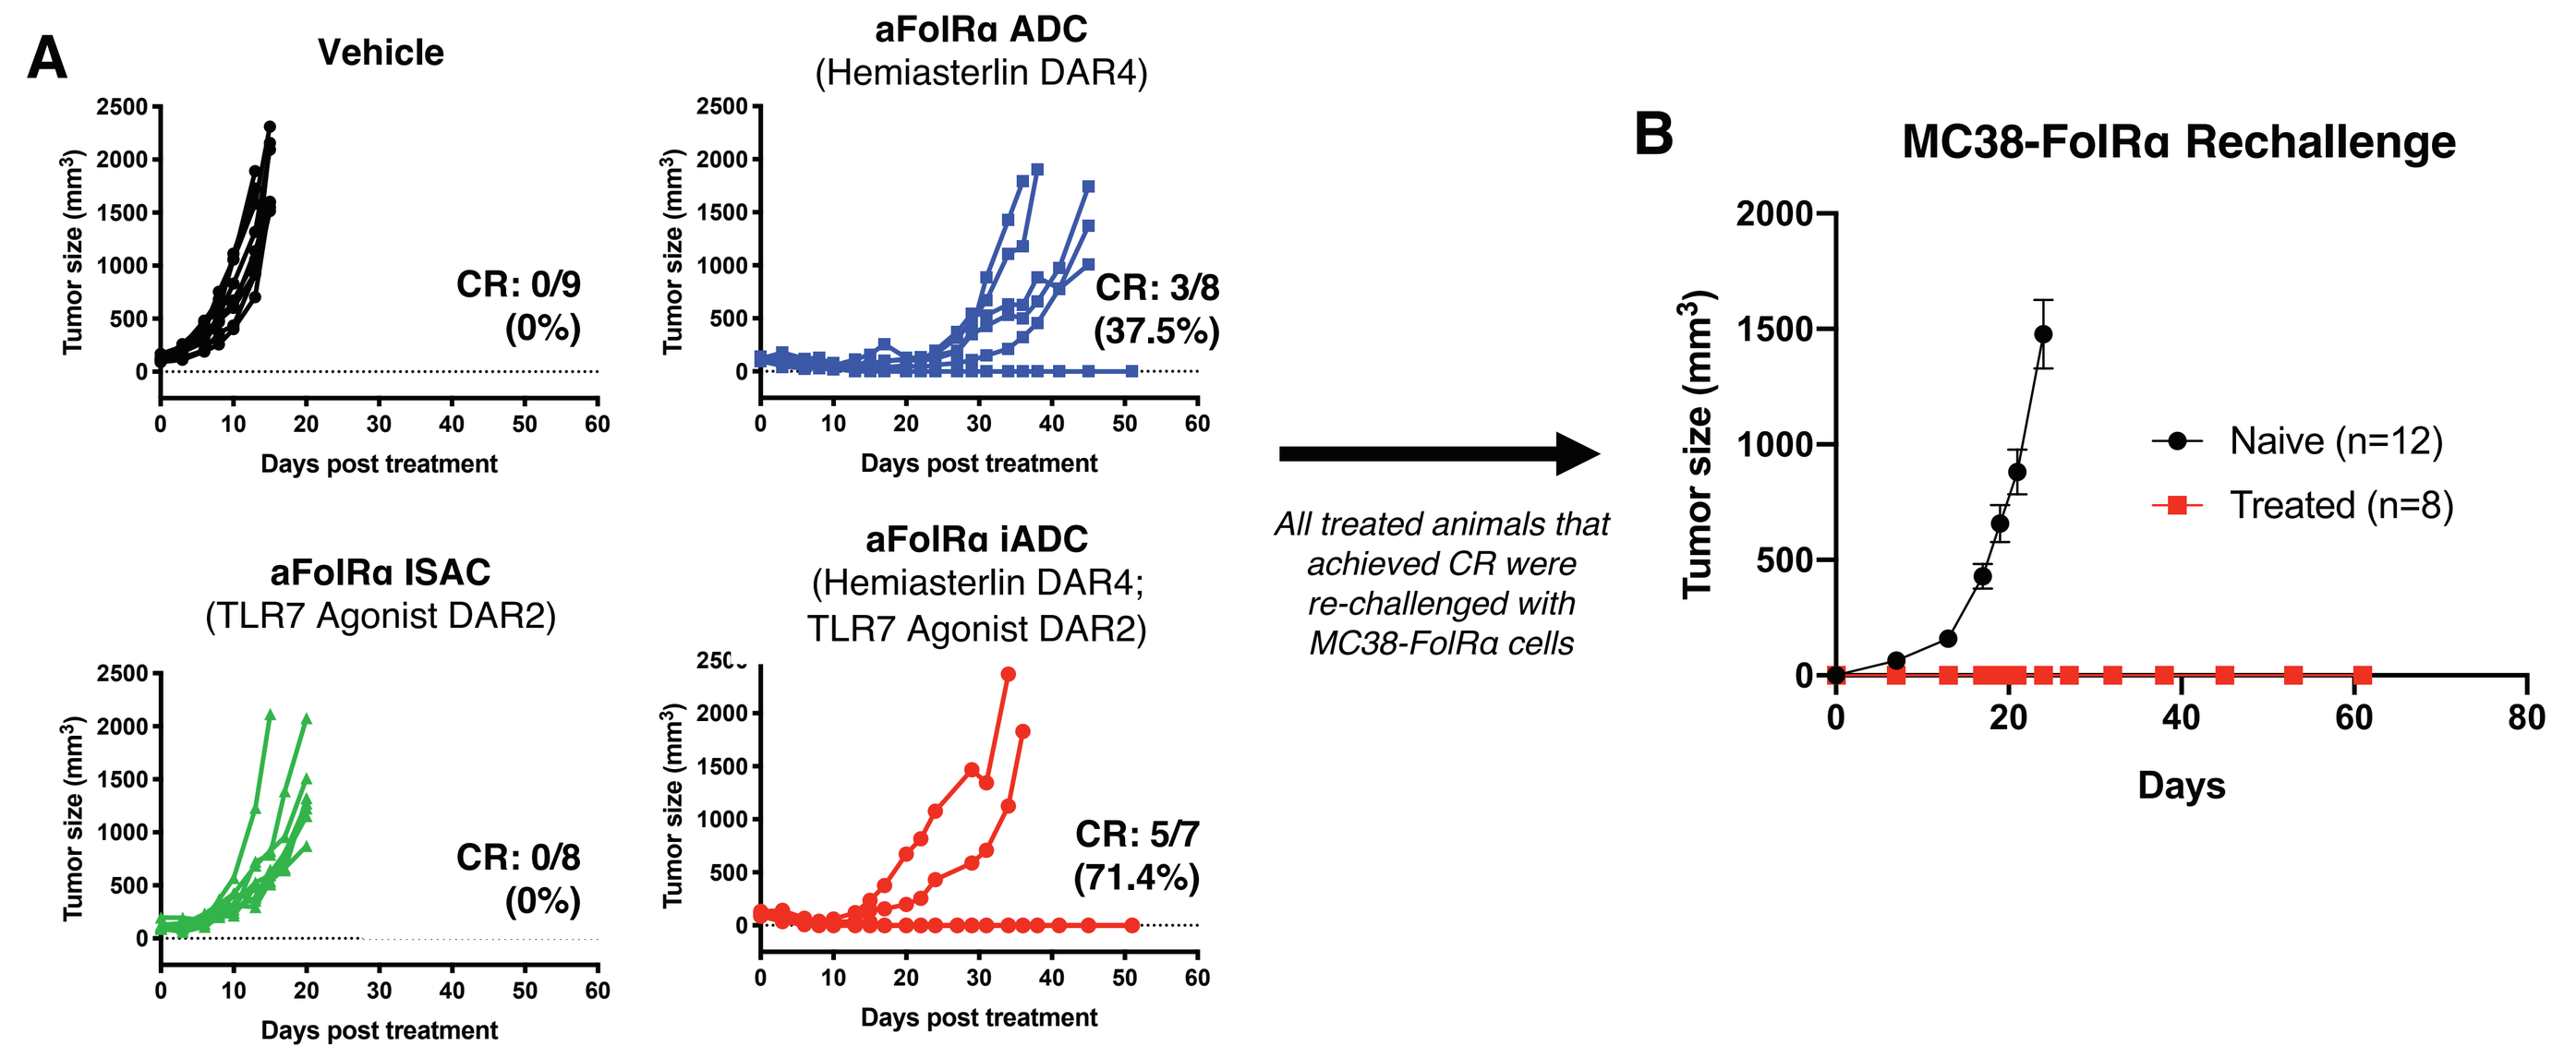
A
Vehicle
CR: 0/9 (0%)
aFolRα ADC (Hemiasterlin DAR4)
CR: 3/8 (37.5%)
aFolRα ISAC (TLR7 Agonist DAR2)
CR: 0/8 (0%)
aFolRα iADC (Hemiasterlin DAR4; TLR7 Agonist DAR2)
CR: 5/7 (71.4%)
Tumor size (mm³)
Days post treatment
All treated animals that achieved CR were re-challenged with MC38-FolRα cells
B
MC38-FolRα Rechallenge
Naive (n=12)
Treated (n=8)
Days

Figure 7. (A) Murine MC38 cells were engineered to express human FolRα (MC38-hFolRα) and implanted into naïve C57BL/6 mice and treated with a single dose of the ADC, ISAC, or iADC at 20 mg/kg. Individual tumor growth curves of the MC38-hFolRα tumors are depicted. Notably, the iADC treatment achieves a greater proportion of complete responses (CR) compared to the ADC treatment (71% vs 38%, respectively). (B) All complete responders remained tumor-free when subsequently rechallenged with MC38-hFolRα cells, indicating acquisition of long-lasting protective immunity.

iADC treatment engages the innate and adaptive immune compartments in MC38-hFolRα tumors

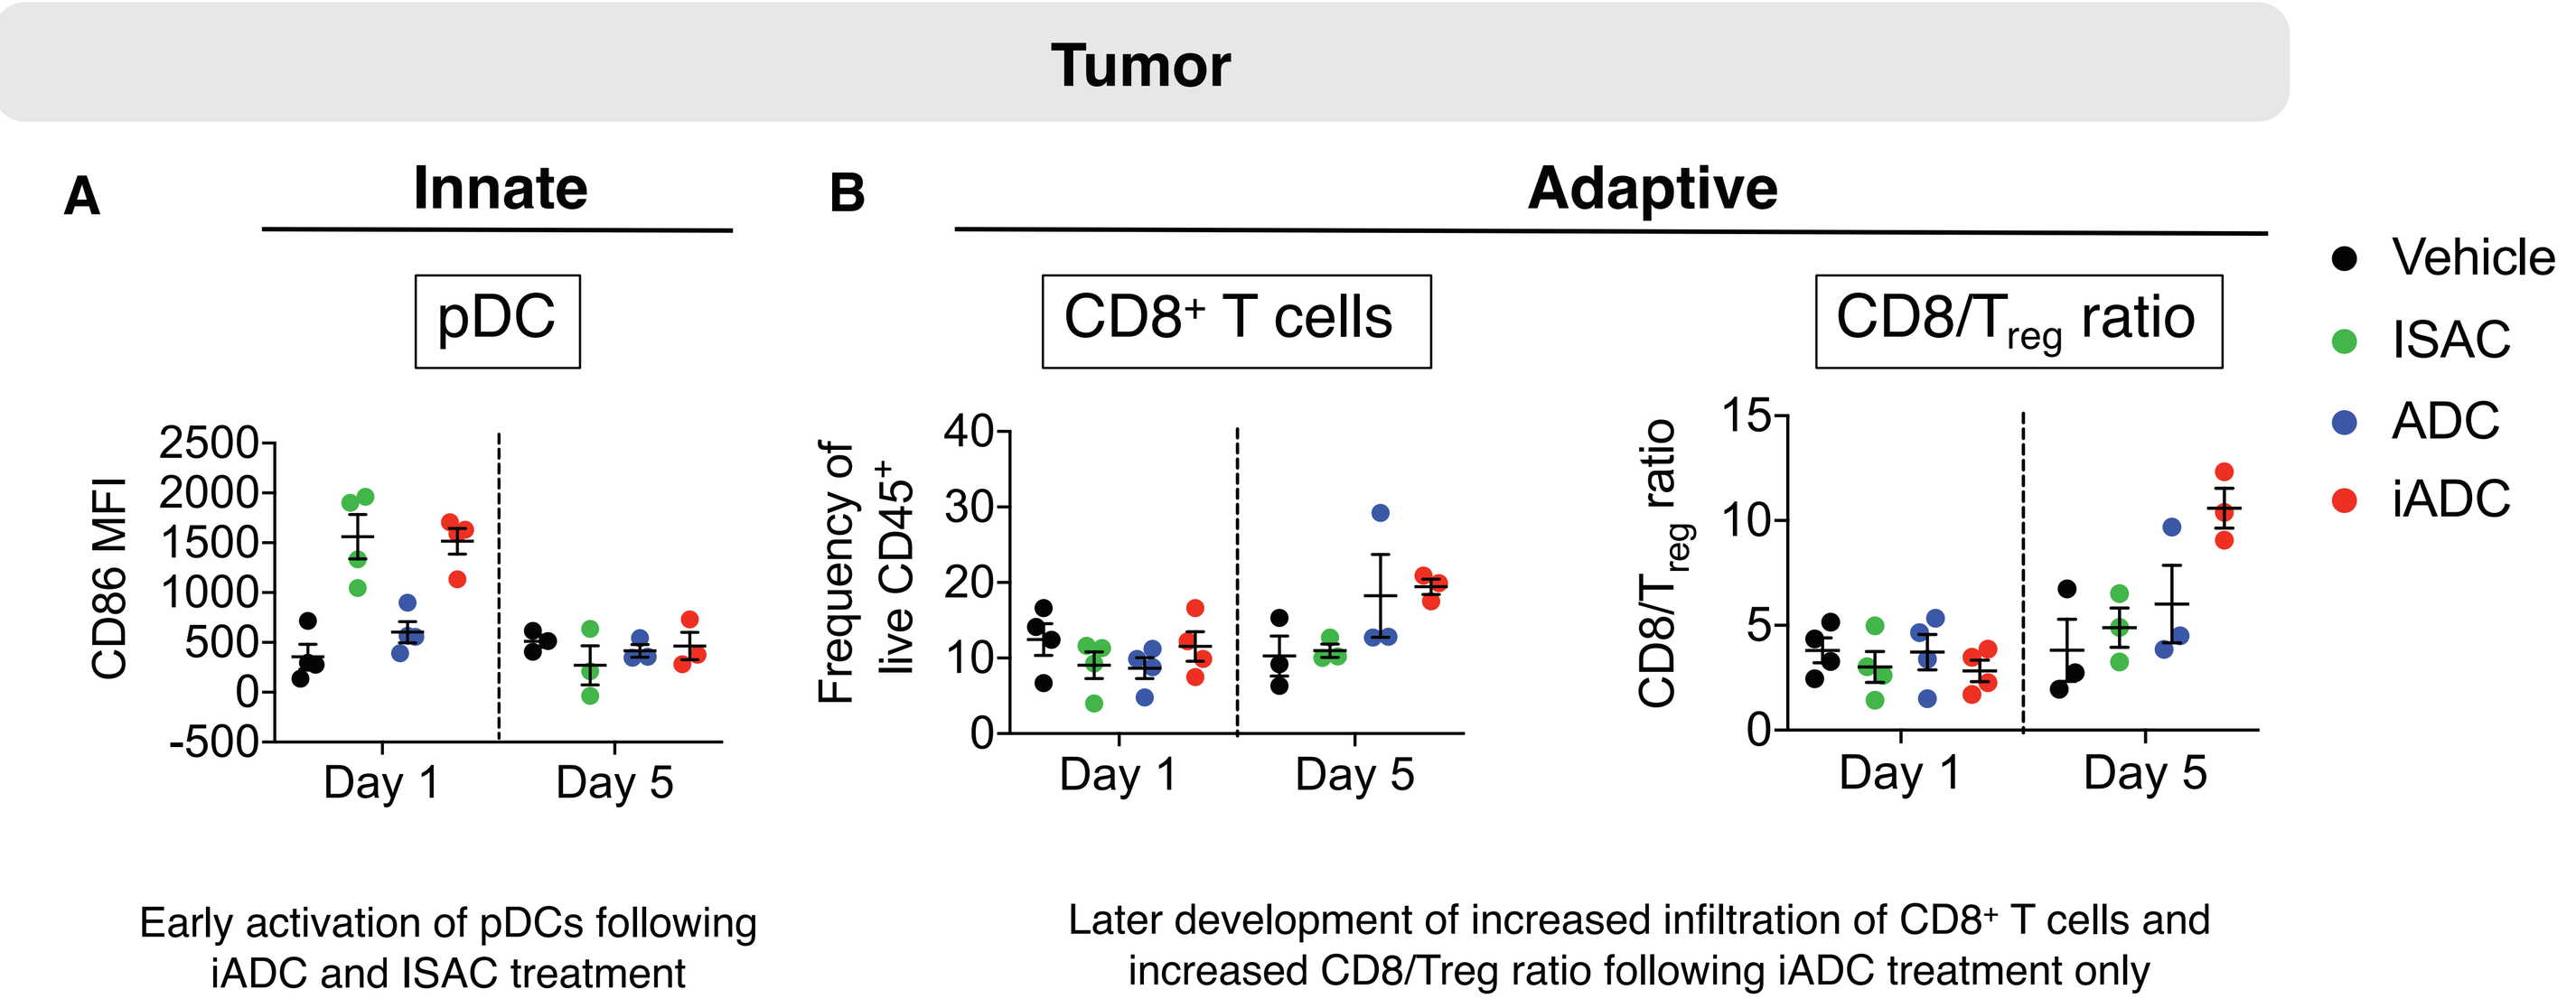
Tumor
A
Innate
pDC
CD86 MFI
B
Adaptive
CD8⁺ T cells
Frequency of live CD45⁺
CD8/Treg ratio
Day 1
Day 5
Vehicle
ISAC
ADC
iADC
Early activation of pDCs following iADC and ISAC treatment
Later development of increased infiltration of CD8⁺ T cells and increased CD8/Treg ratio following iADC treatment only

Figure 8. Flow cytometry analysis of MC38-hFolRα tumors treated with a single dose of the ISAC, ADC, or iADC at 10 mg/kg revealed (A) activation of tumor-infiltrating plasmacytoid dendritic cells (pDCs) following treatment with the ISAC and iADC, but not the ADC, one day after treatment and (B) increased tumoral $CD8^+$ T cell infiltration and CD8/Treg ratios following treatment with the iADC five days post treatment. Thus, a combination of both the cytotoxic payload and the immune agonist is necessary for greater stimulation of the $CD8^+$ T cell response.

Tumoral infiltration of $CD8^+$ T cells is enhanced following iADC therapy

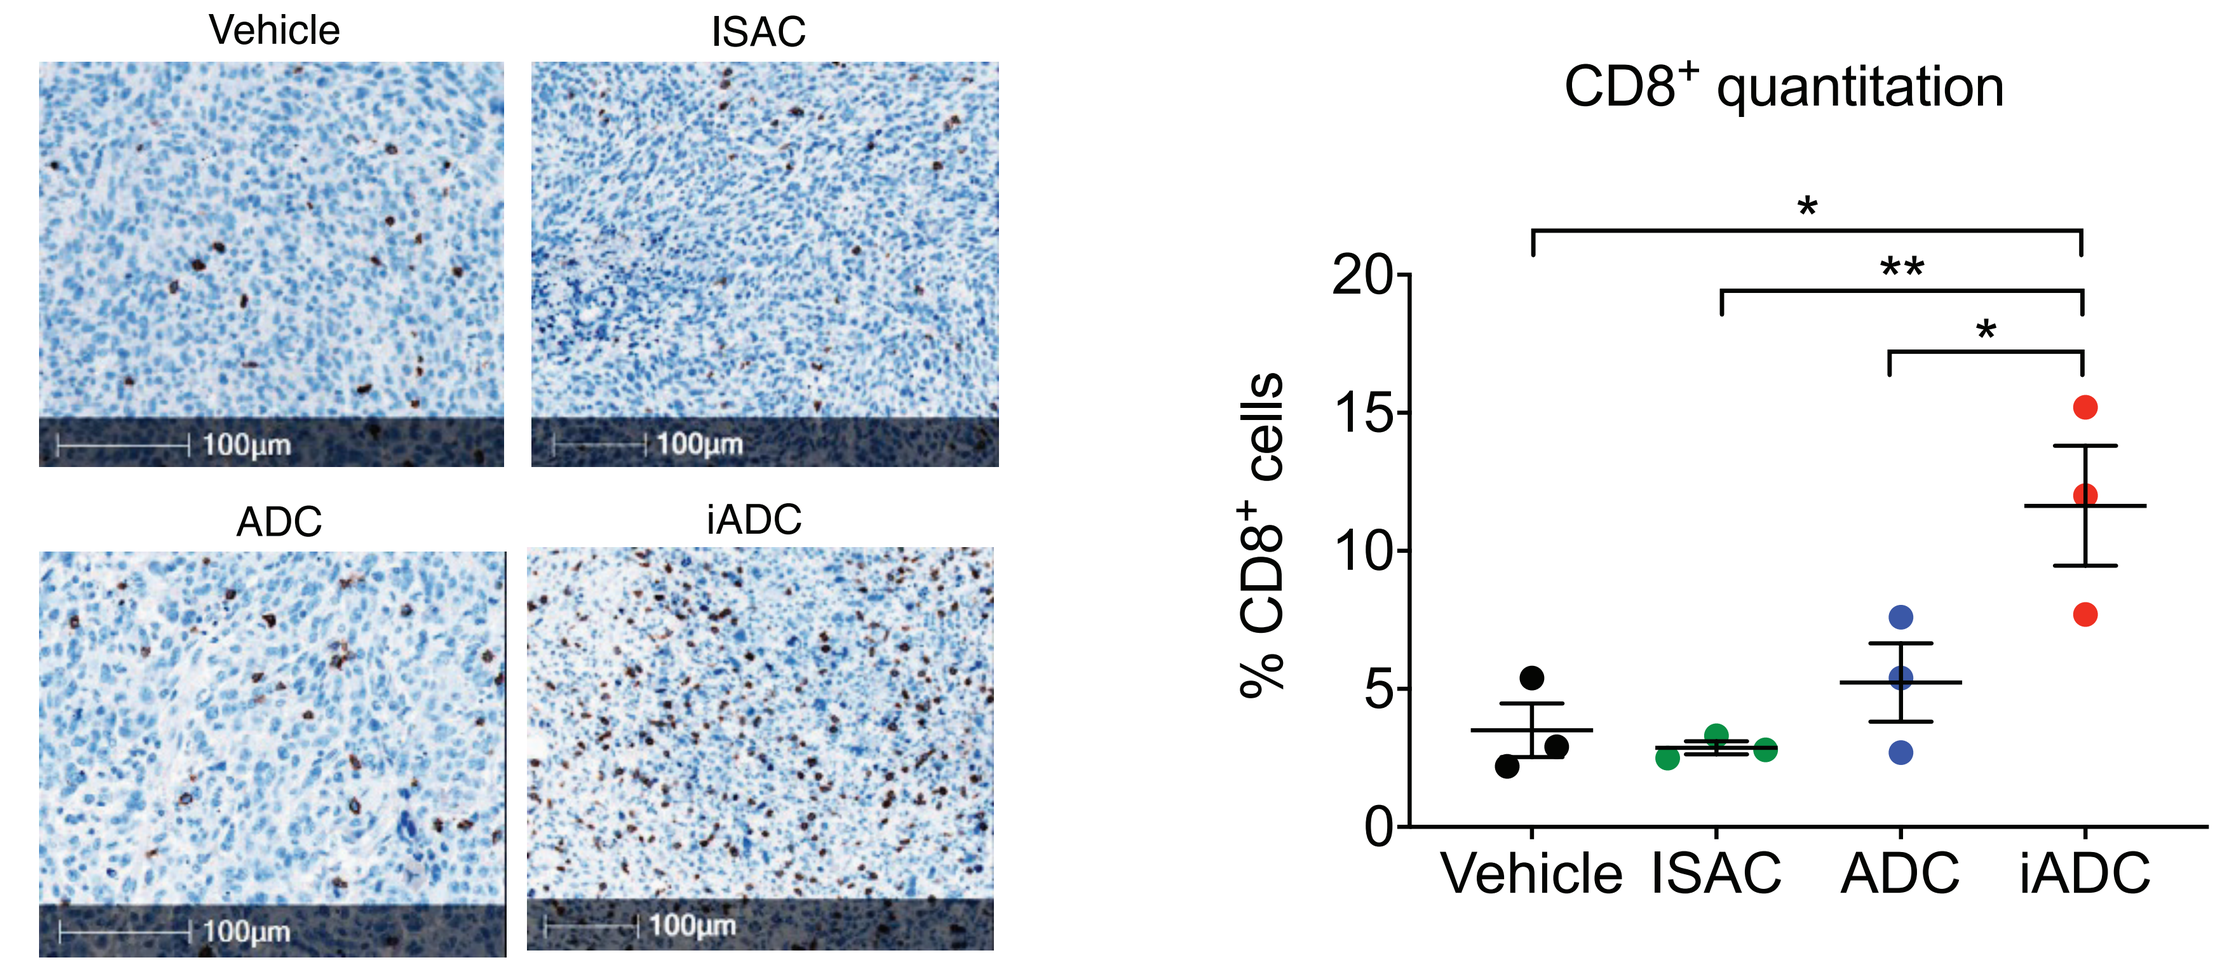
Vehicle
ISAC
ADC
iADC
100µm
CD8⁺ quantitation
% CD8⁺ cells
*
**
*

Figure 9. MC38-hFolRα tumors treated with a single dose of the ISAC, ADC, or iADC at 10 mg/kg were harvested five days post treatment and stained for CD8 and counter-stained with hematoxylin to assess cytotoxic T cell infiltration (left panels). Quantification of $CD8^+$ cells among total cells shows increased $CD8^+$ T cell infiltration in the tumor following only treatment with the iADC (right panel).

Protective anti-tumor immunity induced by iADC treatment is dependent on $CD8^+$ T cells

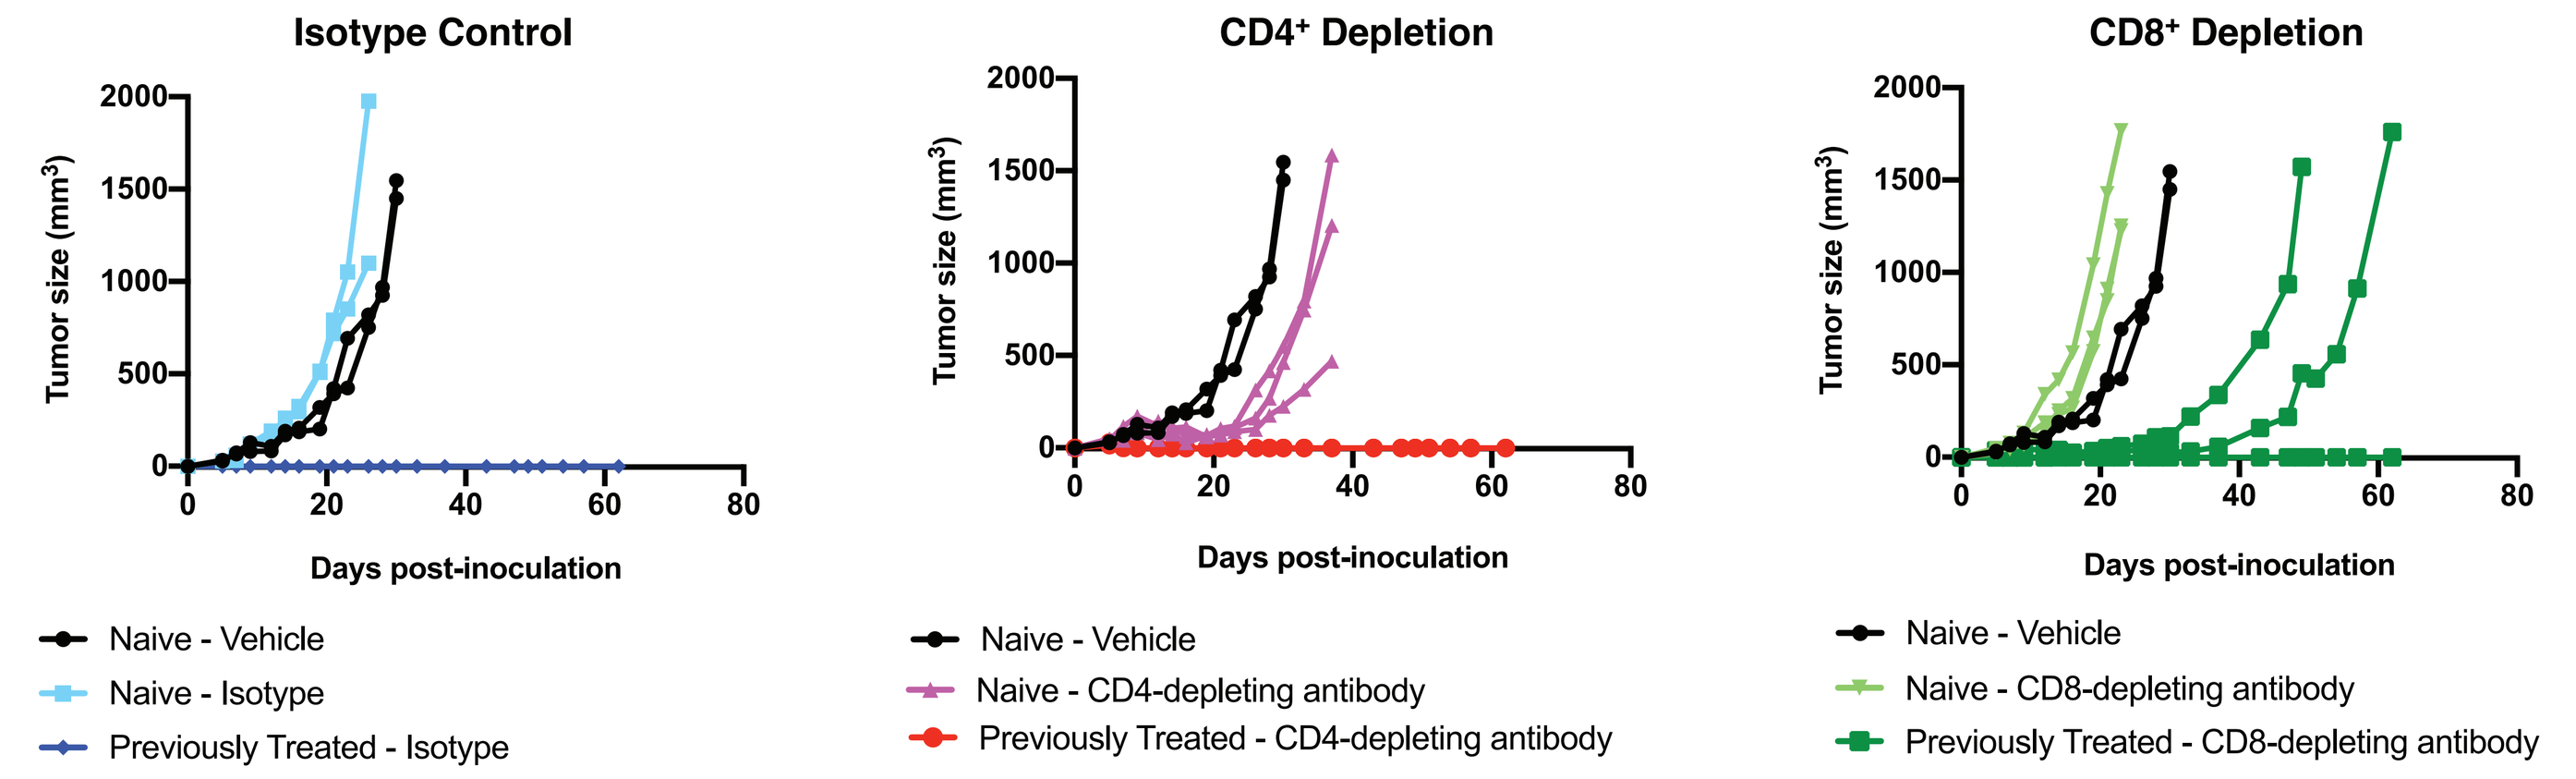
Isotype Control
CD4⁺ Depletion
CD8⁺ Depletion
Tumor size (mm³)
Days post-inoculation
Naive - Vehicle
Naive - Isotype
Previously Treated - Isotype
Naive - CD4-depleting antibody
Previously Treated - CD4-depleting antibody
Naive - CD8-depleting antibody
Previously Treated - CD8-depleting antibody

Figure 10. MC38-hFolRα-bearing mice that achieved complete responses after iADC treatment were rechallenged with MC38-hFolRα and administrated isotype, CD4-depleting, or CD8-depleting antibodies. When $CD8^+$ T cells were depleted, tumor growth was restored after rechallenge in 2/3 animals previously treated with iADC while protective immunity was maintained in all previously treated mice receiving isotype control or the CD4-depleting antibody.

iADC induces durable anti-tumor immunity that is independent of FolRα antigen expression

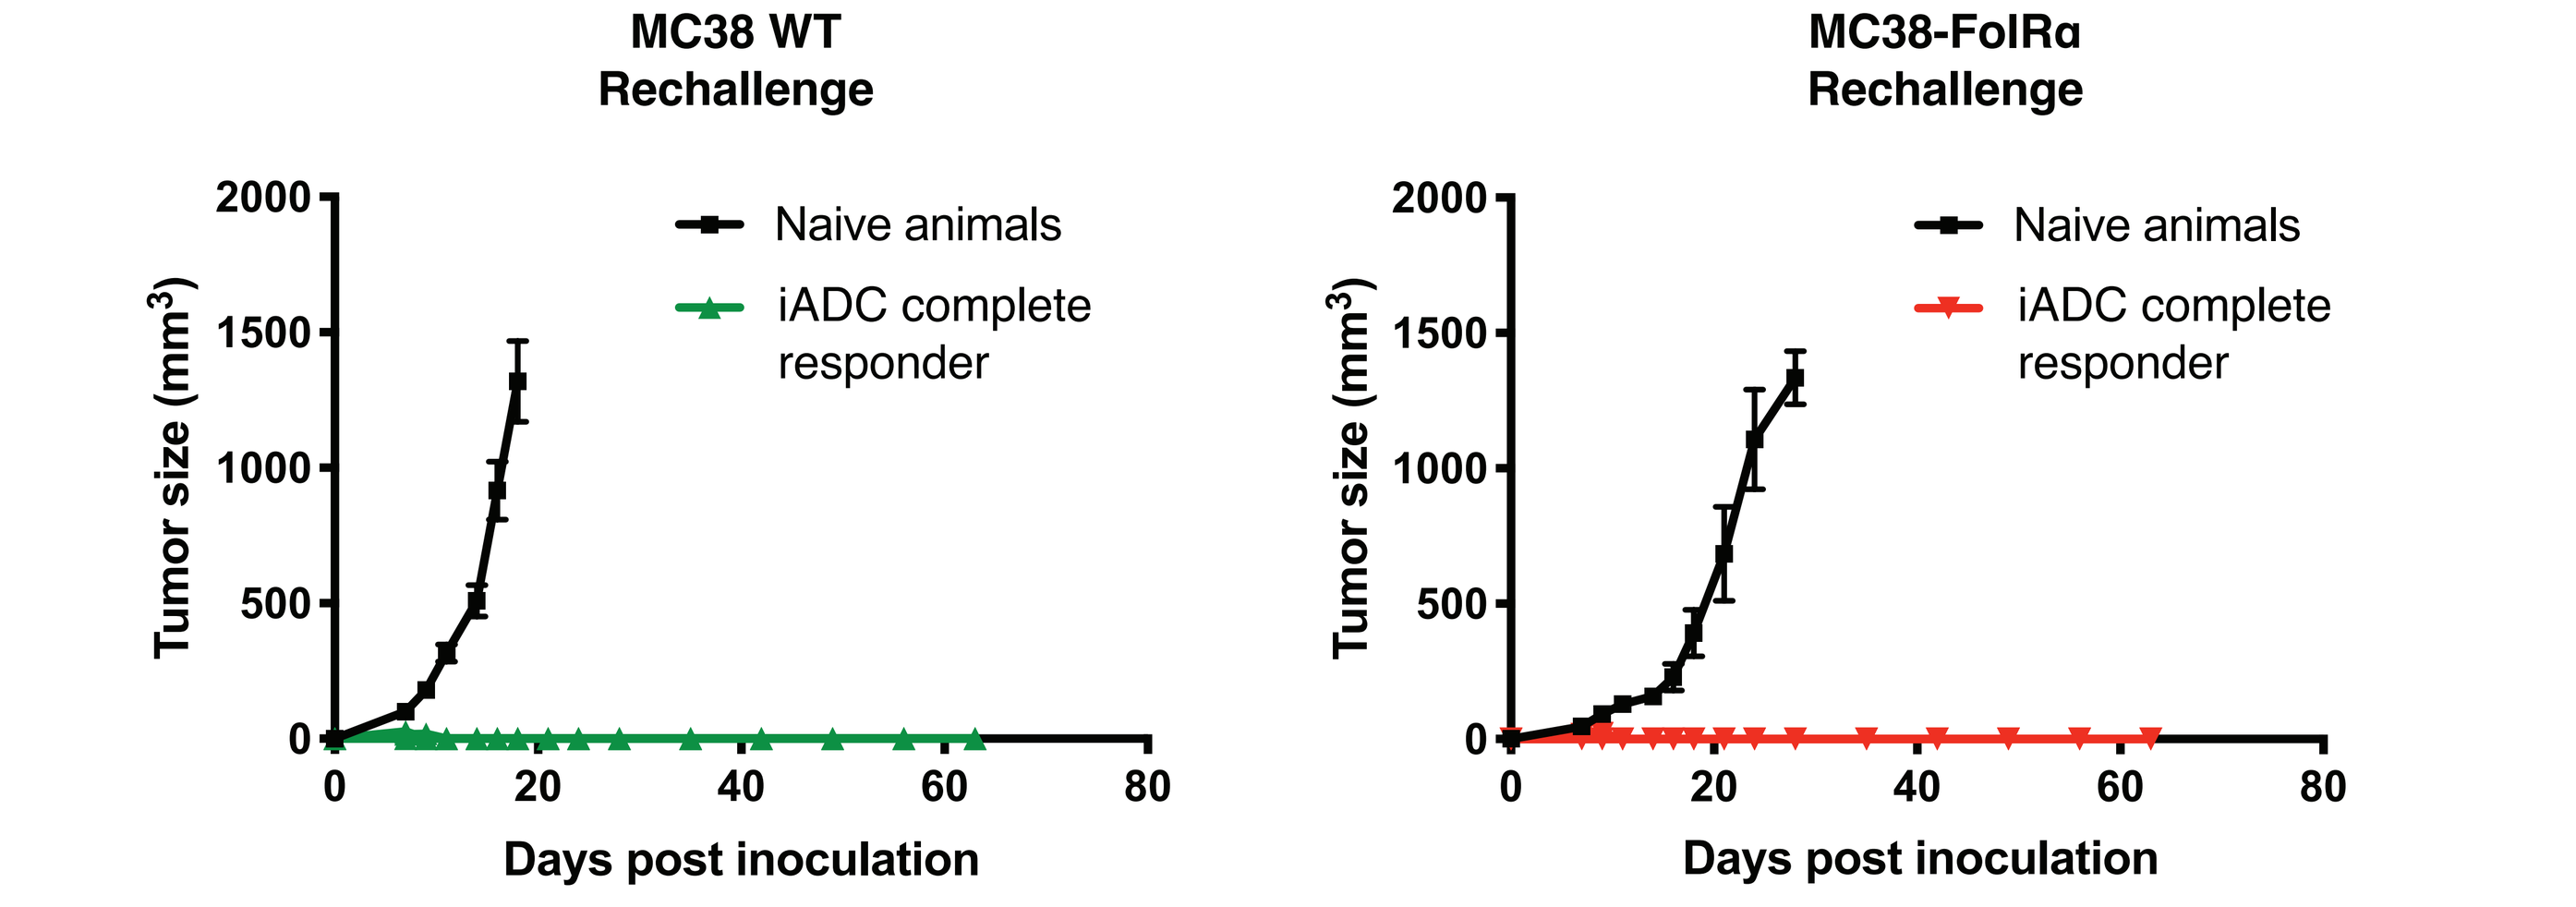
MC38 WT Rechallenge
MC38-FolRα Rechallenge
Tumor size (mm³)
Days post inoculation
Naive animals
iADC complete responder

Figure 11. MC38-hFolRα-bearing mice that achieved complete responses after iADC treatment were rechallenged with either wild-type MC38 or MC38-hFolRα. Complete rejection of wild-type MC38 in addition to MC38-hFolRα indicates protective anti-tumor immunity is established independently of the hFolRα antigen.

Concluding Remarks

- iADC is a novel monotherapy that combines two mechanisms of action to effectively kill the tumor and stimulate a more robust anti-tumor response.
- iADCs can engage the innate and adaptive immune response, increase $CD8^+$ T cell infiltration into tumors, and generate durable and protective immunity.
- iADCs may serve as off-the-shelf therapies that have the potential to work across tumor types by directly killing tumor cells and activating the patient's natural immune response.