Abstract #4849: The anti-ROR1 ADC STRO-003 demonstrates immune-modulating properties that may enhance checkpoint blockade

Andrew McGeehan, Sihong Zhou, Dayson Moreira, Robert Yuan, Indrani Dutta, Jennifer Smith, Millicent Embry, Helena Kiefel, Xiaofan Li, Cuong Tran, Gang Yin, Alice Yam, Kristin Bedard, and Trevor Hallam

SUTRO BIOPHARMA

Summary

- Receptor Orphan Receptor Kinase 1 (ROR1) is an oncofetal transmembrane receptor that is associated with increased invasiveness and resistance of cancer malignancies. In preclinical studies, ROR1 has been implicated in mediating chemoresistance, inducing epithelial-mesenchymal transition (EMT), and driving stem-like properties on tumor initiating cells.
- ROR1 is broadly expressed in solid tumors and hematological malignancies, while having restricted expression in normal adult tissues, thus making it an ideal ADC target. STRO-003 is a potent ROR1-targeted ADC, with site-specific conjugation of a β-glucuronide exatecan linker-payload at a drug-antibody-ratio (DAR) of 8.
- STRO-003 demonstrates potent anti-tumor suppression in preclinical models of breast and lung cancer and significant safety in nonclinical safety studies. STRO-003 was well-tolerated in non-human primates up to 45 mg/kg.
- STRO-003 and its released catabolite exatecan induce markers of immunogenic cell death (ICD) such as HMGB1 release and translocation of calreticulin to the cell surface. Consistent with that finding, STRO-003 additionally induces monocyte activation in a co-culture assay of PBMCs and ROR1-expressing cells.
- Vaccination studies were performed with the exatecan warhead to further explore the significance of STRO-003-induced ICD and protective immunity in vivo. These results demonstrate that tumor cells pre-treated with exatecan undergo potent immunogenic cell death and are capable of mounting a protective immune response *in vivo*, classifying exatecan as a bona fide ICD inducer.
- In line with the observed immunogenic properties, treatment of cold tumors (LLC1-hROR1) with STRO-003 and anti-PD1 showed a trend towards induction of a more inflammatory milieu, supporting that STRO-003 could potentially provide a therapeutic benefit in the context of combination therapy with an immune checkpoint inhibitor.

Our innovative design: STRO-003 is a novel optimized ROR1 ADC, featuring TOPO-1 inhibitors linked with β-glucuronidase cleavable linkers, DAR 8

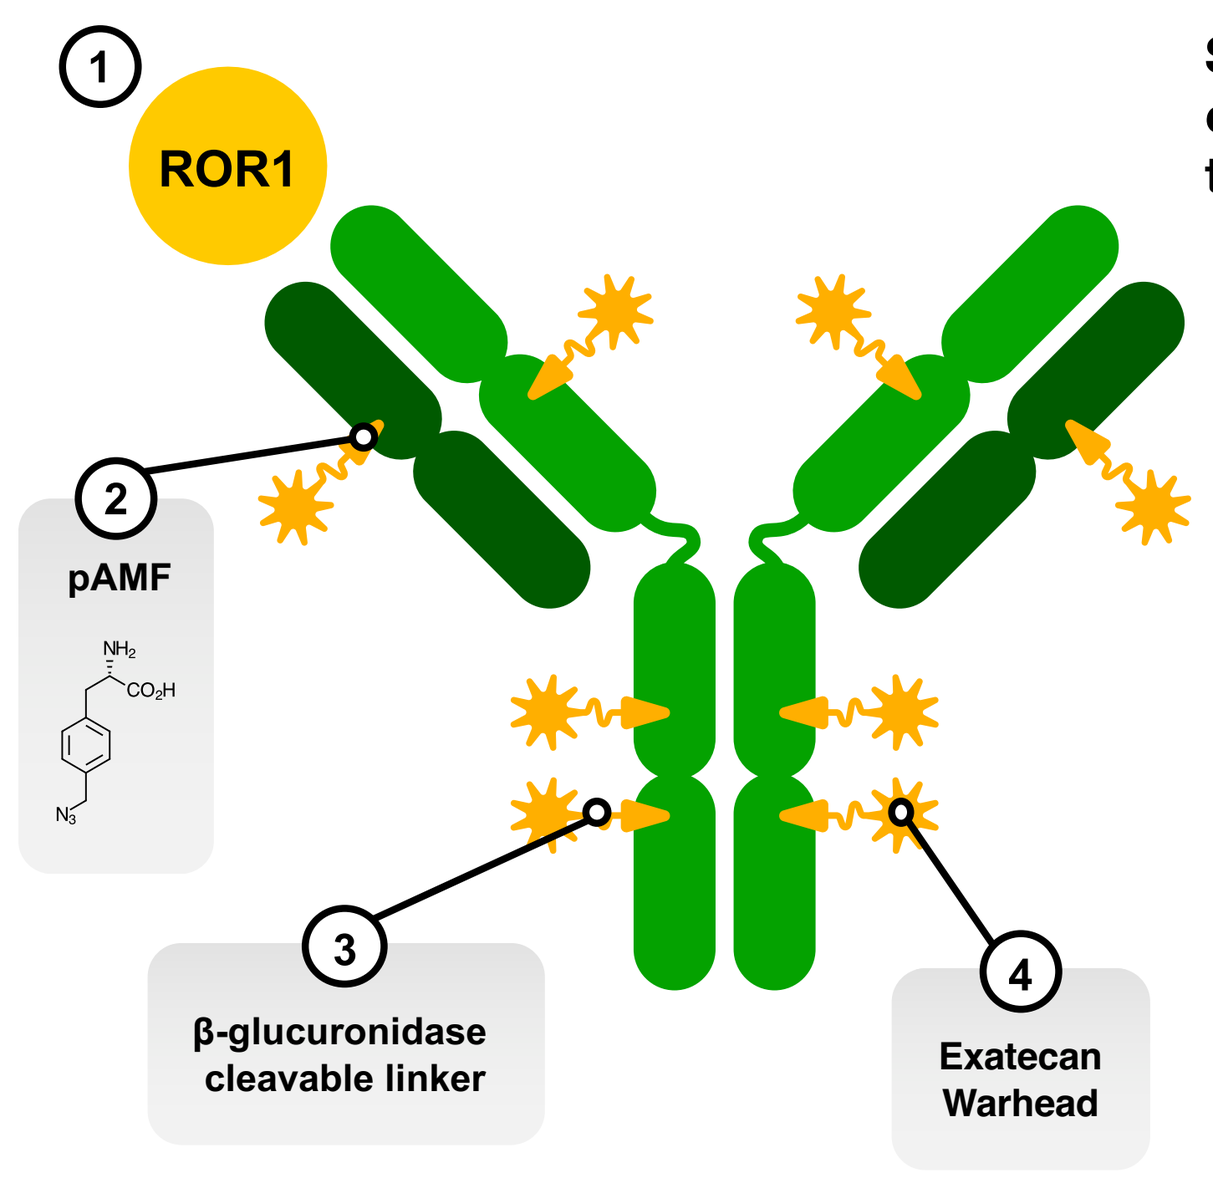

1
ROR1
2
pAMF
3
β-glucuronidase cleavable linker
4
Exatecan Warhead

STRO-003 is a single homogeneous antibody drug conjugate (ADC) with a drug-antibody ratio (DAR) of 8, targeting ROR1 tumor antigen

1. **Targeted ROR1 epitope** is overexpressed in diverse cancers including **hematological and solid tumor indications**
2. **Precisely positioned non-natural amino acids**, p-azidomethyl-L-phenylalanine (pAMF), **to enable DAR8** and optimal conjugation sites for enhanced performance and stability
3. **Stable β-glucuronidase cleavable linkers** demonstrate tumor specificity and encouraging preclinical tolerability. Preclinical data has shown marked **improvement over CatB linkers regarding neutropenia and lung tolerability issues** seen with tubulin and TOPO-1 inhibitors in the clinic
4. **Exatecan warhead inhibits TOPO-1 causing DNA disruption**. It elicits potent tumor cell killing, bystander activity and immunogenic cell death

Figure 1: STRO-003 has potent, ROR1-dependent activity

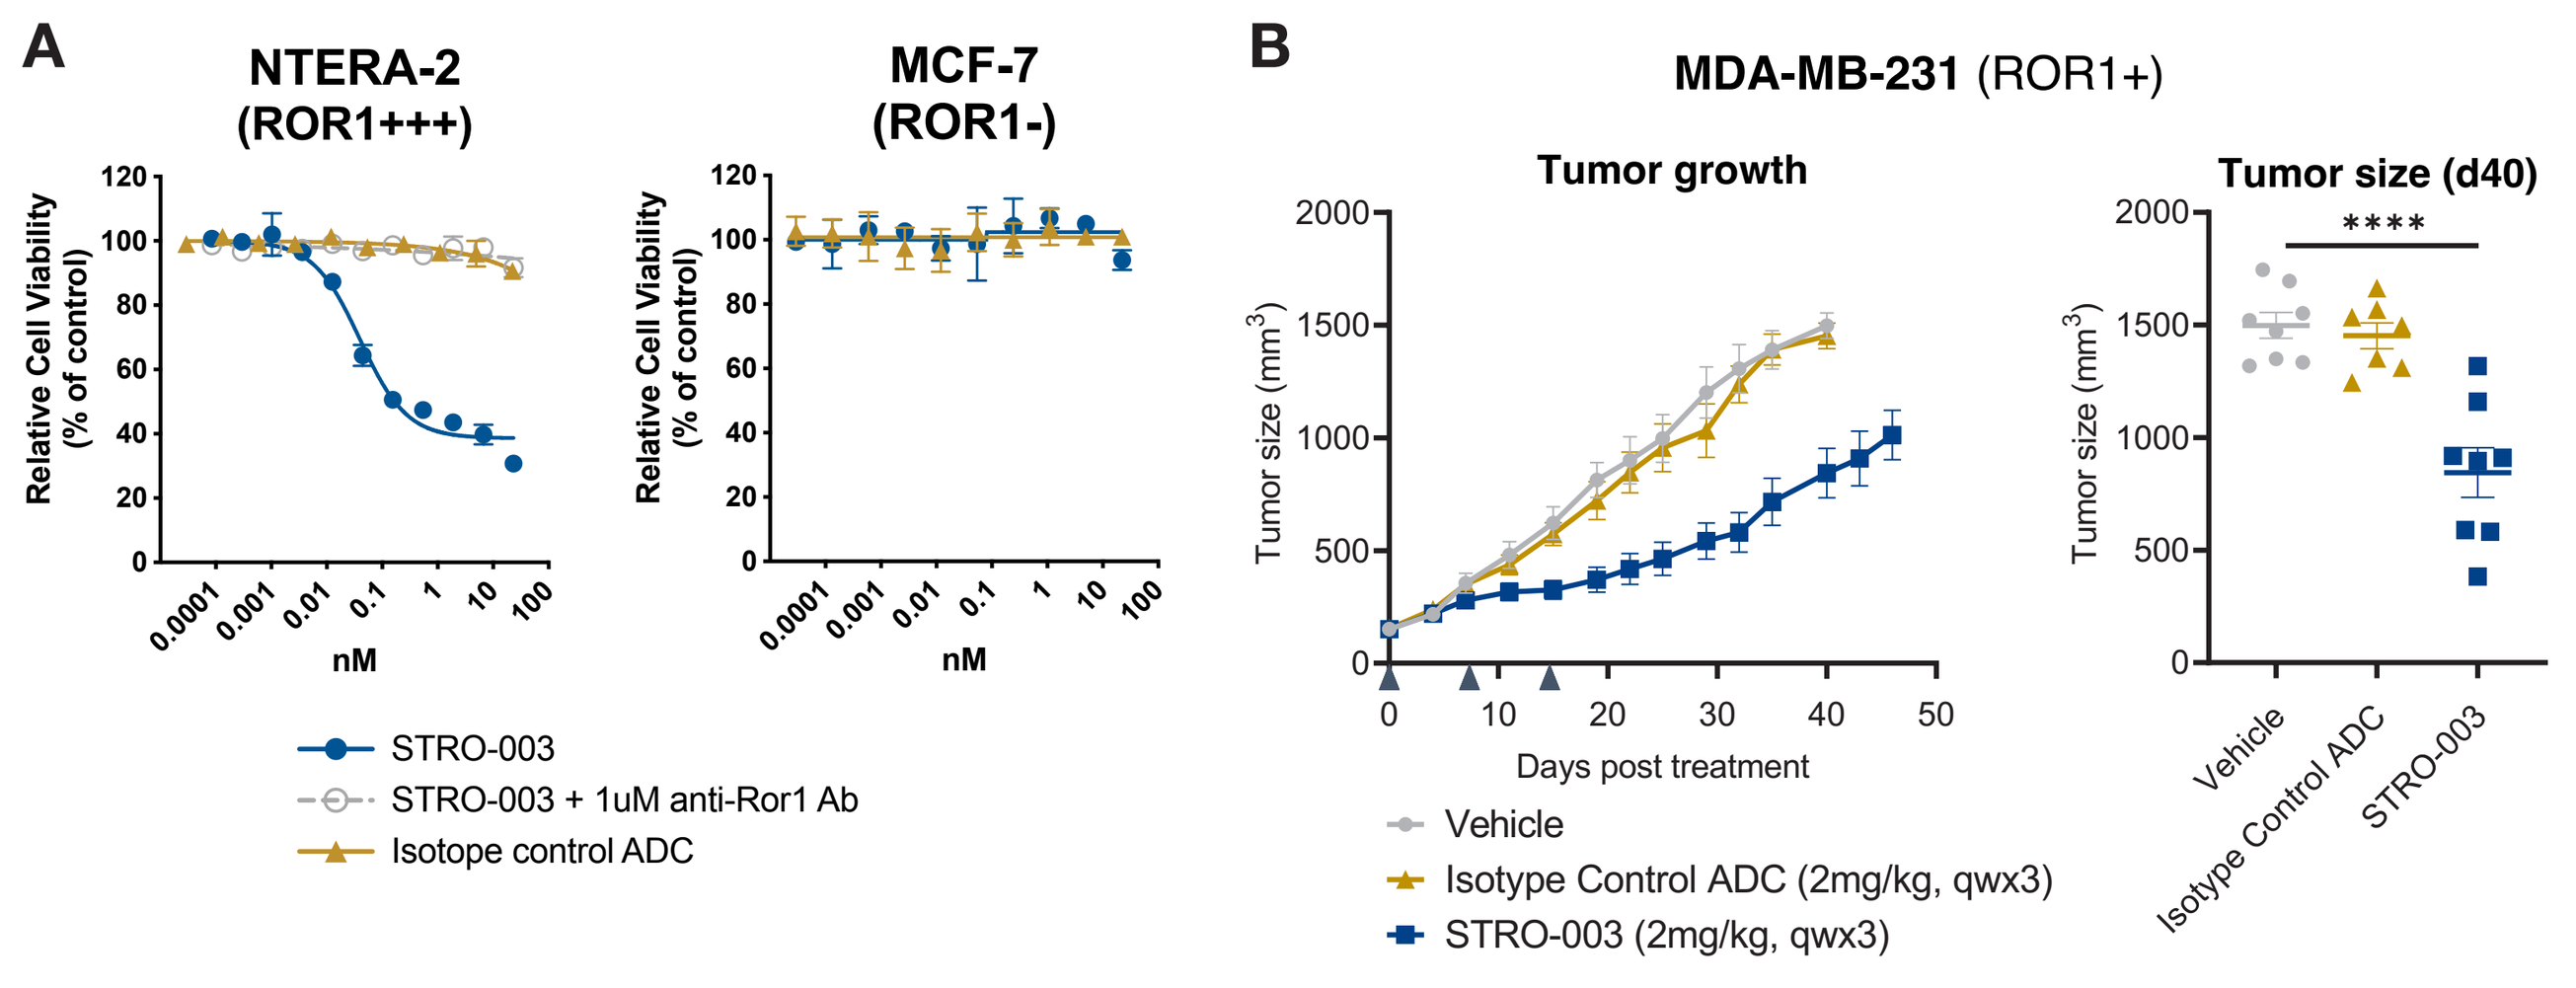

A
NTERA-2 (ROR1+++)
MCF-7 (ROR1-)
Relative Cell Viability (% of control)
nM
STRO-003
STRO-003 + 1uM anti-Ror1 Ab
Isotope control ADC
B
MDA-MB-231 (ROR1+)
Tumor growth
Tumor size (d40)
Tumor size (mm³)
Days post treatment

Vehicle
Isotype Control ADC (2mg/kg, qwx3)
STRO-003 (2mg/kg, qwx3)

Figure 1 (A) STRO-003 showed potent cell killing activity in ROR1 positive NTERA-2 cells, while an isotype ADC control was inactive. Cell killing was specific and could be blocked with a competing anti-ROR1 antibody. No cell killing was observed on ROR1 negative MCF-7 cells. **(B)** STRO-003 elicited potent anti-tumor activity in the ROR1-expressing breast cancer model MDA-MB-231. An isotype ADC control has no anti-tumor activity when tested at the same dose. Blue arrows represent dosing days.

Figure 2: Exatecan and STRO-003 induce hallmarks of immunogenic cell death (ICD)

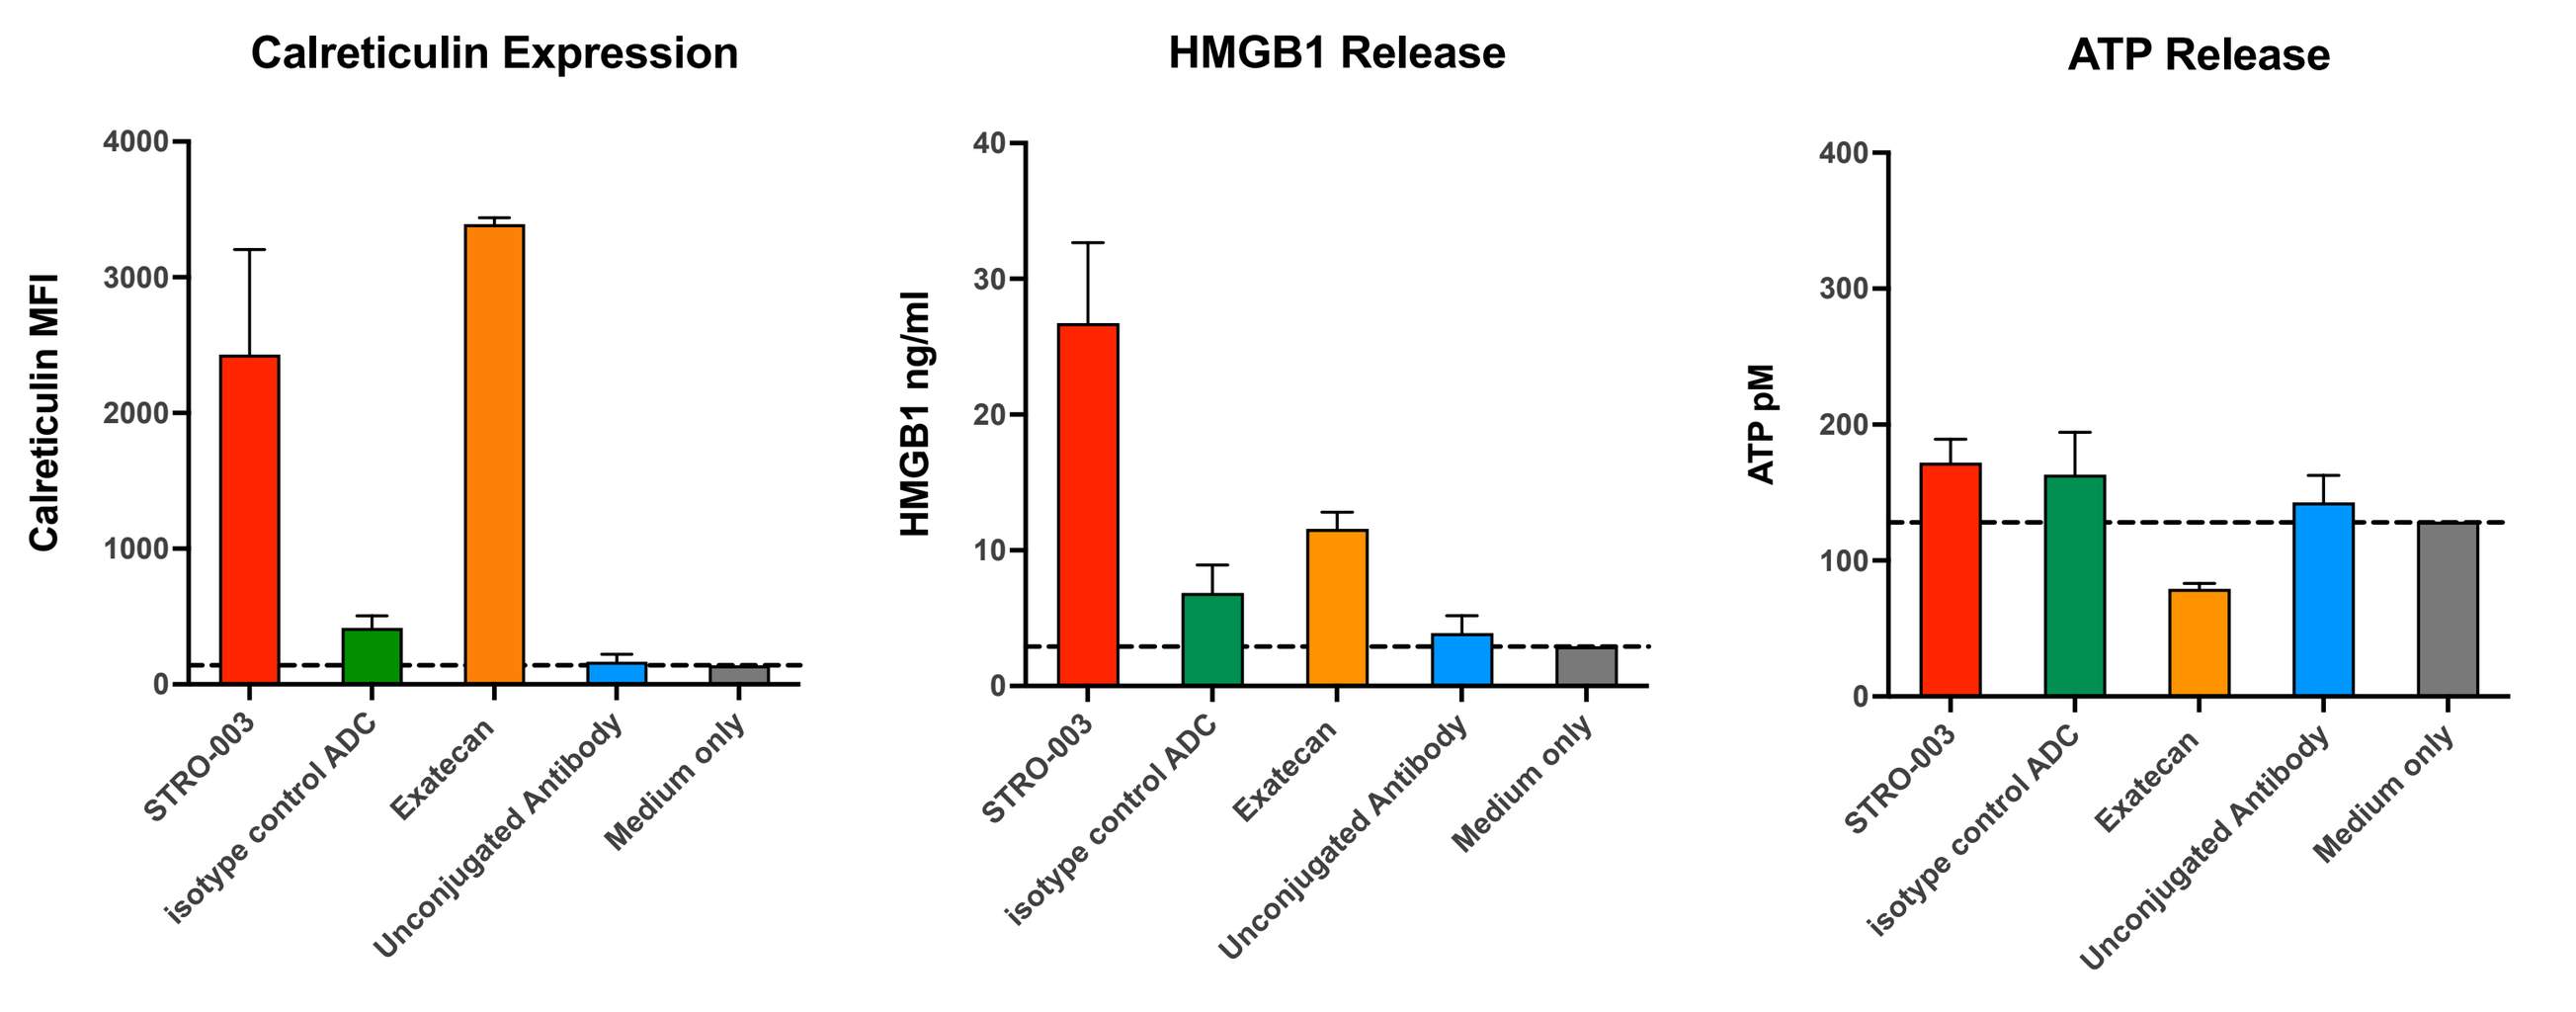

Calreticulin Expression
HMGB1 Release
ATP Release
Calreticulin MFI
HMGB1 ng/ml
ATP pM
STRO-003
isotype control ADC
Exatecan
Unconjugated Antibody
Medium only

Figure 2. Exatecan and STRO-003 treatment induced HMGB1 release and translocation of calreticulin to the cell surface of NTERA-2 cells. An isotype control ADC or an unconjugated anti-ROR1 antibody elicited no effect. Substantial ATP release was not observed, consistent with what others have observed for exatecan-like payloads.

Figure 3: STRO-003 activates monocytes when co-cultured with ROR1-expressing tumor cells

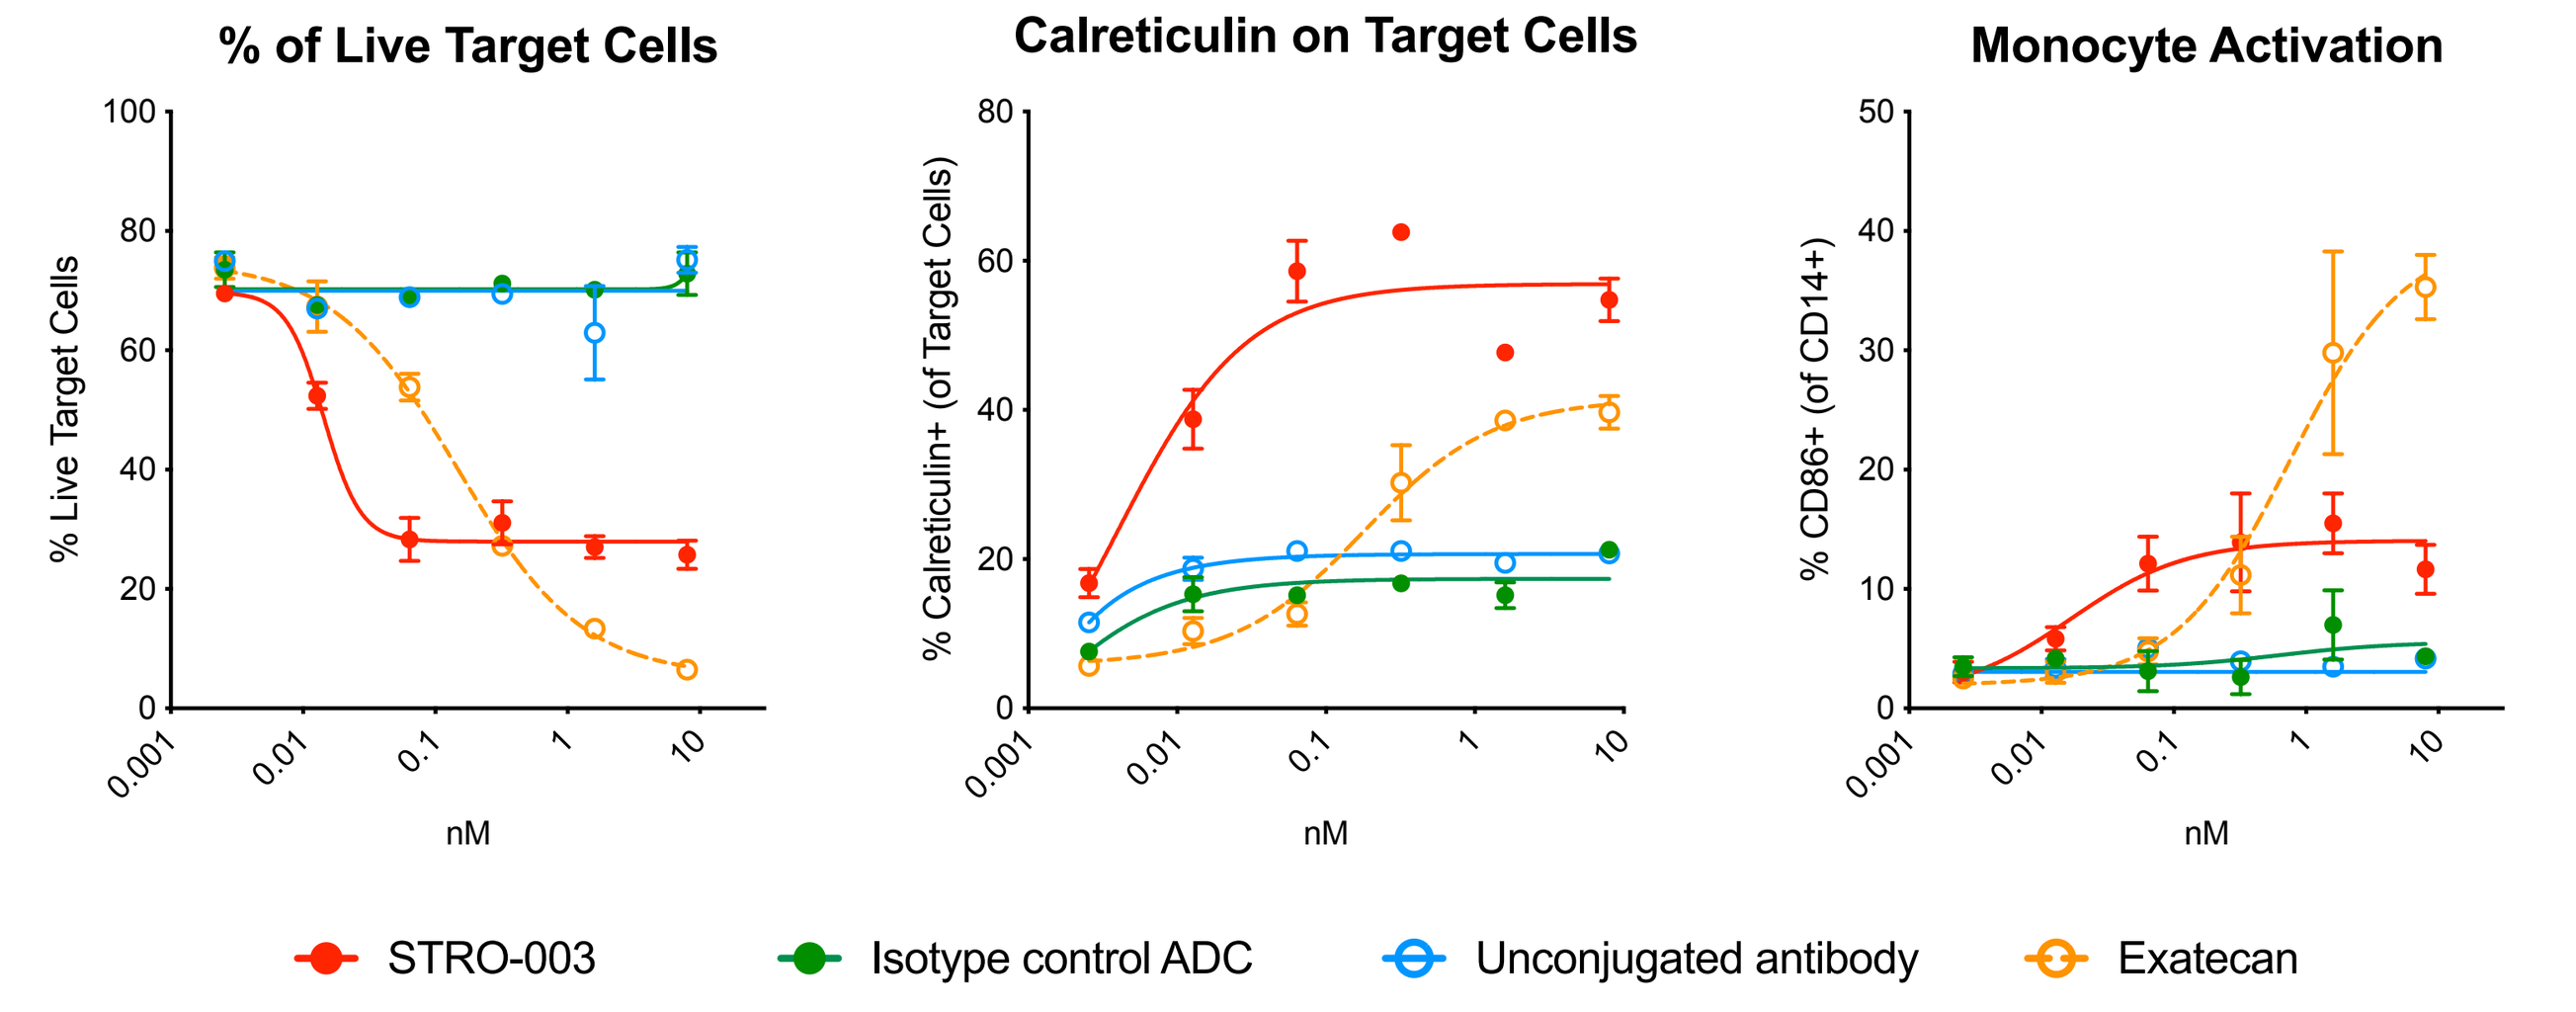

% of Live Target Cells
Calreticulin on Target Cells
Monocyte Activation
% Live Target Cells
% Calreticulin+ (of Target Cells)
% CD86+ (of CD14+)
nM
STRO-003
Isotype control ADC
Unconjugated antibody
Exatecan

Figure 3. STRO-003-treated ROR1-expressing Ntera-2 cells were co-cultured with human PBMCs to evaluate the potential for ICD-induced immune activation. STRO-003 and free exatecan both elicited potent tumor cell killing in this assay, whereas a control ADC or unconjugated anti-ROR1 antibody had no impact. Concomitant with reduced viability, calreticulin could be detected on the cell surface of STRO-003- and exatecan-treated cells, consistent with immunogenic cell death. STRO-003- or exatecan-treated cells potently induced monocyte activation, as demonstrated by the induction of CD86 expression. Unconjugated anti-ROR1 antibody or a non-targeted ADC had no activity.

Figure 4: *In vivo* vaccination is the "gold standard" model for ICD induction

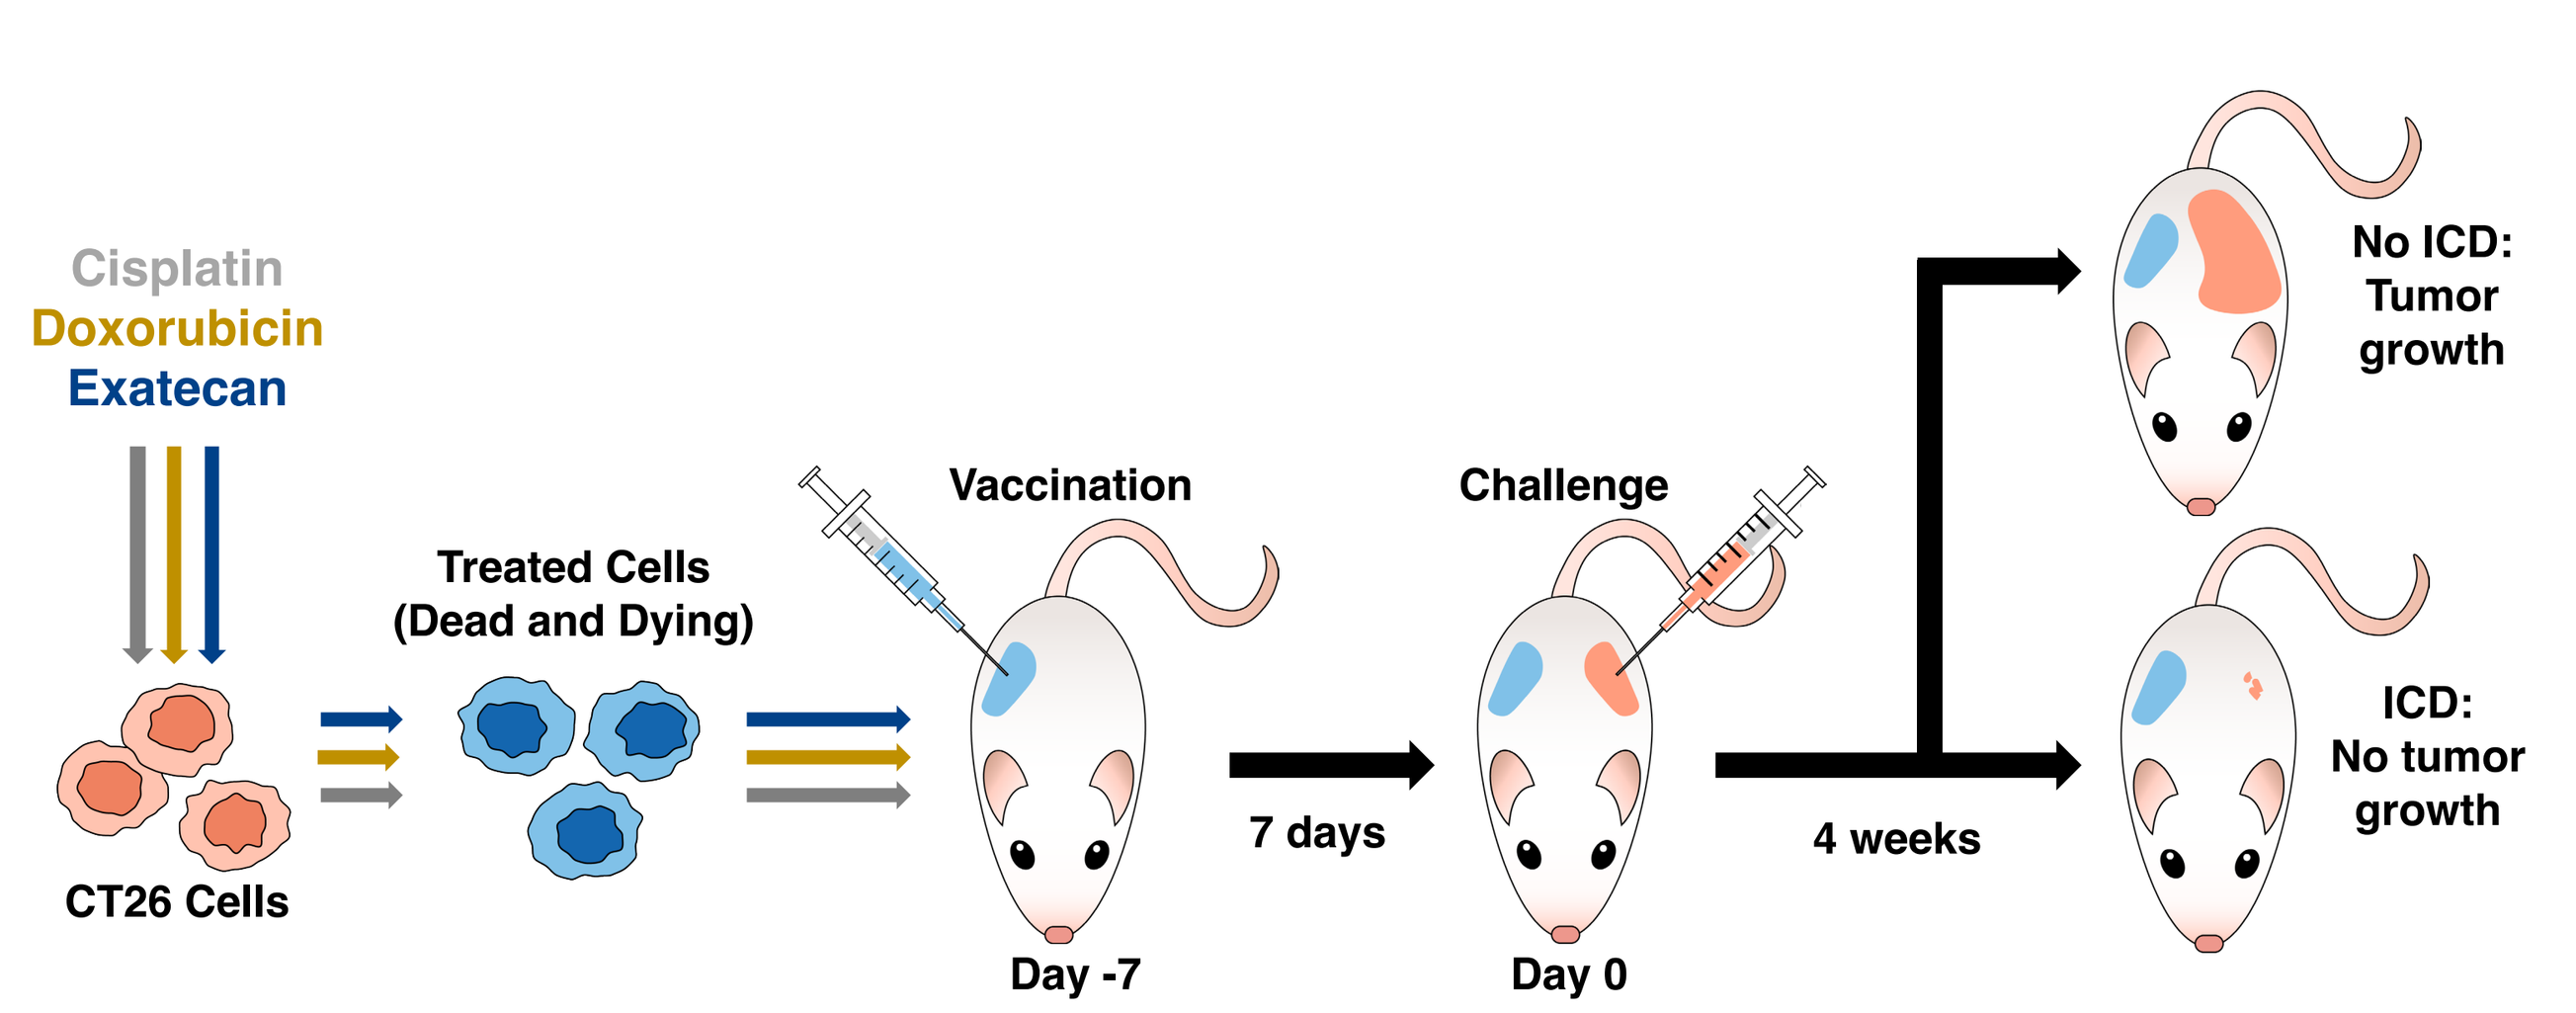

Cisplatin
Doxorubicin
Exatecan
CT26 Cells
Treated Cells (Dead and Dying)
Vaccination
Day -7
7 days
Challenge
Day 0
4 weeks
No ICD: Tumor growth
ICD: No tumor growth

Figure 4. Overview of the "gold standard" ICD vaccination model. CT26 cells were treated in vitro with either doxorubicin, cisplatin, or exatecan such that 50-75% of cells are either dead or undergoing apoptosis. Treated CT26 cells were injected into the right flank of naïve mice. Seven days later, untreated CT26 cells were injected in the opposite flank of vaccinated mice as a challenge and tumor take was monitored for 4 weeks. In a case of a bona fide ICD induction, no tumor growth is observed on the challenge site.

Figure 5: Exatecan exhibits the profile of a bona fide ICD inducer in the "gold standard" vaccination model

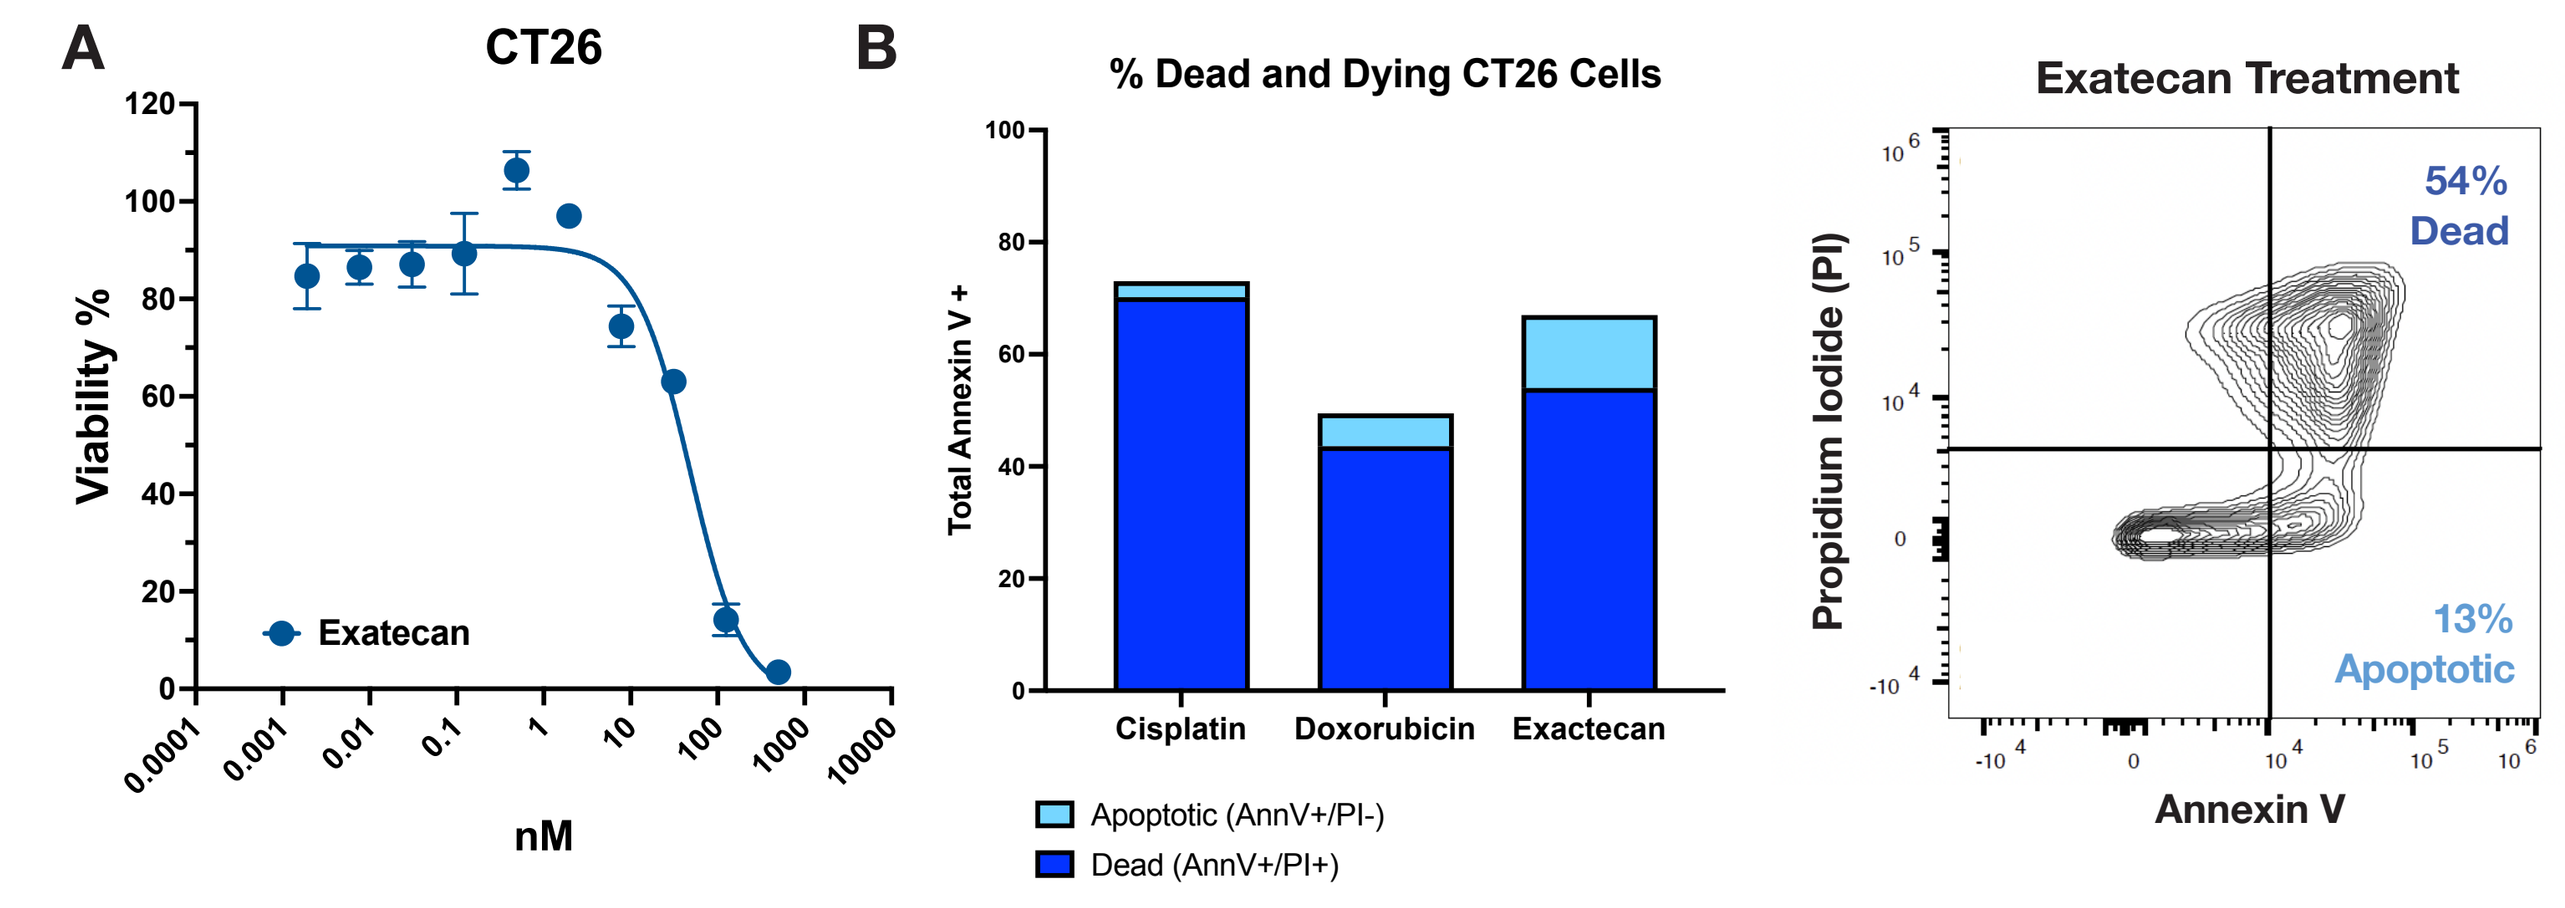

A
CT26
Viability %
Exatecan
nM
B
% Dead and Dying CT26 Cells
Total Annexin V +
Cisplatin
Doxorubicin
Exactecan
Apoptotic (AnnV+/PI-)
Dead (AnnV+/PI+)
Exatecan Treatment
Propidium Iodide (PI)
Annexin V
54% Dead
13% Apoptotic

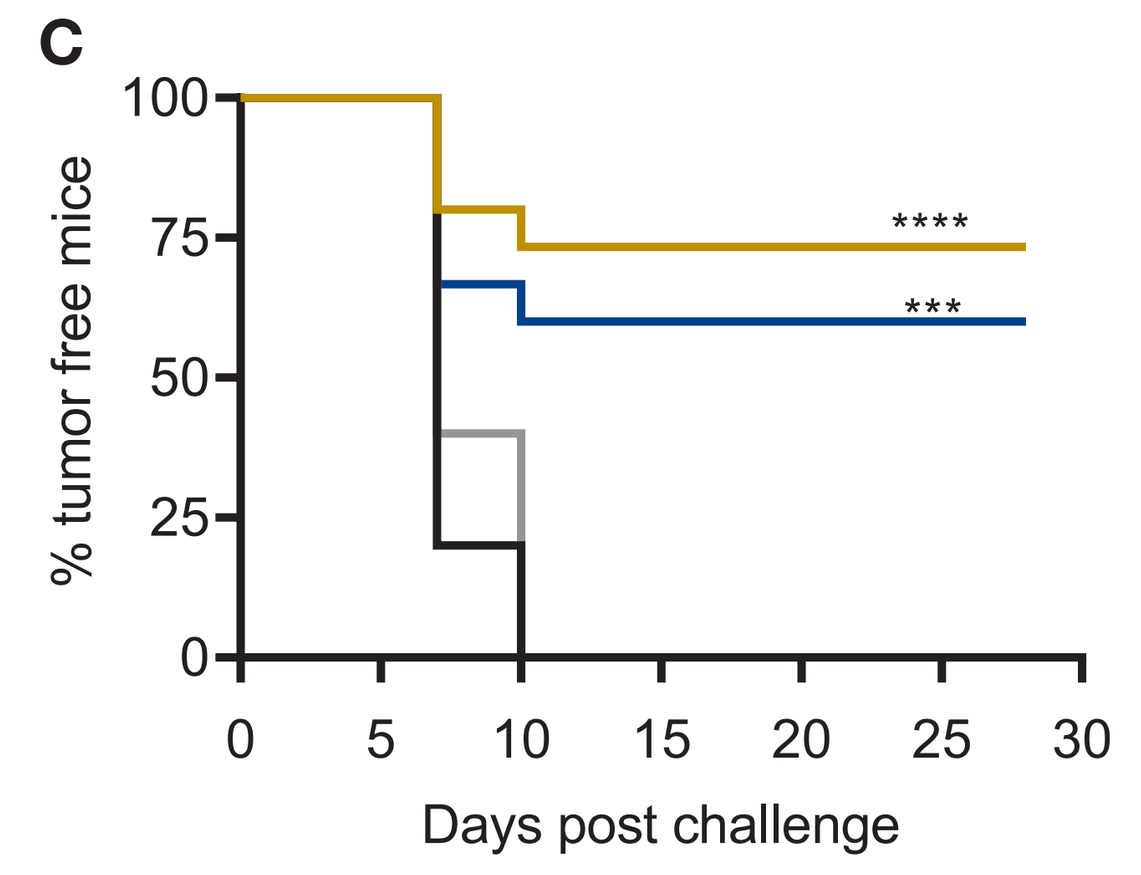

C
% tumor free mice
Days post challenge

Vaccination - CT26 treated with	Group Function	% tumor free	ICD inducer
none	null control	0%	NA
Cisplatin	negative control	0%	N
Doxorubicin	positive control	73%	Y
Exatecan	ICD assessment	60%	Y

Figure 5 (A) Exatecan showed potent cell killing activity against murine CT26 tumor cells. **(B)** Frequency of CT26 cells that are dead or undergoing apoptosis as determined by AnnexinV/PI staining after treatment with cisplatin, doxorubicin, or exatecan. Doxorubicin and cisplatin-treated cells served as positive and negative controls for ICD, respectively. **(C)** Mice were vaccinated with treated CT26 cells, challenged with untreated CT26 cells and monitored for tumor growth. Mice vaccinated with doxorubicin (positive control) and exatecan treated CT26 cells rejected challenge at a rates of 73% and 60%, respectively. 0% of naïve or mice vaccinated with cisplatin (negative control) treated CT26 cells rejected the tumor challenge. Pairwise comparisons to the naïve mouse group were performed using a log-rank test with Bonferroni correction. *** = *p<0.001*; **** = *p<0.0001*.

Figure 6: STRO-003 in combination with anti-PD1 trends towards an inflamed tumor microenvironment in the cold LLC1-hROR1 mouse model

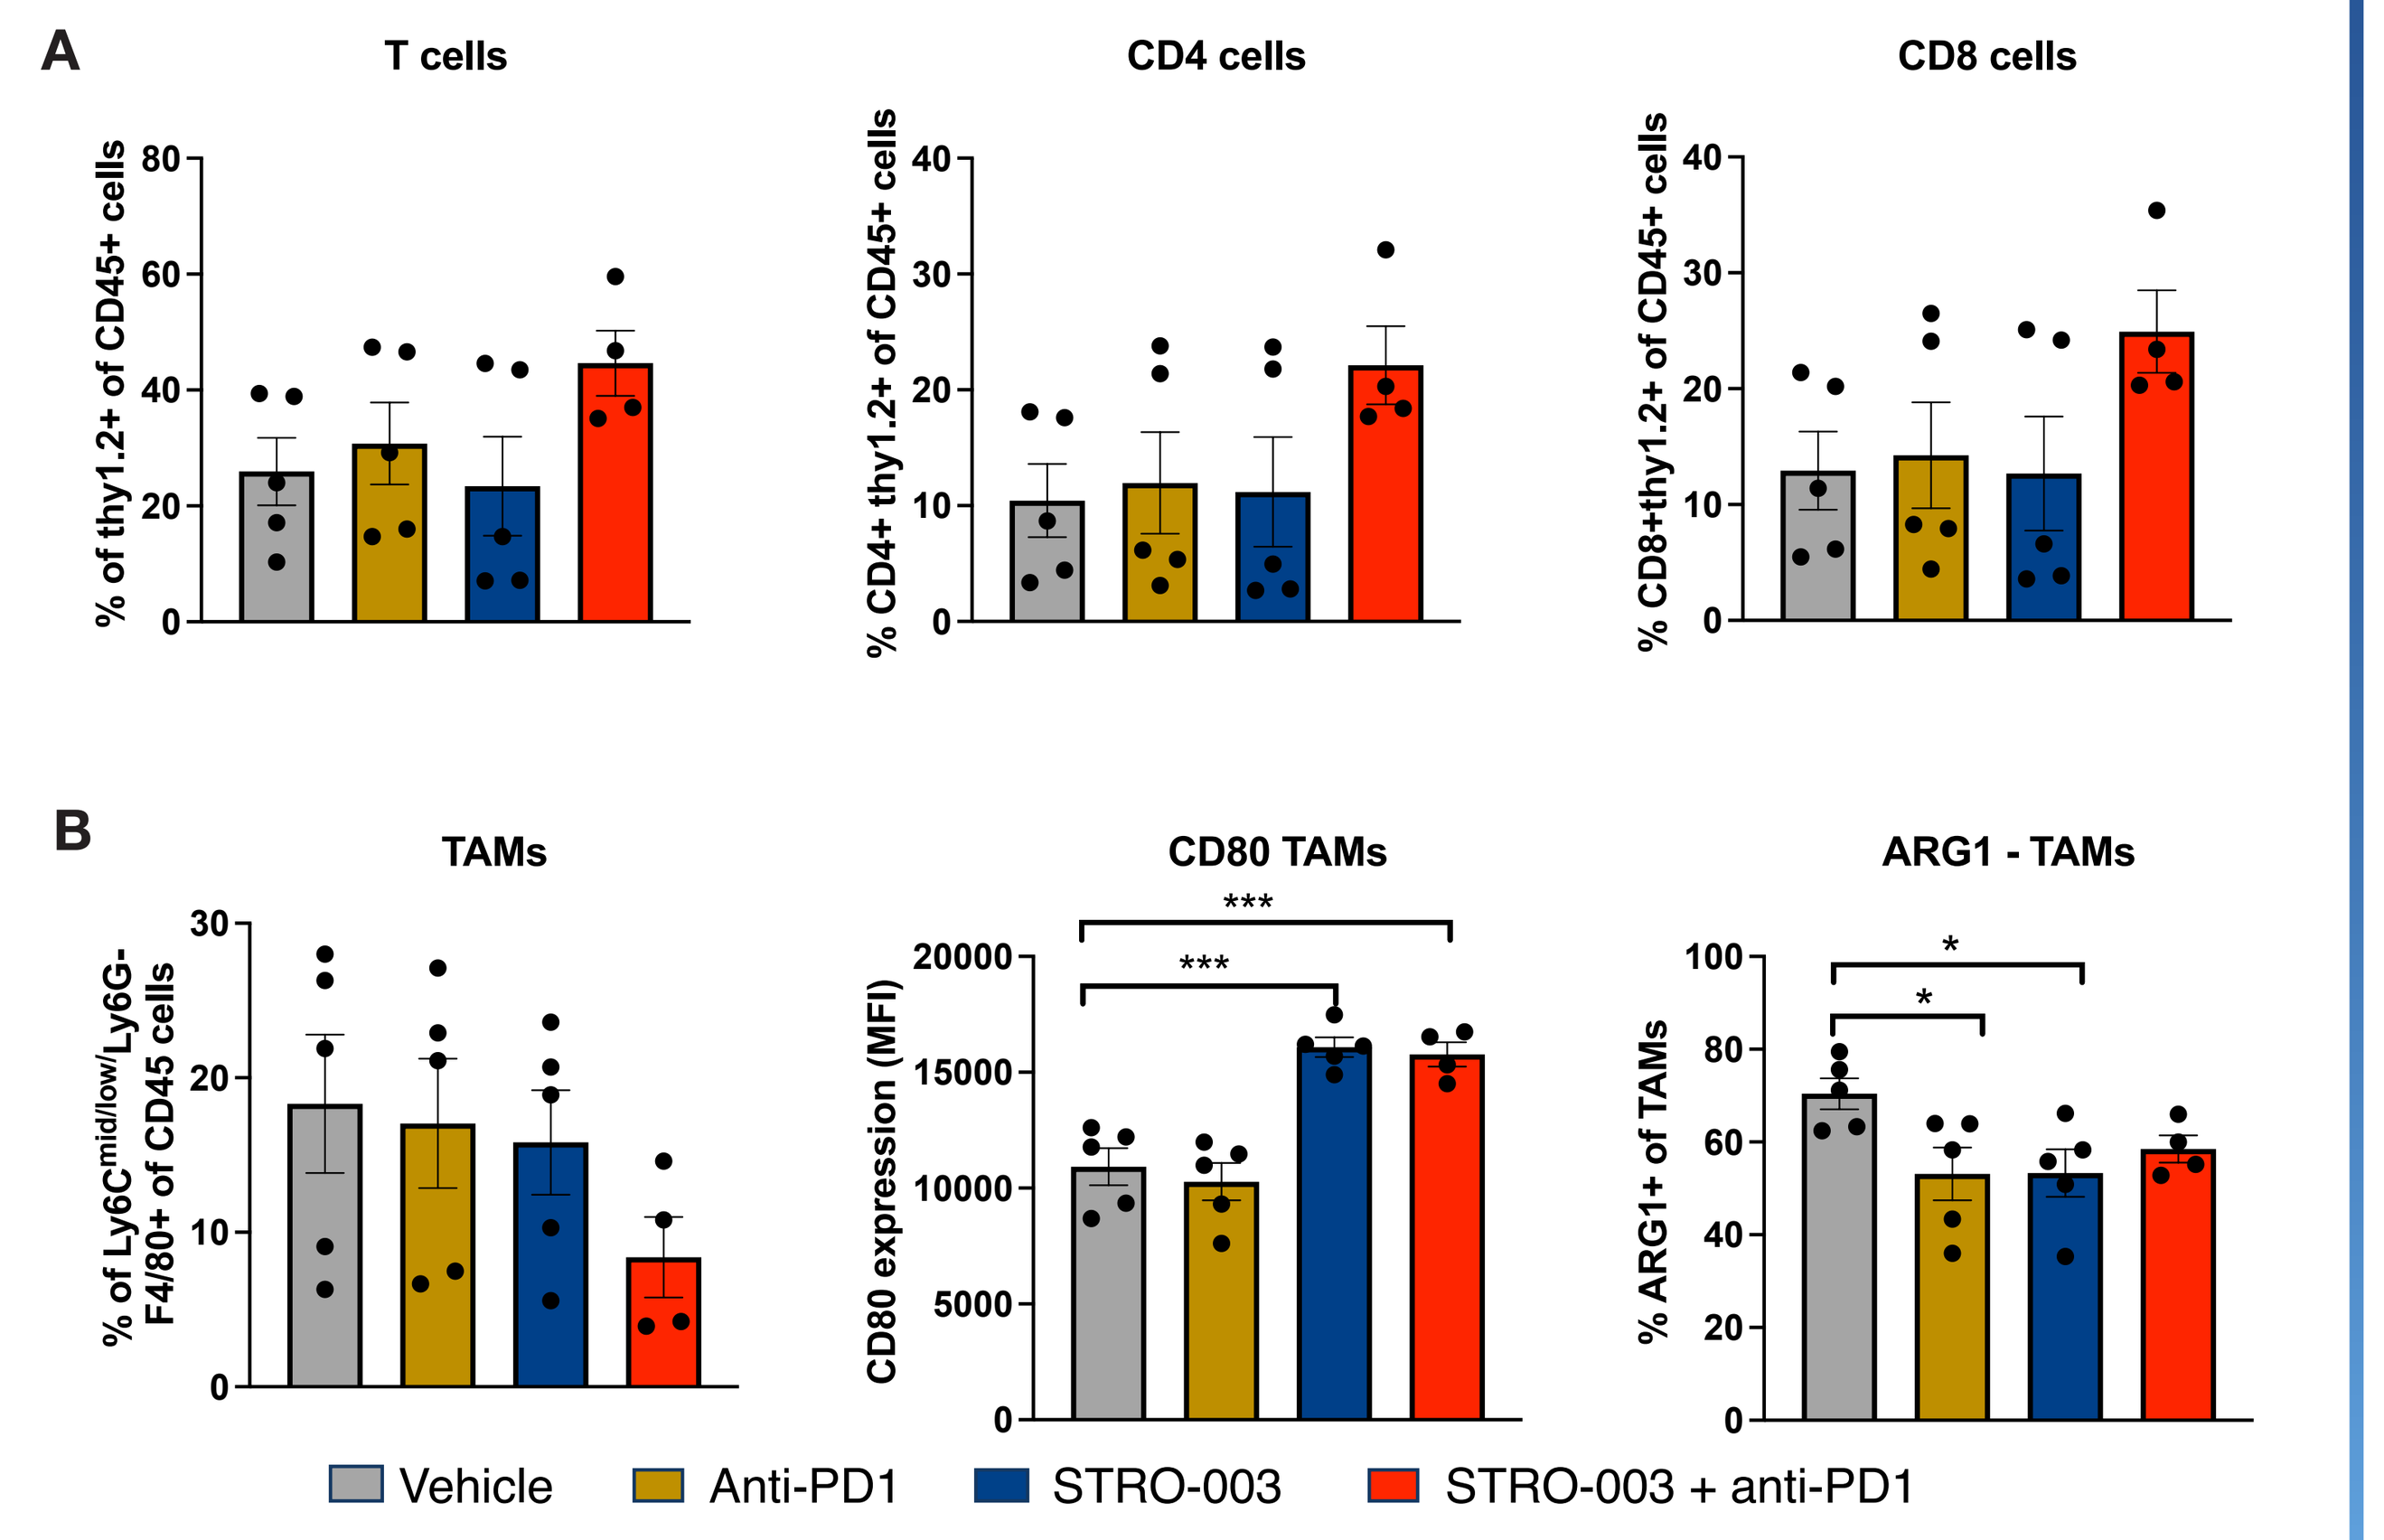

A
T cells
CD4 cells
CD8 cells
% of thy1.2+ of CD45+ cells
% CD4+ thy1.2+ of CD45+ cells
% CD8+thy1.2+ of CD45+ cells
B
TAMs
CD80 TAMs
ARG1 - TAMs
% of Ly6Cmid/low/Ly6G- F4/80+ of CD45 cells
CD80 expression (MFI)
% ARG1+ of TAMs
Vehicle
Anti-PD1
STRO-003
STRO-003 + anti-PD1

Figure 6. Tumors treated with STRO-003 + anti-PD1 combination therapy show a trend towards a more pro-inflammatory tumor microenvironment which can contribute to enhanced anti-tumor immune responses. Tumor-associated T cells **(A)** and tumor-associated macrophages (TAMs) **(B)** were measured by flow cytometry in response to STRO-003 (10 mg/kg, qwx2) + anti-PD1 (8mg/kg, q3dx3) combination treatment in anti-PD1 resistant LLC1-hROR1 tumors. STRO-003 combination with anti-PD1 immune checkpoint blockade shows a trend towards increased T cell infiltration, and reduced frequency of TAMs. Upon treatment, TAMs showed increased surface expression of the co-stimulatory molecule CD80 and reduced expression of Arg1, which suggests a shift from an immunosuppressive to an anti-tumoral phenotype of TAMs.

Figure 7: STRO-003 demonstrates anti-tumor activity in human lung cancer PDX models across a range of ROR1 expression

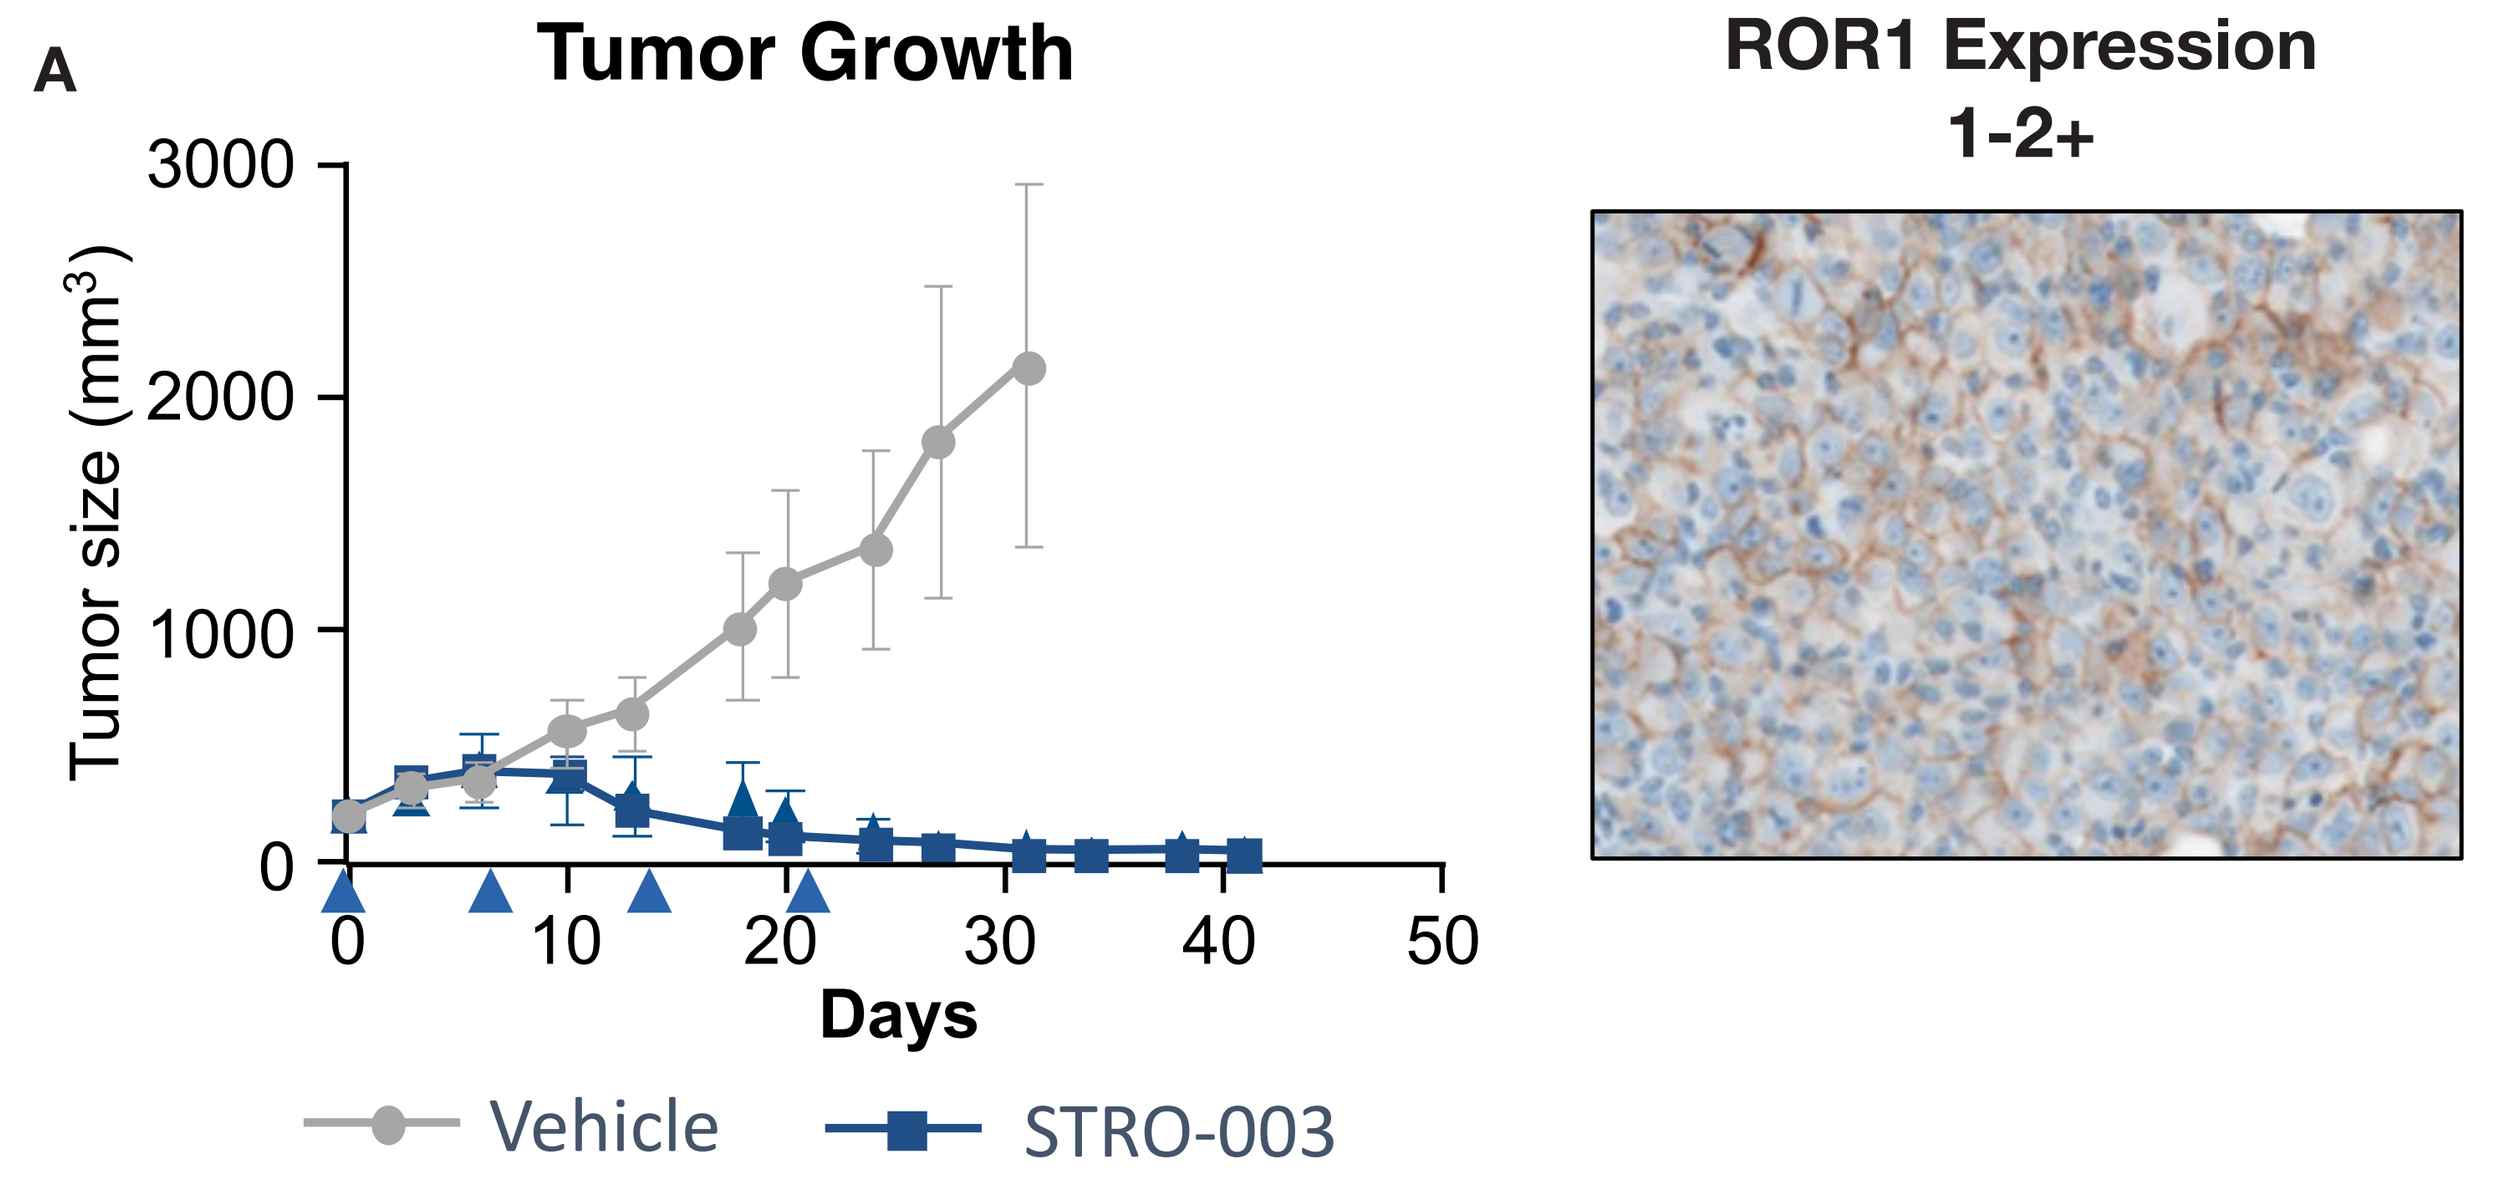

A
Tumor Growth
Tumor size (mm³)
Days
Vehicle
STRO-003
ROR1 Expression 1-2+

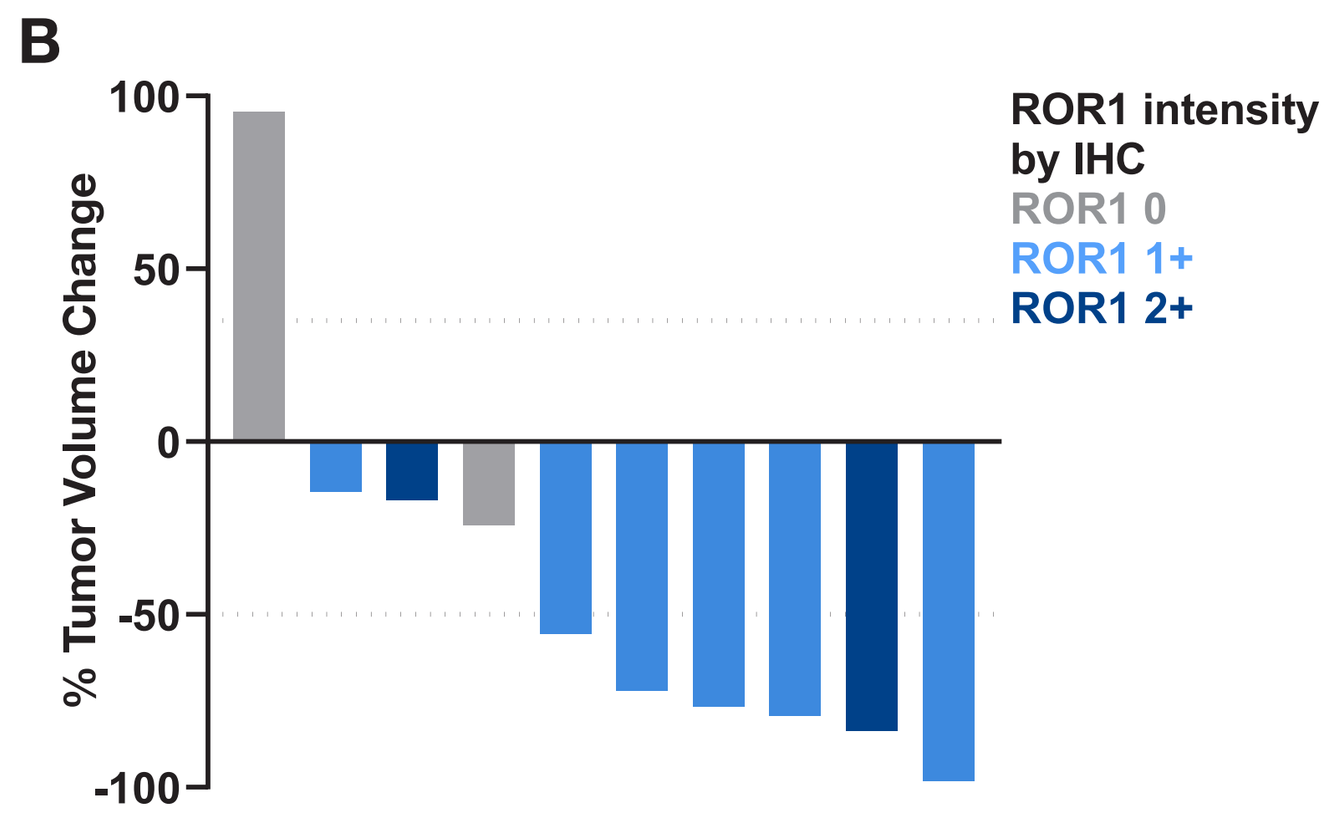

B
% Tumor Volume Change
ROR1 intensity by IHC
ROR1 0
ROR1 1+
ROR1 2+

Overall response based on best response criteria:

Response (tumor volume change)	Vehicle	STRO-003
CR (<-95%)	0%	10%
PR (<-50%)	0%	50%
SD (< 35%)	30%	30%
PD (> 35%)	70%	10%

Figure 7 (A) STRO-003 treatment induced tumor regression in PDX models of NSCLC and demonstrated significant anti-tumor activity compared to vehicle control in 7 out of 8 ROR1 positive NSCLC PDX models. Continued monitoring of the STRO-003 group showed prolonged tumor growth suppression. Representative tumor growth curve and IHC staining indicating the level of ROR1 expression observed in the PDX models are shown. Blue arrows represent dosing days. **(B)** Best response from each NSCLC PDX model was plotted as the greatest reduction in tumor volume observed (percent tumor volume change from start of treatment). Response was categorized based on modified Response Evaluation Criteria In Solid Tumors (mRECIST).

Figure 8: STRO-003 demonstrates anti-tumor activity in human triple negative breast cancer (TNBC) PDX models

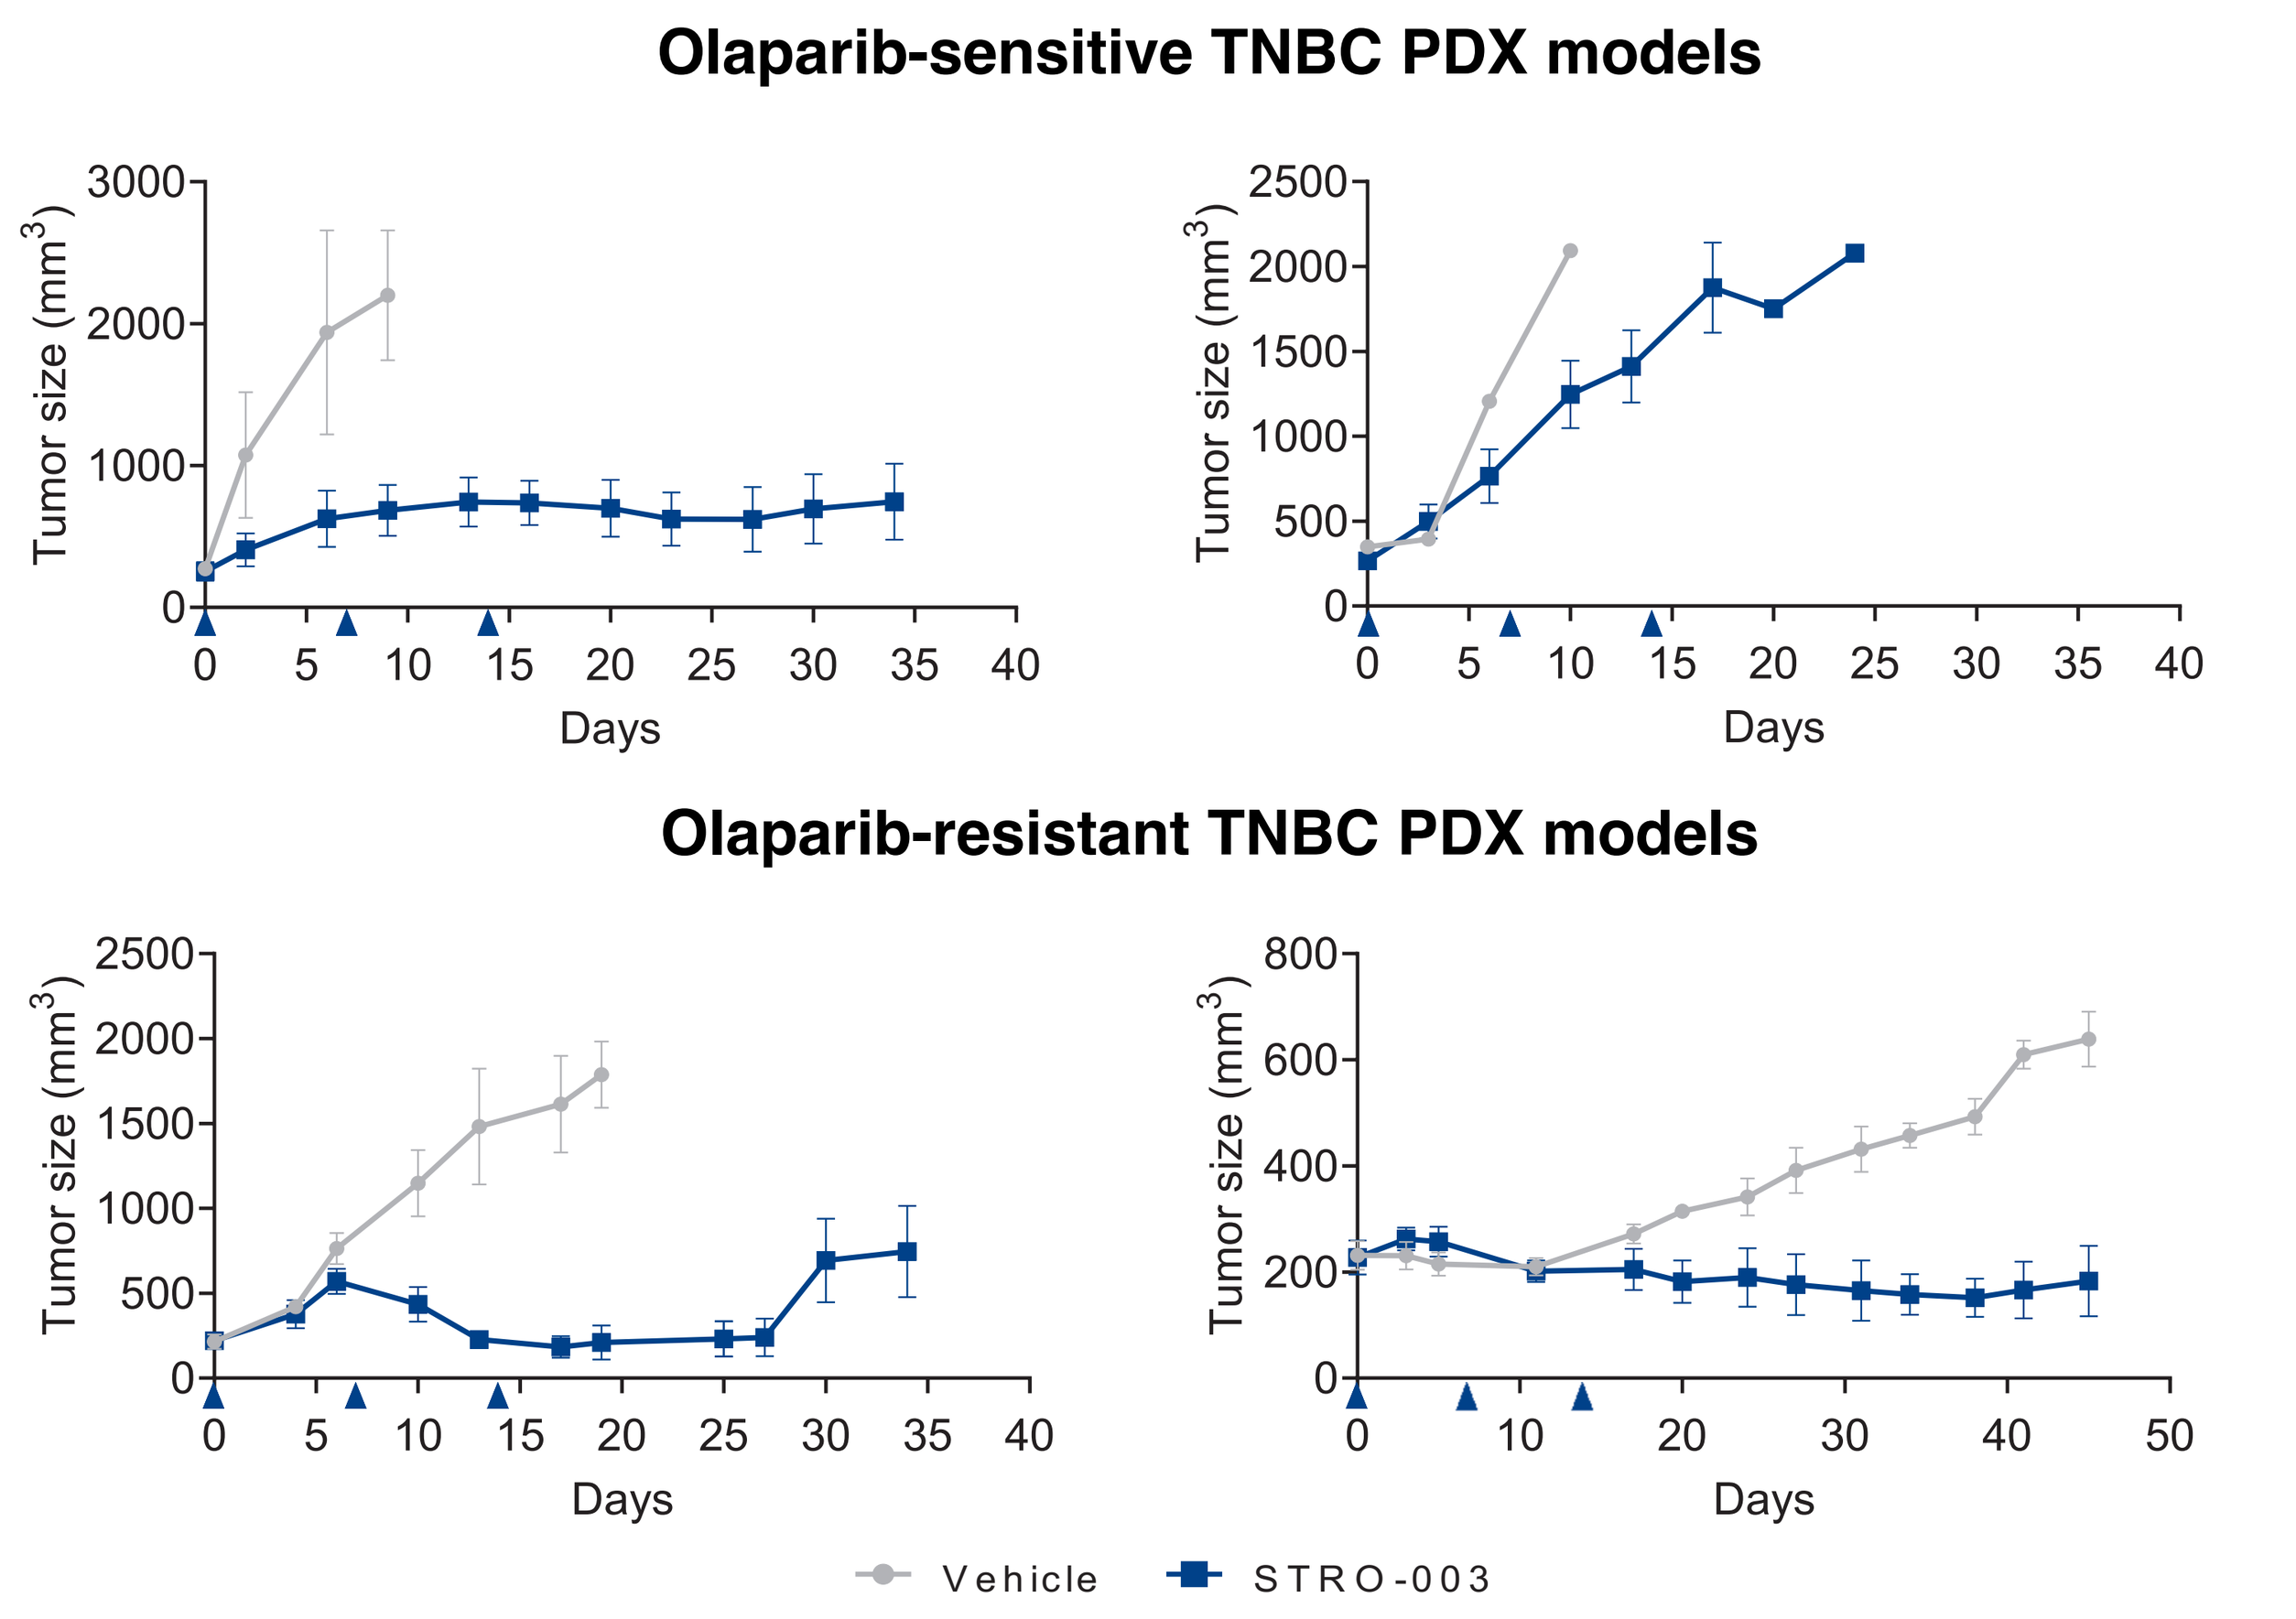

Olaparib-sensitive TNBC PDX models
Olaparib-resistant TNBC PDX models
Tumor size (mm³)
Days
Vehicle
STRO-003

Figure 8. STRO-003 treatment induced tumor growth inhibition in PDX models of TNBC and demonstrated significant anti-tumor activity compared to vehicle control in 3 out of 4 ROR1⁺ models with various levels of ROR1 expression. Models tested were BRCA1 wt. Continued monitoring of the STRO-003 group showed prolonged tumor growth suppression. Blue arrows represent dosing days.