Abstract # 1685

DAR12 HER2-targeting dual-payload antibody-drug conjugate combining a topoisomerase I inhibitor and microtubule inhibitor demonstrates superior efficacy and overcomes resistance to single-payload ADCs in xenograft models

A. Matcham, R. Yuan, B. Vuillemenot, R. Pena, Y. Park, A. Yu, J. Hanson, C. Tran, X. Li, M. Wen, D. Calarese, W. Rubas, G. Yin, K. Bajjuri, G. Xu, A. Yam, H.P. Gerber

SUTRO BIOPHARMA

Introduction

The genetic and phenotypic heterogeneity of human cancers is a primary driver of drug resistance, posing a major challenge to achieving durable therapeutic responses.

To overcome this, Sutro has developed a dual-payload antibody–drug conjugate (dpADC) platform that enables precise co-delivery of two cytotoxic payloads via a single, homogeneous molecule.

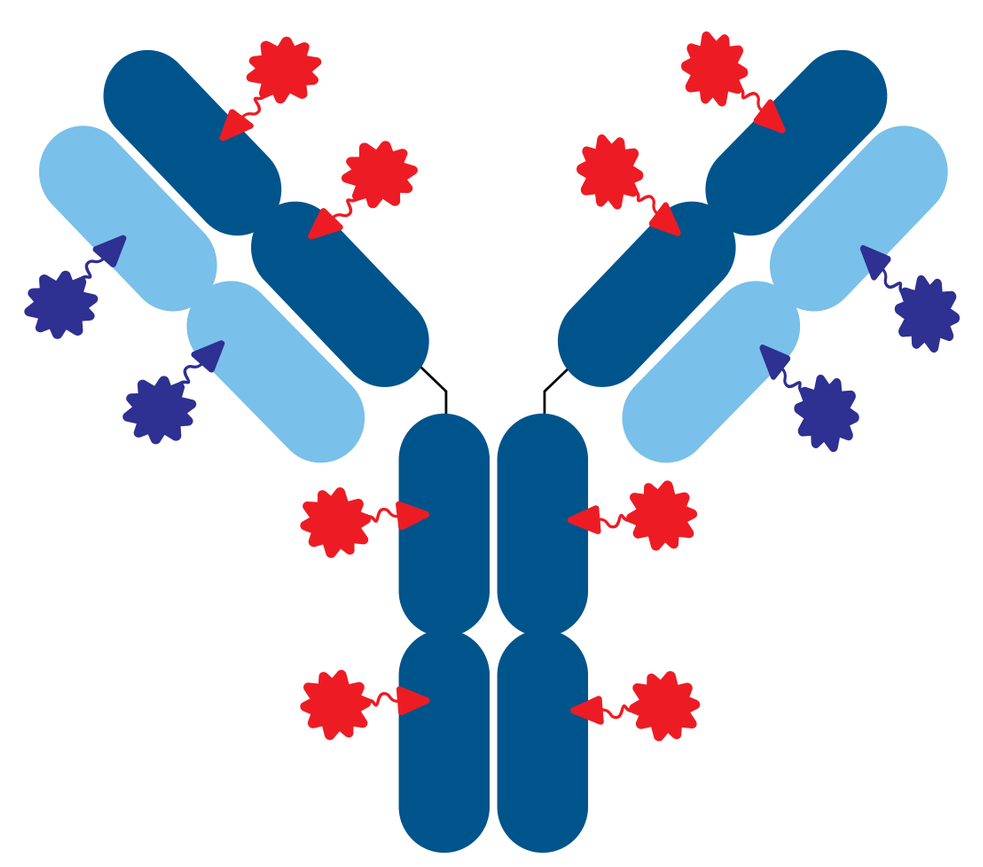

- Can be constructed for a variety of targets, such as PTK7.
- Site-specific conjugation with (8) Topo1i and (4) MTi payloads.
- Tumor-specific Beta-glu linkers

Sutro's dpADC demonstrates greater cell killing than Enhertu

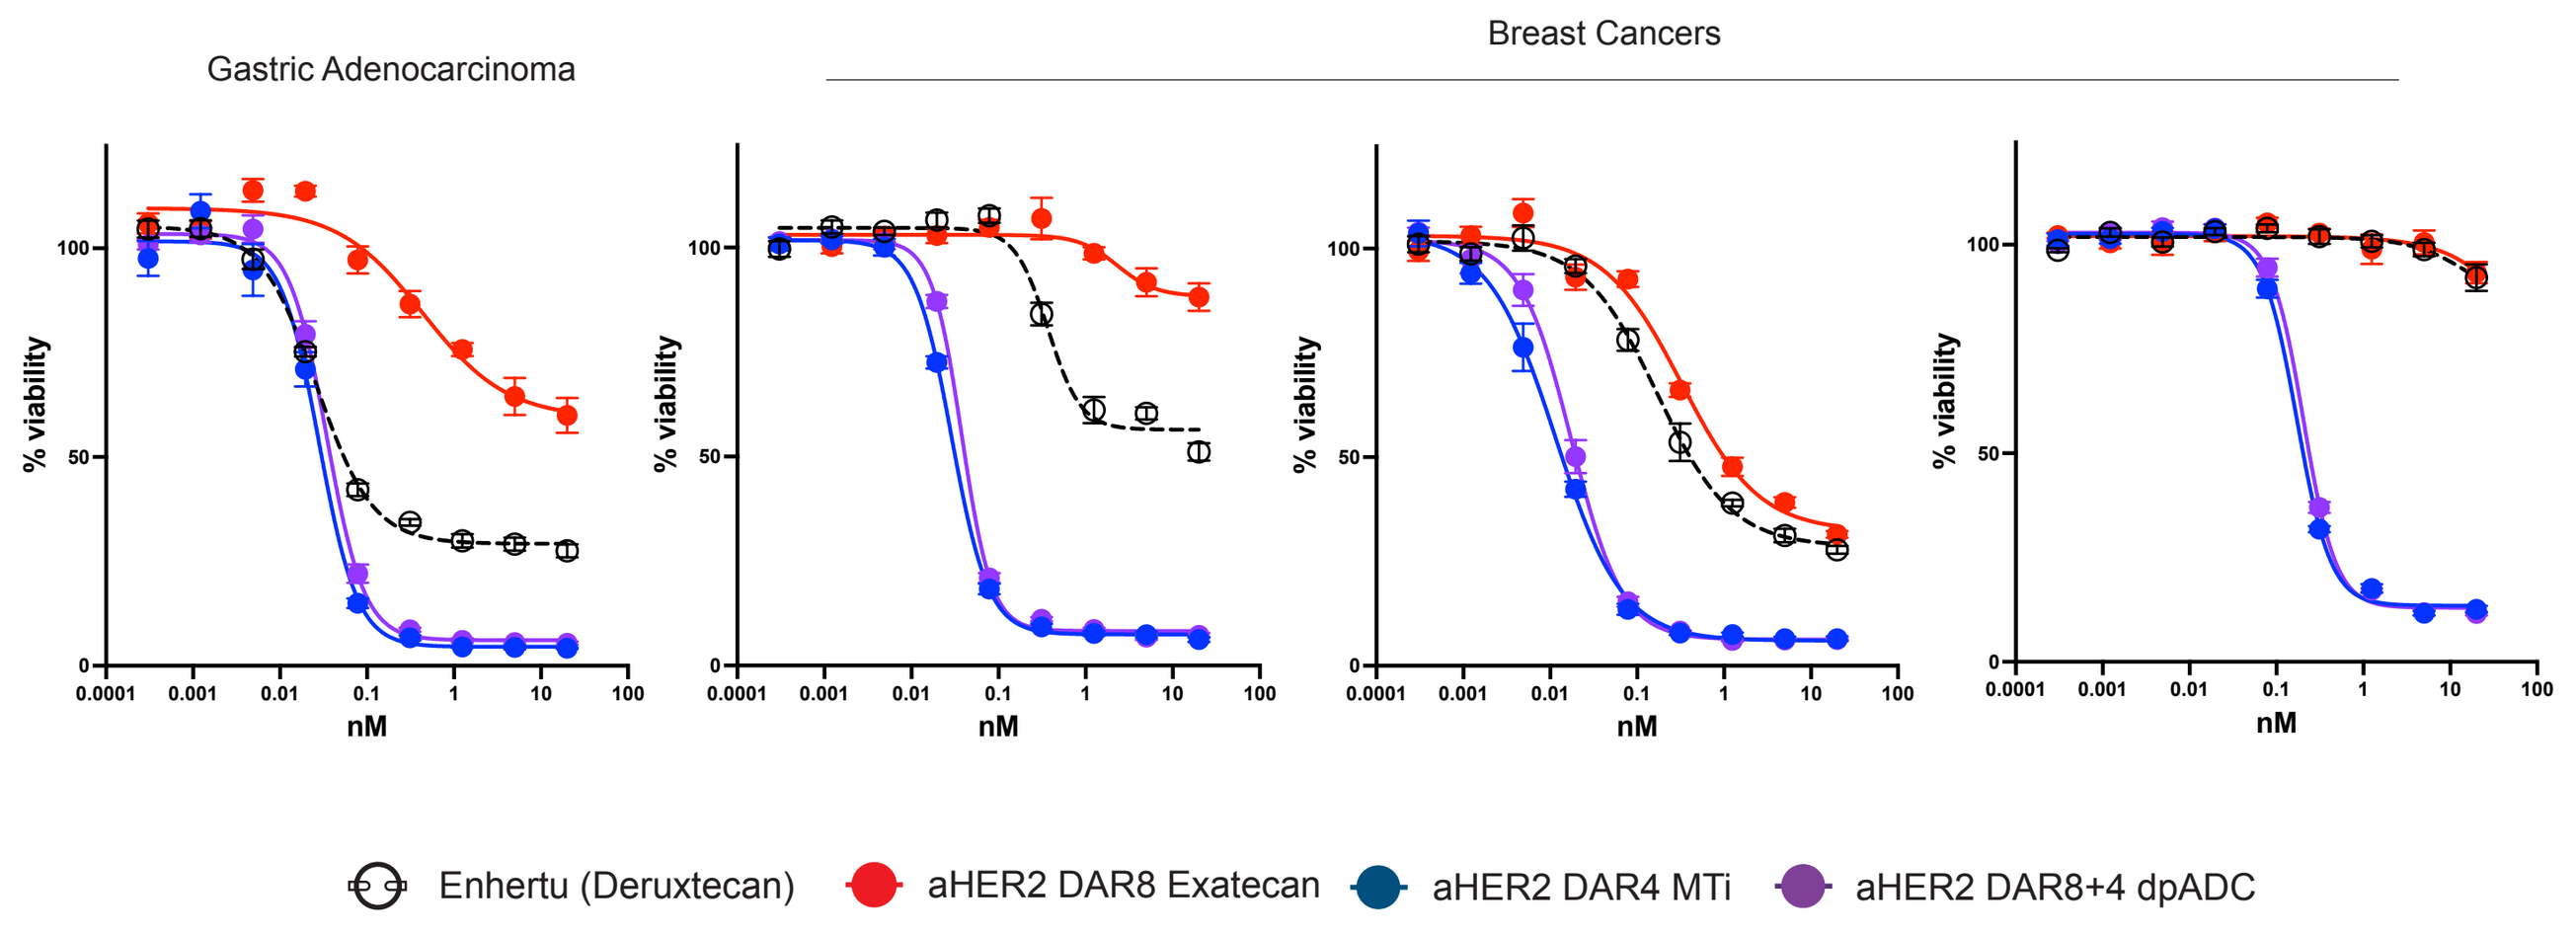
Gastric Adenocarcinoma
Breast Cancers
% viability
nM
Enhertu (Deruxtecan)
aHER2 DAR8 Exatecan
aHER2 DAR4 MTi
aHER2 DAR8+4 dpADC

Figure 1. Cell killing curves of in vitro cultures of a gastric adenocarcinoma and three breast cancer cell lines incubated with Enhertu, anti-HER2 DAR8 Topo1i, anti-HER2 DAR4 MTi, or anti-HER2 DAR8+4 (Topo1+MTi) dpADC.

Sutro's dpADC has stable DAR and favorable PK

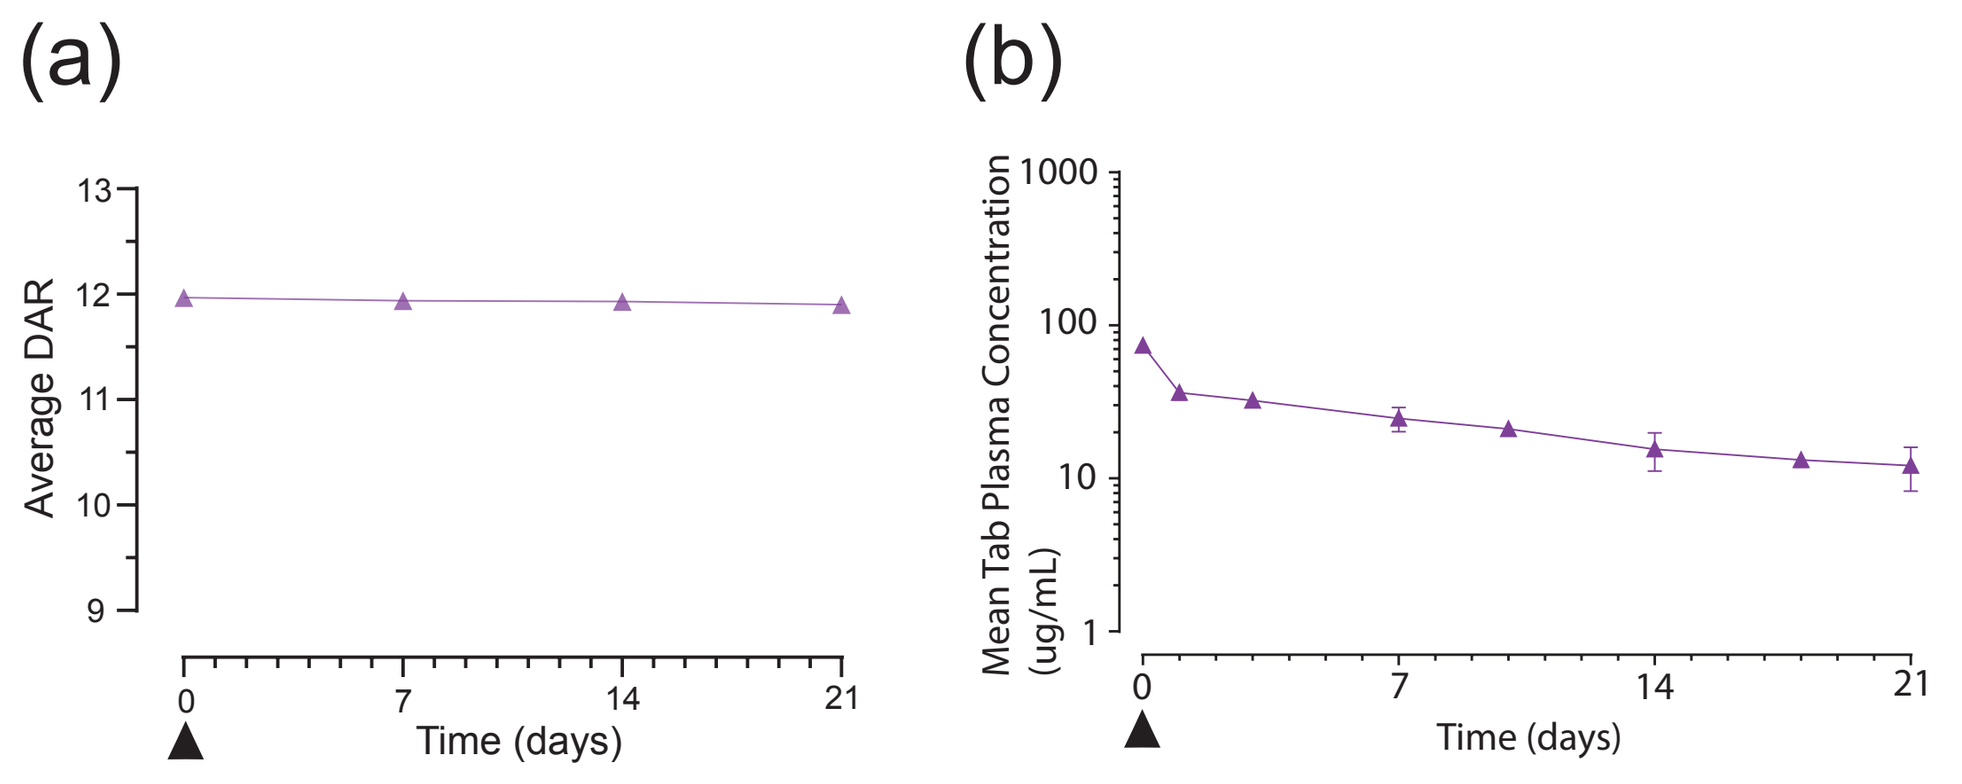
(a)
Average DAR
Time (days)
(b)
Mean Tab Plasma Concentration (ug/mL)
Time (days)

Mean PK Parameters	
AUC 0-t_{last} (d·ug·mL^{-1})	470
C_{max} (ug/mL)	73.9
CL (mL/d/kg)	4.20
V_{ss} (mL/kg)	81.4
$T_{1/2}$ (d)	11.7

Figure 2. Anti-HER2 DAR8+4 Topo1+MTi beta-glu dpADC has stable DAR and favorable PK over 21 days in BL6 mice. (a) DAR analysis over 21 days with a single IV dose in BL6 mice (b) Mean antibody plasma concentration over 21 days with a single IV dose in BL6 mice.

Sutro's dpADC has greater anti-tumor activity compared to single-agent ADCs

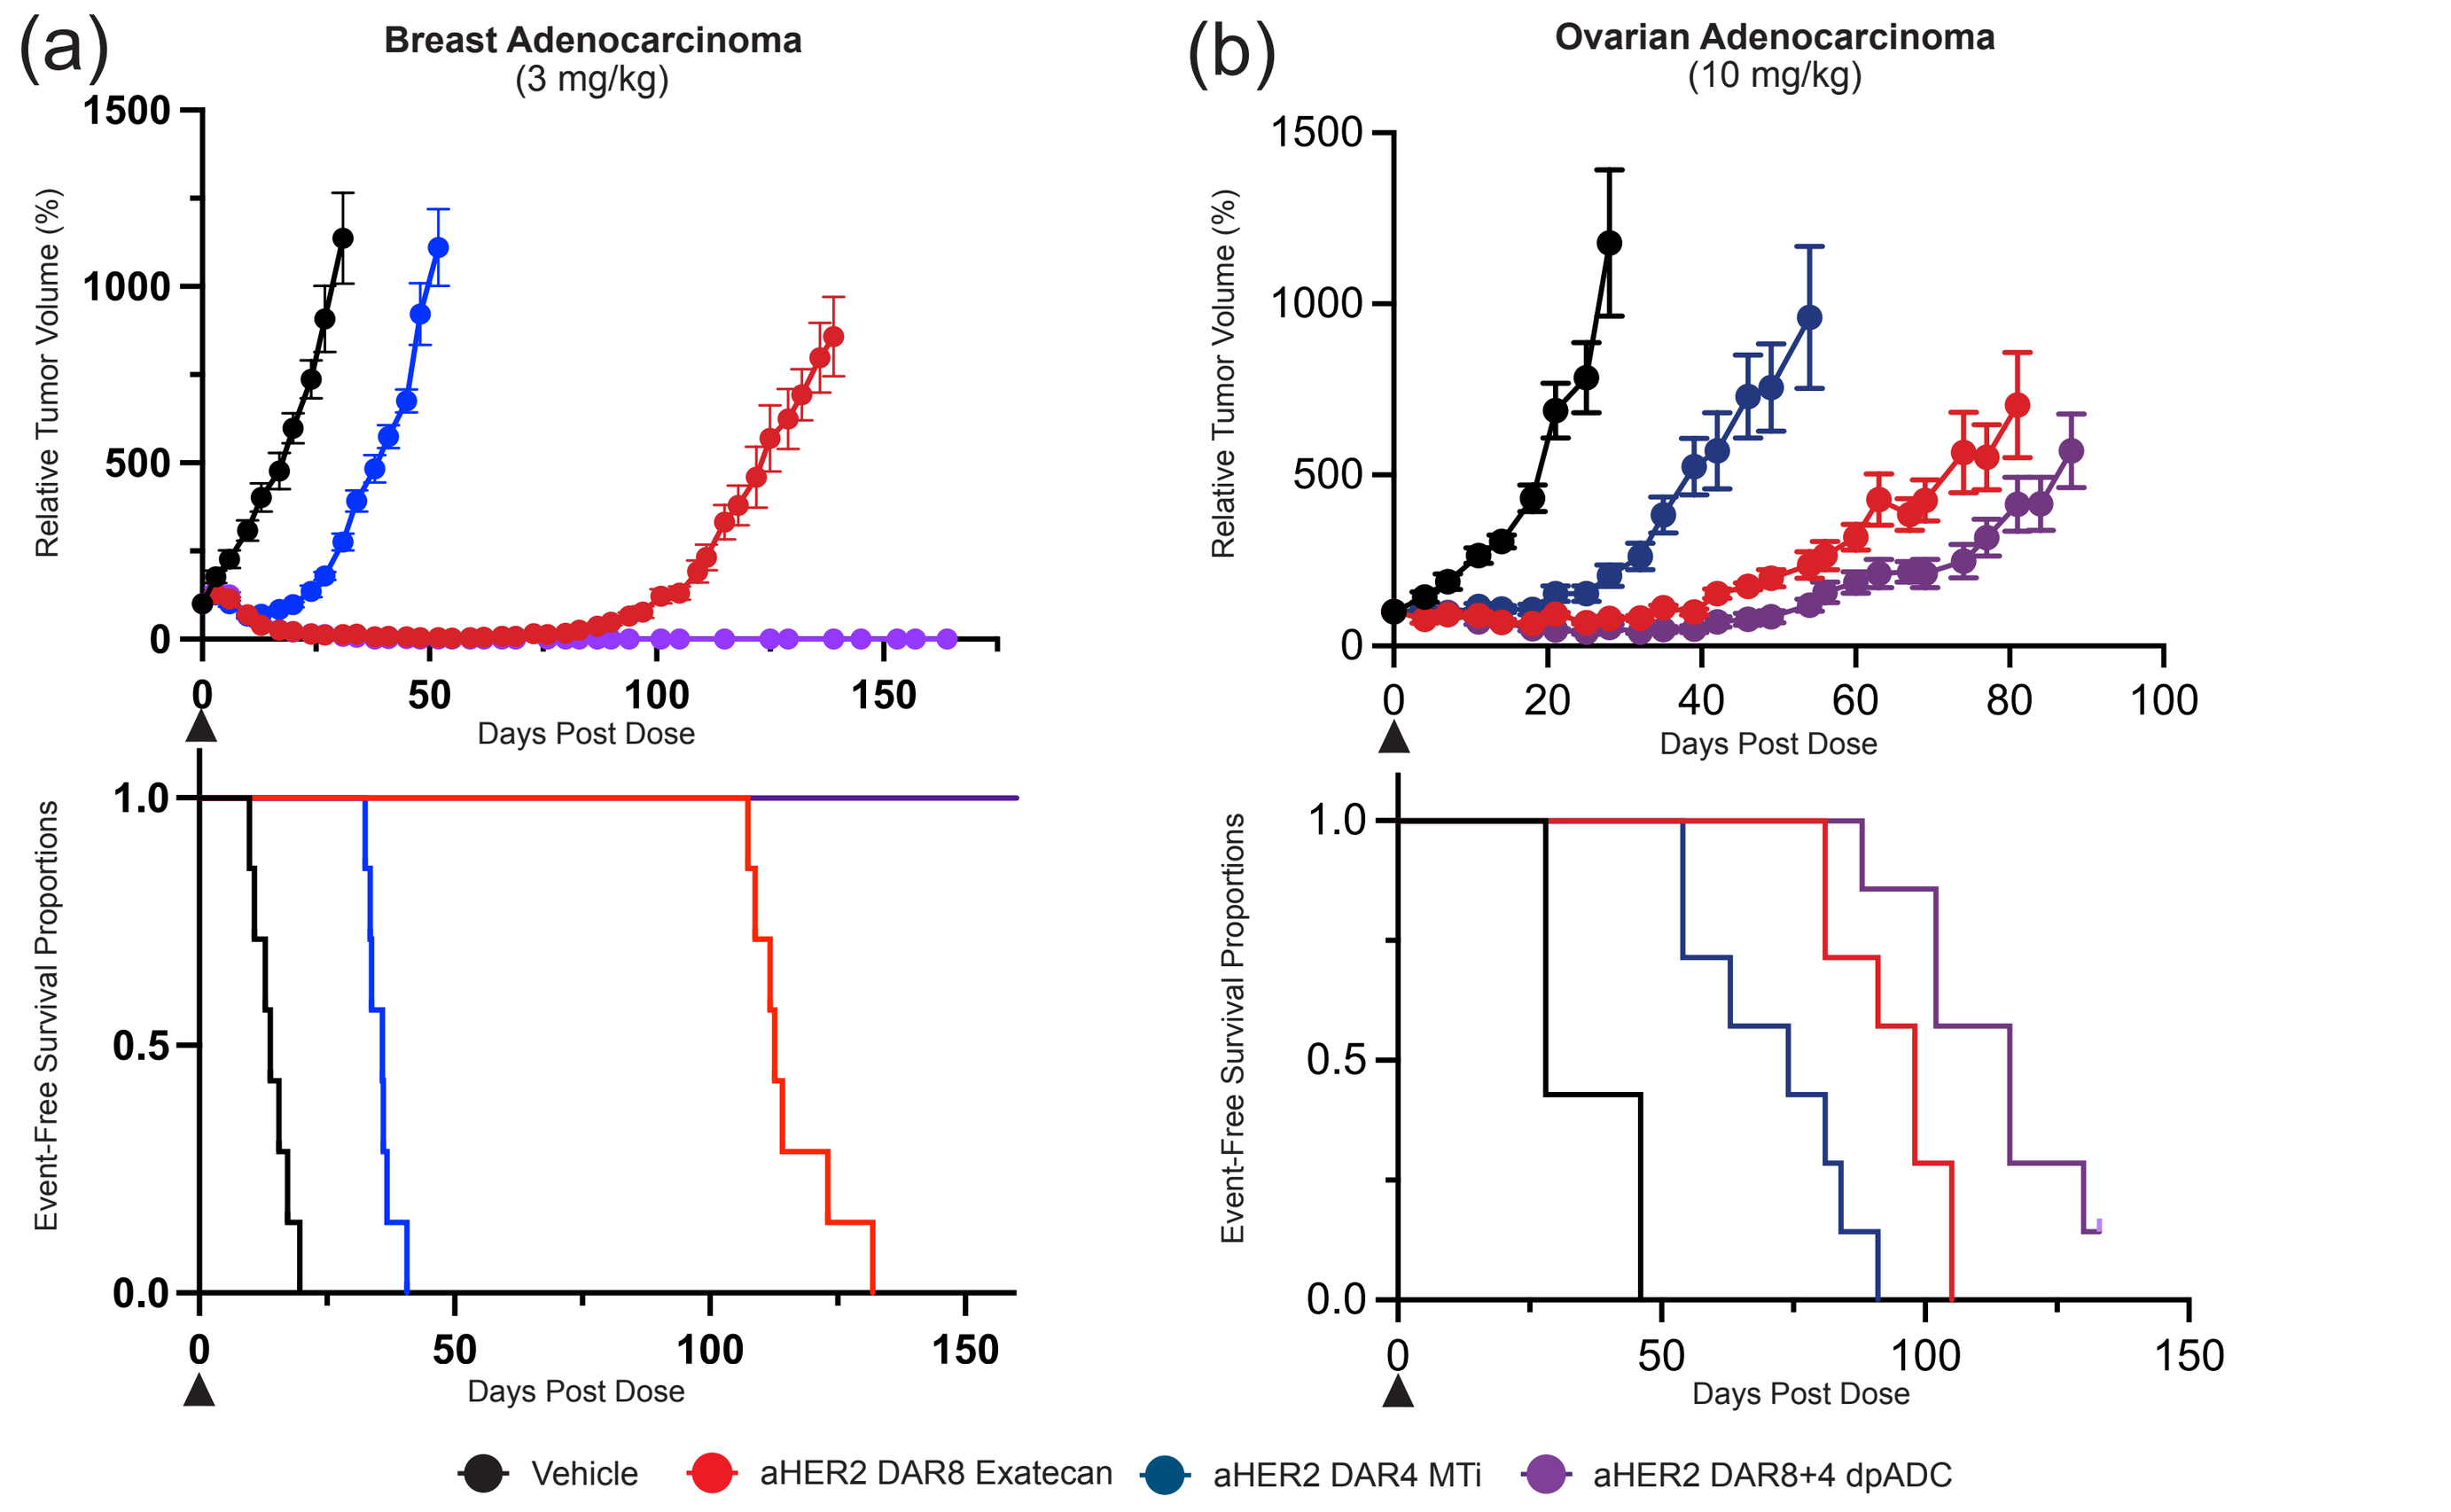
(a)
Breast Adenocarcinoma
(3 mg/kg)
(b)
Ovarian Adenocarcinoma
(10 mg/kg)
Relative Tumor Volume (%)
Event-Free Survival Proportions
Days Post Dose
Vehicle
aHER2 DAR8 Exatecan
aHER2 DAR4 MTi
aHER2 DAR8+4 dpADC

Figure 3. (a) Tumor growth curves (top) and Kaplan-Meier graphs of event-free survival propotions (bottom) of a breast adenocarcinoma CDX in mice treated with a single 3mg/kg IV dose of each test article.
(b) Shows the equivalent graphs for an ovarian adenocarcinoma treated with a single 10mg/kg IV dose of each test article. Event-free survival is defined as the time to tumor volume quadupling (TVQT).

Sutro's dpADC is efficacious in Enhertu-Resistant CDX

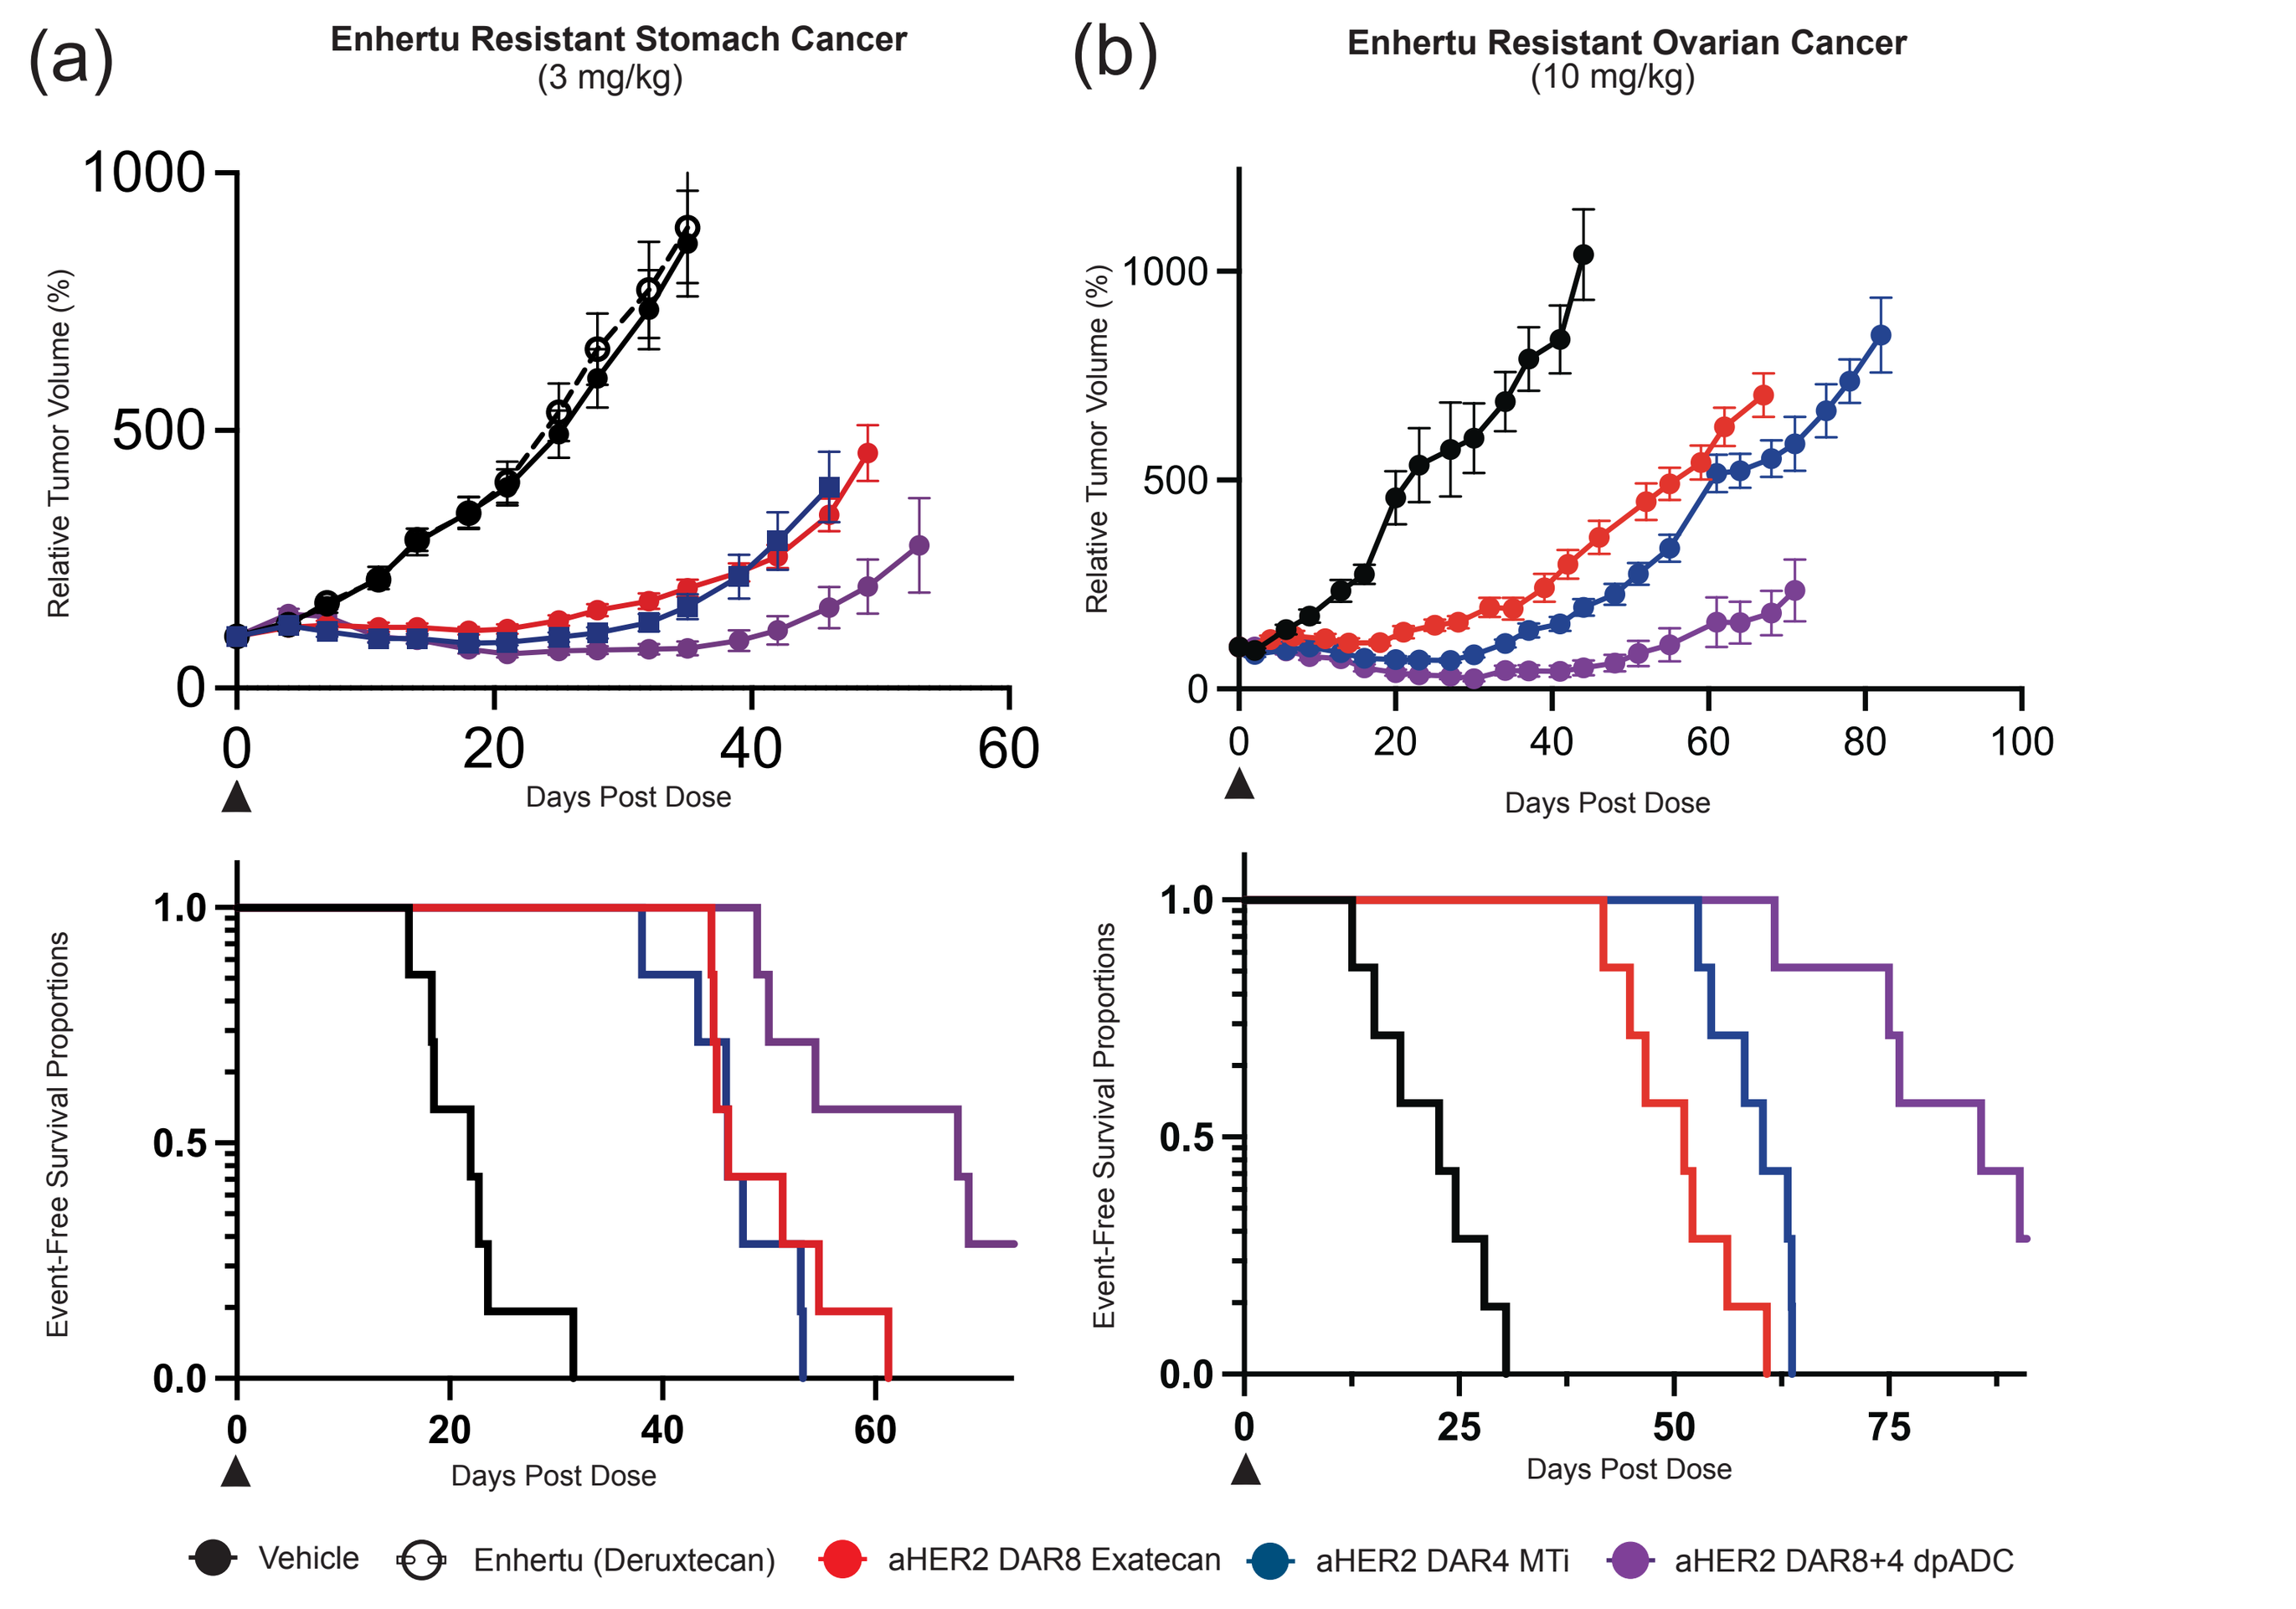
(a)
Enhertu Resistant Stomach Cancer
(3 mg/kg)
(b)
Enhertu Resistant Ovarian Cancer
(10 mg/kg)
Relative Tumor Volume (%)
Event-Free Survival Proportions
Days Post Dose
Vehicle
Enhertu (Deruxtecan)
aHER2 DAR8 Exatecan
aHER2 DAR4 MTi
aHER2 DAR8+4 dpADC

Figure 4: Sutro's dpADC induces tumor regression and greater time to TVQT in Enhertu-resistant xenograft tumor lines in mice. (a) Tumor growth curves (top) and Kaplan-Meier of event-free survival proportion (bottom) of Wuxi's Enhertu-resistant gastric adenocarcinoma dosed with a single 3mg/kg IV dose of each test article. (b) Tumor growth curves (top) and Kaplan-Meier of event-free survival proportion (bottom) of Sutro's Enhertu-resistant ovarian adenocarcinoma dosed with a single 10mg/kg IV dose of each test article. Sutro's dpADC shows greater anti-tumor activity compared to single-payload ADCs in both cell-lines. Sutro's Enhertu-resistant ovarian adenocarcinoma was developed through weekly IV Enhertu dosing of tumors until outgrowth, then tumor collection and cell-line creation.

Sutro's dpADC has greater anti-tumor activity than co-administered single-payload ADCs

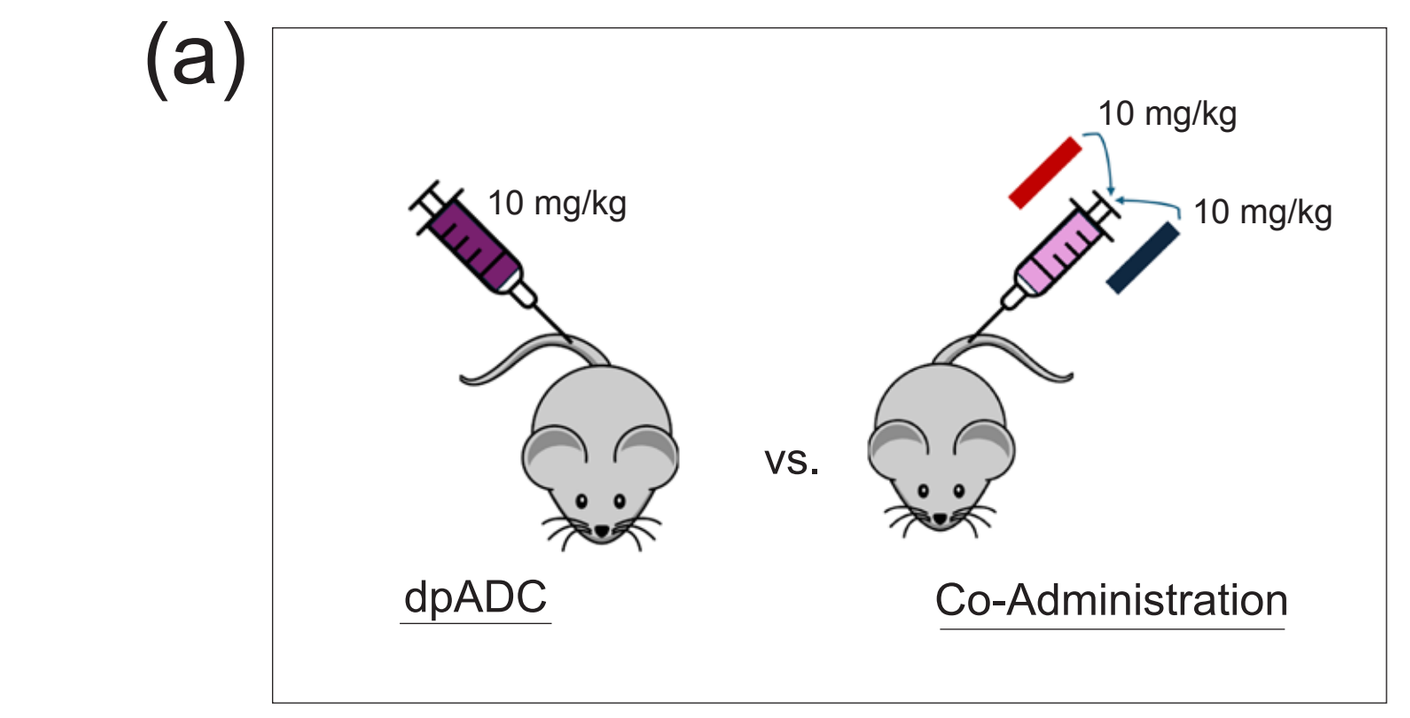
(a)
10 mg/kg
dpADC
vs.
10 mg/kg
10 mg/kg
Co-Administration

Group	Dose	Total Dose
Vehicle	---	---
anti-HER2 DAR8 Topo1i	10 mg/kg	10 mg/kg
anti-HER2 DAR4 MTi	10 mg/kg	10 mg/kg
Co-Administration	10 mg/kg 10 mg/kg	20 mg/kg
anti-HER2 dpADC	10 mg/kg	10 mg/kg

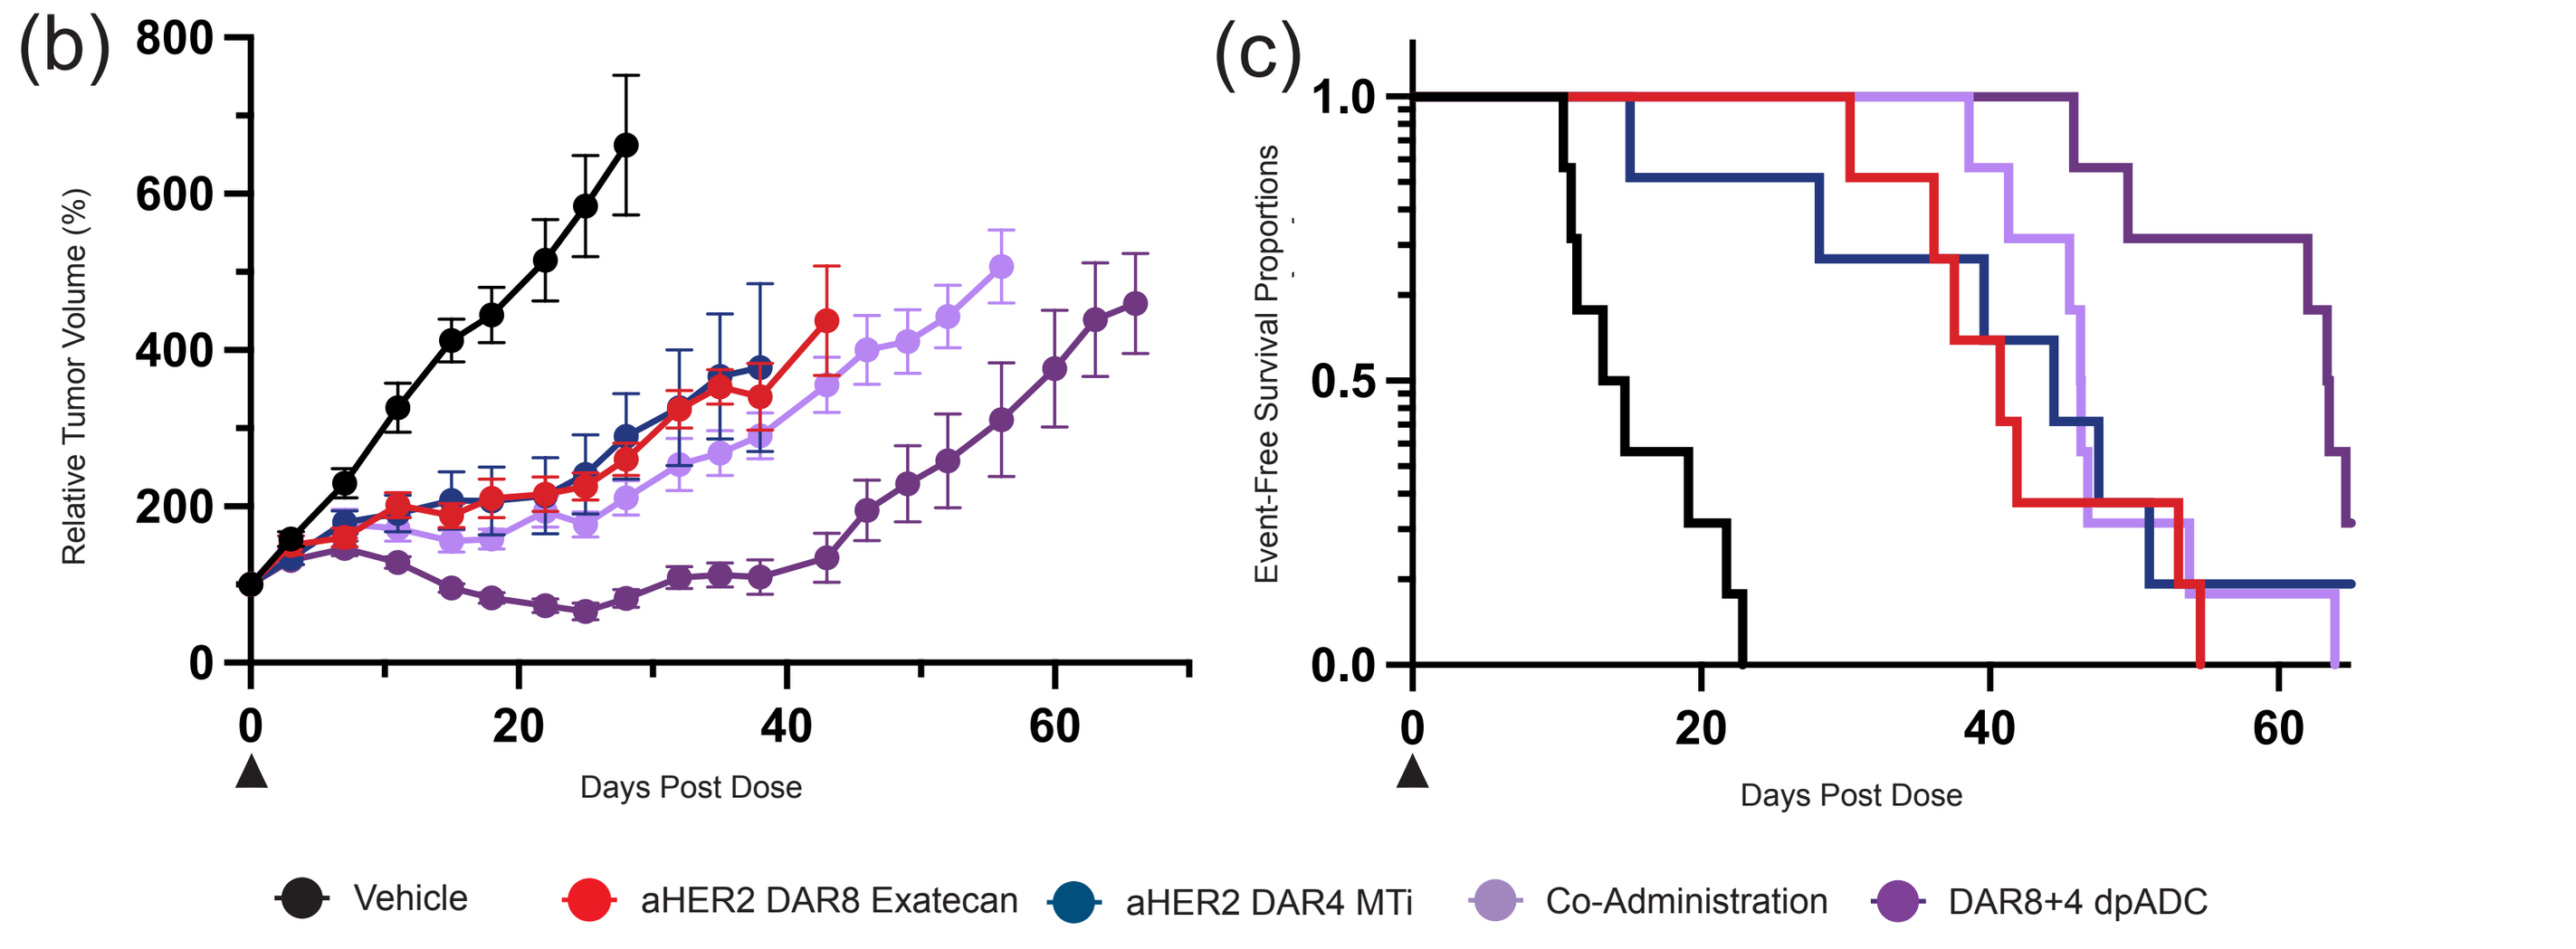
(b)
Relative Tumor Volume (%)
Days Post Dose
(c)
Event-Free Survival Proportions
Days Post Dose
Vehicle
aHER2 DAR8 Exatecan
aHER2 DAR4 MTi
Co-Administration
DAR8+4 dpADC

Figure 5. (a) Schematic of study design for comparing anti-tumor activity of Sutro's aHER2 dpADC (10mg/kg single IV dose) to the co-administration of its component single-agent ADCs (10mg/kg each for a total 20mg/kg single IV dose) in mice. (b) Tumor growth curves of Enhertu-resistant ovarian adenocarcinoma tumors show dpADC induces greater tumor regression and delays tumor growth compared to both single-payload ADCs and than their co-administration. (c) Kaplan-Meier curves for event-free survival proportions show tumors treated with dpADC take longer to reach TVQT than co-administration.

Sutro's dpADC is well tolerated in NHP

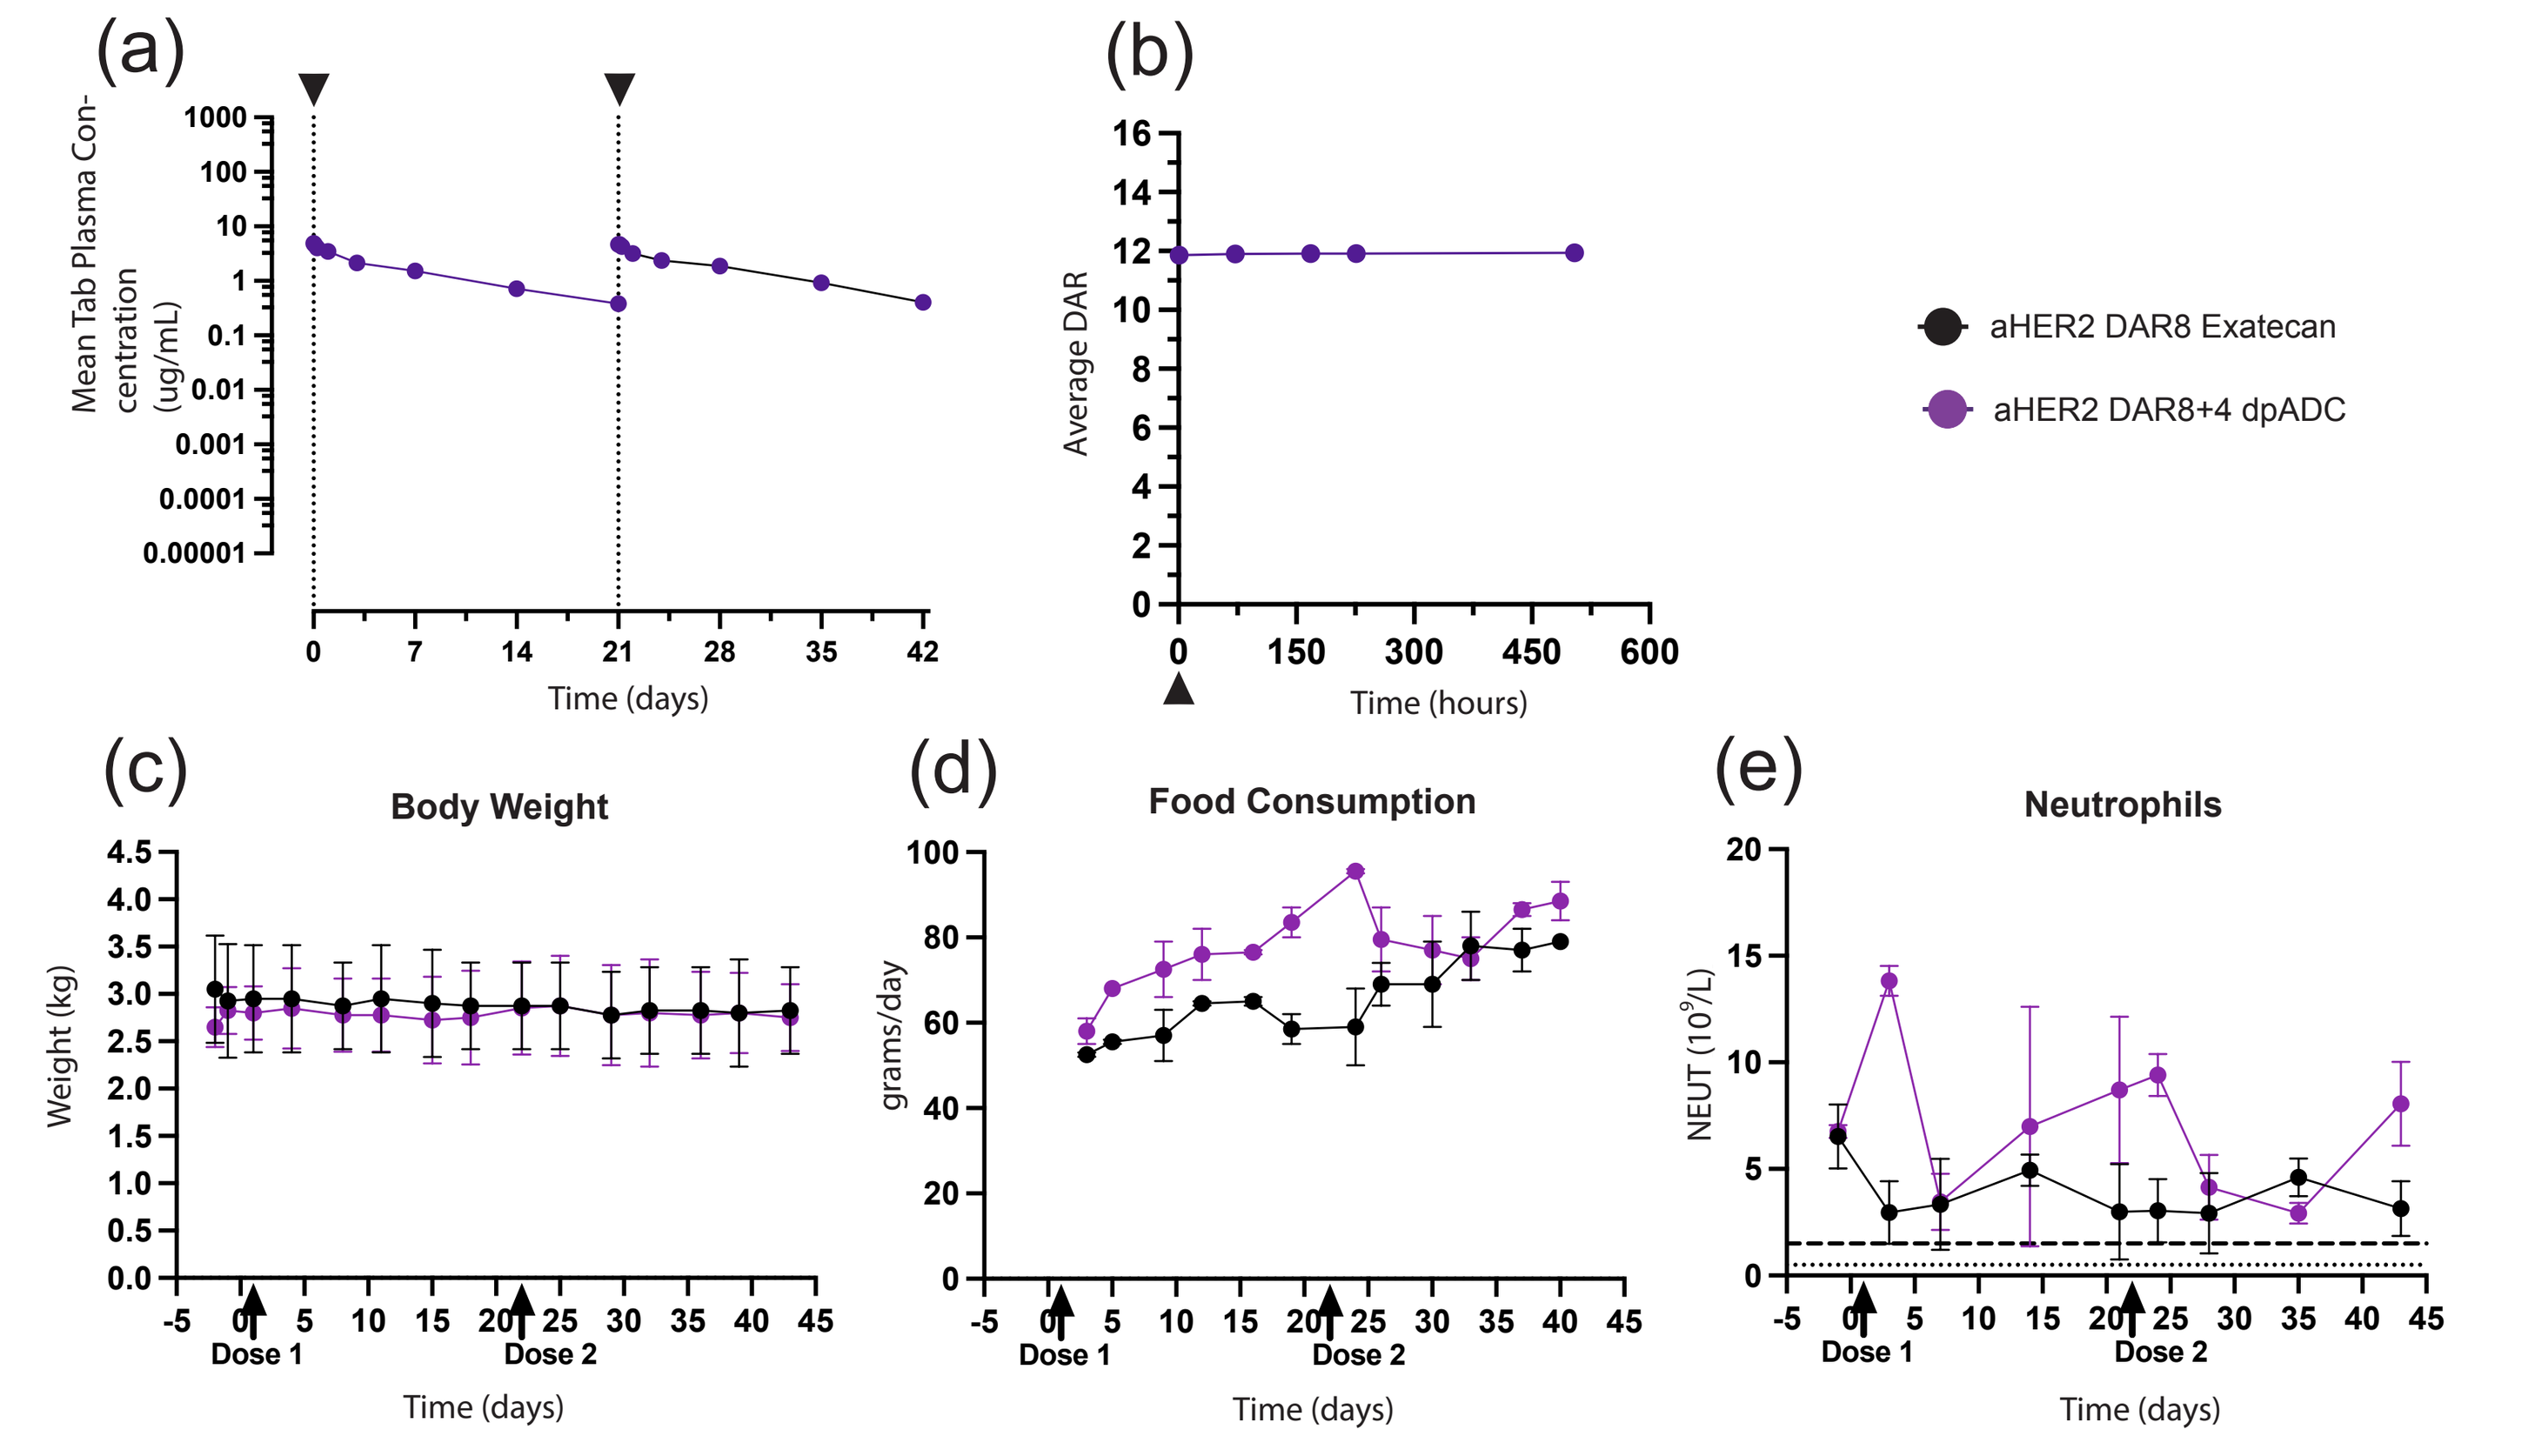
(a)
Mean Tab Plasma Concentration (ug/mL)
Time (days)
(b)
Average DAR
Time (hours)
aHER2 DAR8 Exatecan
aHER2 DAR8+4 dpADC
(c)
Body Weight
Weight (kg)
(d)
Food Consumption
grams/day
(e)
Neutrophils
NEUT (10^9/L)
Dose 1
Dose 2
Time (days)

Figure 6: Sutro's Anti-HER2 DAR8+4 Topo1+MTi dpADC is well tolerated with an HNSTD of 12.5mg/kg, with comparable responses to DAR8 Exatecan ADC. Toxicology analysis consisted of two single 12.5mg/kg IV infusion administered on days 1 and 22 (Q3Wx2) to 1M/1F animals. (a) PK was favorable. (b) DAR was stable. No abnormal changes were observed in (c) body weight or (d) food consumption, and a (e) transient decrease in neutrophils was detected without neutropenia. Minimal microscopic findings in heart, kidney, stomach, skin, thymus, and lung.

Sutro's dpADC overcomes sequential TOPO1 and MTi resistance

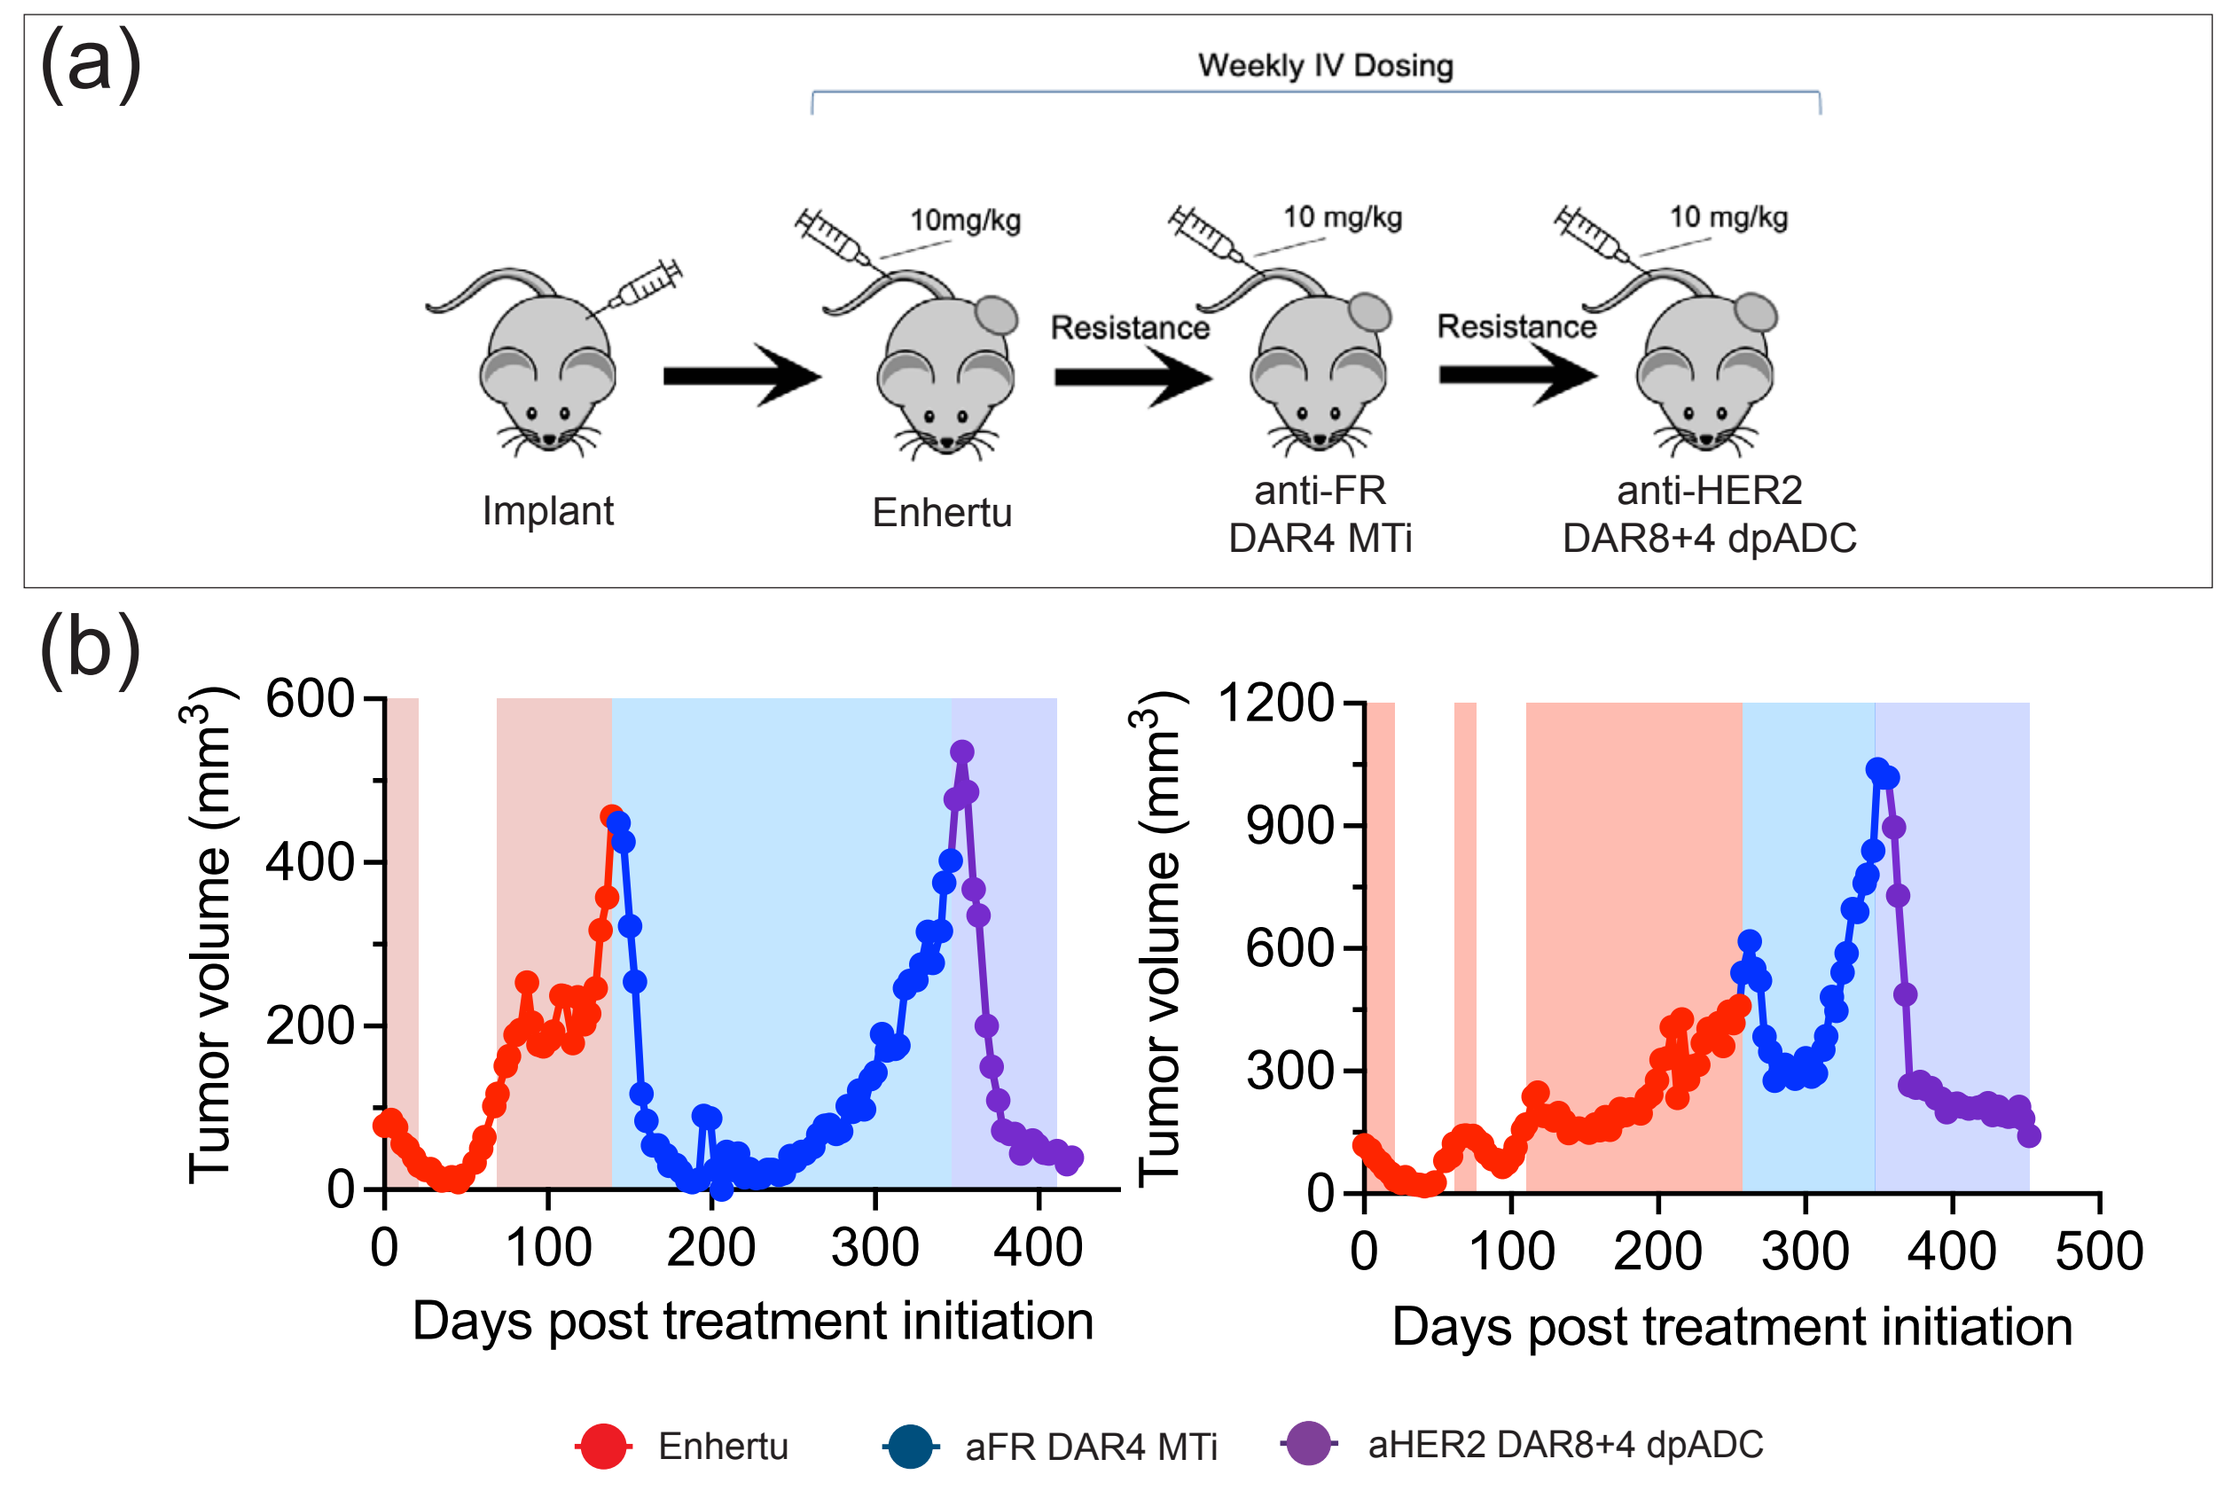
(a)
Weekly IV Dosing
10mg/kg
10 mg/kg
10 mg/kg
Resistance
Resistance
Implant
Enhertu
anti-FR DAR4 MTi
anti-HER2 DAR8+4 dpADC
(b)
Tumor volume (mm^3)
Days post treatment initiation
Enhertu
aFR DAR4 MTi
aHER2 DAR8+4 dpADC

Figure 7:
(a) Schematic of dosing regimen. Ovarian adenocarcinoma tumor-bearing mice were first given weekly IV doses of 10mg/kg Enhertu until tumors were no longer controlled by treatment. Weekly 10mg/kg anti-FR DAR4 MTi IV dose was then administered until the tumor was no longer controlled by treatment. Then 10mg/kg anti-HER2 DAR8+4 (Topo1i+MTi) dpADC was administered IV weekly until mouse was taken-down for non-treatment related reasons.
(b)Tumor growth curves of two individual mice. Colored backgrounds indicate weekly 10mg/kg IV dose of the test article of the respective color.

Conclusions

- Sutro's HER2-targeting DAR8 TOPO1i+ DAR4 MTi dpADC demonstrates favorable pharmacokinetics in vivo, minimal linker-payload loss over a 21-day study, and a 12.5mg/kg HNSTD in primates.
- Sutro's dpADC has greater anti-tumor activity than
 - Enhertu in-vitro and in-vivo.
 - Single-Payload ADCs.
 - Co-administered single-agent ADCs
- Sutro's dpADC has significant anti-tumor activity in Enhertu-resistant tumor cell lines.
- Sutro's dpADC causes tumor regression in tumors that have previously developed resistance to TOPO1i and MTis.

Sutro Biopharma
111 Oyster Point Boulevard
South San Francisco, California
94080

Phone: +1-650-881-6500
Website: www.sutrobio.com